The prevalence and prognostic impact of myosteatosis with and without sarcopenia and its association with age and severity of liver cirrhosis

EASL CONGRESS 2023
Vienna, Austria
21–24 June

Eleni Geladari[1], Theodoros Alexopoulos[2], Meropi Kontogianni[3], Larisa Vasilieva[4], Iliana Mani[5], Roxane Tenta[3], Sofia Manioudaki[6], Vasilios Sevastianos[1], Alexandros Tsigkas[3], Ioannis Vlachogiannakos[2], Alexandra Alexopoulou[5]

[1]Evangelismos General Hospital, 3rd Department of Internal Medicine & Liver Outpatient Clinic, Athens, Greece; [2]Gastroenterology Department, National & Kapodistrian University of Athens, Medical School, Laiko General Hospital, Athens, Greece; [3]Department of Nutrition & Dietetics, School of Health Sciences and Education Harokopio University of Athens, Greece; [4]Alexandra General Hospital, Gastroenterology, Athens, Greece; [5]2nd Department of Internal Medicine & Research Laboratory, National & Kapodistrian University of Athens, Medical School, Hippokration General Hospital, Athens, Greece; [6]Intensive Care Unit, Sismanogleio General Hospital of Athens, Greece

Introduction

1

Myosteatosis (MS) is defined as increased fat infiltration in skeletal muscle and implies impaired muscle quality. MS is different from sarcopenia but part of sarcopenia definition. It is now considered a distinct entity from sarcopenia, but it is also a component of its definition according to the updated European Working Group on Sarcopenia in Older People (EWGSOP-2) criteria. In the setting of liver cirrhosis (LC), MS is associated with minimal and overt hepatic encephalopathy and risk of falls and has been incorporated in MELD as MELD-Sarco-Myo-HE score. Moreover, MS has been implicated in the pre- and post-liver transplantation outcome.

Aim

2

The aim of the present study was to investigate the interplay between muscle quality and quantity and the association of myosteatosis with the severity of liver cirrhosis, other muscle abnormalities such as low muscle mass and performance. The role of myosteatosis in the prognosis of liver disease was also examined.

Method

3

Skeletal muscle index (SMI) and MS measured by computed tomography at third lumbar vertebra, muscle strength evaluated by hand dynamometer and functionality by short physical performance battery (SPPB) were used to diagnose sarcopenia according to latest EWGSOP-2 criteria. MS was defined as muscle radiodensity <41 Hounsfield Units (HU) for dry body mass index <24.9 kg/m^2 and <33 HU for ≥25 kg/m^2. Patients' demographics, severity of LC and 360-day mortality were recorded.

Conclusions

4

In patients with LC, MS alone is present in earlier stage of LC and younger age and may imply a prodromal phase of muscle degeneration before the development of sarcopenia. MS is present in all but 3 patients with sarcopenia. Furthermore, sarcopenic patients with low SMI are older and have more advanced liver disease compared to sarcopenic with MS but normal SMI, implying that MS predates the decline of muscle strength and performance. Longitudinal data are required to draw solid conclusions about the significance of MS in patients with LC.

References

5

1. Cruz-Jentoft AJ, et al. Sarcopenia: revised European consensus on definition and diagnosis. Age Aging 2019;48:16-31.
2. Ebadi M, et al. Skeletal Muscle Pathological Fat Infiltration (Myosteatosis) Is Associated with Higher Mortality in Patients with Cirrhosis. Cells. 2022;11:1345.
3. Correa-de-Araujo R,, et al. Myosteatosis in the Context of Skeletal Muscle Function Deficit: An Interdisciplinary Workshop at the National Institute on Aging. Front Physiol. 2020 Aug 7;11:963
4. Bhanji RA, et al. Myosteatosis and sarcopenia are associated with hepatic encephalopathy in patients with cirrhosis. Hepatol Int. 2018;12:377-86.

