EASL The Home of Hepatology

Digital pathology with artificial intelligence analyses reveal new dynamics of treatment-induced fibrosis regression in nonalcoholic steatohepatitis

PO-889

Scan to download a copy of this poster

N.V. Naoumov[1], D. Brees[1], J. Loeffler[1], E. Chng[2], Y. Ren[2], P. Lopez[1], D. Tai[2], A. J. Sanyal[3], S. Lamle[1]

[1]Novartis Pharma AG, Basel, Switzerland, [2]HistoIndex Pte. Ltd, Singapore, [3]Virginia Commonwealth University School of Medicine, Richmond, United States

INTRODUCTION

- Liver fibrosis is a tissue repair response to liver injury and reflects the dynamic balance between fibrogenesis and fibrolysis.
- The architectural patterns and the quantity of hepatic fibrosis have been established as key prognostic determinants for clinical outcomes in nonalcoholic steatohepatitis (NASH).[1]
- Current scoring systems are static, based on fibrosis progression, and do not capture the full spectrum of fibrosis dynamics in NASH.[2,3] There is a need for quantification of perisinusoidal fibrosis, and greater granularity in assessing fibrosis regression.
- The second harmonic generation/two-photon excitation fluorescence (SHG/TPEF) microscopy provides a standardised and reproducible quantification of NASH fibrosis on a linear scale, as well as fine details of the collagen fibres.[4,5]

AIM

The aim of this exploratory analysis was to gain in-depth understanding of liver fibrosis regression and the relation to steatosis reduction after treatment with tropifexor (TXR), a non-bile acid farnesoid X receptor (FXR) agonist, in patients with histologically proven NASH participating in the FLIGHT-FXR study (NCT02855164).

METHODS

- Unstained sections from 198 paired liver biopsies (baseline [BL] and end of treatment [EOT]) from 99 patients with NASH (fibrosis stage F2–F3) who received placebo (PBO; n=34), TXR 140 µg (n=37), or TXR 200 µg (n=28) for 48 weeks were examined.
- Hepatic fat (at BL and EOT) was measured by magnetic resonance imaging-proton density fat fraction (MRI-PDFF).
- The Clinical Research Network (CRN) scoring system was used to determine the components of nonalcoholic fatty liver disease (NAFLD) Activity Score and fibrosis stage by a central pathologist, blinded for treatment and other examinations.
- SHG/TPEF microscopy with Artificial Intelligence (AI) analyses were used to quantify hepatic fat (qSteatosis®) and liver fibrosis (qFibrosis®) in the entire specimen, which included 128 collagen and 45 steatosis parameters.[4] Changes in septa morphology and septa parameters (area, length, width), number of fine collagen fibres, and zonal fibrosis distribution within liver lobules were also quantitated. Furthermore, fibrosis changes around the fat vacuoles were measured concomitantly.

RESULTS

Figure 1. Changes in steatosis after treatment with TXR (BL to EOT) as assessed with a) AI digital quantitation (qSteatosis), b) MRI-PDFF, and c) CRN steatosis score

