Investigation of linerixibat 40 mg BID for cholestatic pruritus of primary biliary cholangitis (PBC); further data from the Phase 2b GLIMMER study to support the Phase 3 GLISTEN study

Poster No. 3606

Fettiplace J[1], Swift B[2], Zhang S[3], von Maltzahn R[1], McLaughlin MM[3]

[1]GSK, London, UK; [2]GSK, Research Triangle Park, NC, USA; [3]GSK, Collegeville, PA, USA

Background and Aims

- Cholestatic pruritus (itch) is frequent in primary biliary cholangitis (PBC), occurring in approximately 75% of patients during the course of their disease.[1] Patients with cholestatic pruritus have a substantially impaired quality of life (QoL), with impacts on sleep, mental and emotional wellbeing.[1-6] Severe pruritus limits daily life activities and causes fatigue, depression and even suicidal tendencies.[7]
- Current therapies used for PBC do not treat cholestatic pruritus; there are few treatment options available to patients and these are often ineffective, with limited clinical evidence to support their use.[8,9]
 - Due to the limited number of therapies approved for cholestatic pruritus, the definition of a meaningful within-person change in itch has not been determined.
- Therapies that reduce serum bile acids are under investigation for the treatment of cholestatic pruritus associated with PBC.
 - Linerixibat (GSK233067) is a minimally absorbed selective small-molecule inhibitor of the ileal bile-acid transporter (IBAT) that reduces absorption of bile acids in the terminal ileum and increases faecal bile acid excretion.[10,11]
- GLIMMER (NCT02966834) was a double-blind, randomised, placebo-controlled, Phase 2b dose-response study of linerixibat for patients with PBC and moderate-to-severe pruritus.[12]
- Here, we focus on the group of patients who received linerixibat 40 mg twice daily (BID), the dose selected for further evaluation in the ongoing Phase 3 GLISTEN study (NCT04950127).[13]
 - To further assess the efficacy of linerixibat 40 mg BID, data from GLIMMER were reanalysed to assess several potential itch responder definitions.
 - We also present additional efficacy and safety data with linerixibat 40 mg BID as well as changes in circulating bile acids and other exploratory biomarkers, evaluated based on the mode of action (MoA) of linerixibat.

Methods

Study design

- GLIMMER enrolled patients with PBC and pruritus graded as ≥4 on a 0–10 numeric rating scale (NRS).[12]
- Following a 4-week single-blind placebo period, patients with NRS ≥3 were randomised to receive placebo or linerixibat (double blind) for 12 weeks, followed by a further 4-week single-blind placebo period (**Figure 1A**).

Figure 1. GLIMMER study design and derivation of daily, weekly and monthly itch and sleep interference scores