Contact information

6

Corresponding author: **Alexandra Alexopoulou**, 2nd Department of Internal Medicine National & Kapodistrian University of Athens, Medical School, Hippokration General Hospital, Athens, Greece, email: alexopou@ath.forthnet.gr, Tel 2132088179

Results

194 consecutive patients were included [age 61 (IQR 52-68); 66.5% male; MELD 10.5 (7.7-16); 60.3% with decompensated LC; 43.8%, 22.7% and 33.5% with alcoholic, viral and other etiology, respectively]. Patients were classified in four groups according to presence of MS and sarcopenia. Neither MS nor sarcopenia was diagnosed in 25.3% (**group A**), MS without sarcopenia in 30.9% (**group B**), sarcopenia according to low handgrip and presence of MS in 17.5% (**group C**) and sarcopenia according to low handgrip, presence of MS and low SMI in 26.3% (**group D**). MS was present in all but 3 cases with sarcopenia, who were not included in the analysis. There was a significant ascending order across the groups A, B, C and D in age [56 (50-63.5), 57.5 (51.2-66), 62.5 (57-69.5) and 67 (59-72.5) years, respectively, p<0.001] and severity of LC as it was documented by the increasing rate of decompensated cirrhosis [32.7%, 58.3%, 73.5% and 80.4%, respectively, p<0.001] and similarly by MELD (p<0.001) and Child Pugh values (p<0.001). In contrast, there was a significant descending order across the groups in SMI (p<0.001) and SPPB (p<0.001) values (**Table 1**). Viral etiology was more prevalent in Group A and alcoholic etiology in C or D (sarcopenia groups) (p=0.038). No difference in gender was demonstrated across the groups.

Table 1. Comparison of demographics and clinical characteristics among 4 groups according to the extent of muscle abnormalities

	Group A (N=49)	Group B (N=60)	Group C* (N=34)	Group D* (N=51)	P
Age	56.0 (50.0-63.5)	57.5 (51.25-66.00)	62.5 (57.0-69.5)	67.00 (59.00-72.50)	< 0.001
Gender (Male, N, %)	37 (75.5)	40 (66.7)	17 (50.00)	35 (68.62)	0.109
Dry BMI	26.67 (25.61-29.98)	24.4 (22.04-28.83)	28.15 (24.08-34.05)	23.28 (20.80-26.41)	<0.001
Aetiology (N, %):					0.038
- Alcoholic	20 (40.81)	24 (40)	17 (50.00)	24 (47.05)	
- Viral	17 (34.69)	17 (28.33)	6 (17.64)	4 (7.80)	
- Other	12 (24.48)	19 (31.66)	11 (32.45)	23 (45.09)	
Decompensated cirrhosis (N, %)	16 (32.65)	35 (58.33)	25 (73.52)	41 (80.39)	< 0.001
MELD score	9.0 (7.0-10.75)	11.0 (7.00-14.0)	10.0 (7.00-15.75)	15.0 (11.50-21.50)	< 0.001
Child-Pugh score	5.0 (5.0-7.0)	7.0 (5.0-9.0)	7.0 (6.0-8.0)	9.0 (7.0-10.0)	< 0.001
Handgrip strength	32.0 (24.50-40.0)	34.00(30.0-38.00)	18.50 (15.50-27.25)	20.0 (14.0-25.0)	< 0.001
SMI	54.20 (49.72-57.48)	46.84(42.25-53.03)	51.03 (44.38-57.82)	40.50 (31.57-45.54)	< 0.001
SPPB	12.0 (10.0-12.0)	11.0 (10.0-12.0)	9.0 (6.75-11.0)	8.00 (3.75-10.00)	< 0.001

*Groups C and D had sarcopenia according to the updated EWGSOP-2 criteria; SMI, skeletal mass index; SPPB, short physical performance battery test; Kruskal Wallis test (P) and x^2 test were used for continuous and categorical variables, respectively

Table 2. Hazard ratios for death according to the extent of muscle abnormalities. Crude and adjusted values by the use of Cox's regression analysis