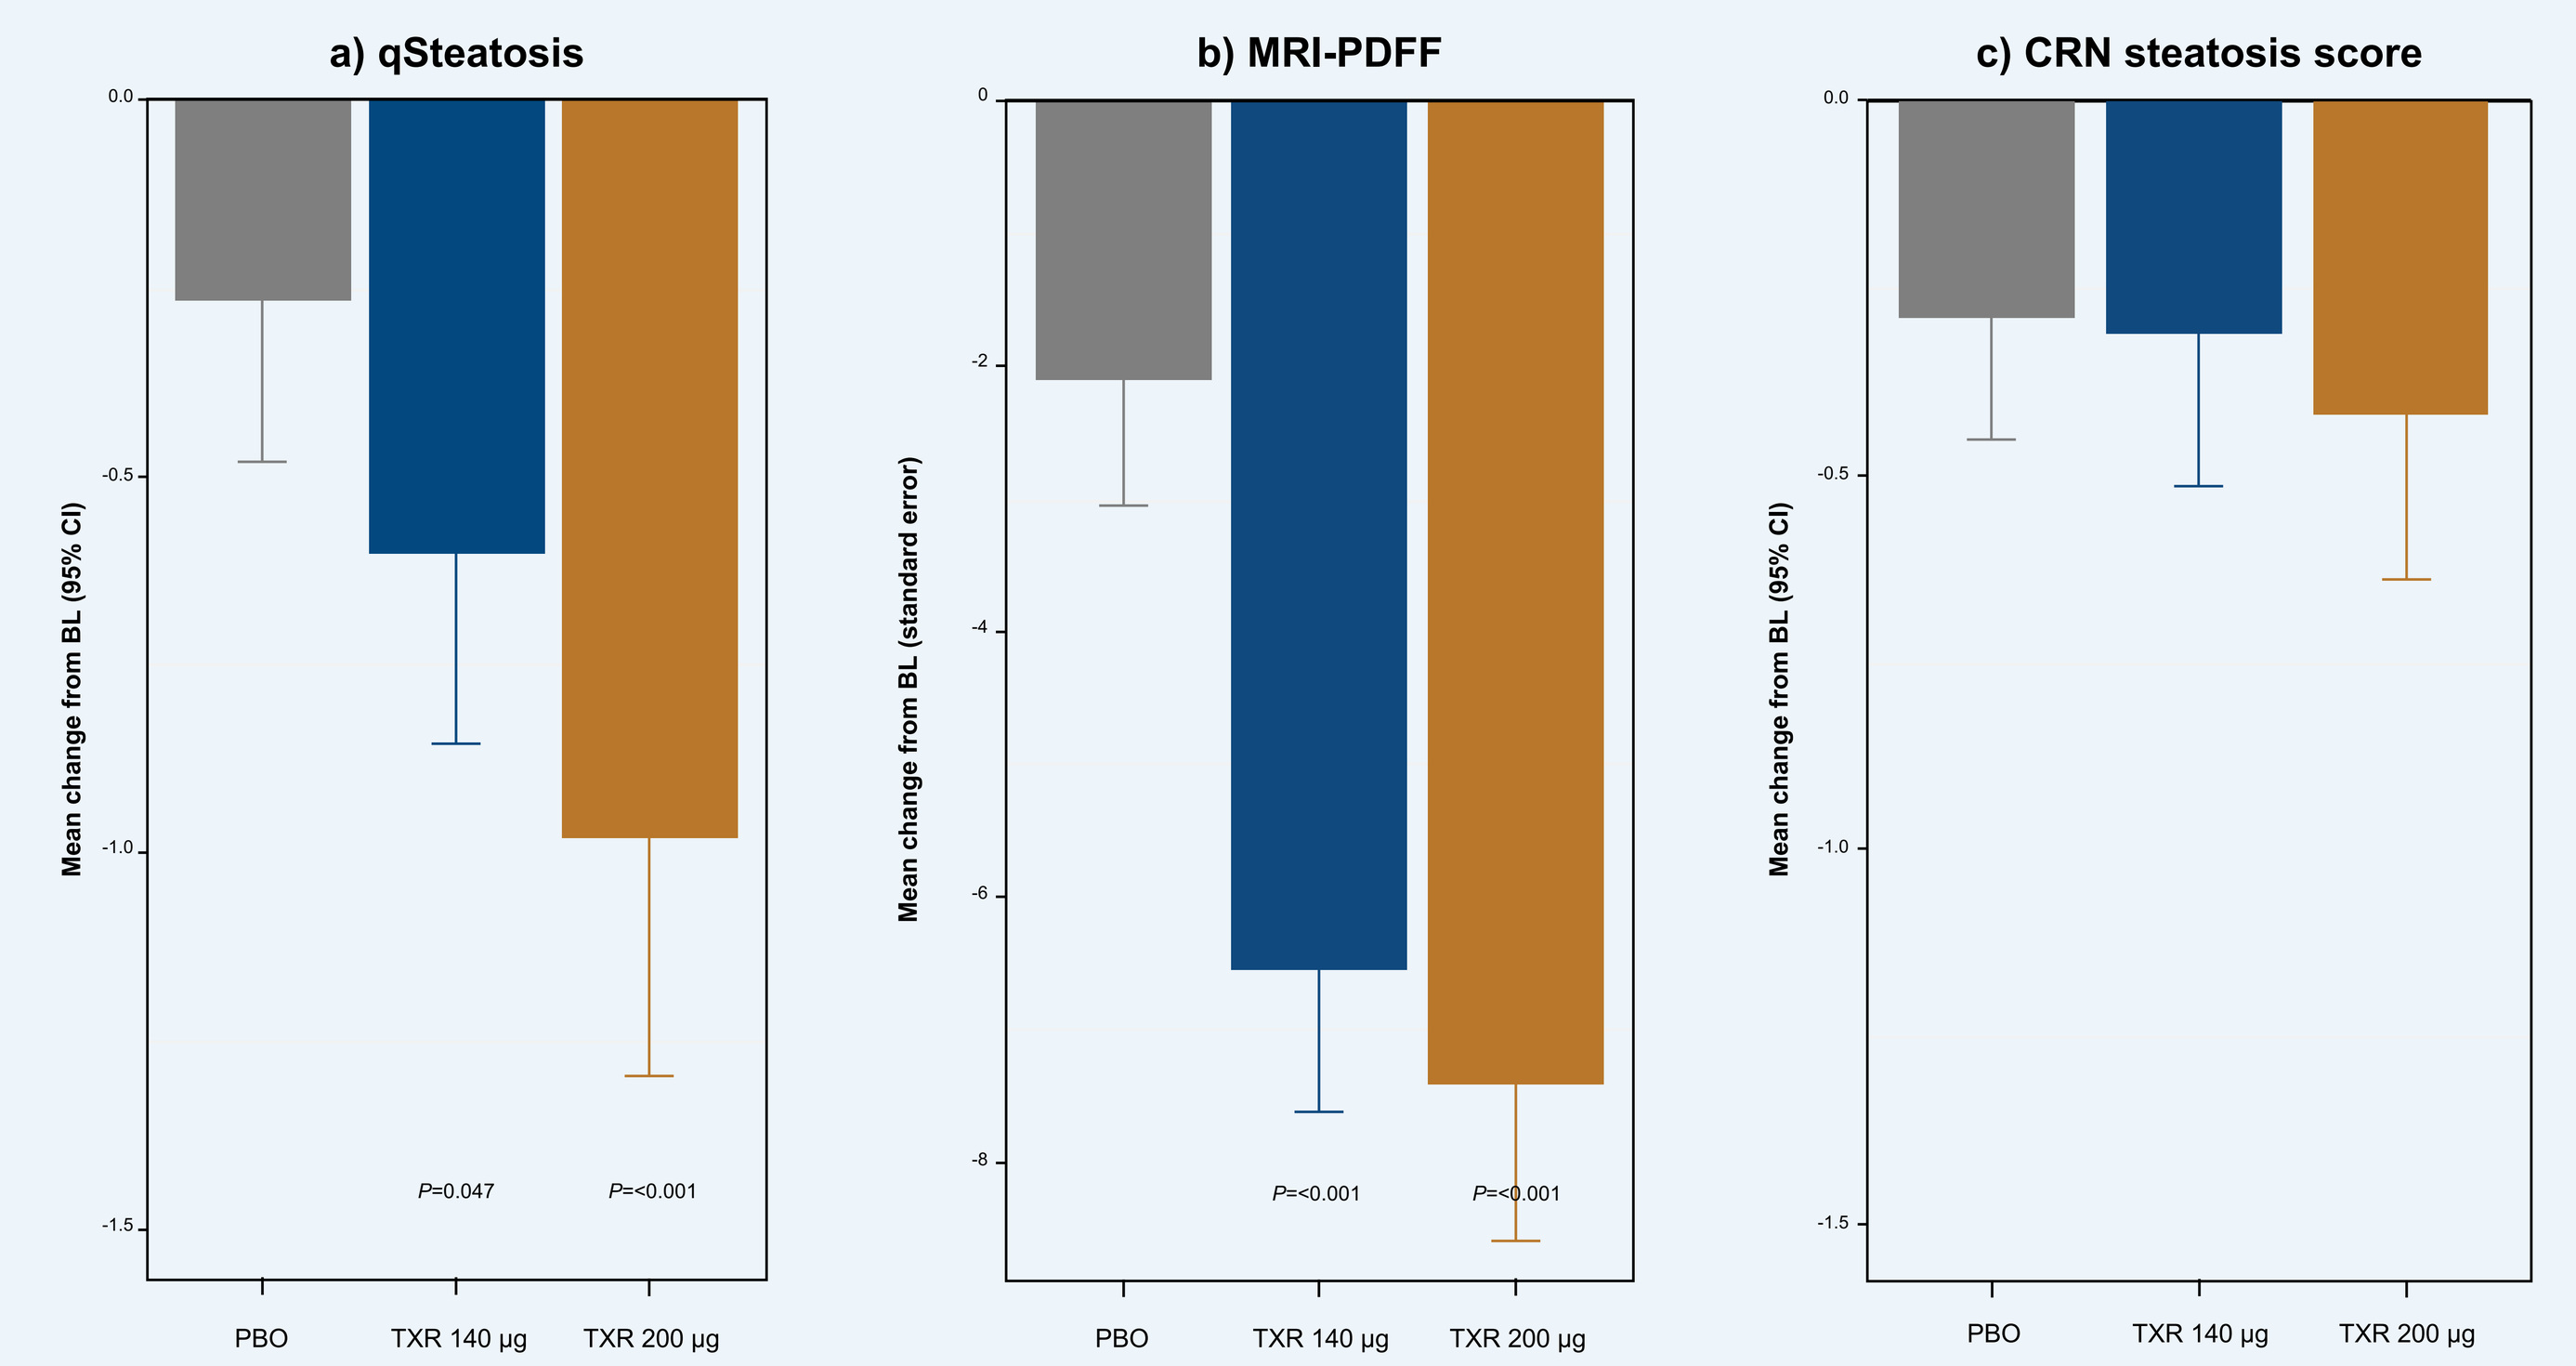
a) qSteatosis
b) MRI-PDFF
c) CRN steatosis score
Mean change from BL (95% CI)
Mean change from BL (standard error)
P=0.047
P=<0.001
PBO
TXR 140 µg
TXR 200 µg