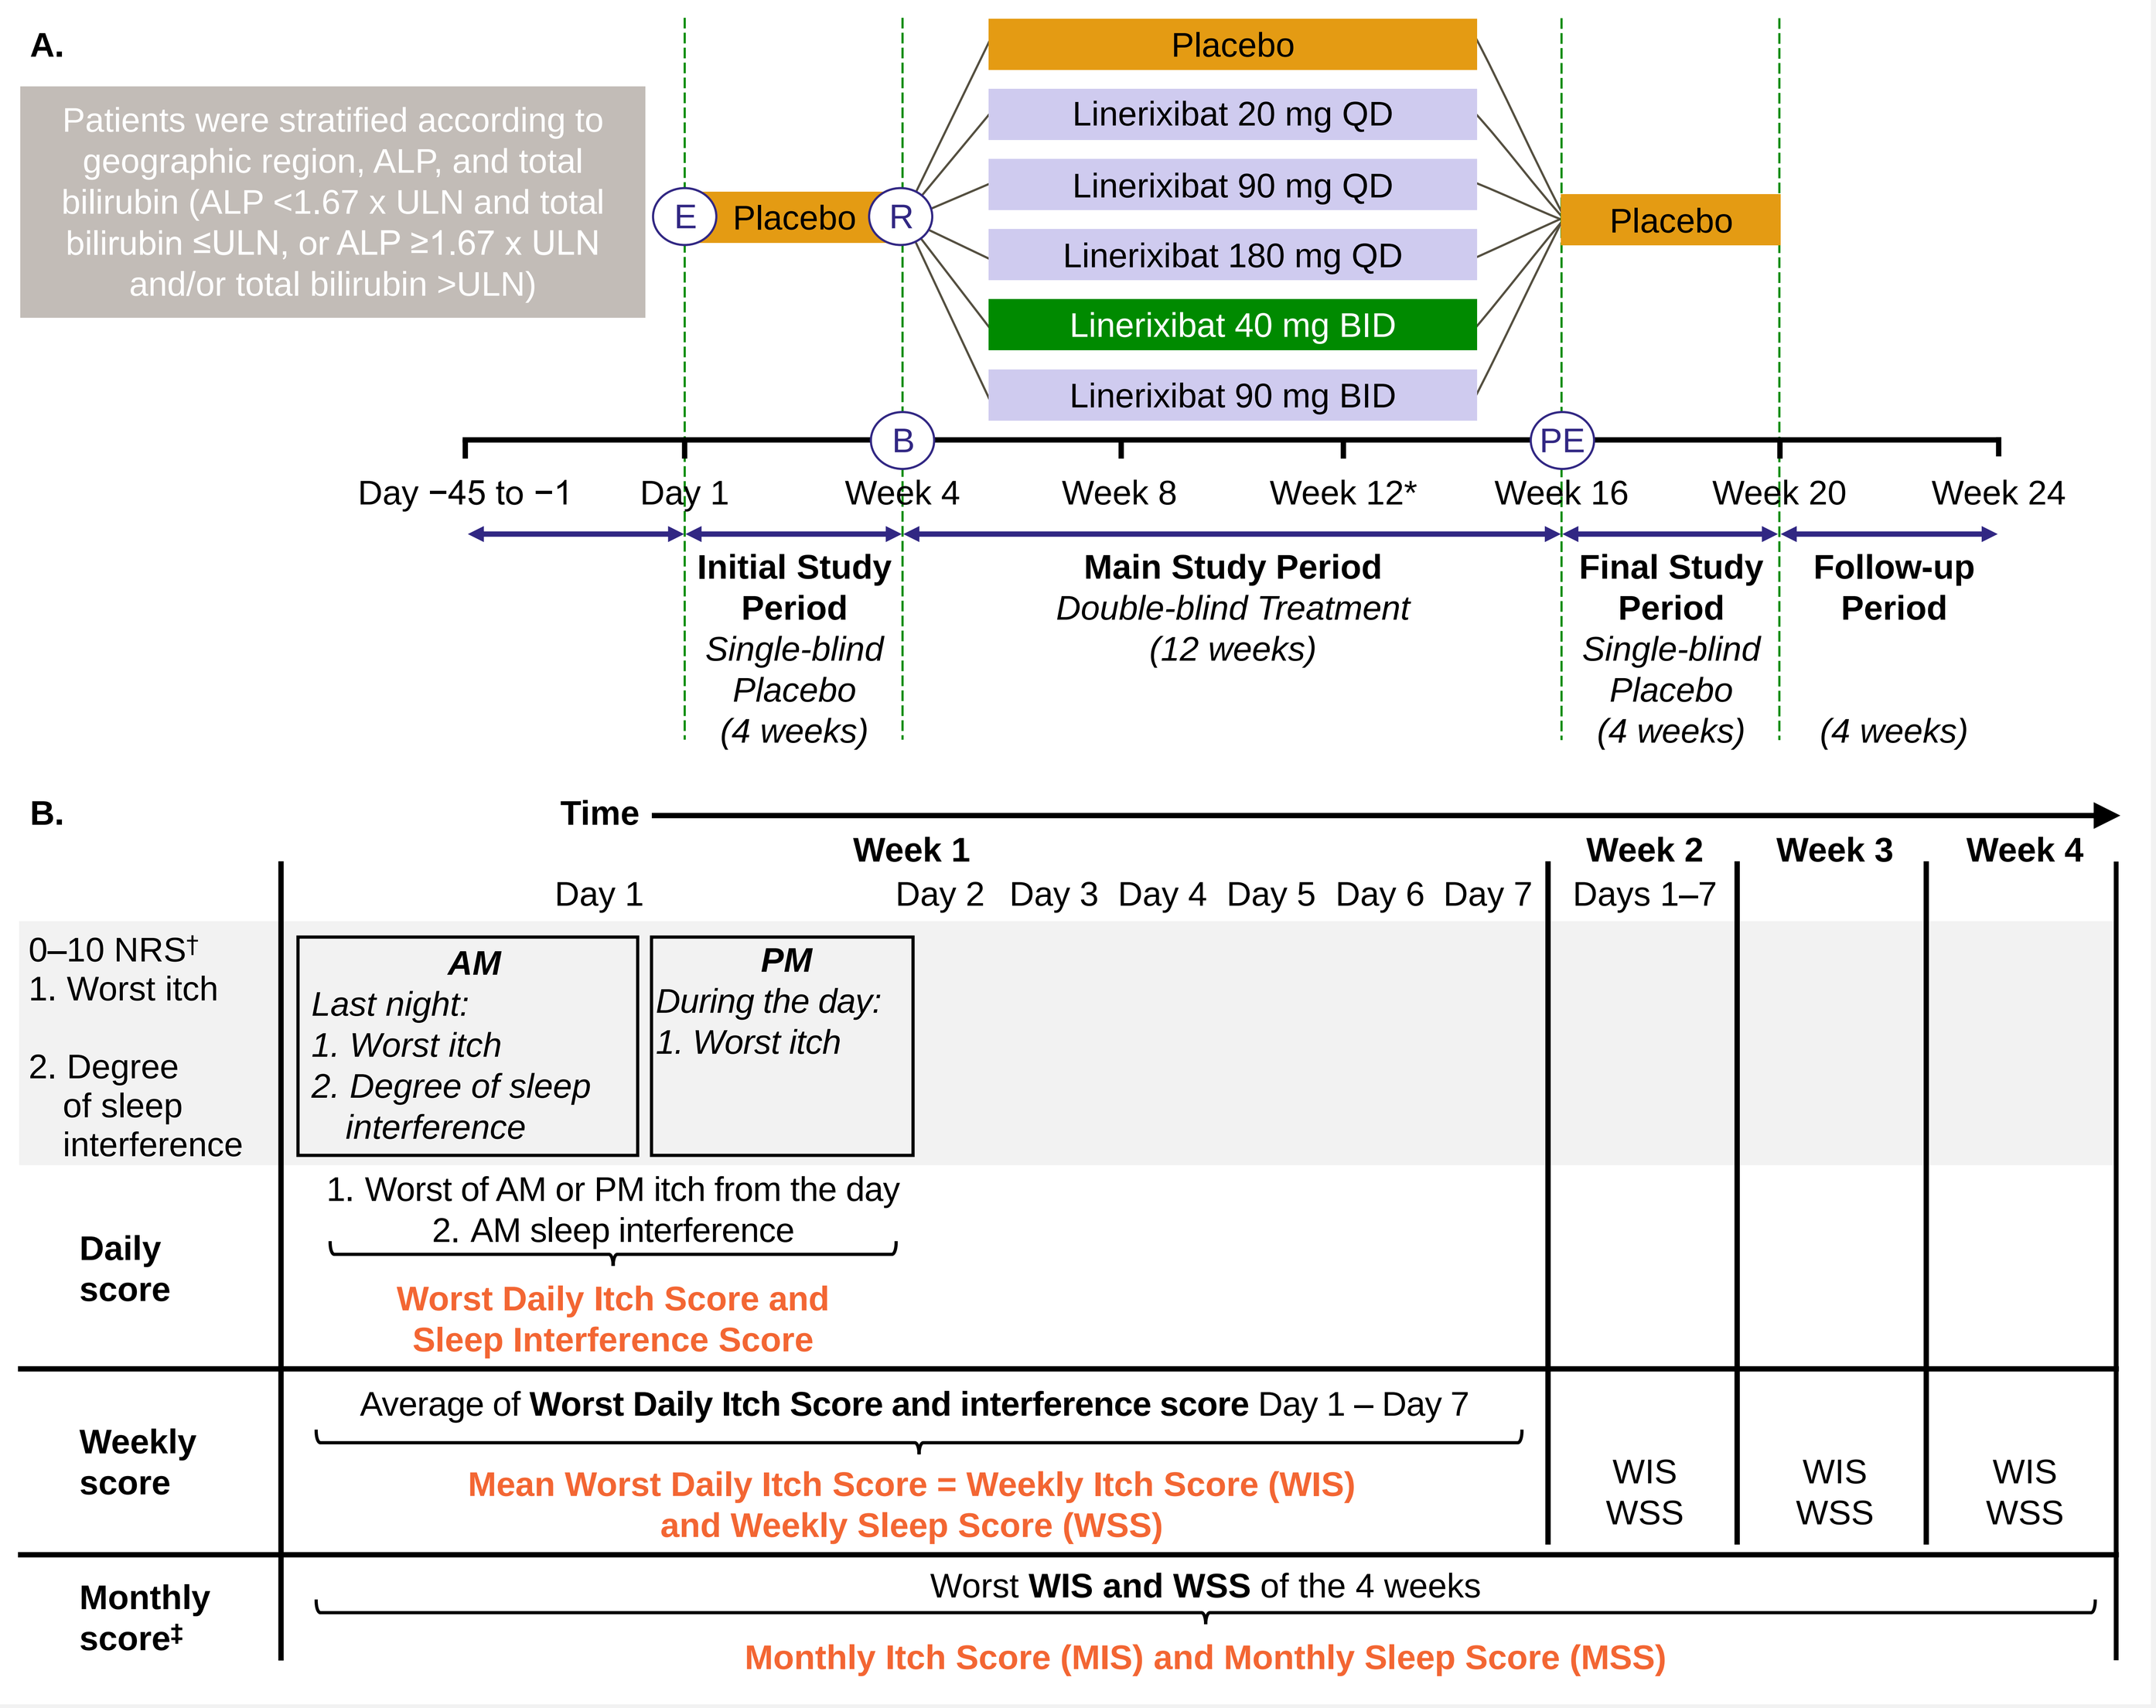
A.
Patients were stratified according to geographic region, ALP, and total bilirubin (ALP <1.67 x ULN and total bilirubin ≤ULN, or ALP ≥1.67 x ULN and/or total bilirubin >ULN)
E
Placebo
R
Placebo
Linerixibat 20 mg QD
Linerixibat 90 mg QD
Linerixibat 180 mg QD
Linerixibat 40 mg BID
Linerixibat 90 mg BID
Placebo
B
PE
Day −45 to −1
Day 1
Week 4
Week 8
Week 12*
Week 16
Week 20
Week 24
Initial Study Period
Single-blind Placebo
(4 weeks)
Main Study Period
Double-blind Treatment
(12 weeks)
Final Study Period
Single-blind Placebo
(4 weeks)
Follow-up Period
(4 weeks)
B.
Time
Week 1
Week 2
Week 3
Week 4
Day 1
Day 2
Day 3
Day 4
Day 5
Day 6
Day 7
Days 1–7
0–10 NRS†
1. Worst itch
2. Degree of sleep interference
AM
Last night:
1. Worst itch
2. Degree of sleep interference
PM
During the day:
1. Worst itch
Daily score
1. Worst of AM or PM itch from the day
2. AM sleep interference
Worst Daily Itch Score and Sleep Interference Score
Weekly score
Average of Worst Daily Itch Score and interference score Day 1 – Day 7
Mean Worst Daily Itch Score = Weekly Itch Score (WIS) and Weekly Sleep Score (WSS)
WIS
WSS
WIS
WSS
WIS
WSS
Monthly score‡
Worst WIS and WSS of the 4 weeks
Monthly Itch Score (MIS) and Monthly Sleep Score (MSS)

