The therapeutic potential of αv integrins in liver fibrosis

S R. Rahman[1,2,3], A J. Bennett[1,2,3], J I. Grove[2,3], J A. Roper[4], K T. Pun [4], G P. Aithal[2,3]

[1]FRAME Alternatives Laboratory, University of Nottingham, Nottingham, UK [2]NIHR Nottingham Biomedical Research Centre, Nottingham University Hospitals NHS Trust and University of Nottingham, Nottingham, UK. [3]Nottingham Digestive Diseases Centre, University of Nottingham, Nottingham, UK. [4]Novel Human Genetics Research Unit, GlaxoSmithKline, Stevenage, UK. **Contact information:** syedia.rahman@nottingham.ac.uk

Introduction

- Liver fibrosis occurs in most chronic liver diseases[1], affects an estimated 844 million people worldwide and accounts for 2 million deaths per year[2].
- Hepatic stellate cells (HSC) are the primary effector cells in liver fibrosis.
- HSCs secrete inactive transforming growth factor β (TGFβ) which is then activated by αv integrins, leading to fibrosis.
- αv integrins can form heterodimers with β1, 3, 5, 6 or 8 subunits.
- Specific inhibitors which target αv integrins and their particular β subunit have shown to reduce fibrosis progression in mouse models[3].
- **The role of integrins in human liver disease is currently unclear[3].**

Aims

- To detect integrin expression in human liver samples from fibrotic and non-fibrotic samples.
- To assess integrin gene and protein expression in activated human HSCs.
- To test capability of subtype selective integrin inhibitors to inhibit TGFβ activation in activated human HSCs.

Methods

- Tissue expression of integrins in fibrotic and non-fibrotic liver was examined using RNAscope®[4] in biopsies from patients with non-alcoholic steatohepatitis with bridging fibrosis (F3)[5].
- HSCs were isolated from human liver tissue obtained with full ethical approval following hepatic resection at QMC Nottingham and integrin gene and protein expression was measured using qPCR and western blot.
- HSCs were co-cultured with mink lung epithelial cells (MLEC) transformed to stably express firefly luciferase under the control of a TGFβ-sensitive portion of the plasminogen activator inhibitor-1 (PAI-1) promoter. These TGFβ reporter cells were used to detect levels of active TGFβ[6] and was used to test a β1 integrin inhibitor, compound 8 (C8)[7].

Conclusions

- There was higher β1 integrin expression on fibrotic human liver tissue compared with non-fibrotic and was gene and protein expression of integrins αv, β1, 3 and 5 in activated HSCs with β6 integrin not detected.
- C8, a β1 integrin inhibitor, successfully inhibited TGFβ activation in a dose-dependent manner.
- **These results show the potential of β1 integrin specifically being a treatment target for liver fibrosis.**

References

1. Bataller, R., 2005. Journal of Clinical Investigation, 115(4), pp.1100-1100.
2. Marcellin P, Kutala B. Liver International. 2018;38:2-6.
3. Rahman et al. Liver International. 2022;42(3):507-521.
4. Wang F et al. J Mol Diagn. 2012;14(1):22-29.
5. Angulo P et al. Hepatology. 2007;45(4):846-854.
6. Abe M et al. Analytical Biochemistry. 1994;216(2):276-284.
7. Reed N et al. Science Translational Medicine. 2015;7(288).

Acknowledgements

The views expressed are those of the authors and not necessarily those of the NHS, NIHR, or the Department of Health and Social Care. This work was supported by the University of Nottingham, FRAME, NIHR Nottingham BRC and GSK. We thank the FRAME lab for technical assistance and MLECs were a gift from Professor Daniel Rifkin (New York University Medical Center, New York). The human biological samples were sourced ethically and their research use was in accord with the terms of the informed consents under an IRB/EC approved protocol.

Results

αv and β1 integrin mRNA expression in fibrotic and non-fibrotic liver tissue

- Fibrotic and non-fibrotic human liver tissue sections had similar expression of αv integrin (Fig 1A, B).
- Fibrotic human liver tissue sections had higher expression β1 integrin compared to non-fibrotic (Fig 1C, D).
- RNAscope® data indicates a higher level of β1 than αv integrin in fibrotic tissue confirmed through the dual staining (Fig 1E).
- There was little β3 integrin expression with ≤ 1 focus per field detected in fibrotic and non-fibrotic liver tissue (not shown).

