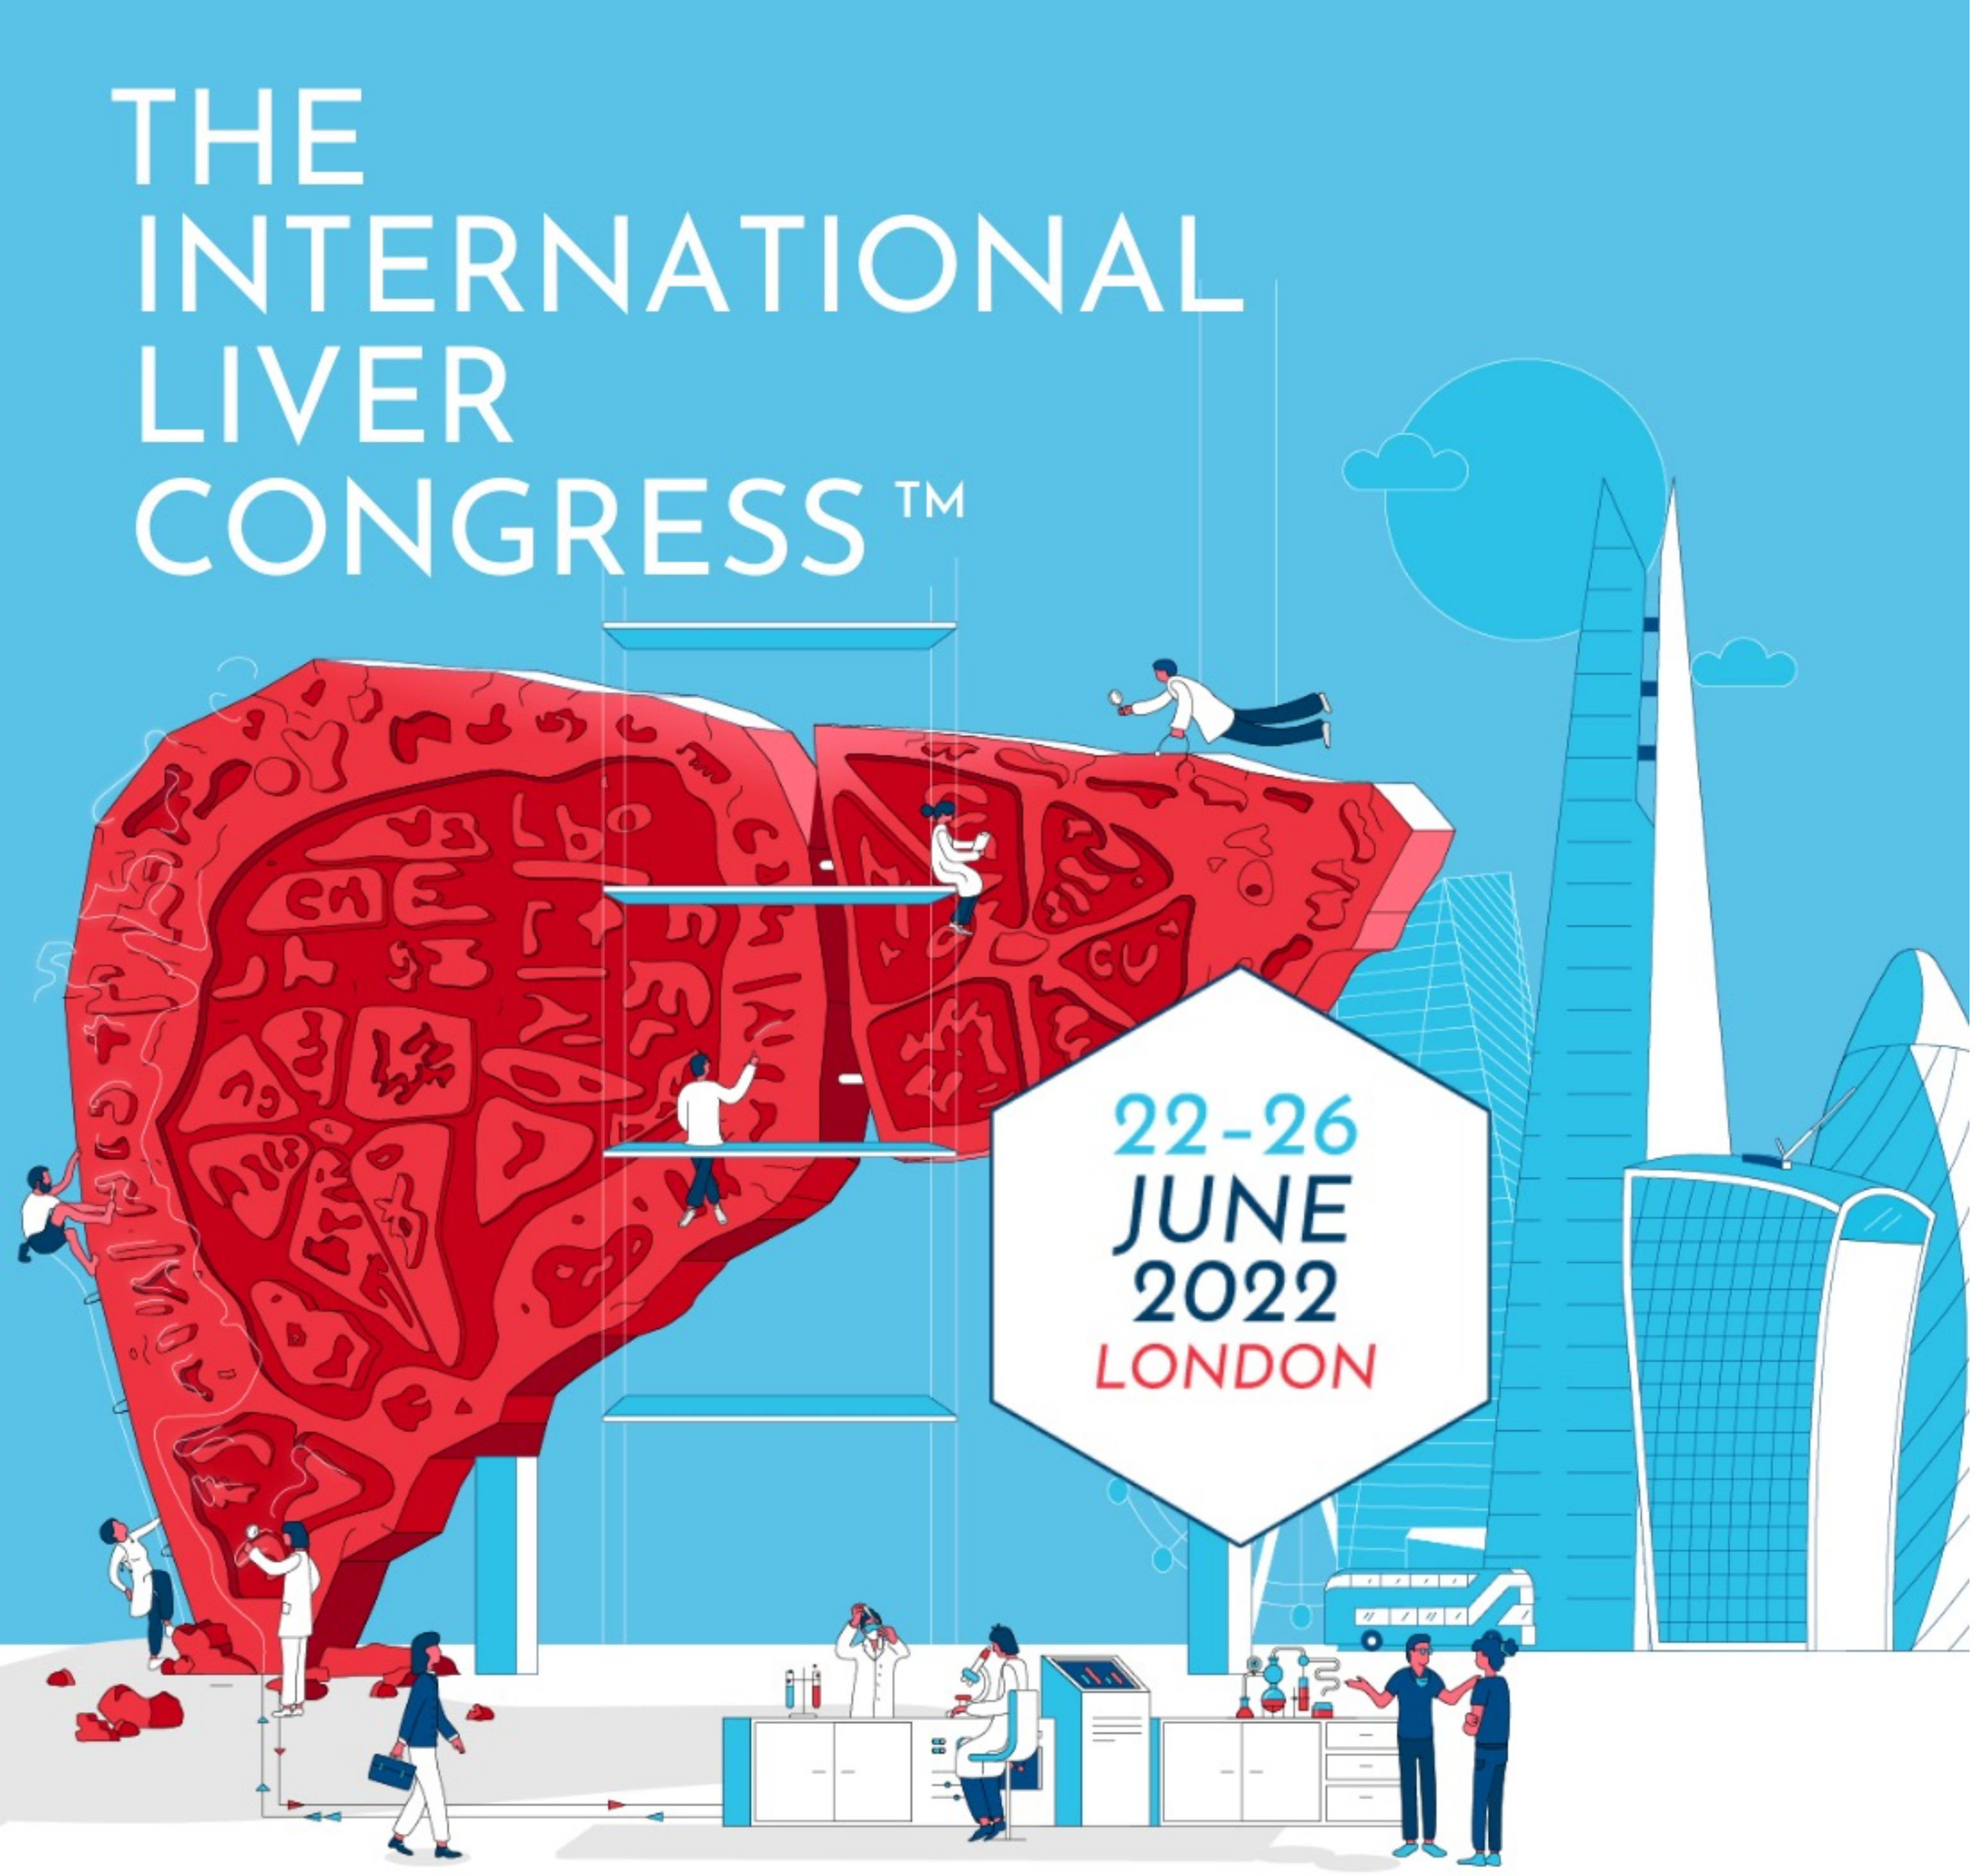
THE INTERNATIONAL LIVER CONGRESS™
22-26 JUNE 2022 LONDON

