Ocular screening of children with HIV/AIDS in an AIDS care center

Authors: Sudharshan Sridharan[1], Kalpa Negiloni[1], Kavitha Kalaivani[1], Suresh Kumar[2], Jeevitha Ravichandran[1]

Affilations: 1.Medical Research Foundation, Sankara Nethralaya , Chennai

2. Institute of Child Health and Hospital for Children, Chennai

Introduction

- Ocular manifestations of HIV/AIDS in children is rarely reported
- Most reports are from tertiary care eye institutions care
- These may not be representative of the actual occurrence
- Not many studies on ocular manifestations in children population with HIV/AIDS
- Important to assess effectiveness of highly active anti retroviral therapy (HAART) on prevention of ocular opportunistic infections (OIs) in children
- **Aim of this study** was to analyse the clinical profile and correlation of both ocular and systemic findings in children with HIV/AIDS in a paediatric HIV/AIDS care centre

Methodology

- Prospective study
- Details of complete ophthalmic examination performed in children with HIV/AIDS through an eye screening programme at a pediatric HIV centre and systemic details were noted and analysed

Results

- 239 children with Mean age-11±4 years (boys-61%), with/without visual complaints included
- Of these, 14% had ocular lesions attributable to HIV/AIDS.
 - Stereopsis was reduced(<600"arc) in 14 children
 - Defective colour vision(7%)
 - Schirmer test<10mm in 17% and TBUT<10 seconds in 24%
- Moderate to profound visual impairment in 0.8%
- Ocular manifestations
 - Anterior segment -12%
 - Posterior segment-10%
- Significantly, no ocular manifestations were noted in children affected through blood transfusion(n=4,1.8%) and IV drug use(IDU n=2,0.9%). However, in those with vertical transmission from mother(n=209), 12.9%(n=29) had ocular involvement
- Statistically, no risk of ocular manifestations in children was noted based on age(p=0.727), Schirmers(p=0.456), TBUT(p=0.58), CD4 at the start of ART(p=0.348) or during screening(p=0.363)