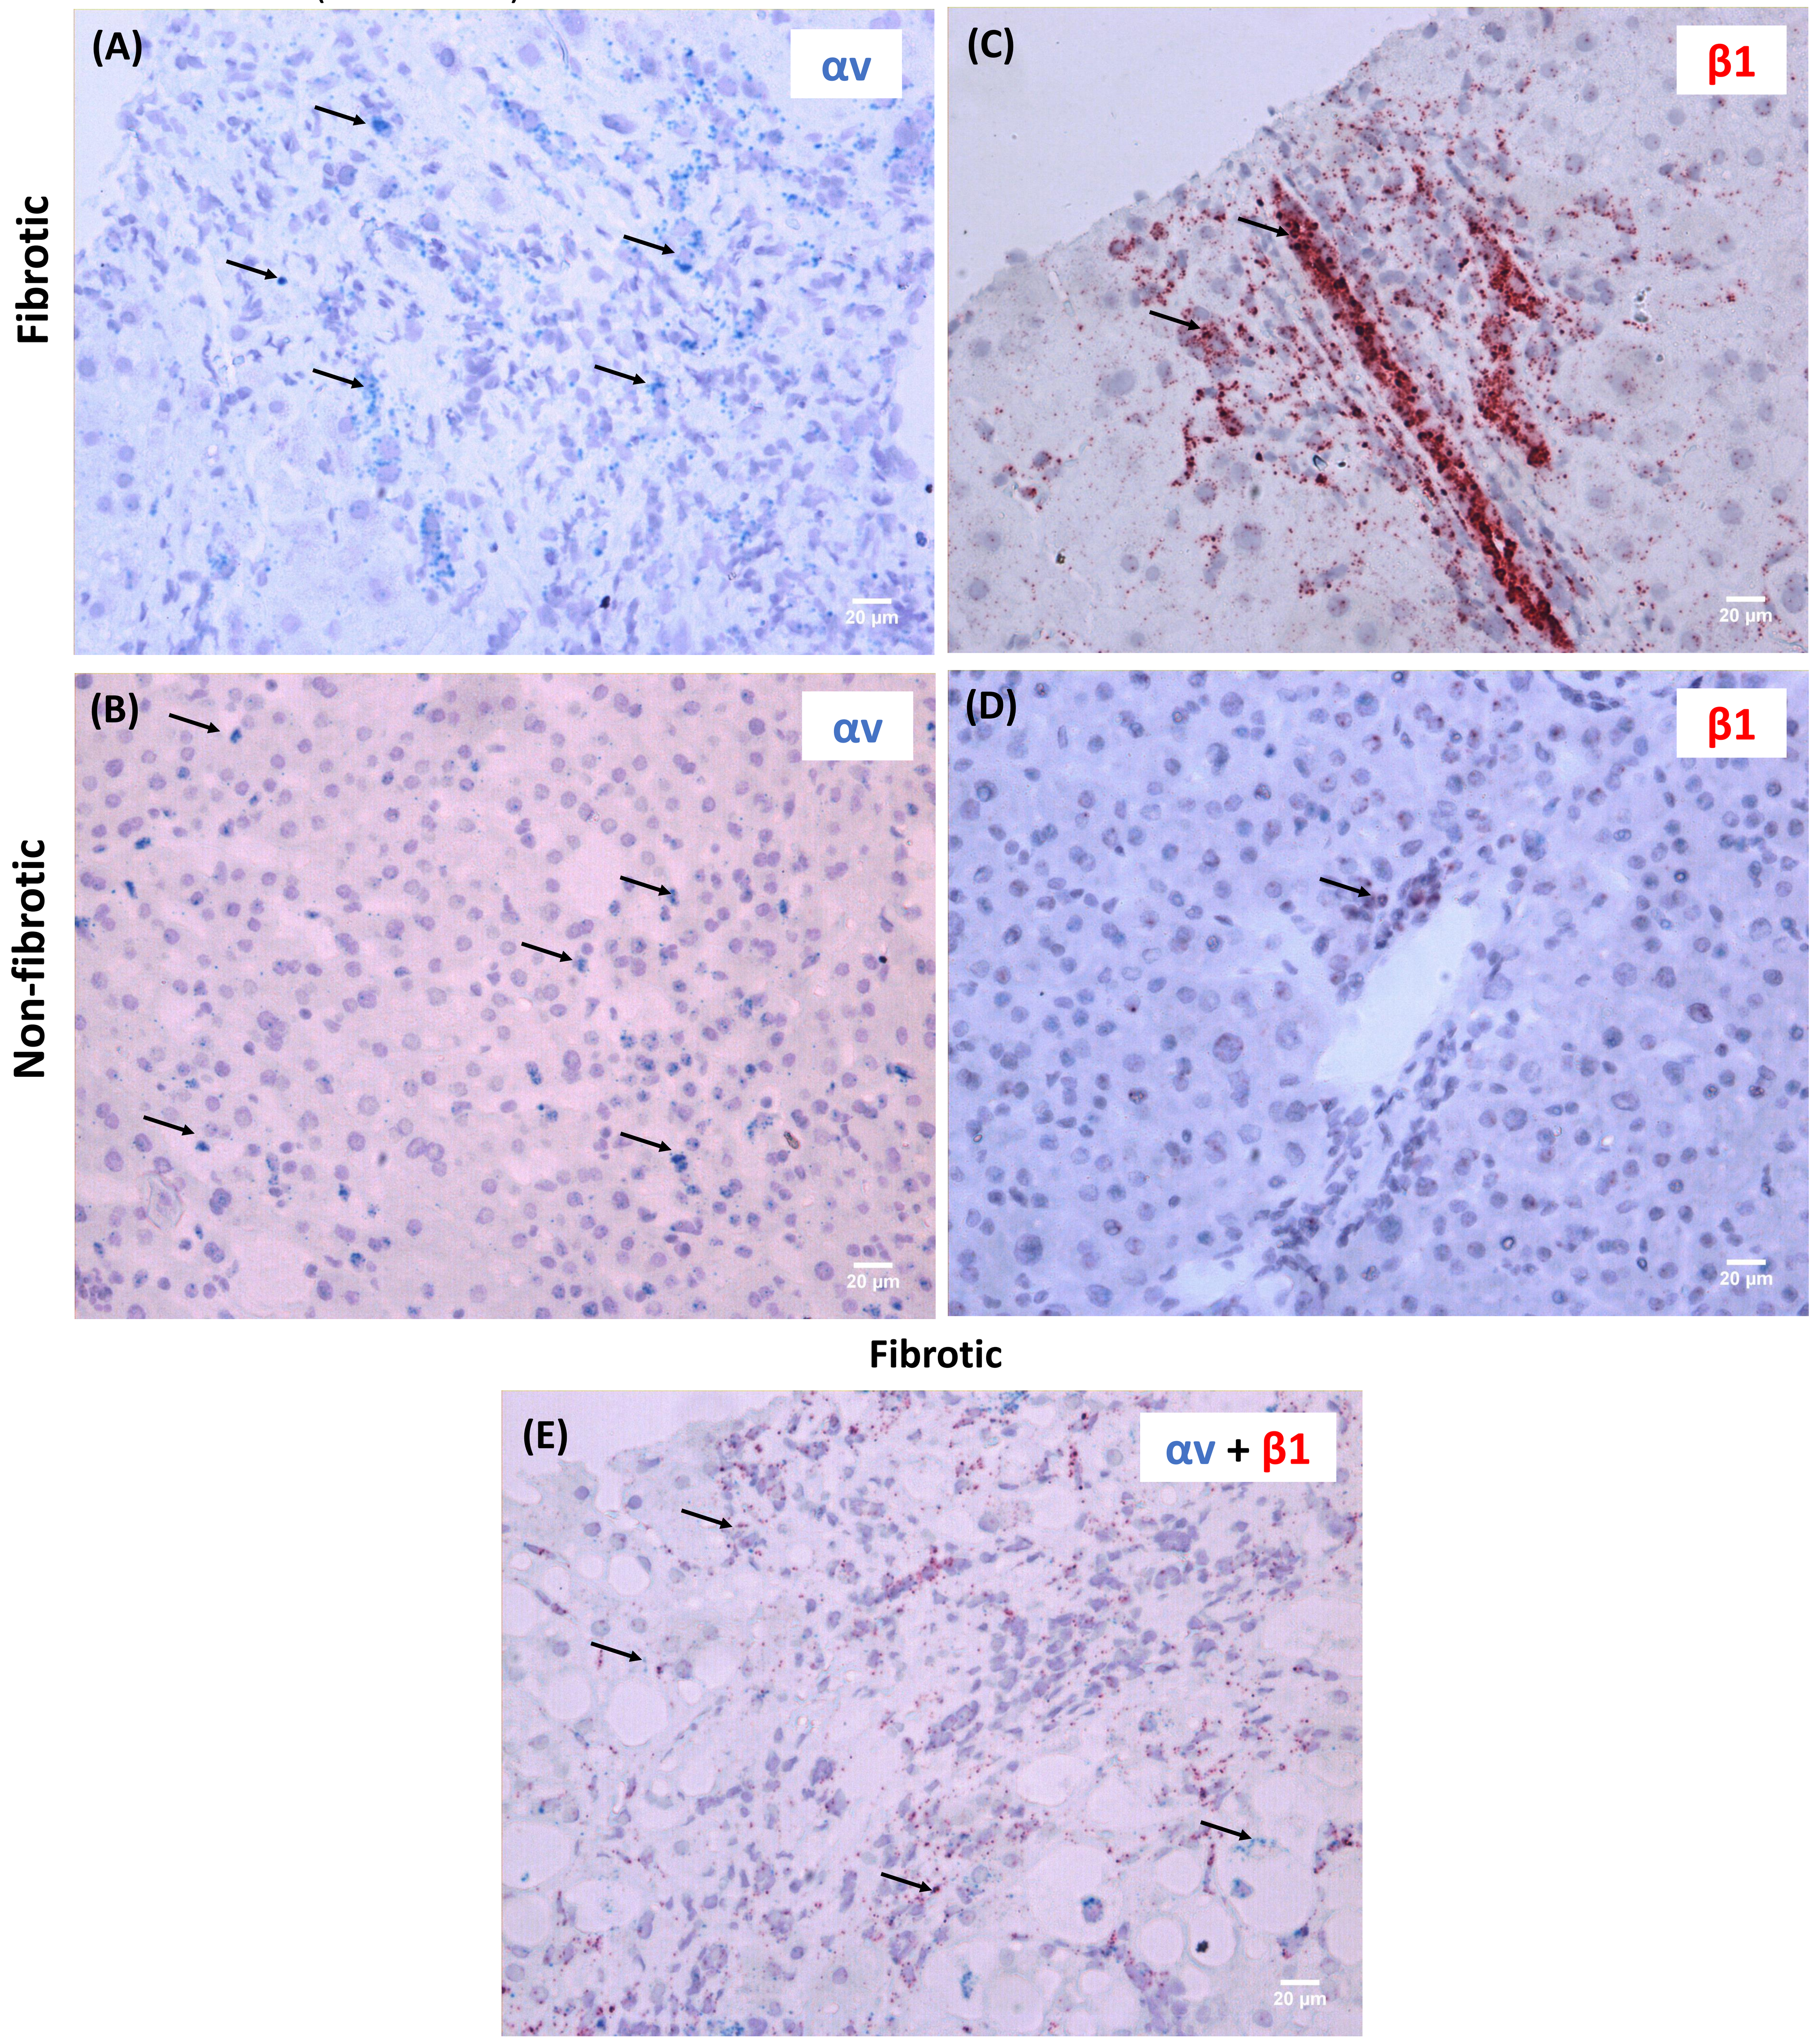
Fibrotic
Non-fibrotic
(A)
αv
(C)
β1
(B)
αv
(D)
β1
Fibrotic
(E)
αv + β1
20 µm