*Interim changes (halting recruitment to the linerixibat 20 mg QD group and creating a linerixibat 40 mg BID group) were made at Week 12. †Itch NRS: 0 represents 'no itching', 10 represents 'worst imaginable itching'. Sleep interference NRS: 0 represents 'did not interfere', 10 represents 'completely interfered'. ‡Derived post hoc. ALP, alkaline phosphatase; B, baseline; E, enrolment; PE, primary endpoint; R, randomisation; QD, once daily; ULN, upper limit of normal.

Assessment of itch

- Patients assessed worst itch twice daily on a 0–10 NRS and recorded in an eDiary.
- **Figure 1B** outlines how worst daily itch score, mean worst daily itch (MWDI) and monthly itch score (MIS) were calculated.
- The proportion of patients with a specified change in MIS at Week 16 versus baseline were compared post hoc in patients receiving placebo or linerixibat 40 mg BID using an empirical cumulative distribution function (eCDF) graph.

Additional endpoints

- Sleep interference was assessed as described in **Figure 1B**.
- Health-related QoL (HRQoL) was assessed at all visits from initial study period (Day 1) to the final study period (Week 20) using PBC-40.
- Safety and tolerability were assessed by the incidence of adverse events (AEs), clinical laboratory parameters, electrocardiograms, vital signs and the Gastrointestinal Symptom Rating Scale.
- Changes in total serum bile acids (TSBA) were quantified post hoc using an enzymatic assay consistent with that used in the Phase 2a study.[11] Mean change from baseline to Week 16 was compared post hoc using a mixed model repeated measures analysis (MMRM).
- Changes in biomarkers including the bile acid synthesis biomarker C4 (7α-hydroxy-4-cholesten-3-one), fibroblast-growth factor 19 (FGF-19), and autotaxin were assessed at baseline and Weeks 4, 8, 12, 16 and 20.

Results

- The intention-to-treat population comprised 147 patients. Baseline characteristics for patients receiving placebo (N=36) and linerixibat 40 mg BID (N=23) are presented in **Table 1**.