ELIMINATE – interim results of an Austrian HCV macro-elimination project

C. Schwarz[1,2,3 *], D. Bauer[1,2 *], L. Dorn[4 *], M. Jachs[1,2], L. Hartl[1,2], H. Holzmann[5], N. Pfisterer[1,2,6], B. Hennlich[6], F. Mayer[7], R. Schmidt[7], A. Voill-Glaninger[8], W. Hübl[9], M. Willheim[10], K. Köhrer[11], D. Chromy[12], S. Jansen-Skoupy[13], S. Tomez[14], W. Krugluger[14,15], C. Madl[6,16], M. Schwarz[1,2,3], L. Brinkmann[1,2,3], L. Burghart[1,2,3], G. Weidinger[17], F. Riedl[4], H. Laferl[18], C. Wenisch[18], A. Aburaia[19], C. Sebesta[19,20], D. Schmid[21], T. Reiberger (Corr.)[1,2 #], A. Maieron[4 #], M. Gschwantler[3,16 #]

[1] Division of Gastroenterology and Hepatology, Department for Medicine III, Medical University of Vienna, Vienna, Austria; [2] Vienna HIV & Liver Study Group, Medical University of Vienna, Vienna, Austria; [3] Department for Internal Medicine IV, Klinik Ottakring, Vienna, Austria; [4] Department for Internal Medicine II, University Clinic St. Pölten, St. Pölten, Austria; [5] Center for Virology, Medical University of Vienna, Vienna, Austria; [6] Department for Internal Medicine IV, Klinik Landstraße, Vienna, Austria; [7] Clinical Institute for Laboratory Medicine, Medical University of Vienna, Vienna, Austria; [8] Central Laboratory and Blood Bank, Klinik Landstraße, Vienna, Austria; [9] Central Laboratory, Klinik Ottakring, Vienna, Austria; [10] Clinical Institute for Laboratory Medicine, University Clinic St. Pölten, St. Pölten, Austria; [11] Institute for Medical-Chemical and Molecularbiological Laboratory Diagnostics with Blood Depot, Landesklinikum Wiener Neustadt, Wiener Neustadt, Austria; [12] Department for Dermatology, Medical University of Vienna, Vienna, Austria; [13] Institute for Laboratory Diagnostics, Klinik Favoriten, Vienna, Austria; [14] Institute for Laboratory Medicine with Blood Depot, Klinik Donaustadt, Vienna, Austria; [15] Institute for Laboratory Medicine and Blood Depot, Klinik Floridsdorf, Vienna, Austria; [16] Sigmund Freud University, Vienna, Austria; [17] Department for Internal Medicine, Gastroenterology and Hepatology, Landesklinikum Wiener Neustadt, Wiener Neustadt, Austria; [18] Department for Internal Medicine IV, Klinik Favoriten, Vienna, Austria; [19] Department for Internal Medicine and Gastroenterology, Klinik Floridsdorf, Vienna, Austria; [20] Department for Internal Medicine II, Klinik Donaustadt, Vienna, Austria; [21] Austrian Agency for Health and Food Safety (AGES), Vienna, Austria.

** shared first authorship, # shared last authorship*

1 Background & Aims

The WHO has declared the elimination of HCV until 2030 a global public health goal. We present the preliminary results of an Austrian macro-elimination approach aiming to provide linkage to care and treatment initiation for persons who are recalled based on a “last-positive” HCV-RNA PCR result. Importantly, many of these lost-to-follow-up patients do not belong to known HCV risk groups and are, thus, often not recognized by targeted HCV screening/elimination projects.

2 Method

First, we identified patients with a “last-positive” HCV-RNA PCR result between 2010-2020 in laboratories of Eastern Austria (Burgenland, Lower Austria, Vienna) and (i) described their demographic characteristics. Subsequently, (ii) a systematic recall of these patients to the respective HCV treatment centers was performed, and (iii) HCV-DAA was initiated and SVR was monitored. Here we report the interim results of this ELIMINATE project including data/results from eight centers.

3 Funding

This study was supported by GILEAD.

4 Results

Overall, 22682 patients underwent HCV-RNA PCR testing, 11216 (49.4%) showed a positive HCV-RNA result at any given time, and in 6223 (27.4%) patients the last available HCV-RNA result was positive (suggesting persisting HCV-viremia) *(Fig. 1)*.