Figure 1

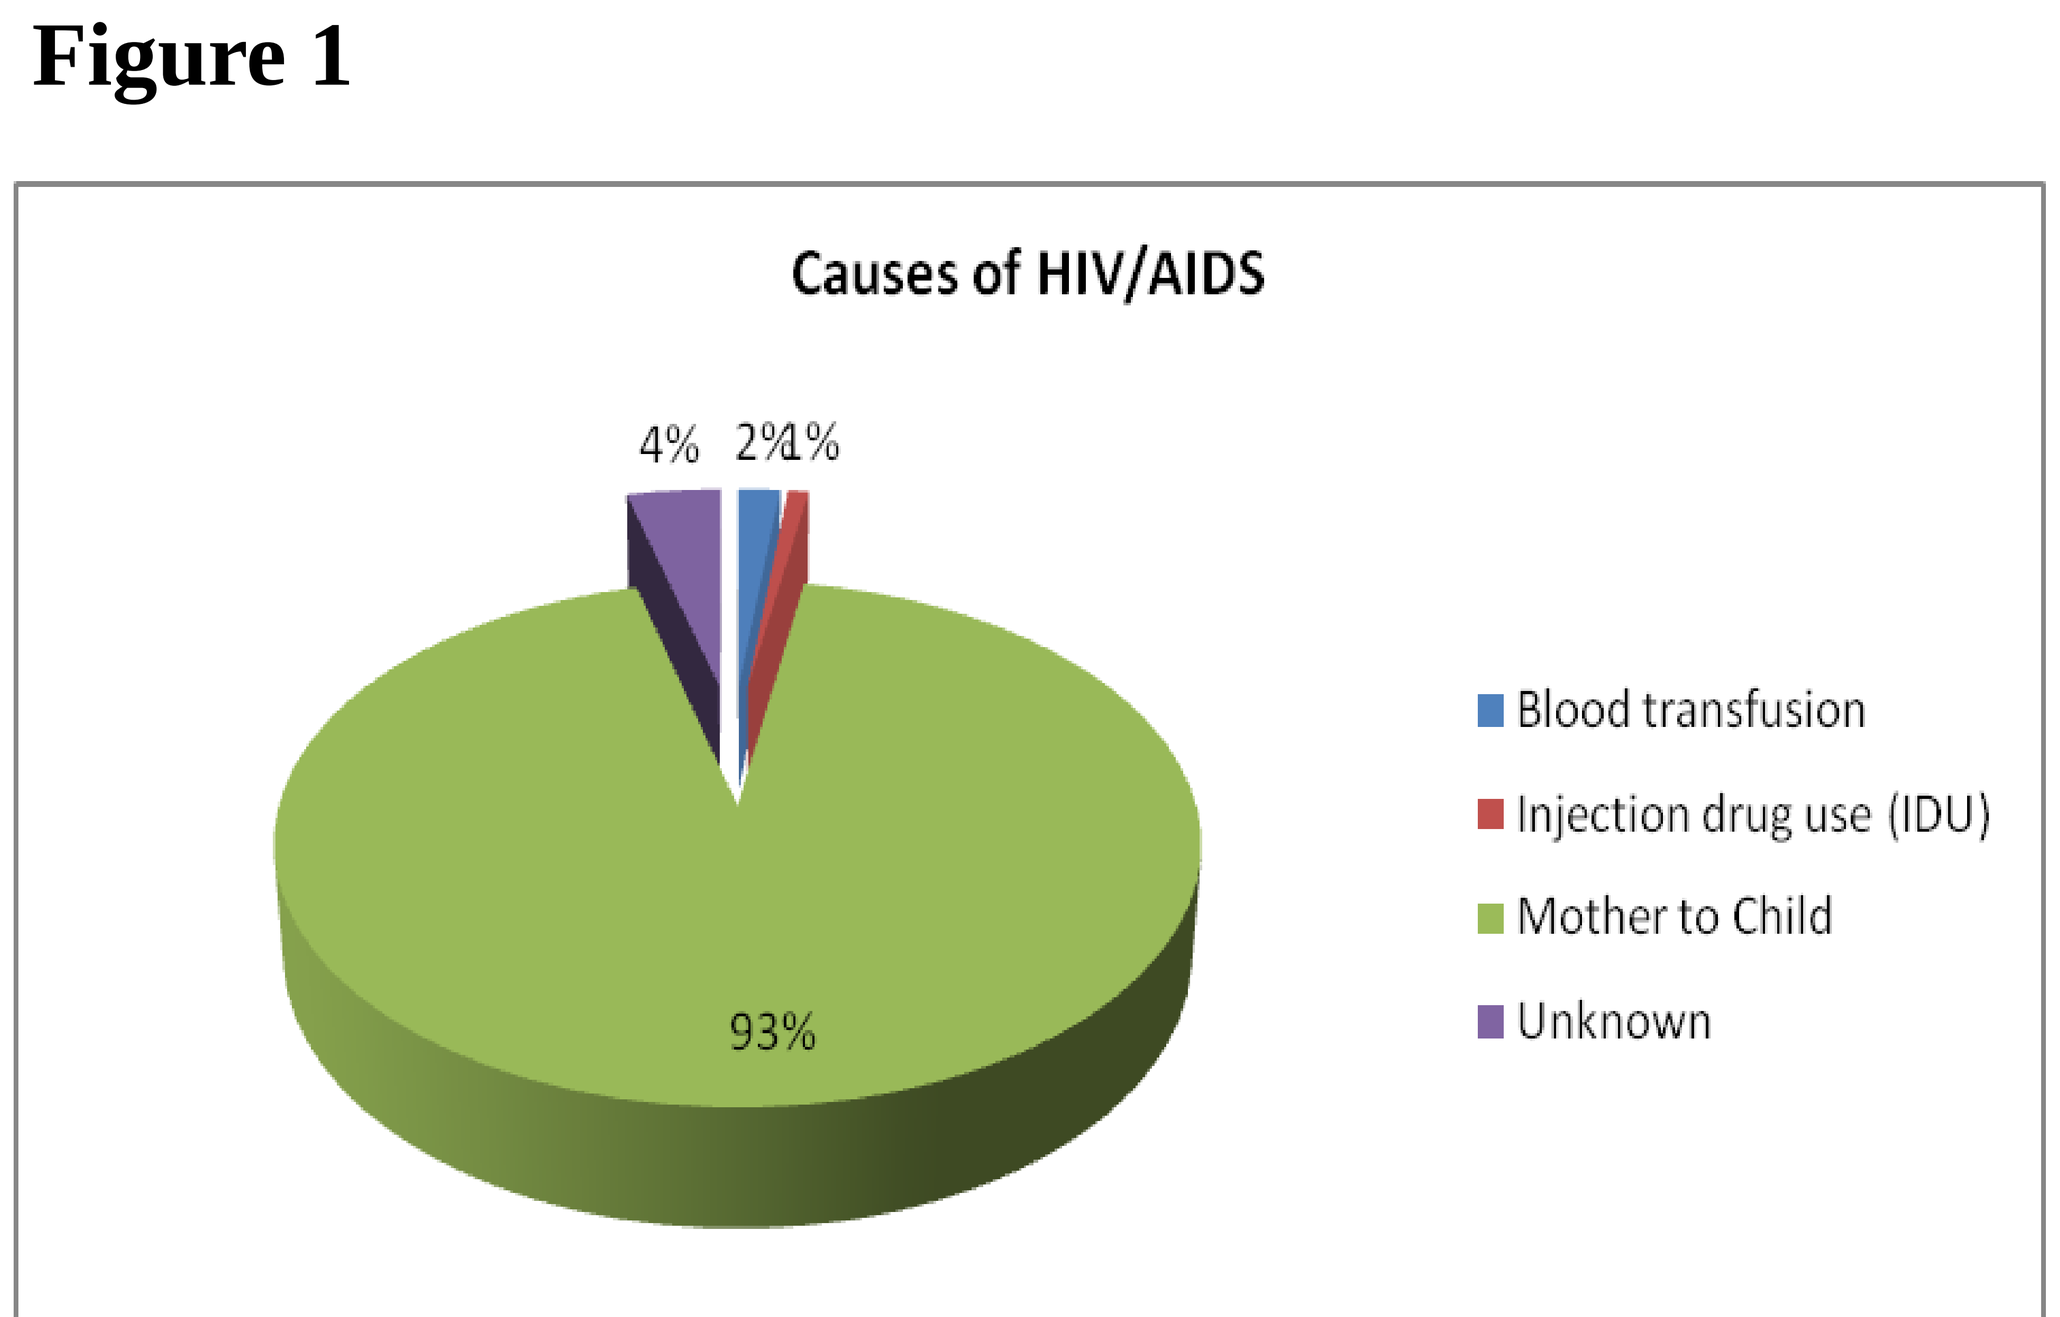
Causes of HIV/AIDS
4%
2%
1%
93%
Blood transfusion
Injection drug use (IDU)
Mother to Child
Unknown