Table 1. Baseline characteristics for placebo and linerixibat 40 mg BID dose groups

	Placebo (N=36)	Linerixibat 40 mg BID (N=23)
Female, n (%)	34 (94)	22 (96)
Age, mean (SD) years	54 (11)	56 (11)
Race, n (%)		
White	26 (72)	16 (70)
Japanese, East Asian or South-east Asian	8 (22)	4 (17)
Other	2 (6)	3 (13)
Baseline itch severity, n (%)		
Mild (<4)	11 (31)	7 (30)
Moderate (≥4–<7)	14 (39)	13 (57)
Severe (≥7–10)	11 (31)	3 (13)
Baseline WIS, mean (SD)	5.6 (2.0)	5.1 (1.5)
Baseline WSS, mean (SD)	4.0 (2.8)	3.2 (2.2)
Concomitant UDCA, n (%)	32 (89)	21 (91)
Previous bile acid binding resin, n (%)	9 (25)	9 (39)
Concomitant pruritus medications, n (%)		
Any	15 (42)	7 (30)
Antihistamines	7 (19)	4 (17)
Antihistamine + other*	1 (3)	1 (4)

*Other = nalfurafine, naltrexone and sertraline, rifampicin or sertraline. SD, standard deviation; UDCA, ursodeoxycholic acid.

Efficacy

Itch relief

- In the pre-specified primary endpoint of change from baseline at Week 16, linerixibat 40 mg BID reduced MWDI score by −1.13 compared with placebo (95% confidence interval [CI]: −2.29, 0.03).
- In a post hoc subgroup analysis of only those patients with moderate-to-severe pruritus, linerixibat 40 mg BID reduced MWDI score by −1.64 at Week 16 compared with placebo (95% CI: −3.19, −0.10).
- In the eCDF graph for MIS (post hoc), clear separation of the curves was observed for the linerixibat 40 mg BID and placebo groups for a wide range of MIS responder threshold values at Week 16 (**Figure 2**).
 - The largest differences between the curves were observed between thresholds of −3 to −2, where the cumulative percentages were more than 20% greater in the linerixibat group compared with placebo.

Figure 2. Cumulative percentage of patients with specified change from baseline in monthly itch score (MIS) at Week 16