Fig. 1

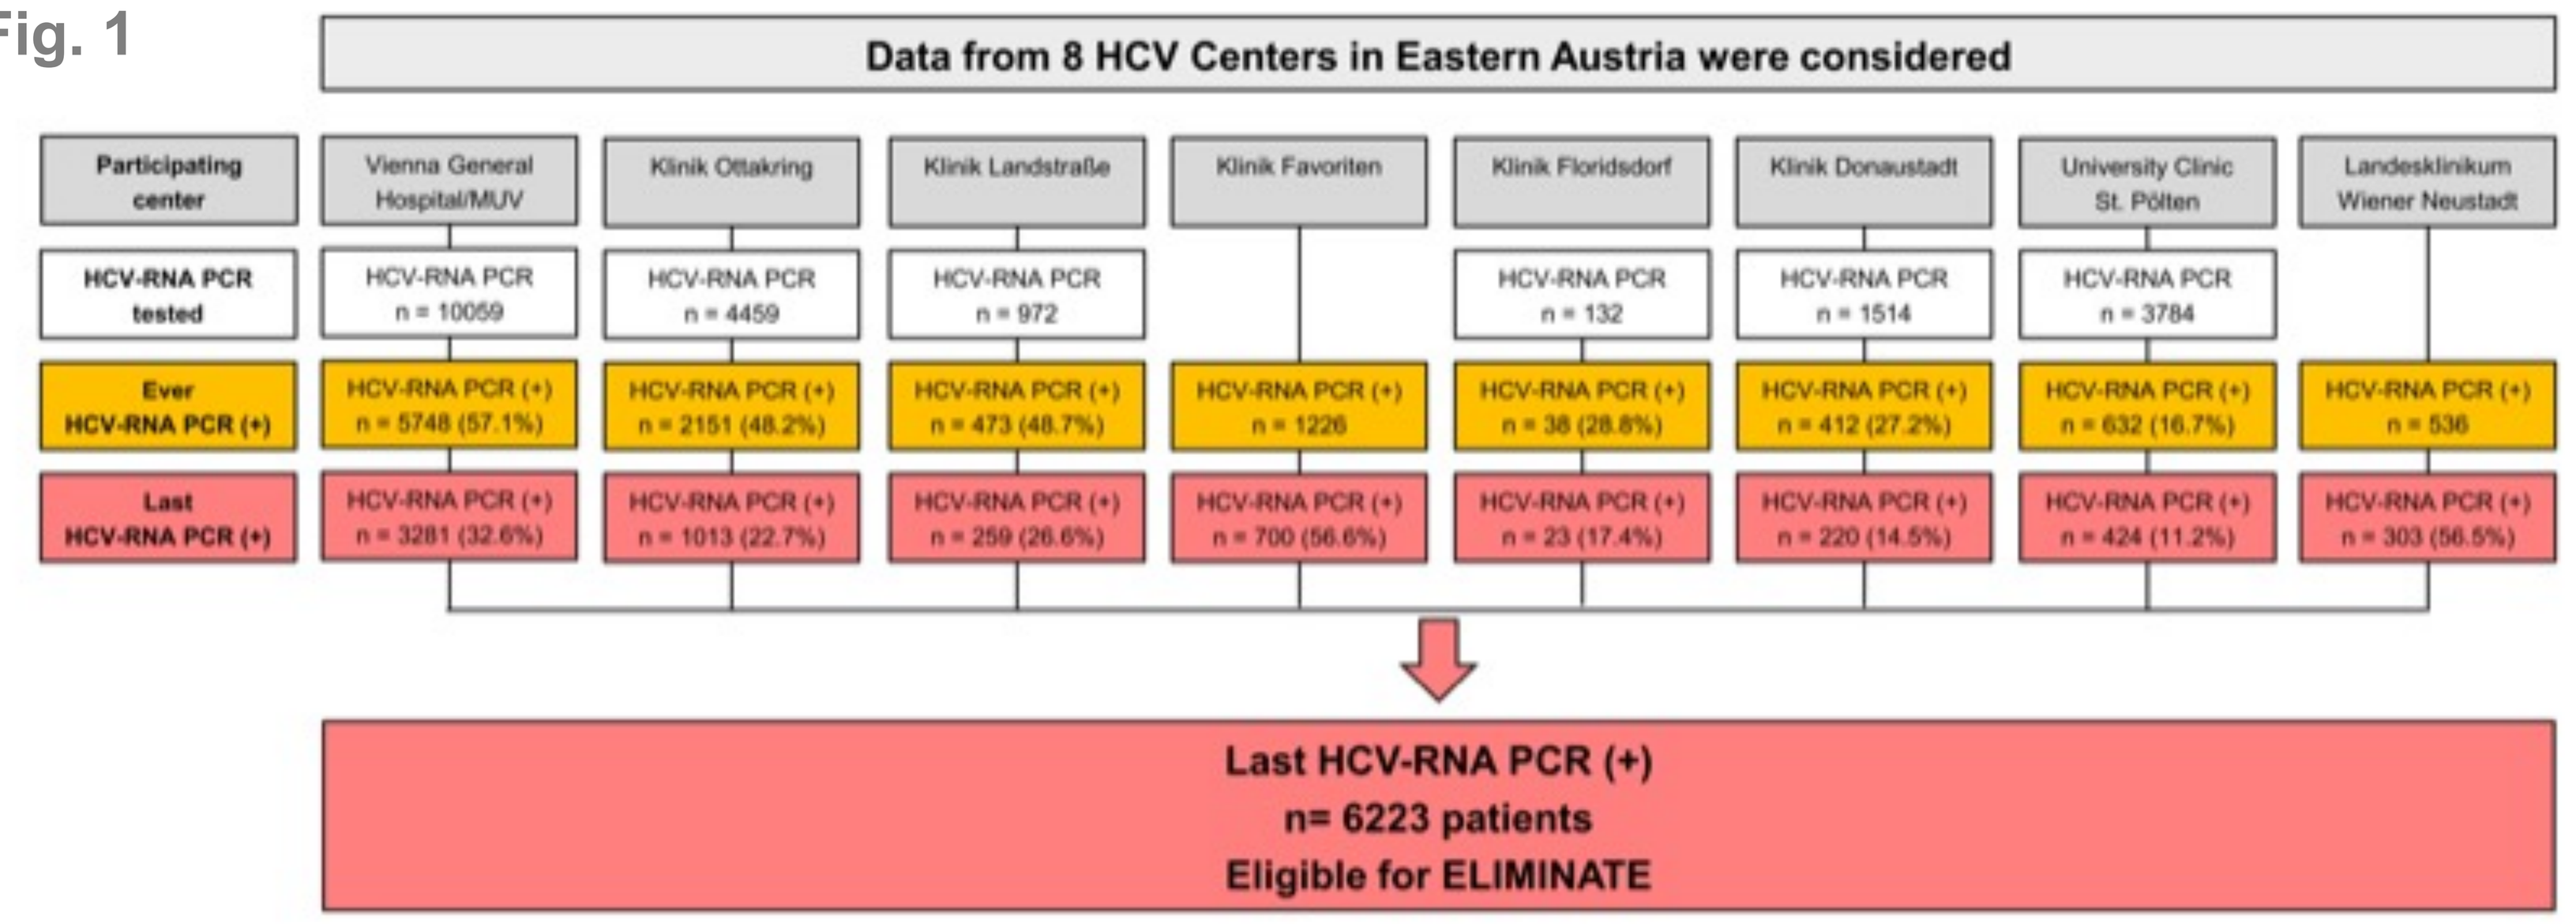
Data from 8 HCV Centers in Eastern Austria were considered
Participating center
Vienna General Hospital/MUV
Klinik Ottakring
Klinik Landstraße
Klinik Favoriten
Klinik Floridsdorf
Klinik Donaustadt
University Clinic St. Pölten
Landesklinikum Wiener Neustadt
HCV-RNA PCR tested
HCV-RNA PCR n = 10059
HCV-RNA PCR n = 4459
HCV-RNA PCR n = 972
HCV-RNA PCR n = 132
HCV-RNA PCR n = 1514
HCV-RNA PCR n = 3784
Ever HCV-RNA PCR (+)
HCV-RNA PCR (+) n = 5748 (57.1%)
HCV-RNA PCR (+) n = 2151 (48.2%)
HCV-RNA PCR (+) n = 473 (48.7%)
HCV-RNA PCR (+) n = 1226
HCV-RNA PCR (+) n = 38 (28.8%)
HCV-RNA PCR (+) n = 412 (27.2%)
HCV-RNA PCR (+) n = 632 (16.7%)
HCV-RNA PCR (+) n = 536
Last HCV-RNA PCR (+)
HCV-RNA PCR (+) n = 3281 (32.6%)
HCV-RNA PCR (+) n = 1013 (22.7%)
HCV-RNA PCR (+) n = 259 (26.6%)
HCV-RNA PCR (+) n = 700 (56.6%)
HCV-RNA PCR (+) n = 23 (17.4%)
HCV-RNA PCR (+) n = 220 (14.5%)
HCV-RNA PCR (+) n = 424 (11.2%)
HCV-RNA PCR (+) n = 303 (56.5%)
Last HCV-RNA PCR (+)
n= 6223 patients
Eligible for ELIMINATE