	Univariate analysis		Multivariate analysis	
	HR (95% CI)	P	HR (95% CI)	P
Age (per 1 year)	1.040 (1.006-1.075)	0.022	1.020 (0.984-1.057)	0.275
Gender (♂ vs ♀)	0.917 (0.465-1.811)	0.804	0.800 (0.394-1.622)	0.536
Myosteatosis alone (Group B)	Reference group		Reference group	
Myosteatosis combined by low handgrip (Group C*)	1.537 (0.547-4.318)	0.415	1.397 (0.492-3.693)	0.530
Myosteatosis combined by low handgrip and low skeletal muscle index (Group D*)	3.505 (1.603-7.663)	0.002	3.097 (1.338-7.169)	0.008

*Groups C and D had sarcopenia according to the updated EWGSOP-2 criteria

Considering the Kaplan-Meier curve at 360 days, in groups exhibiting myosteatosis alone (group B) or with myosteatosis plus more extended muscle abnormalities (C and D) (no patient died in group A), patients of group D had a higher mortality rate compared to B (log rank P=0.001) but not C (log rank P=0.068) (log rank P=0.002 in overall) (**Fig. 1**). According to the Cox's regression analysis, after adjustment for age and gender, patients in Group C had similar risk of death to those of Group B (reference group) (p=0.530). However, mortality was 3 times higher in those of group D compared to B [HR 3.097 (1.338-7.169), P=0.008] (**Table 2**).

Figure 1. Kaplan Meier survival curves for the 3 muscle phenotypes (myosteatosis alone (group B); myosteatosis combined by low handgrip strength (group C); myosteatosis combined by low handgrip strength and low skeletal mass index (group D) (log rank P=0.002)