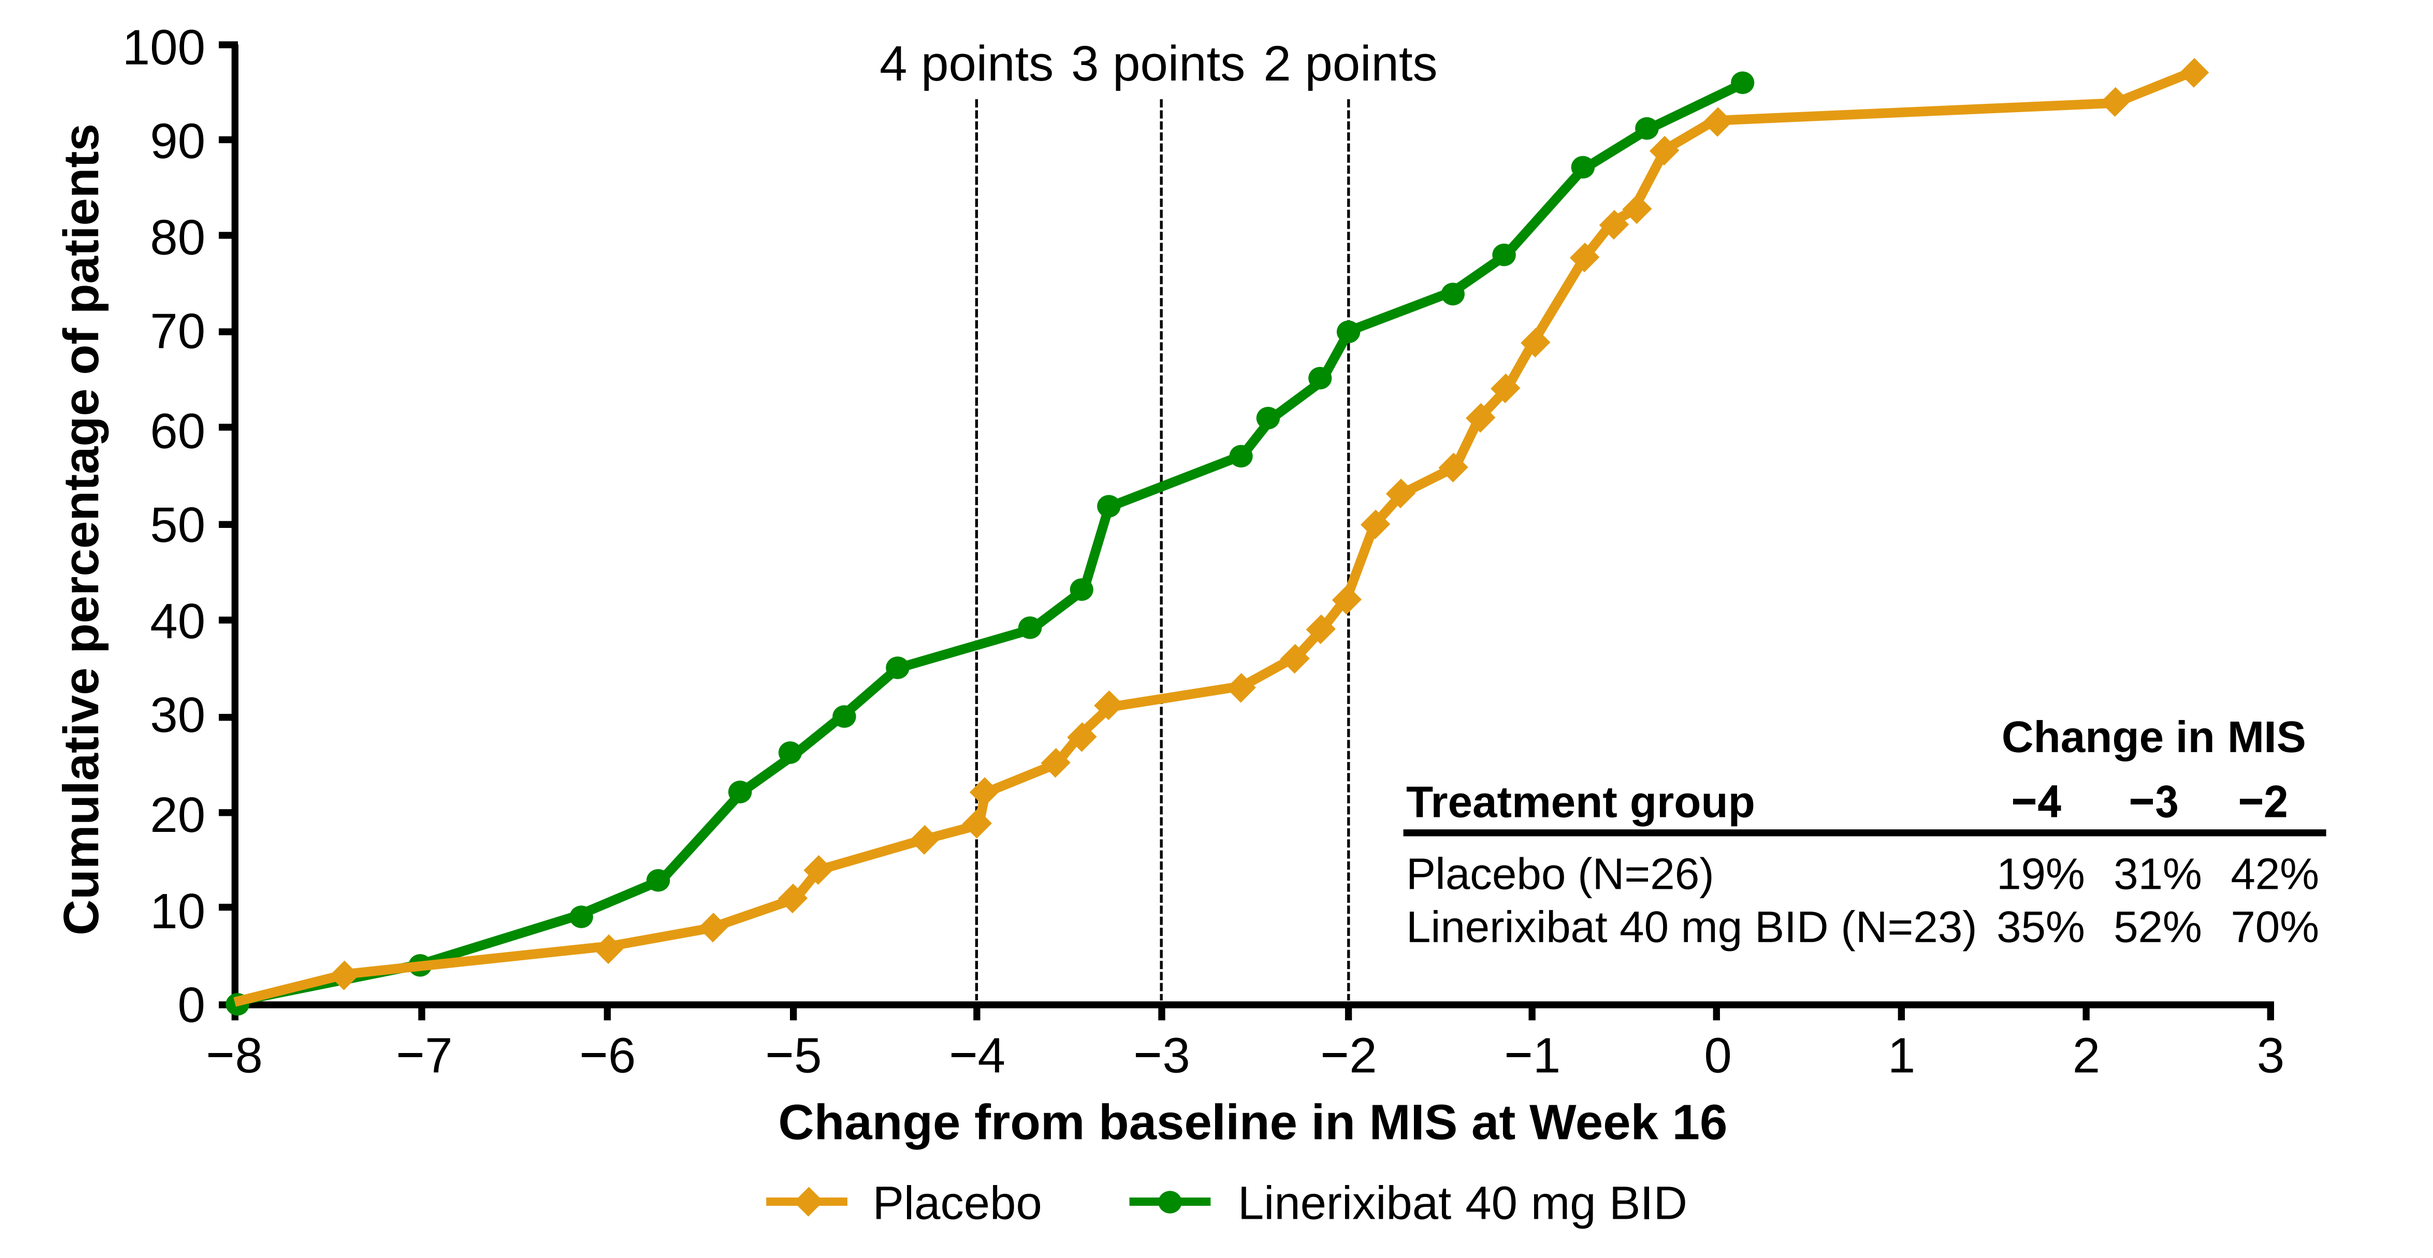
Cumulative percentage of patients
100
90
80
70
60
50
40
30
20
10
0
4 points 3 points 2 points
−8 −7 −6 −5 −4 −3 −2 −1 0 1 2 3
Change from baseline in MIS at Week 16
Change in MIS
Treatment group −4 −3 −2
Placebo (N=26) 19% 31% 42%
Linerixibat 40 mg BID (N=23) 35% 52% 70%
Placebo
Linerixibat 40 mg BID

Additional efficacy endpoints

- In patients receiving linerixibat 40 mg BID, a statistically significant change from baseline in weekly sleep score (WSS) was observed at Week 16 (least squares [LS] mean change: −2.35 [95% CI: −3.19, −1.50). However, this change was not significantly different compared with placebo (LS mean change: −0.96 [95% CI: −2.03, 0.12).
- In relation to HRQoL, a statistically significant change from baseline was observed in the PBC-40 social and emotional domains for patients receiving linerixibat 40 mg BID (LS mean change: −3.1 [95% CI: −5.1, −1.2] and −1.4 [95% CI: −2.2, −0.5], respectively). These changes were not statistically significant compared with placebo (LS mean change: −2.4 [95% CI: −4.8, 0.1] and −0.8 [95% CI: −1.9, 0.3], respectively.