Fig. 1: Flowchart of the screened population.
HCV-RNA PCR results by participating center (acquired 2008-2020). For Klinik Favoriten (Vienna) and Landesklinikum Wiener Neustadt (Lower Austria), the total number of HCV-RNA PCR tests performed during the observation period was not available, therefore calculations were based on the number of persons with "Ever HCV-RNA PCR (+)“.
Abbreviations: HCV, hepatitis C virus; RNA, ribonucleic acid; PCR, polymerase chain reaction; MUV, Medical University of Vienna.

For this interim report, 2252/6223 HCV-RNA PCR(+) patients were evaluated: 215 (9.5%) were linked to HCV-care *(Fig. 2A, B)*. 82 (3.6%) underwent liver disease evaluation and in 56 (2.5%) DAA treatment was initiated. In 38 (1.7%) sustained virologic response was documented.

Main reasons for the suboptimal cascade of care included invalid contact data (687; 30.5%) or patients' unavailability at the time of recall (424; 18.8%) and death (429; 19.0%). Successful DAA therapy at other centers had already been conducted in 497 patients (22.1%).

Fig. 2A

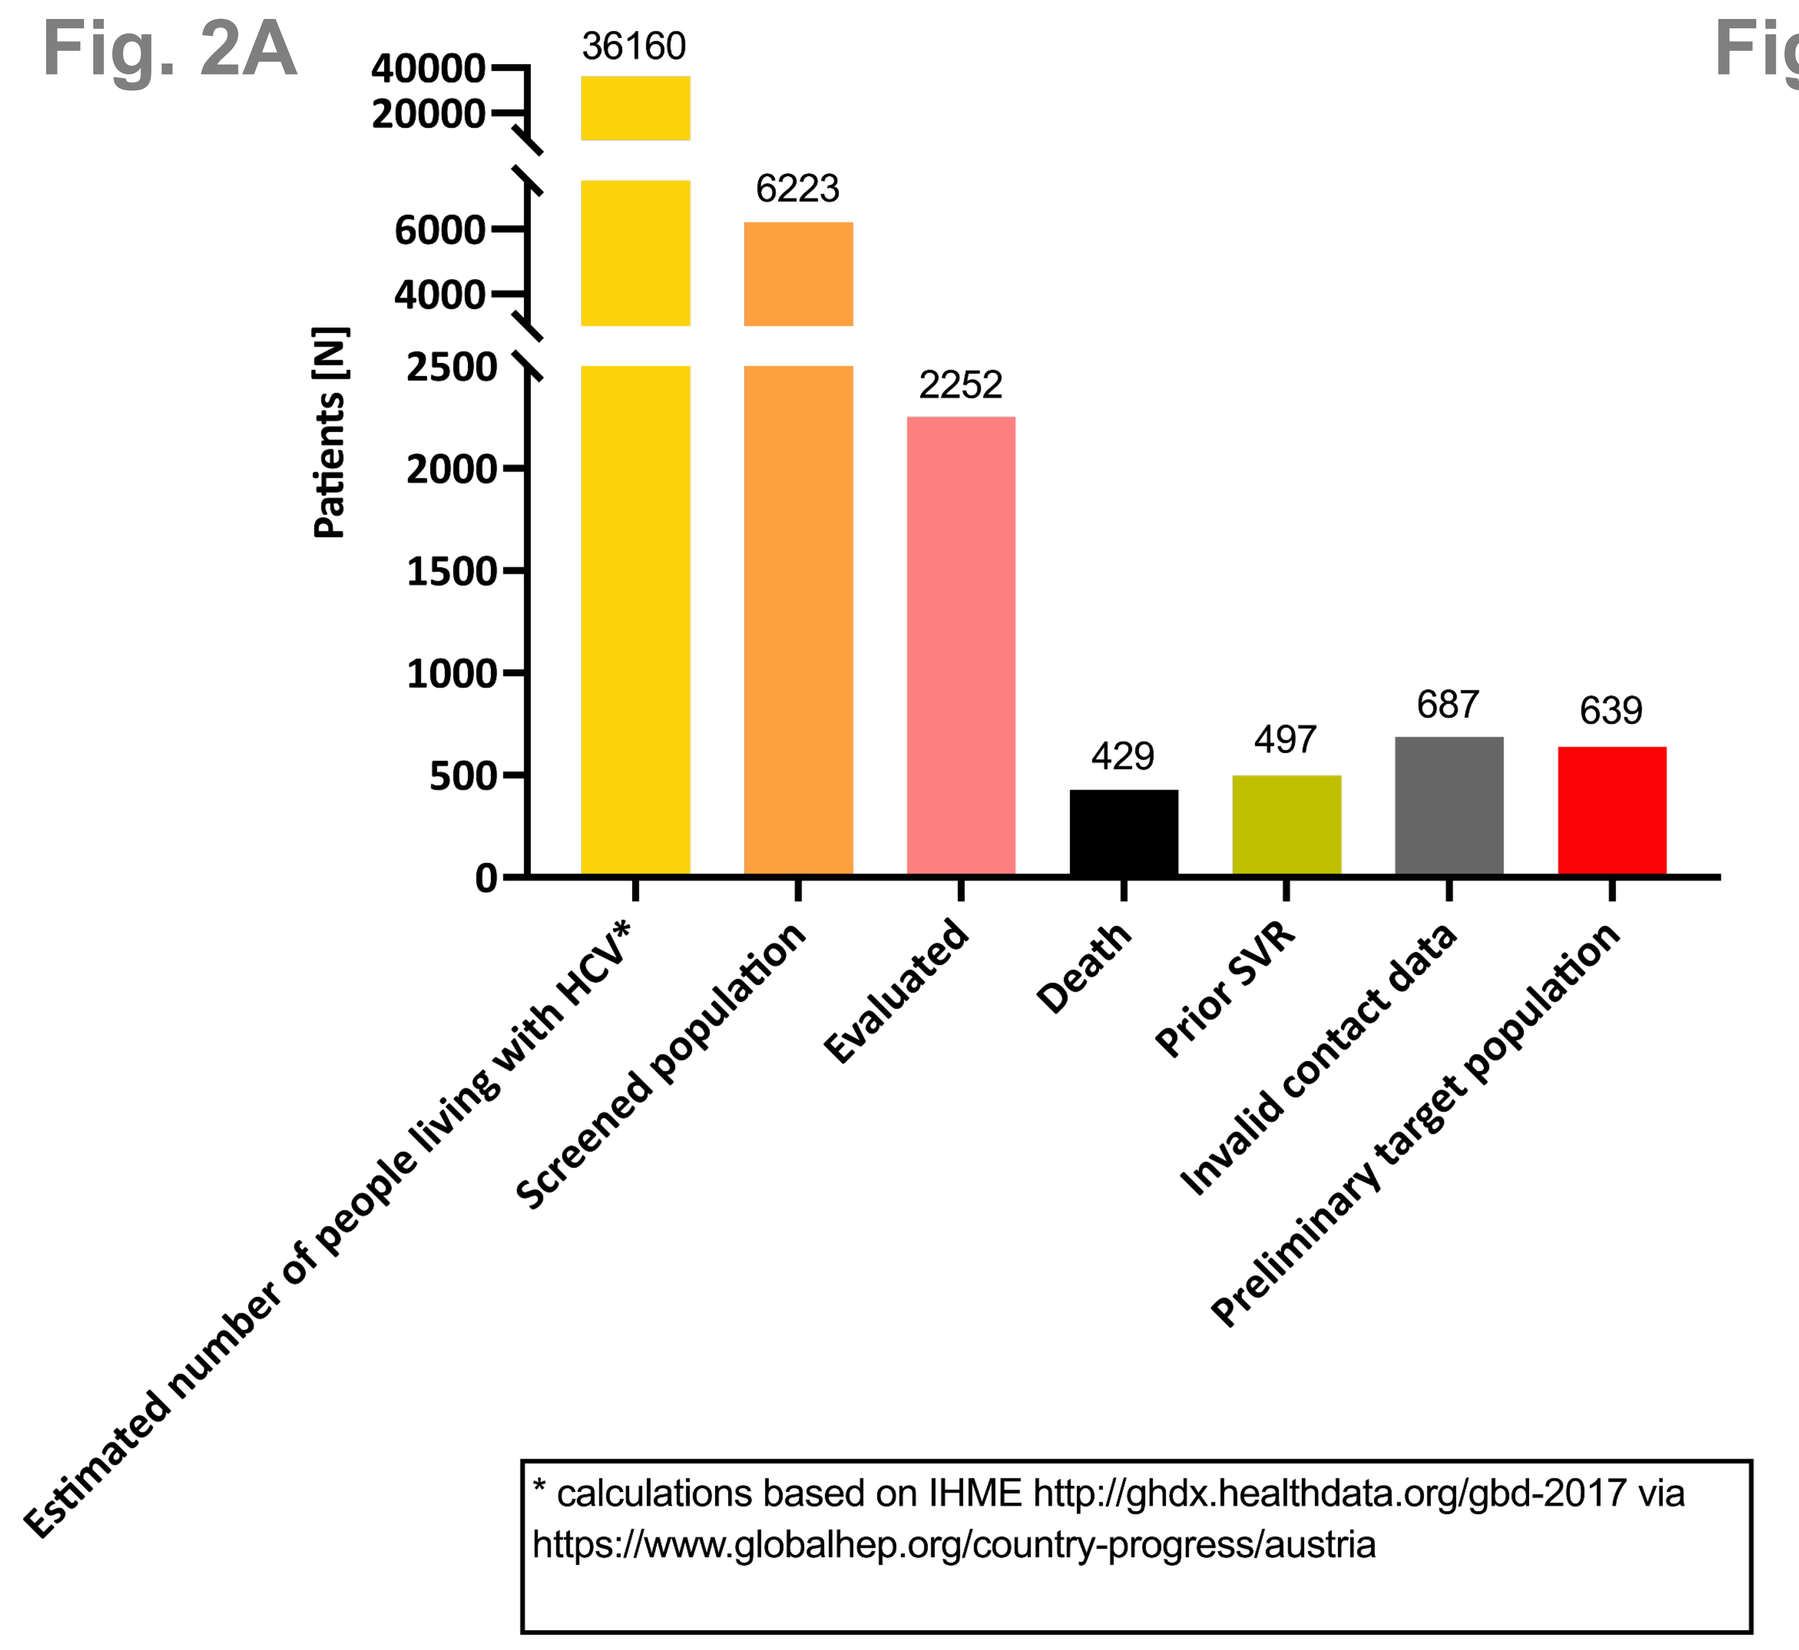
Patients [N]
40000
20000
6000
4000
2500
2000
1500
1000
500
0
36160
6223
2252
429
497
687
639
Estimated number of people living with HCV*
Screened population
Evaluated
Death
Prior SVR
Invalid contact data
Preliminary target population
* calculations based on IHME http://ghdx.healthdata.org/gbd-2017 via https://www.globalhep.org/country-progress/austria