BL, baseline; CRN, clinical research network; EOT, end of treatment; p-values not corrected for multiplicity; MRI-PDFF, magnetic resonance imaging derived proton density fat fraction; TXR, tropifexor

- Digital quantitation of hepatic fat (qSteatosis) showed a dose-dependent reduction (mean change: PBO, –0.25; TXR 140 µg, –0.6 [*P*=0.047]; TXR 200 µg, –0.95 [*P*<0.001]), which was in line with the changes detected by MRI-PDFF. There were no notable changes in the CRN steatosis score (by central pathologist).
- At an individual level, there were strong correlations between the qSteatosis data with the MRI-PDFF: at Week 48 (r=0.851), as well as for the liver fat reduction from BL to EOT (r=0.709).

Figure 2. Digital quantification of overall liver fibrosis (qFibrosis) at BL and at the EOT reveals fibrosis regression in a greater proportion of patients than conventional microscopy

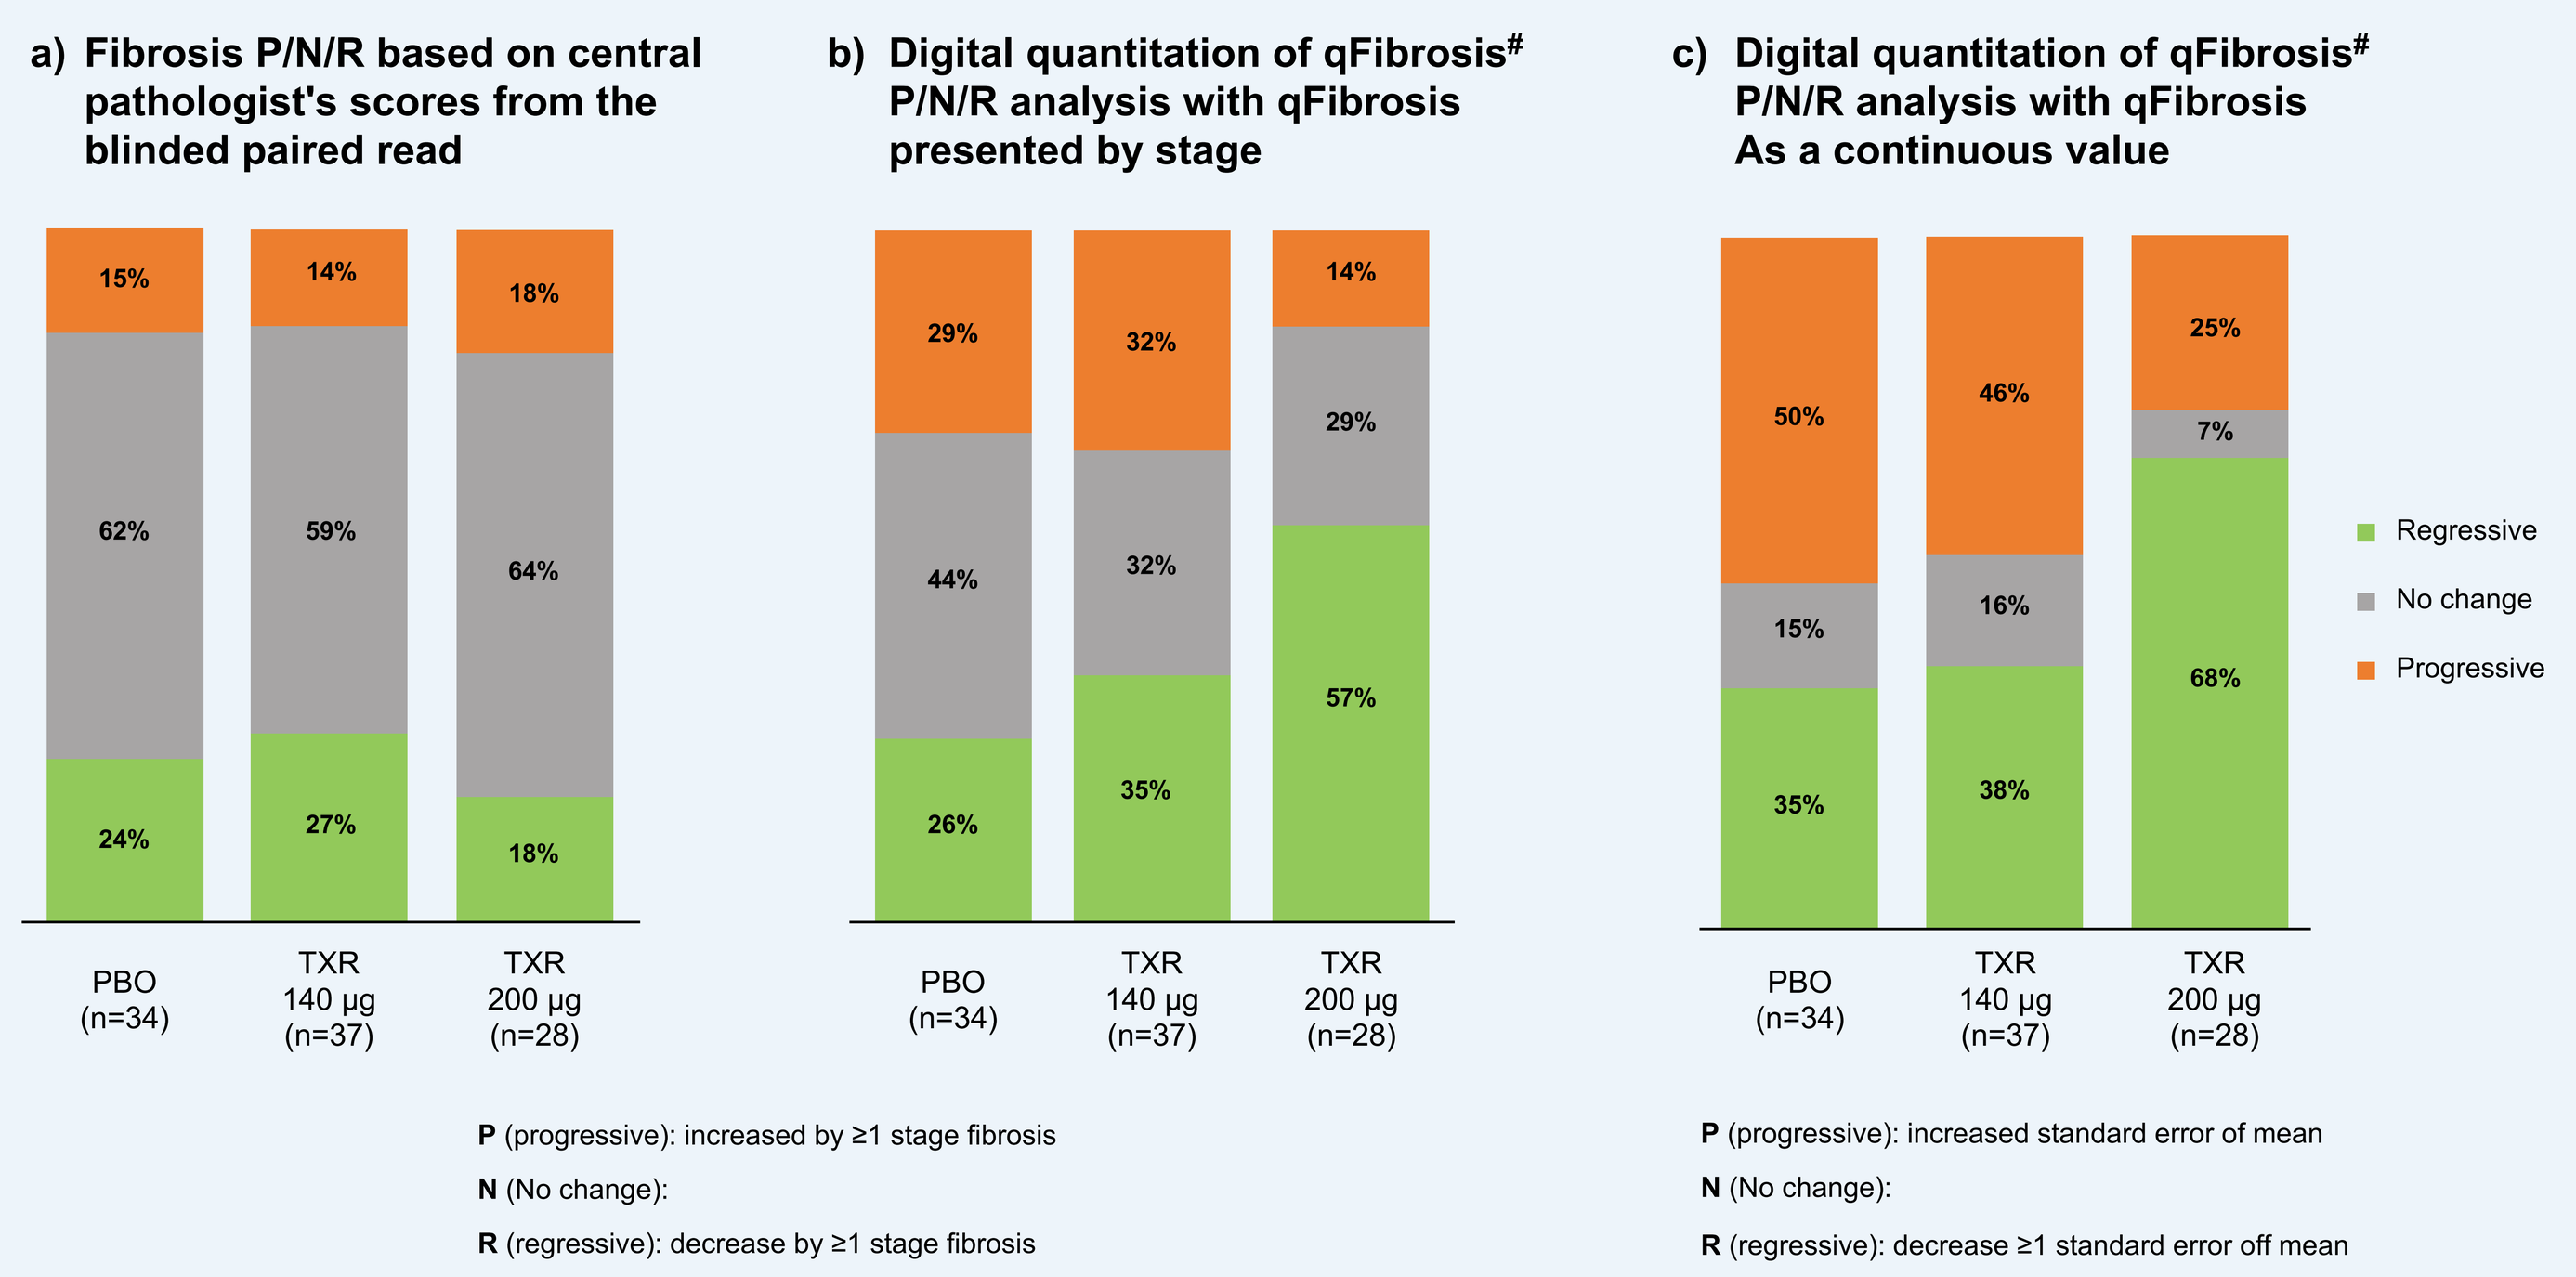
a) Fibrosis P/N/R based on central pathologist's scores from the blinded paired read
b) Digital quantitation of qFibrosis# P/N/R analysis with qFibrosis presented by stage
c) Digital quantitation of qFibrosis# P/N/R analysis with qFibrosis As a continuous value
PBO (n=34)
TXR 140 µg (n=37)
TXR 200 µg (n=28)
Regressive
No change
Progressive
P (progressive): increased by ≥1 stage fibrosis
N (No change):
R (regressive): decrease by ≥1 stage fibrosis
P (progressive): increased standard error of mean
N (No change):
R (regressive): decrease ≥1 standard error off mean

The proportion of pts in each study group are categorized as: P–progressive; N–no change; R–regressive, according to liver fibrosis evaluation by stage or as a continuous value. #qFibrosis model based on the scores from FLIGHT-FXR central reader.
BL, baseline; FXR, farnesoid X receptor; n, number of patients; PBO, placebo; TXR, tropifexor

- Assessment of liver fibrosis by AI digital pathology (qFibrosis) – expressed either by stage (decrease by ≥1 stage fibrosis: PBO, 26%; TXR 140 µg, 35%; and TXR 200 µg, 57%), or as a continuous value (decrease ≥1 standard error of mean: PBO, 35%; TXR 140 µg, 38%; and TXR 200 µg, 68%) showed an increased proportion of patients with fibrosis regression in the TXR groups compared with PBO.