Safety

- There were no deaths or serious AEs attributed to linerixibat during the study.
- The most common AEs are summarised in **Table 2.**
- Diarrhoea was the mostly commonly reported with linerixibat 40 mg BID, consistent with the mechanism of action (MoA). Fatigue and pruritus (the most common symptoms of PBC) were more frequent in the placebo group.

Table 2. Summary of the most frequent on-treatment* AEs (≥5% of patients; safety population)

Preferred term, n (%)	Placebo (N=36)	Linerixibat 40 mg BID (N=23)
Diarrhoea	4 (11)	12 (52)
Abdominal pain	3 (8)	2 (9)
Myalgia	0	2 (9)
Sinusitis	2 (6)	2 (9)
Upper RTI	2 (6)	2 (9)
Fatigue	3 (8)	1 (4)
Lower RTI	2 (6)	0
Muscle spasms	2 (6)	0
Pruritus	2 (6)	0

*On-treatment defined as the period from the date of first randomised study treatment (Week 4) until the last date of randomised study treatment (Week 16) + 2 days. Data ordered according to descending frequency in the linerixibat 40 mg BID group; AEs occurring in ≥5% of patients in either treatment group are shown. RTI, respiratory tract infection.

Pharmacodynamic biomarkers

- In the group of patients receiving linerixibat 40 mg BID, mean (SD) TSBA levels decreased by −6.94 µM (17.5) in comparison with baseline [18.55 µM (21.9)] following 12 weeks of treatment (Week 16; **Figure 3A**).
 - MMRM analysis showed a statistically significant decrease of 39% (p=0.0001) from baseline and 37% (p=0.0030) versus placebo.

Figure 3. Mean change from baseline in serum concentrations of biomarkers over time: (A) TSBA, (B) C4, (C) FGF-19, and (D) autotaxin