Figure 1: Fibrotic (F3) and non-fibrotic human liver tissue stained with haemotoxylin and RNAscope® probe detecting (A & B) αv (blue) and (C & D) β1 (red) integrin with (E) dual staining of αv and β1 (blue and red respectively). n = 5 for fibrotic and non-fibrotic liver tissue. n = 2 for dual staining.

Gene and protein expression of αv, β1, β3, β5 integrins in activated human HSCs with gene expression of β8 integrin

- Activated HSCs had gene expression of αv, β1, β3, β5 and β8 integrins and protein expression of αv, β1, β3 and β5 integrins (Fig 2).
- There was no gene or protein expression of β6 integrin detected in activated HSCs.

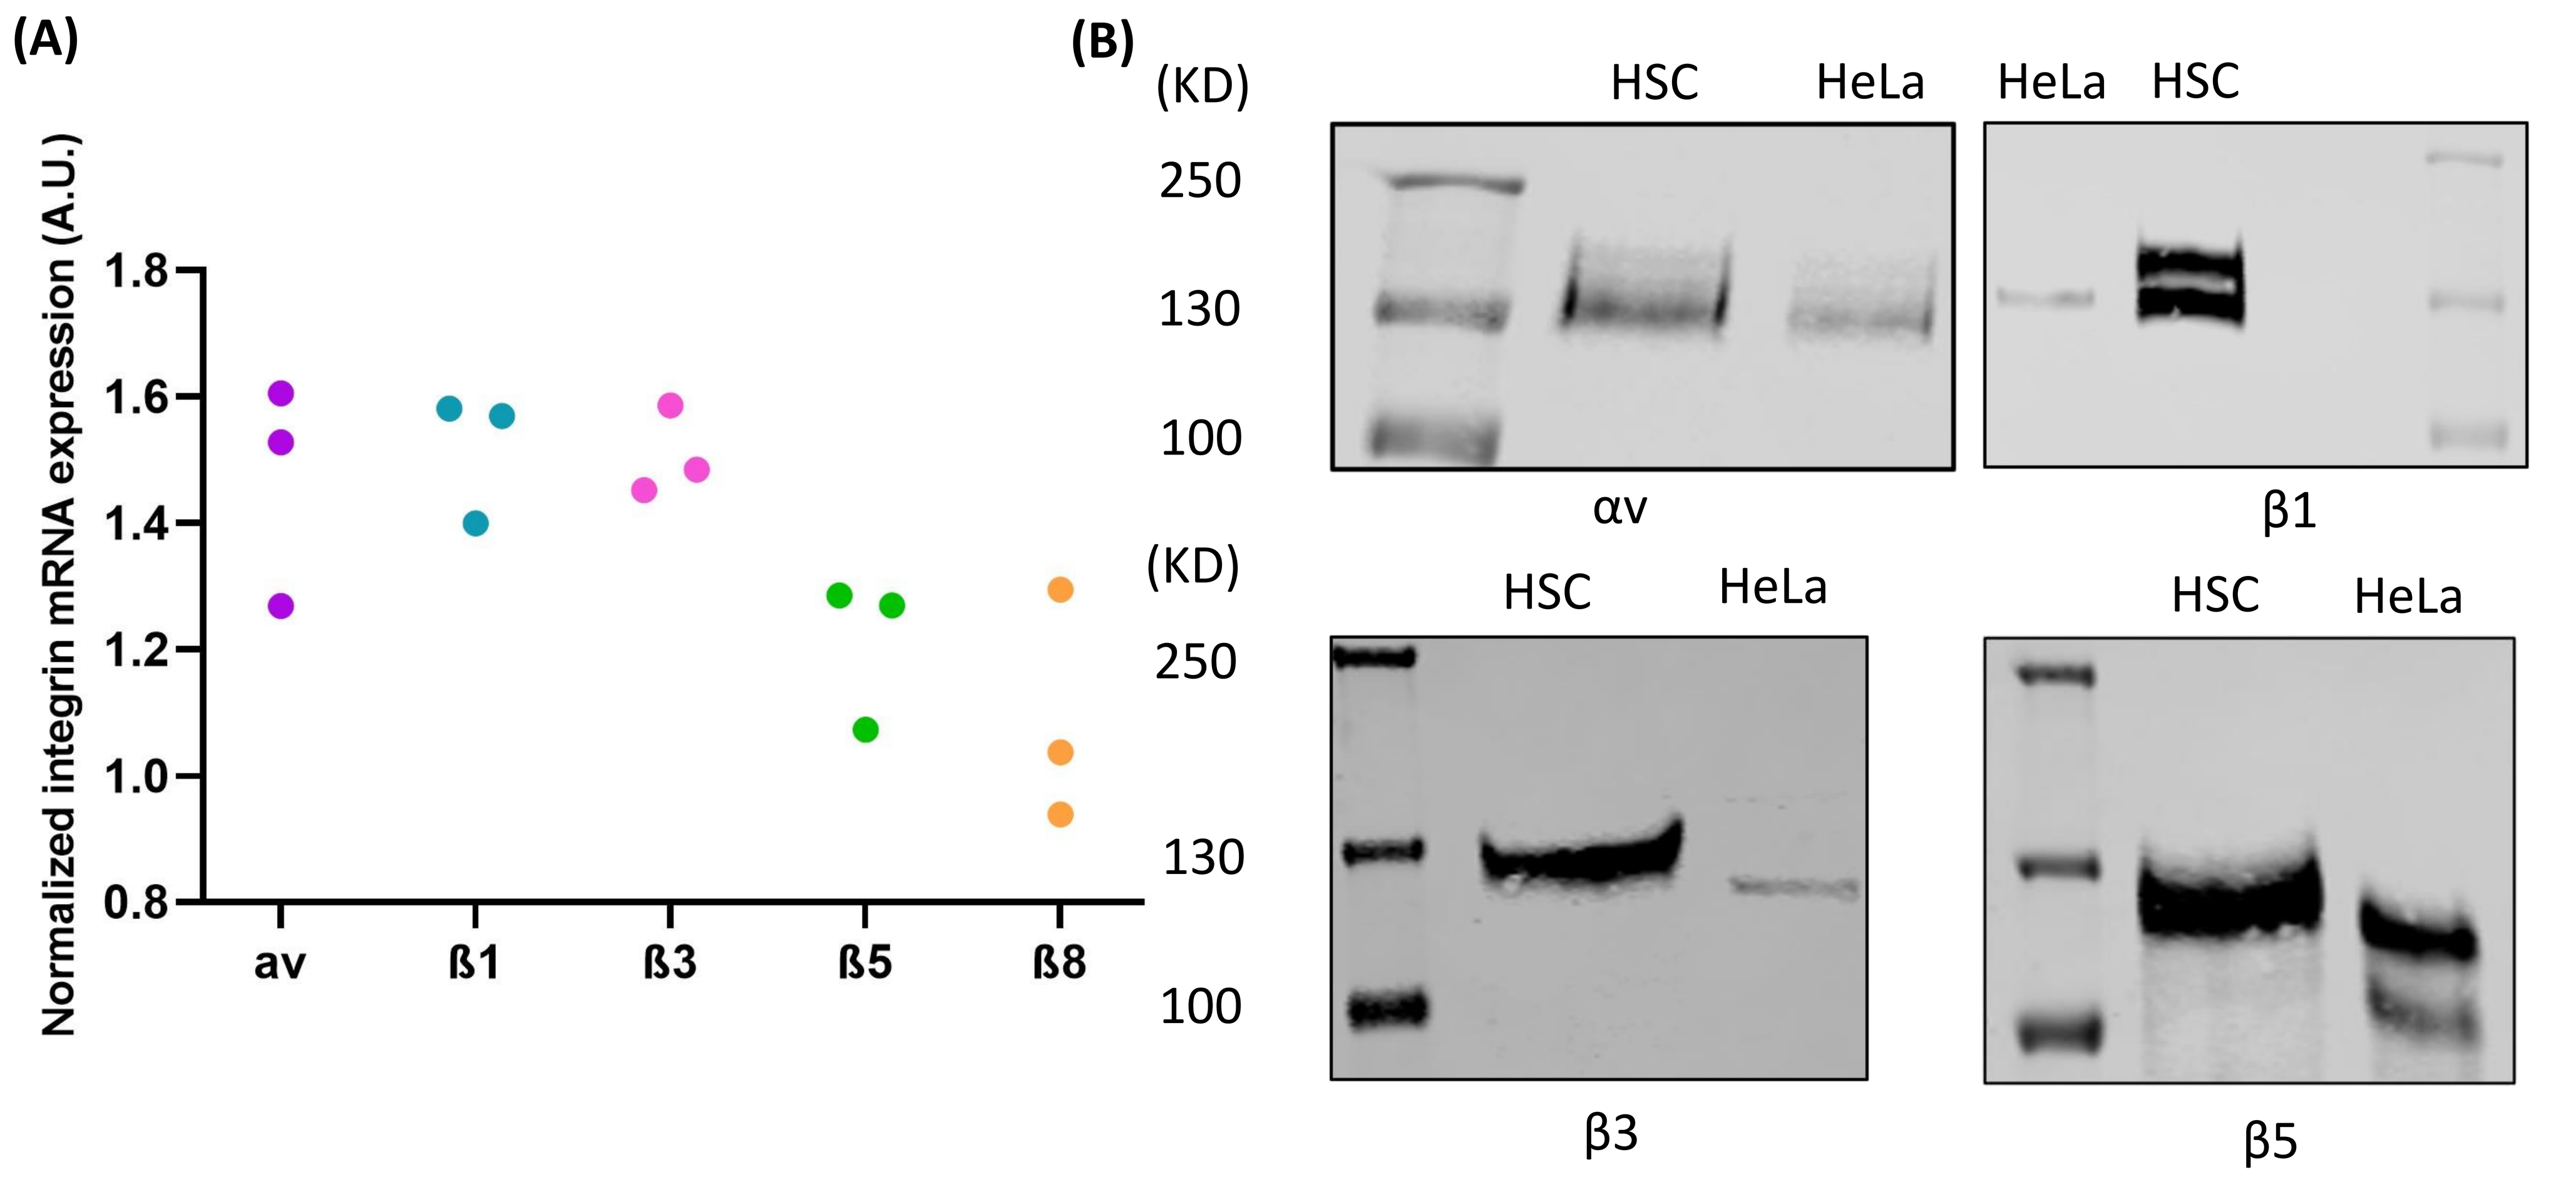
(A)
Normalized integrin mRNA expression (A.U.)
1.8
1.6
1.4
1.2
1.0
0.8
av
ß1
ß3
ß5
ß8
(B)
(KD)
250
130
100
HSC
HeLa
HeLa
HSC
αv
β1
HSC
HeLa
HSC
HeLa
β3
β5