Figure 2

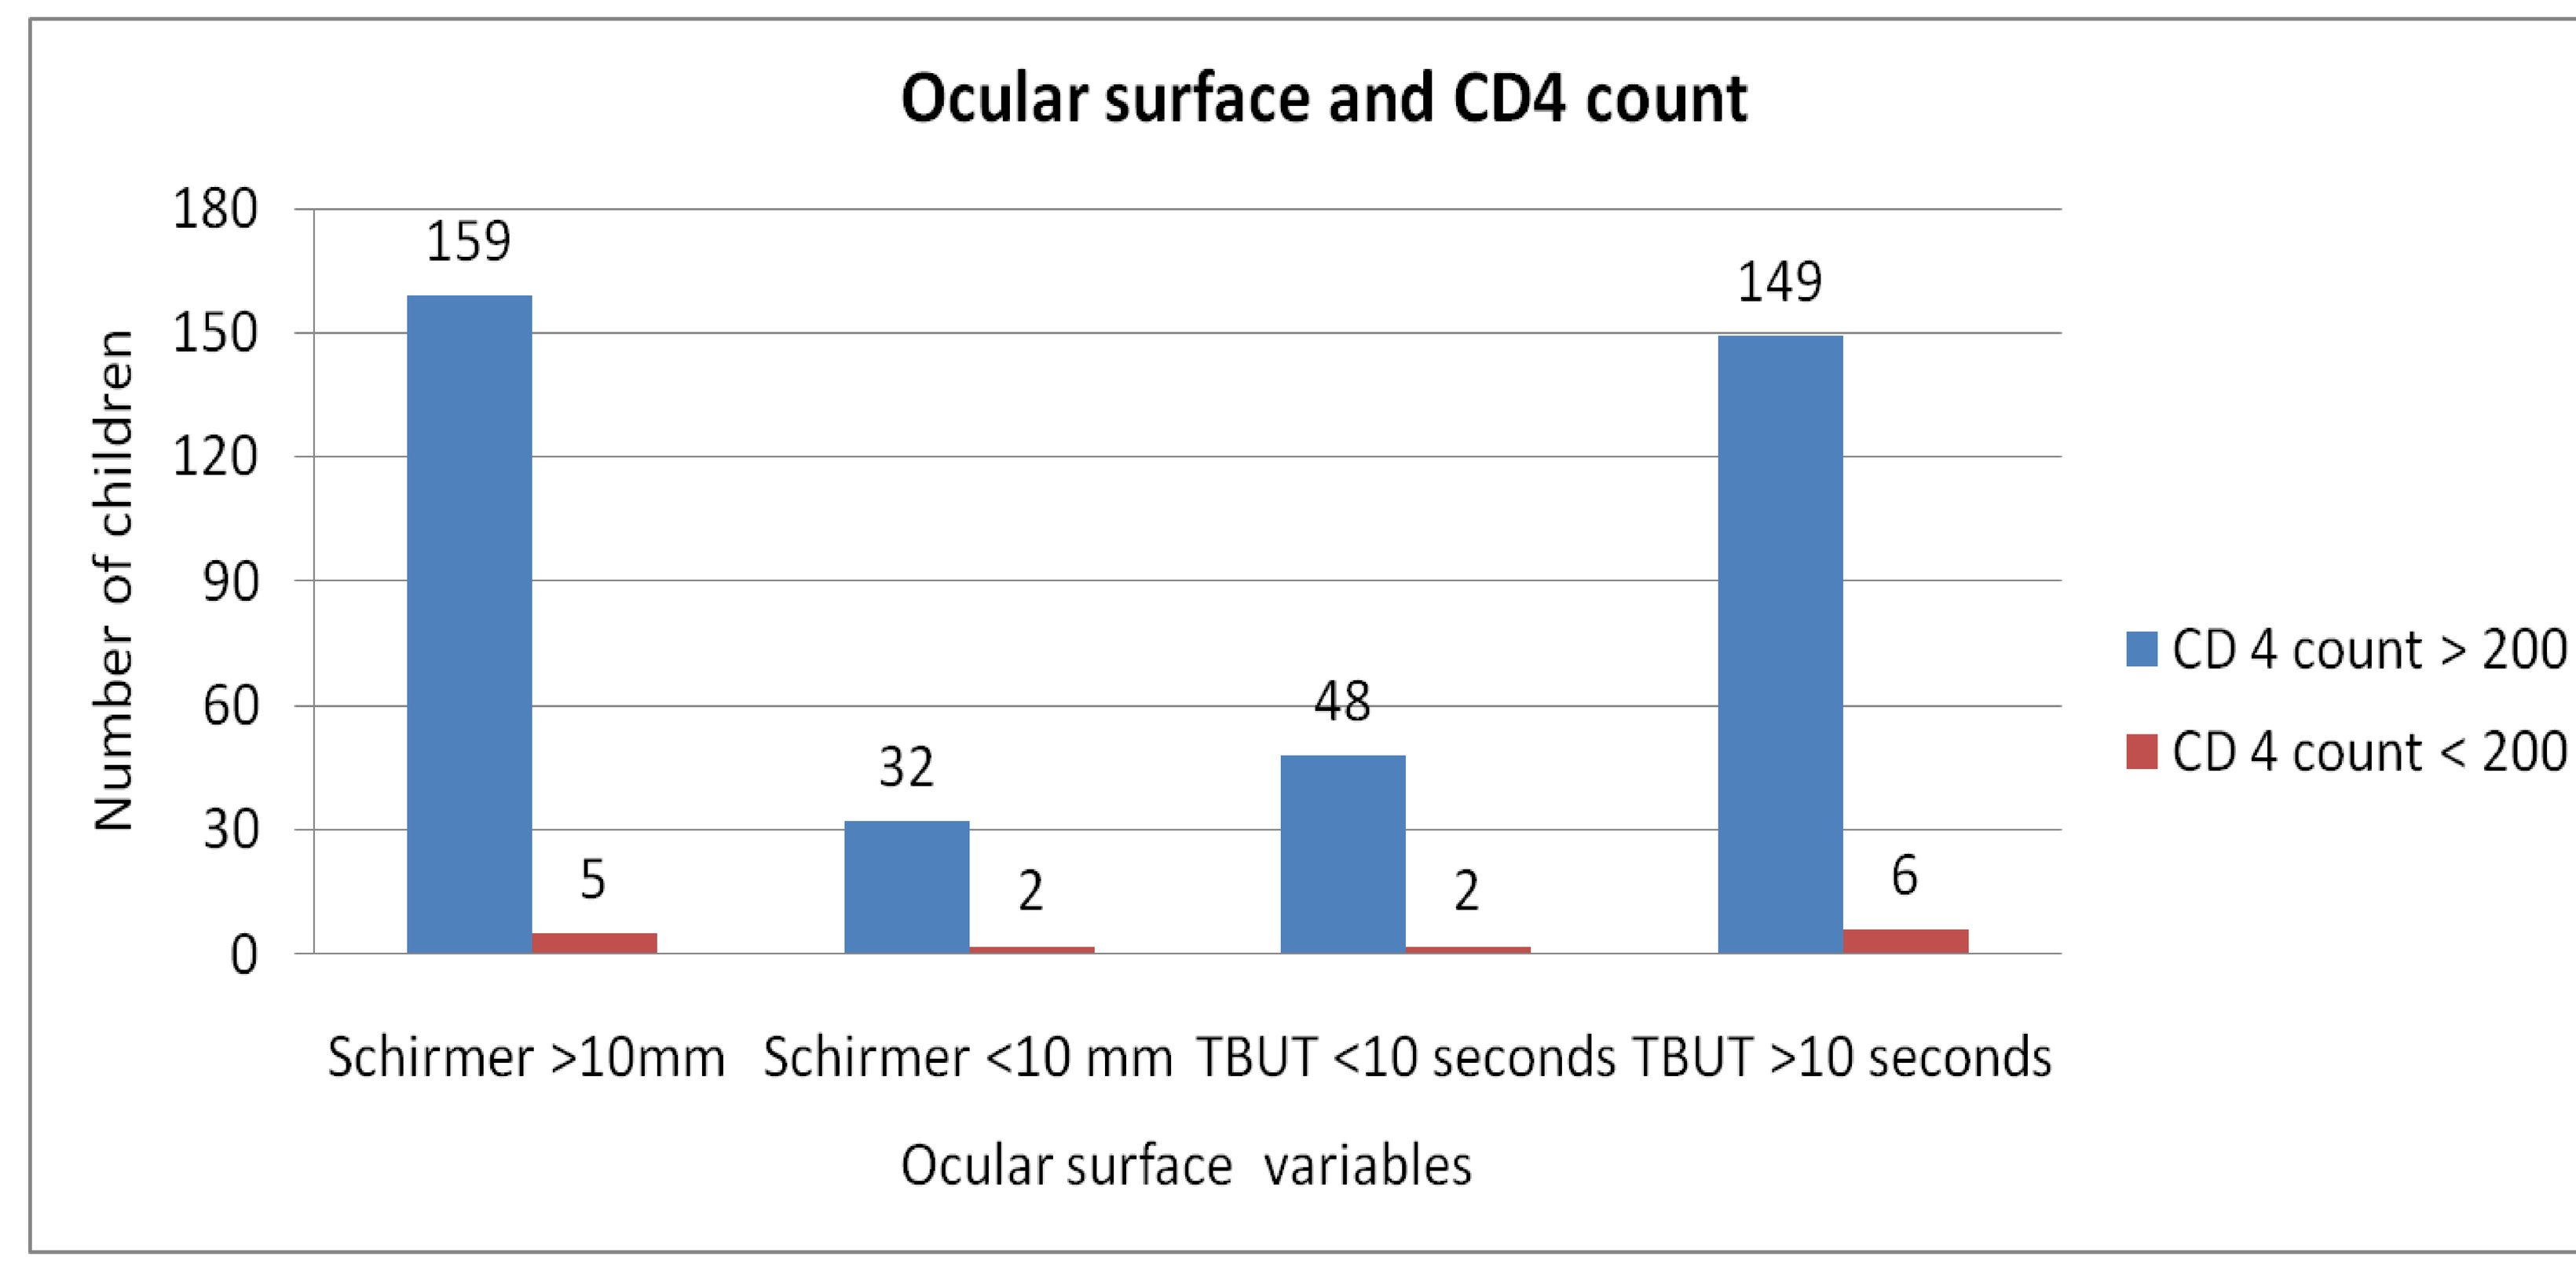
Ocular surface and CD4 count
Number of children
180
150
120
90
60
30
0
159
5
32
2
48
2
149
6
Schirmer >10mm
Schirmer <10 mm
TBUT <10 seconds
TBUT >10 seconds
Ocular surface variables
CD 4 count > 200
CD 4 count < 200