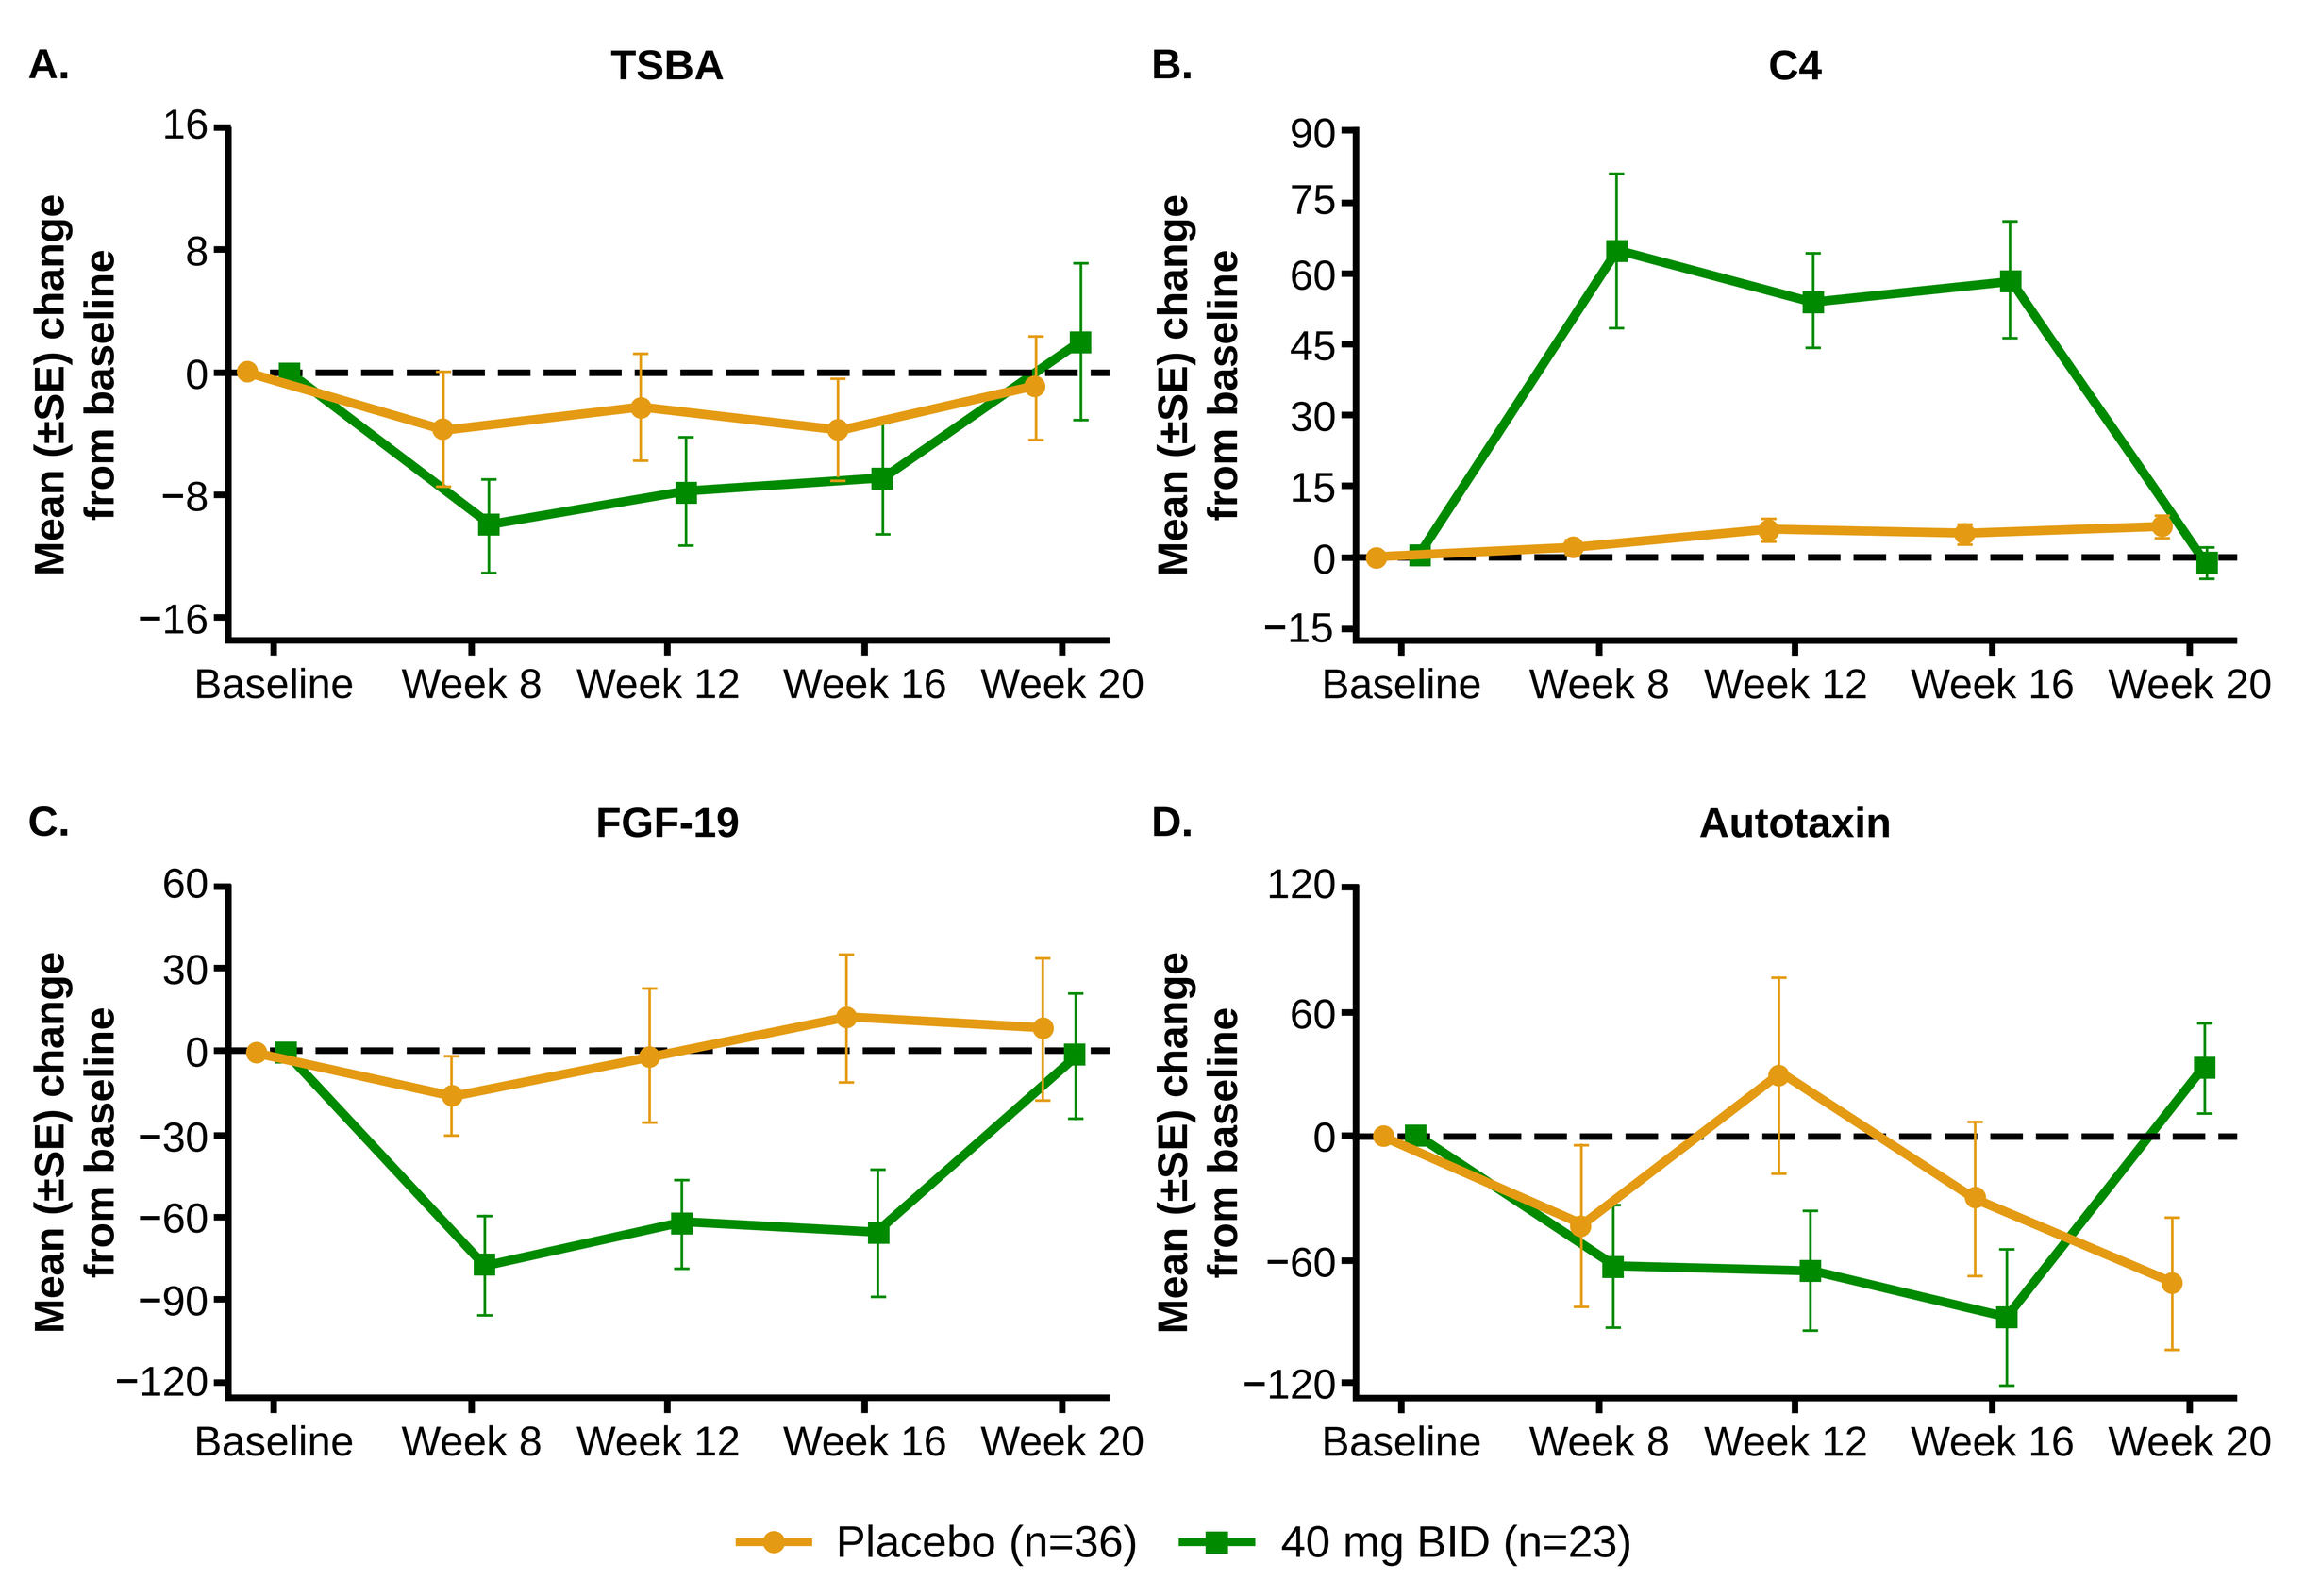
A.
TSBA
B.
C4
C.
FGF-19
D.
Autotaxin
Mean (±SE) change from baseline
Baseline
Week 8
Week 12
Week 16
Week 20
Placebo (n=36)
40 mg BID (n=23)