Figure 3. Digital quantification of fibrosis in different zones of the liver lobule at BL and EOT reveals marked fibrosis reduction in the perisinusoidal areas

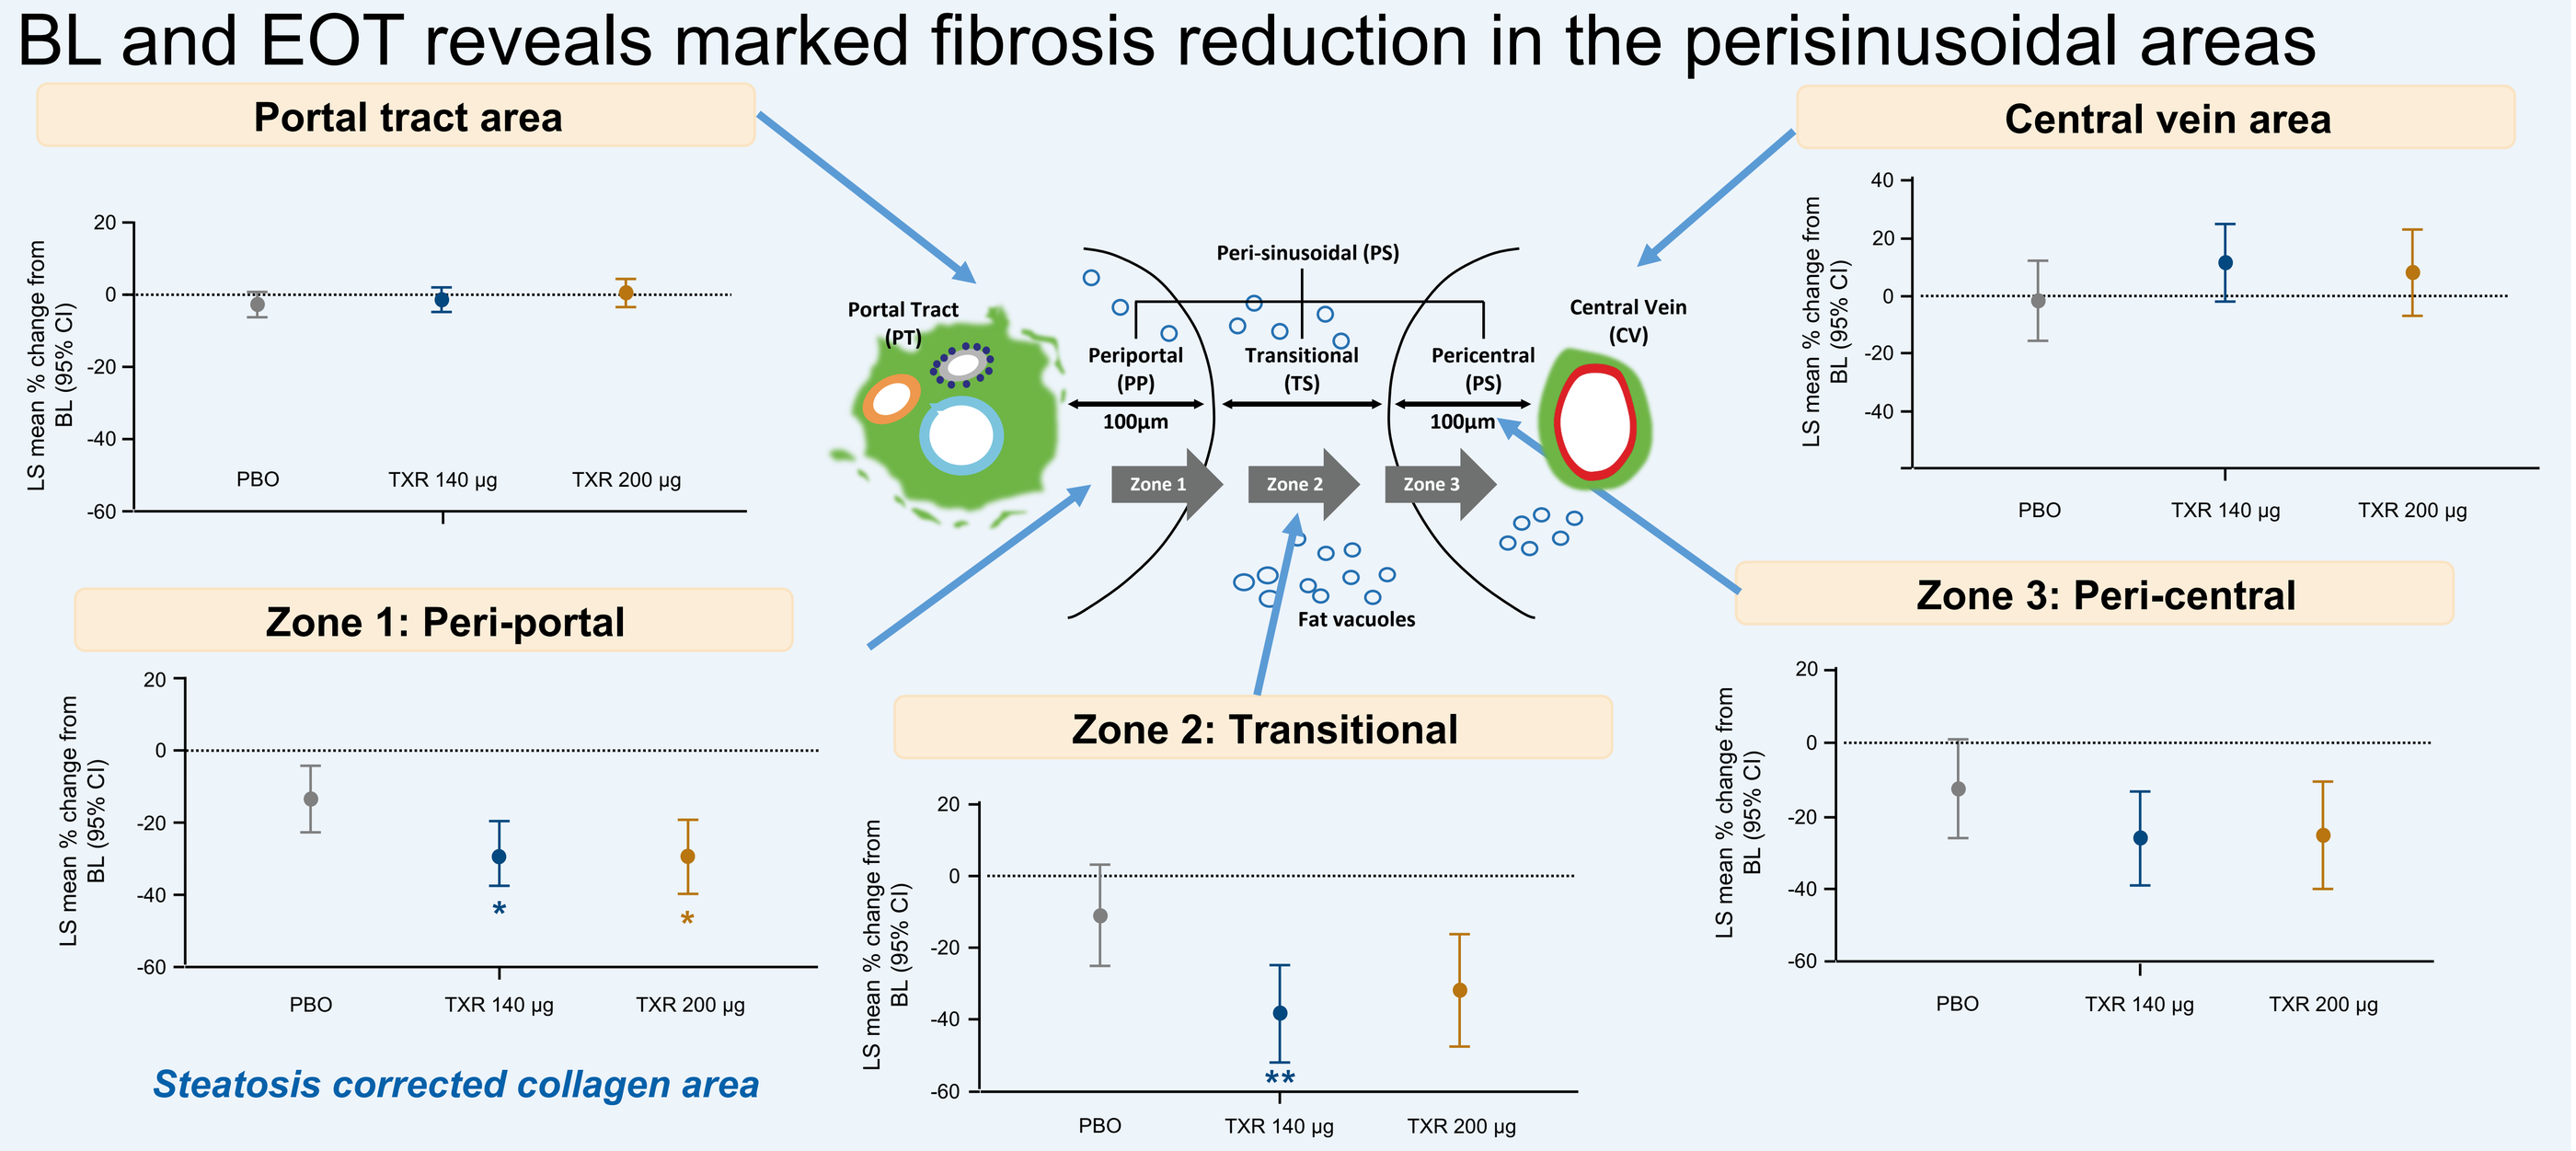
Portal tract area
Central vein area
Zone 1: Peri-portal
Zone 2: Transitional
Zone 3: Peri-central
Portal Tract (PT)
Peri-sinusoidal (PS)
Periportal (PP)
Transitional (TS)
Pericentral (PS)
Central Vein (CV)
100µm
Fat vacuoles
LS mean % change from BL (95% CI)
PBO
TXR 140 µg
TXR 200 µg
Steatosis corrected collagen area