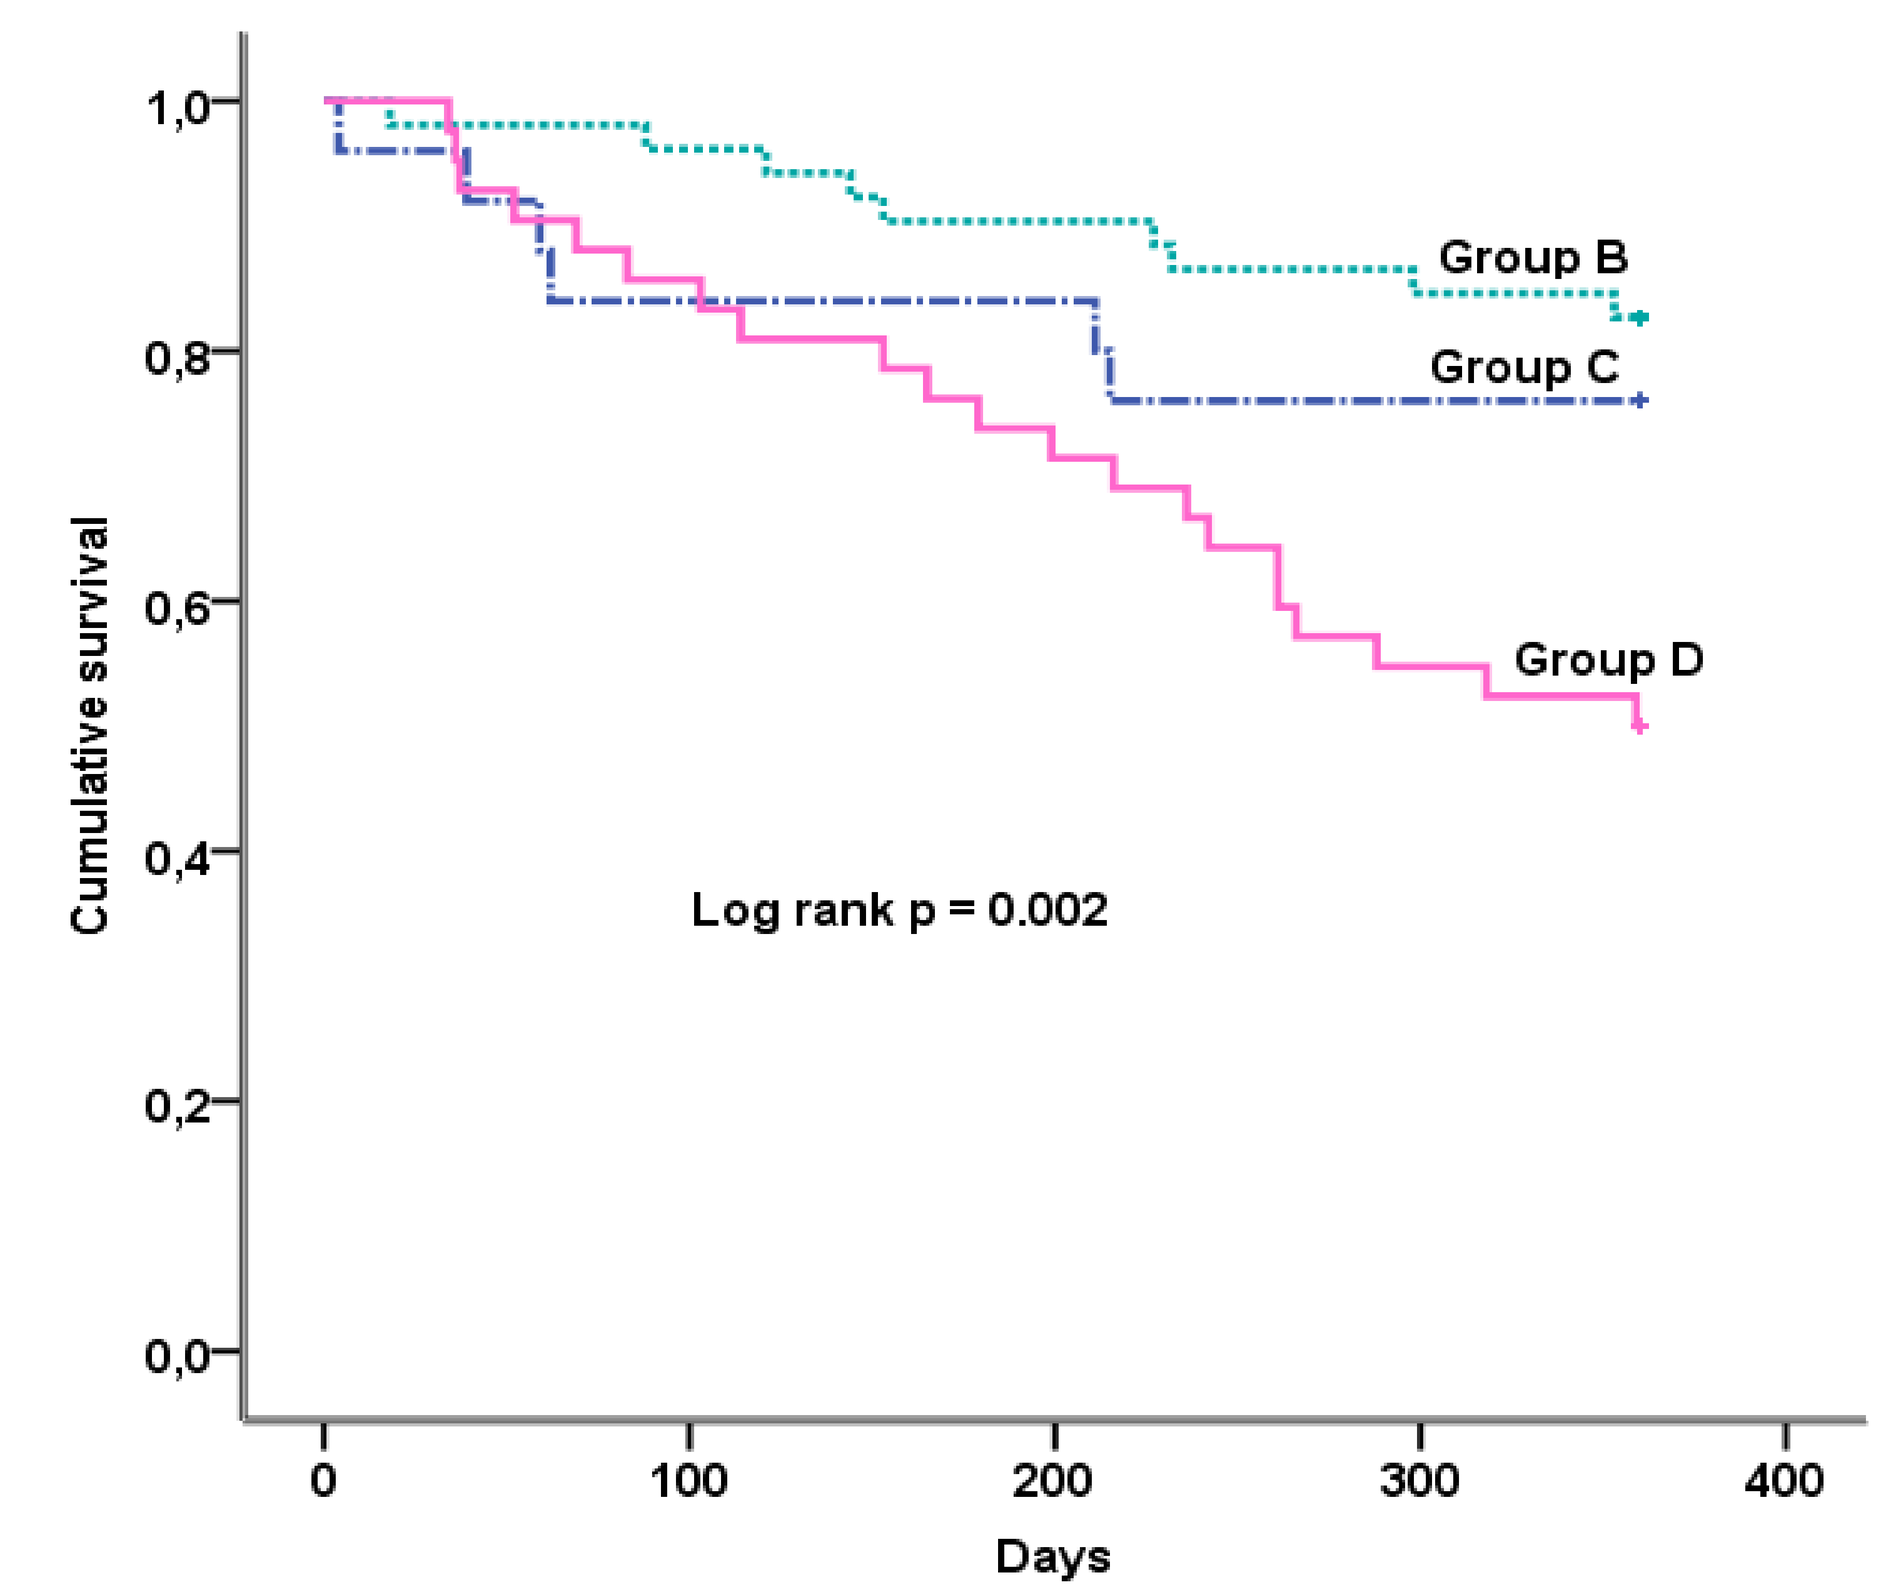
Cumulative survival
1,0
0,8
0,6
0,4
0,2
0,0
Group B
Group C
Group D
Log rank p = 0.002
0
100
200
300
400
Days

PosterSessionOnline
Scan to download the poster
EASL CONGRESS 2023 Vienna, Austria 21–24 June
DOI: 10.3252/pso.eu.EASLCONGRESS.2023
Cirrhosis and complications
Alexandra Alexopoulou
WED--362
EASL2023