Error bars represent SE. SE, standard error.

- Linerixibat 40 mg BID was also associated with changes in other biomarkers compared with both baseline and placebo.
 - At Week 16, there was a significant increase from baseline and versus placebo in the pharmacodynamic biomarker C4 (**Figure 3B**).
 - At the same time point, levels of FGF-19 (**Figure 3C**) and autotaxin (**Figure 3D**) were also significantly reduced compared with baseline and versus placebo.

Conclusions

- In comparison with placebo, the proportion of patients with a change from baseline in monthly itch score at Week 16 was greater in patients receiving linerixibat 40 mg BID over a range of responder threshold values and especially between thresholds of −3 to −2, where the cumulative percentages were more than 20% greater in the linerixibat group compared with placebo.
- As previously reported,[12] and further highlighted here for the 40 mg BID group, linerixibat led to significant improvements from baseline in sleep, and in the emotional and social domains of the PBC-40, with a favourable safety profile.
- Consistent with linerixibat's MoA of inhibiting reuptake of bile acids, a reduction in TSBA was observed over the course of 12 weeks of linerixibat 40 mg BID treatment.
- Linerixibat 40 mg BID is being studied in the ongoing confirmatory Phase 3 GLISTEN trial.
 - Responder thresholds of 2-, 3- and 4-point improvements in MIS compared with baseline are key secondary endpoints of this study.
 - In addition, the impact of linerixibat 40 mg BID on sleep and QoL measures will be assessed in the larger study population enrolled in GLISTEN.

References

1. Hegade S, et al. *Clin Gastroenterol Hepatol* 2019;17:1379–87.
2. Younossi ZM, et al. *J Gastroenterol* 2000;95:497–502.
3. Montagnese S, et al. *Liver Int* 2013;33:203–09.
4. Hegade VS, et al. *Frontline Gastroenterol* 2016;7:158–66.
5. Jin XY, Khan TM. *J Formos Med Assoc* 2016;115:689–702.
6. Hönig S, et al. *J Hepatol 2018*;68:S216.
7. Tajiri K, Shimizu Y. *World J Gastroenterol* 2017;23:3418–26.
8. Rudic JS, et al. *Cochrane Database Syst Rev* 2012;12:CD000551.
9. Nevens, F et al. *N Engl J Med* 2016;375:631–43.
10. Al-Dury S, Marschall HU. *Front Pharmacol* 2018;9:931.
11. Hegade VS, et al. *Lancet* 2017;389:1114–23.
12. Levy C, et al. AASLD – The Liver Meeting Digital Experience 2020; poster LP38.
13. https://clinicaltrials.gov/ct2/show/NCT04950127.

Disclosures

- This study was funded by GlaxoSmithKlimne (GSK Study 201000; NCT02966834).
- The presenting author, James Fettiplace, declares the following real or perceived conflicts of interest during the last 3 years in relation to this presentation: employee and shareholder of GSK.
- **BS**, **SZ**, **RvM** and **MMM** are employees of GSK and hold GSK shares.
- Editorial support (in the form of writing, layout assistance, collating authors' comments for each draft, and grammatical editing) was provided by Gemma Corr, of Fishawack Indicia Ltd, UK, part of Fishawack Health, and was funded by GSK.

Presented at International Liver Congress™ 2022 (European Association for the Study of the Liver), 22–26 June 2022, London, United Kingdom

PosterSessionOnline.com
Scan to download the poster

THE INTERNATIONAL LIVER CONGRESS™
DOI: 10.3252/pso.eu.ILC2022.2022
Immune-mediated and cholestatic diseases
James Fettiplace
THU-485
ILC2022