P*=0.021; *P*=0.006 (no multiplicity correction).
BL, baseline; CI, confidence interval; EOT, end of treatment; LS; least squares; PBO, placebo; TXR, tropifexor

- Zonal analyses of qFibrosis show marked reduction in peri-sinusoidal fibrosis (Zone 1, 2, and 3), but not in the portal tracts.

Figure 4. Colocalisation analysis of qSteatosis and qFibrosis changes as result of TXR treatment

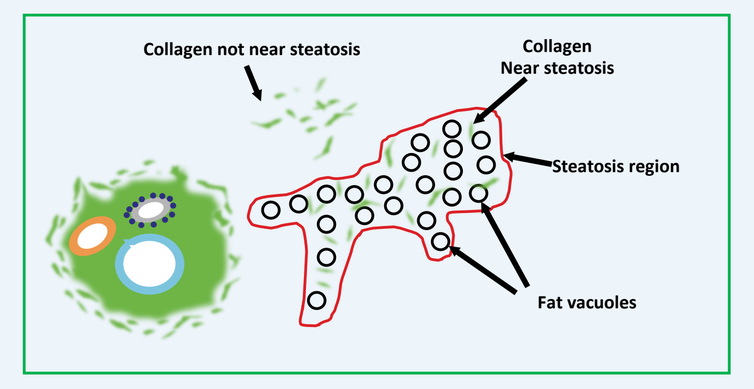
Collagen not near steatosis
Collagen Near steatosis
Steatosis region
Fat vacuoles

This cartoon illustrates the approach for selecting the area for concomitant quantification of steatosis and fibrosis in the same area of the liver lobule. Collagen near steatosis is the collagen in the area within 14 µm around fat vacuoles (which is half the size of a normal hepatocyte). Multiple areas like this were analysed in each liver biopsy.