Table 1:Risk factors for ocular manifestations

Factor		Ocular manifestations NO	YES	OR (unadjusted)	p value
Age	<= 5 years	16 (7.1%)	2 (0.9%)	0.763 (0.167, 3.494)	0.727
	>= 6 years	177 (79%)	29 (12.9%)	1	
Gender	Boys	110 (49.1%)	24 (10.7%)	2.587 (1.064, 6.292)	0.036
	Girls	83 (37.1%)	7 (3.1%)		
CD4 stages – start of ART	>500/mm3	120 (53.6%)	15 (6.7%)		
	350 − 499/mm3	26 (11.6%)	4 (1.8%)	1.231 (0.378, 4.012)	0.731
	200 −349/mm3	27 (12.1%)	7 (3.1%)	2.074 (0.771, 5.579)	0.148
	<200/mm3	20 (8.9%)	5 (2.2%)	2.0 (0.654, 6.113)	0.224
CD4 stages – start of ART	<200/mm3	20 (8.9%)	5 (2.2%)	1.663 (0.574, 4.817)	0.348
	>200/mm3	173 (77.2%)	26 (11.6%)		
CD4 stages – screening time	<200/mm3	6 (2.7%)	2 (0.9%)	2.149 (0.414, 11.163)	0.363
TB	Yes	24 (10.9%)	7 (3.2%)	2.005 (0.78, 5.157)	0.149
	No	165 (75%)	24 (10.9%)		

Conclusions

- Asymptomatic ocular manifestations attributable to HIV/AIDS occur in children even while on HAART even without any active systemic disease.
- These can lead to which irreversible vision loss if left untreated
- VKC and Dry eyes are relatively more common in children with HIV/AIDS
- Drug related ocular side effects is on the rise
- Ocular manifestations noted in HIV/AIDS care centres are different than those presenting to tertiary eye care centres
- Ocular screening should be part of management protocol of all children with HIV/AIDS

Reference

1. Biswas J, Kumar AA, George AE, Madhavan HN, Kumarasamy N, Mothi SN, Solomon S.Ocular and systemic lesions in children with HIV. Indian J Pediatr. 2000Oct;67(10):721-4. PubMed PMID: 11105421.
2. Yonaba C, Kalmogho A, Sondo KA, Nacoulma M, Okengo K, Ouédraogo F, Zoungrana C, Kabore A, Kam L. Ocular Manifestations among HIV Infected Children in Ouagadougou, Burkina Faso. Open Journal of Pediatrics. 2016 May 20;6(02):185.
3. Mathebula SD, Makunyane PS. Ocular surface disorder among HIV and AIDS patients using antiretroviral drugs. Afr Vision Eye Health. 2019;78(1), a457. https://doi.org/10.4102/ aveh. v78i1.457
4. Agarwal S, Srinivasan B, Iyer G, Sudharshan S, Kalaivani K. Vernal keratoconjunctivitis in human immunodeficiency virus – The possible role of T-helper 1–T-helper 2 shift. Indian J Ophthalmol 2018;66:1004-6

IOIS 2019 105--P

PosterSession

Sudharshan SRIDHARAN

DOI: 10.3252/pso.eu.IOIS2019.2019

Poster presented at: IOIS International Ocular Inflammation Society

PosterSessionOnline.com