Figure 2: Expression determined by qRTPCR of (A) αv, β1, β3, β5 and β8 integrins via specific primer-probe sets in activated HSCs normalised to β-actin and (B) western blot using anti-αv, β1, β3 and β5 integrins antibodies. HeLa cells were used as a control. n = 1.

Integrin blocking antibodies to αv, β1 and β3 integrins and a β1 integrin inhibitor, C8 inhibited TGFβ activation in human HSCs co-culture

- Integrin blocking antibodies to αv, β1 and β3 integrins inhibited TGFβ activation with increased active TGFβ detected when co-cultured with HSCs (Fig 3A, B, C).
- Integrin blocking antibodies to β5 and β6 integrin did not effect TGFβ activity (not shown).
- C8, a β1 integrin inhibitor showed TGFβ inhibition in a concentration-dependent manner with an IC_{50} of ~1nM (Fig 3D).

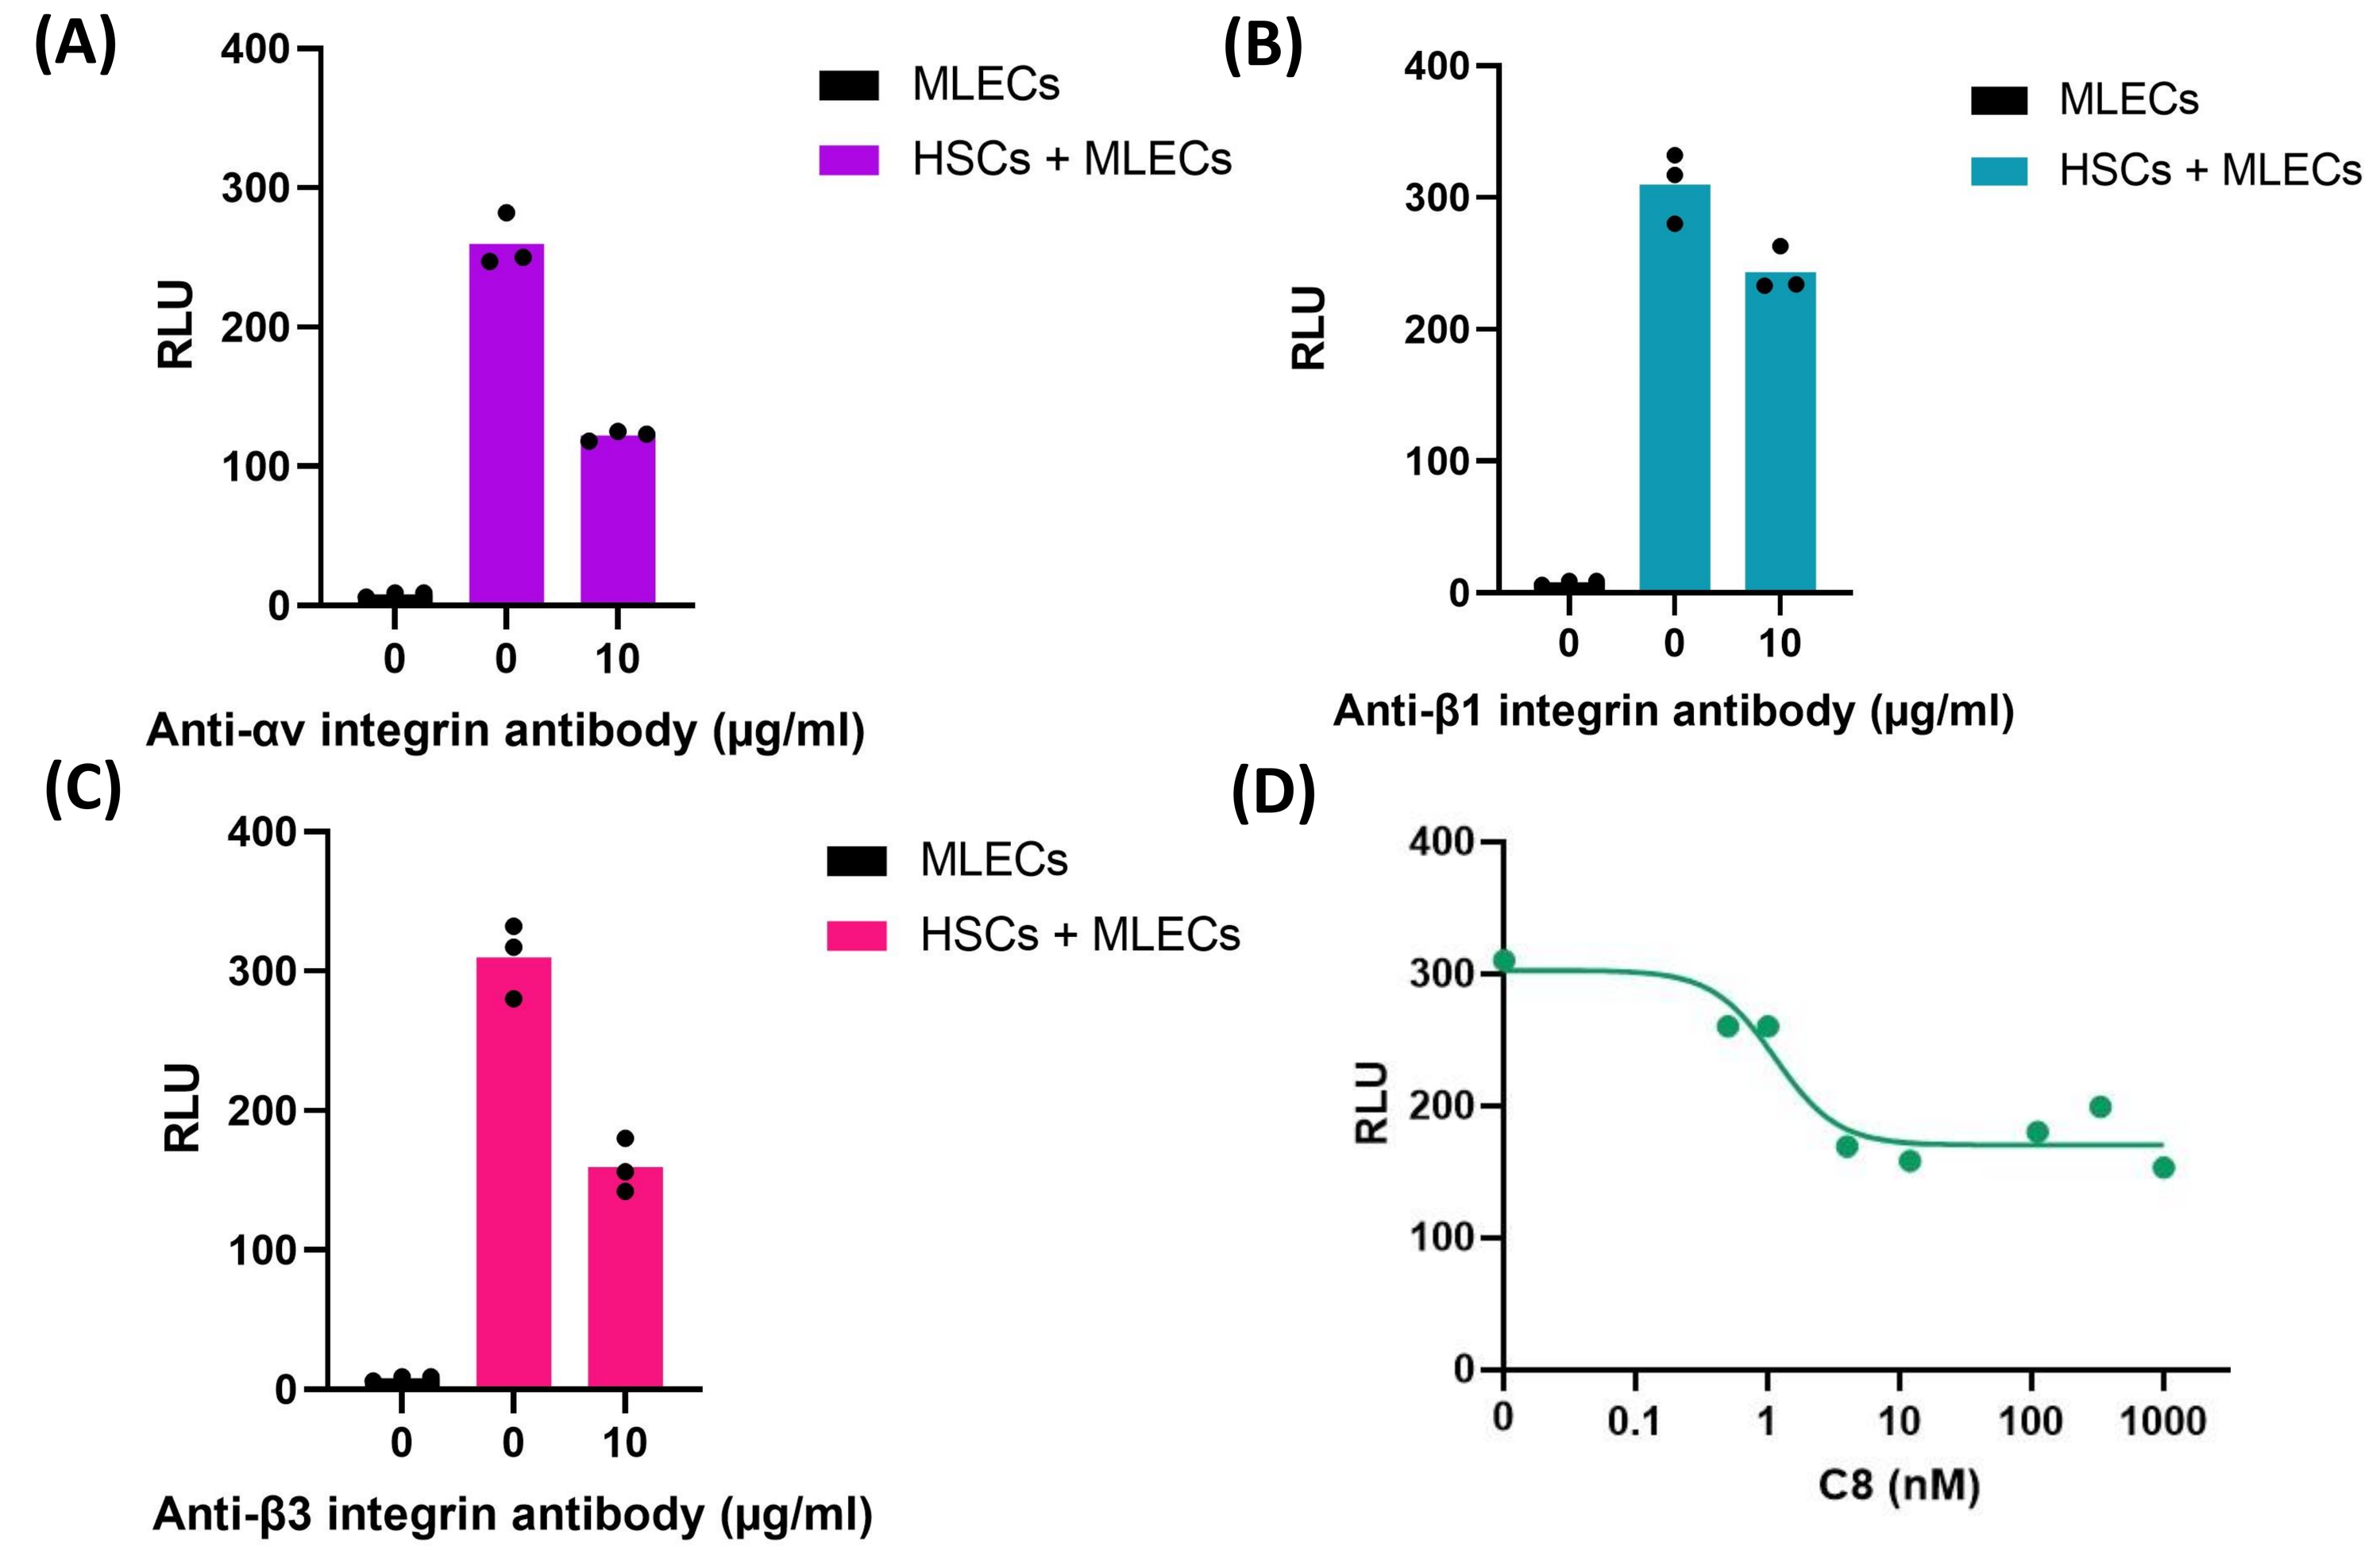
(A)
RLU
400
300
200
100
0
0
0
10
Anti-αv integrin antibody (µg/ml)
MLECs
HSCs + MLECs
(B)
Anti-β1 integrin antibody (µg/ml)
(C)
Anti-β3 integrin antibody (µg/ml)
(D)
0
0.1
1
10
100
1000
C8 (nM)

Figure 3: Luciferase reporter quantification of TGFβ activation following addition of integrin blocking antibodies to (A) αv (B) β1 (C) β3 integrins (D) C8, a β1 integrin inhibitor in co-culture of HSCs and MLECs. C8 had one outlier omitted. The experiment was performed in triplicate from one donor. Baseline active TGFβ was detected in MLECs cultured alone and with HSCs. n = 1. RLU, relative light unit.

PosterSessionOnline.com
Scan to download the poster
THE INTERNATIONAL LIVER CONGRESS™
DOI: 10.3252/pso.eu.ILC2022.2022
Basic Science
Syedia Rahman
FRI--151
ILC2022