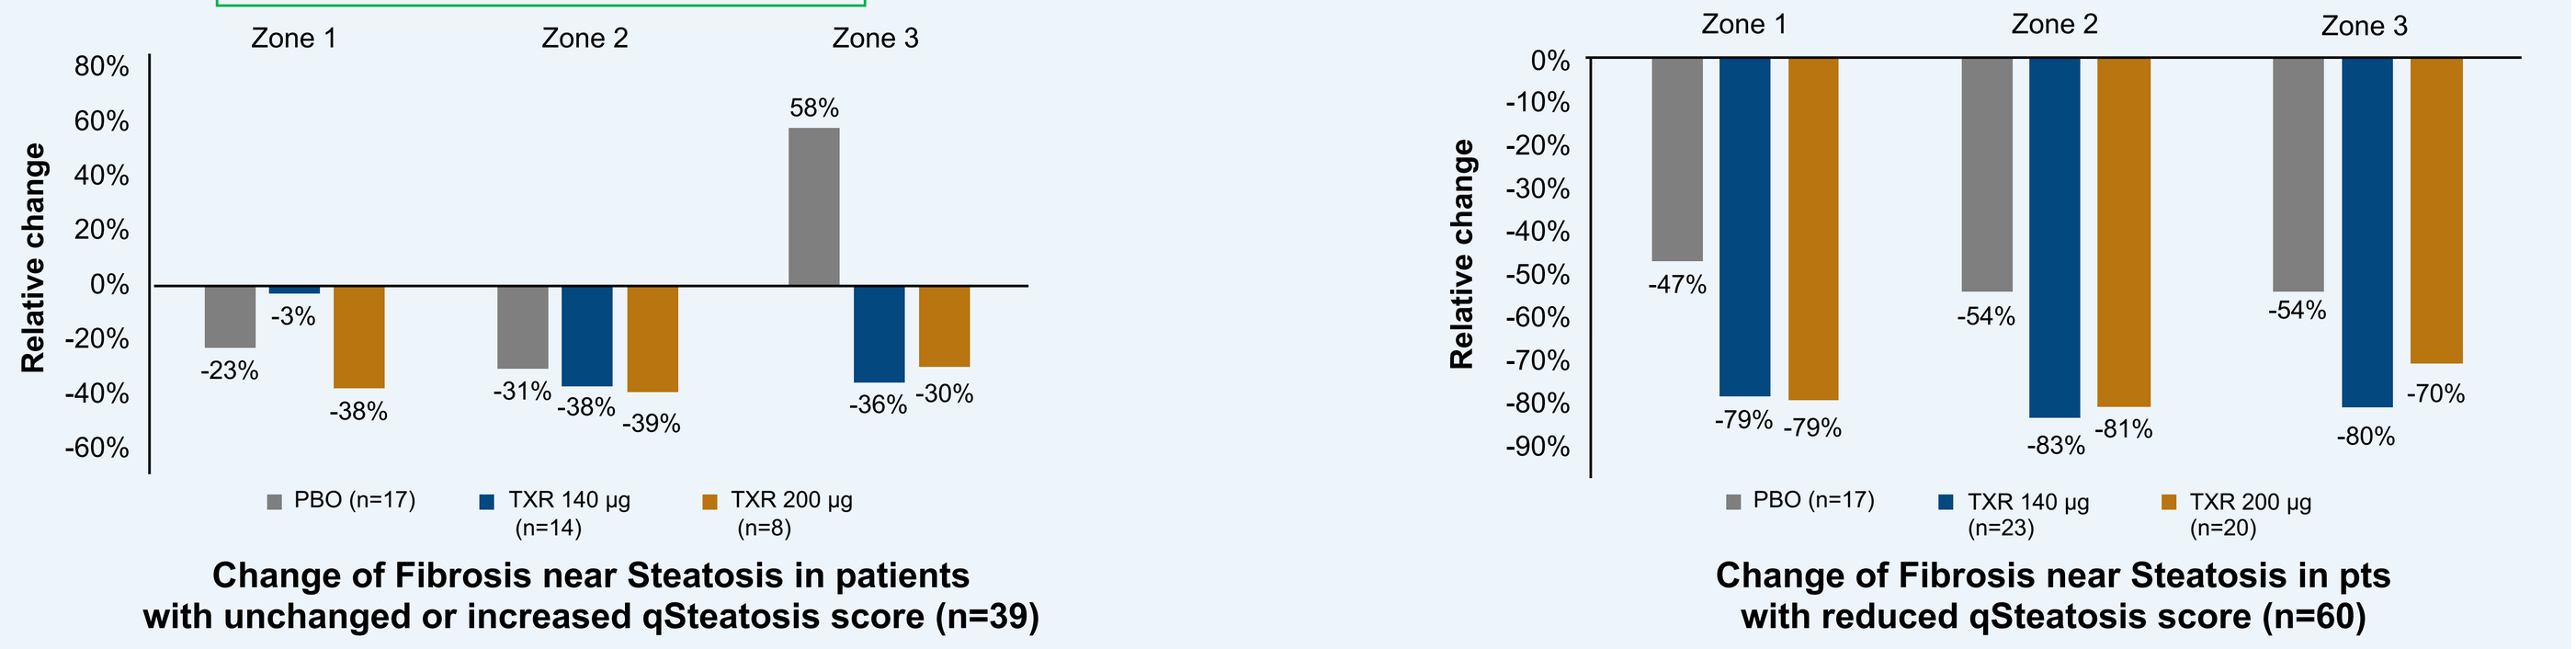
Zone 1
Zone 2
Zone 3
Relative change
PBO (n=17)
TXR 140 µg (n=14)
TXR 200 µg (n=8)
Change of Fibrosis near Steatosis in patients with unchanged or increased qSteatosis score (n=39)
PBO (n=17)
TXR 140 µg (n=23)
TXR 200 µg (n=20)
Change of Fibrosis near Steatosis in pts with reduced qSteatosis score (n=60)

PBO, placebo; TXR, tropifexor

- Patients with unchanged or increased qSteatosis (**left panel**) showed only mild reduction of nearby fibrosis and there was 58% increase of Zone 3 fibrosis in the PBO group.
- Patients with reduced qSteatosis (**right panel**) showed marked reduction by 70–80% of fibrosis across the perisinusoidal area (Zones 1, 2, and 3).

Figure 5. Digital quantification of septal fibrosis reveals reduction in collagen deposits within septa between BL and Week 48 with TXR treatment