Fig. 2B

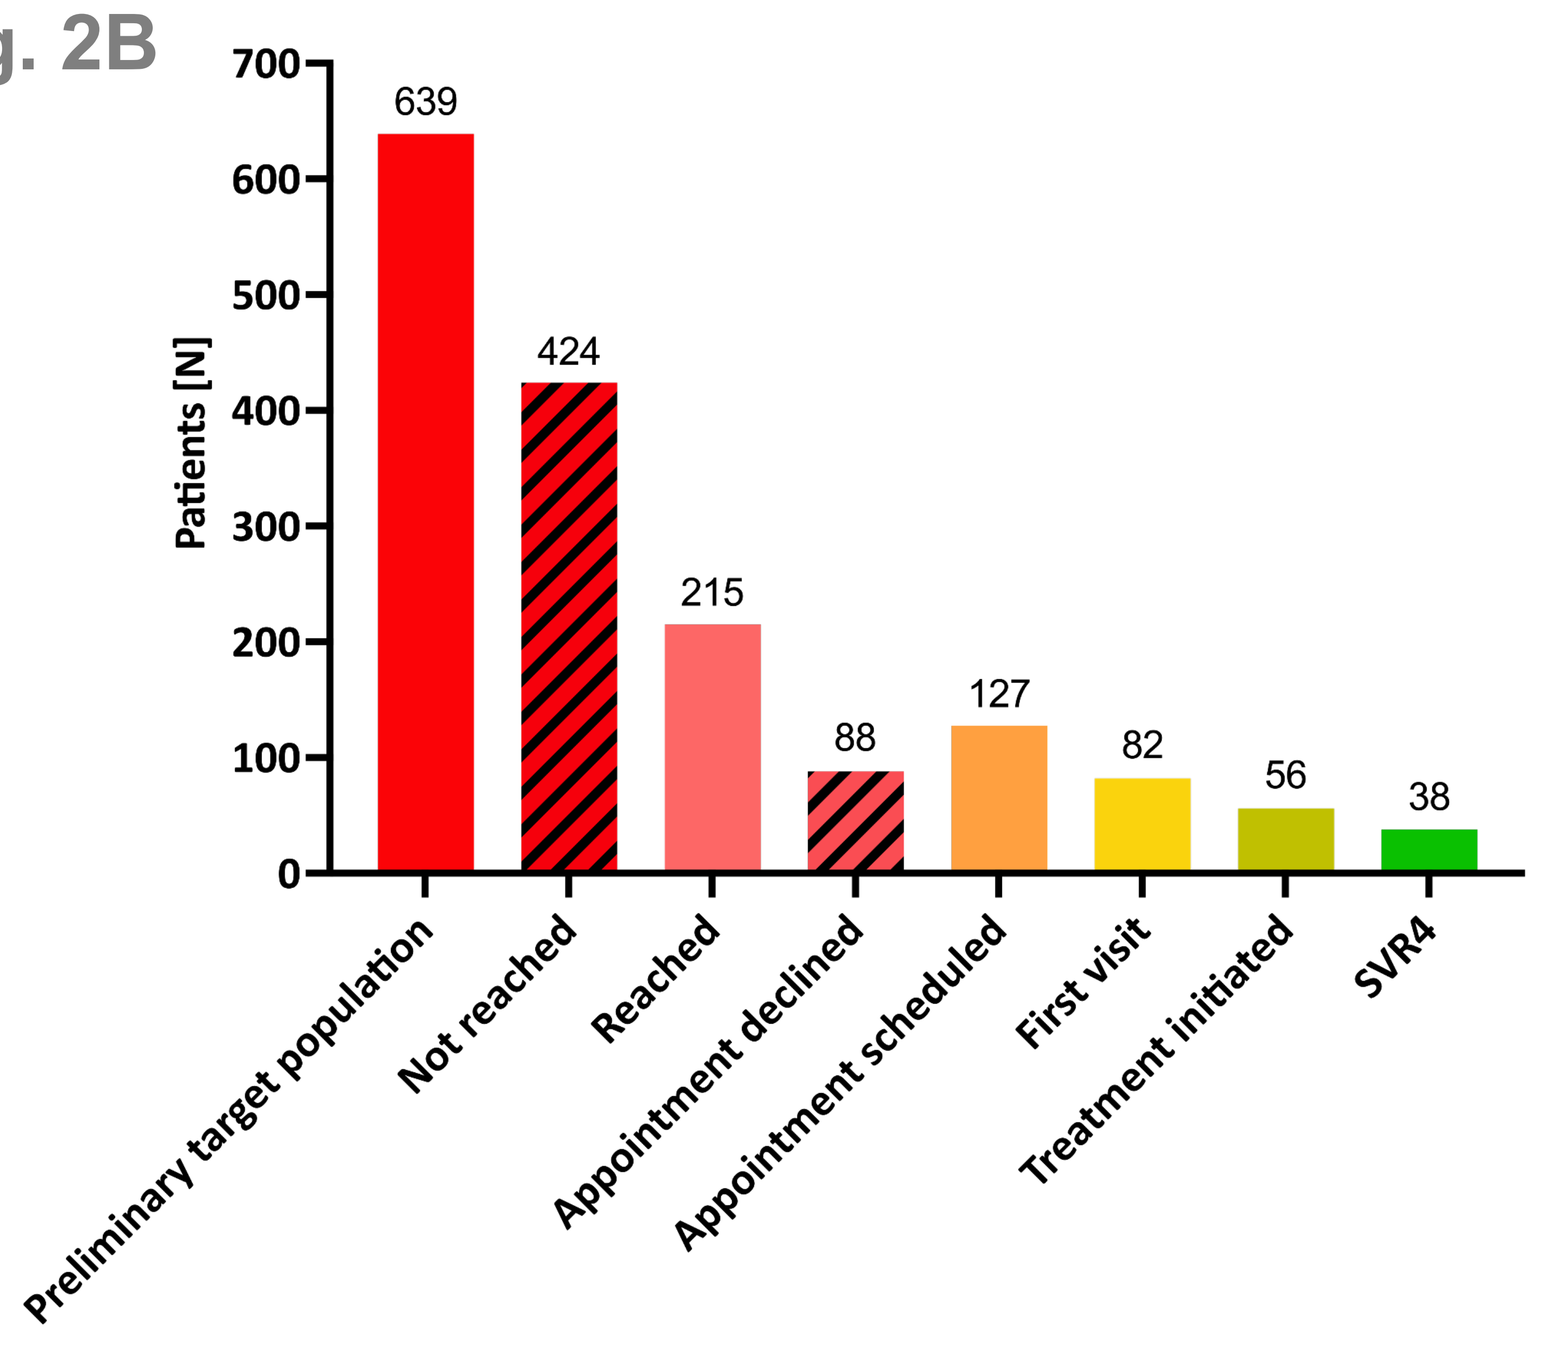
Patients [N]
700
600
500
400
300
200
100
0
639
424
215
88
127
82
56
38
Preliminary target population
Not reached
Reached
Appointment declined
Appointment scheduled
First visit
Treatment initiated
SVR4

Fig. 2: Preliminary workup of the screened population.
Evaluation of the "screened population" and assessment of the "target population" in the context of the estimated number of people living with HCV and the current progress of the ELIMINATE project at the time of preliminary data analysis.
- 2A: Determination of the preliminary "target population".
- 2B: Cascade of care based on the preliminary "target population".
Abbreviations: HCV, hepatitis C virus; SVR, sustained virologic response; LTFU, lost to follow-up.

5 Conclusions

This lab record-based ELIMINATION project identified a considerable number of 6223 HCV patients with potential persisting viremia. Invalid contact data (30.5%) and pre-treatment death (19.0%) represent major problems/barriers. Importantly, at this interim stage 215 (9.5%) patients were successfully linked to care and 56 (2.5%) patients started DAA therapy.

6 Contact information

Corresponding author: Thomas Reiberger (thomas.reiberger@meduniwien.ac.at)

PosterSessionOnline
Scan to download the poster
THE INTERNATIONAL LIVER CONGRESS™
DOI: 10.3252/pso.eu.ILC2022.2022
Viral hepatitis
Caroline Schwarz
FRI--309
ILC2022