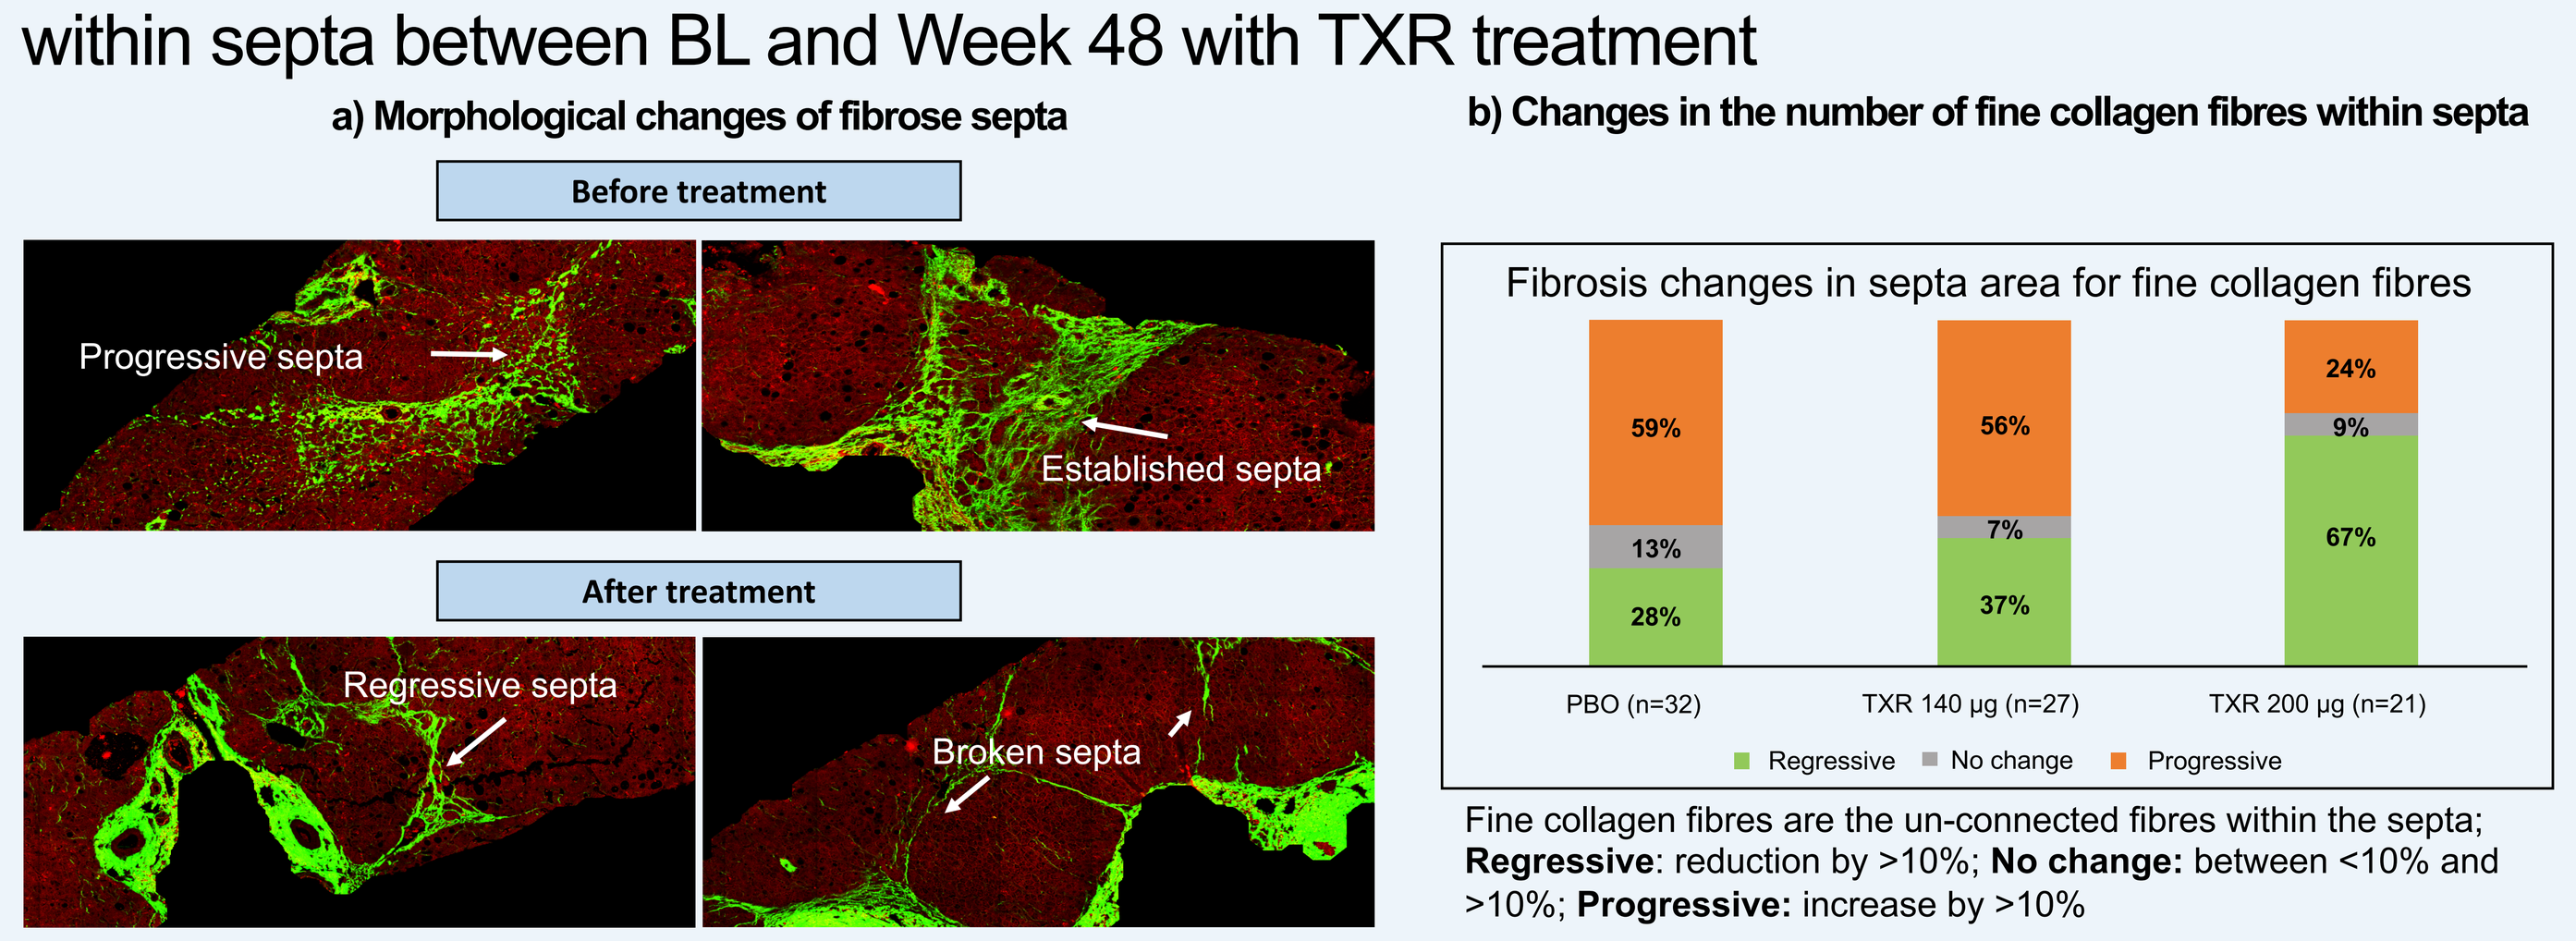
a) Morphological changes of fibrose septa
b) Changes in the number of fine collagen fibres within septa
Before treatment
Progressive septa
Established septa
After treatment
Regressive septa
Broken septa
Fibrosis changes in septa area for fine collagen fibres
PBO (n=32)
TXR 140 µg (n=27)
TXR 200 µg (n=21)
Regressive
No change
Progressive

Fine collagen fibres are the un-connected fibres within the septa; **Regressive**: reduction by >10%; **No change:** between <10% and >10%; **Progressive:** increase by >10%

BL, baseline; PBO; placebo; TXR, tropifexor

- Progressive septa are wide, with loose connective tissue, invading adjacent parenchyma; Regressive septa are thin, compact with sharp borders (as described previously).[6]
- The proportion of patients with regressive fibrosis changes, i.e. decreased number of fine collagen fibres within septa, was much greater after treatment with TXR 200 µg compared with the other groups.

Figure 6. Digital quantification of septa parameters in pts with NASH CRN F3 fibrosis (at BL) after TXR treatment: changes from BL to Week 48

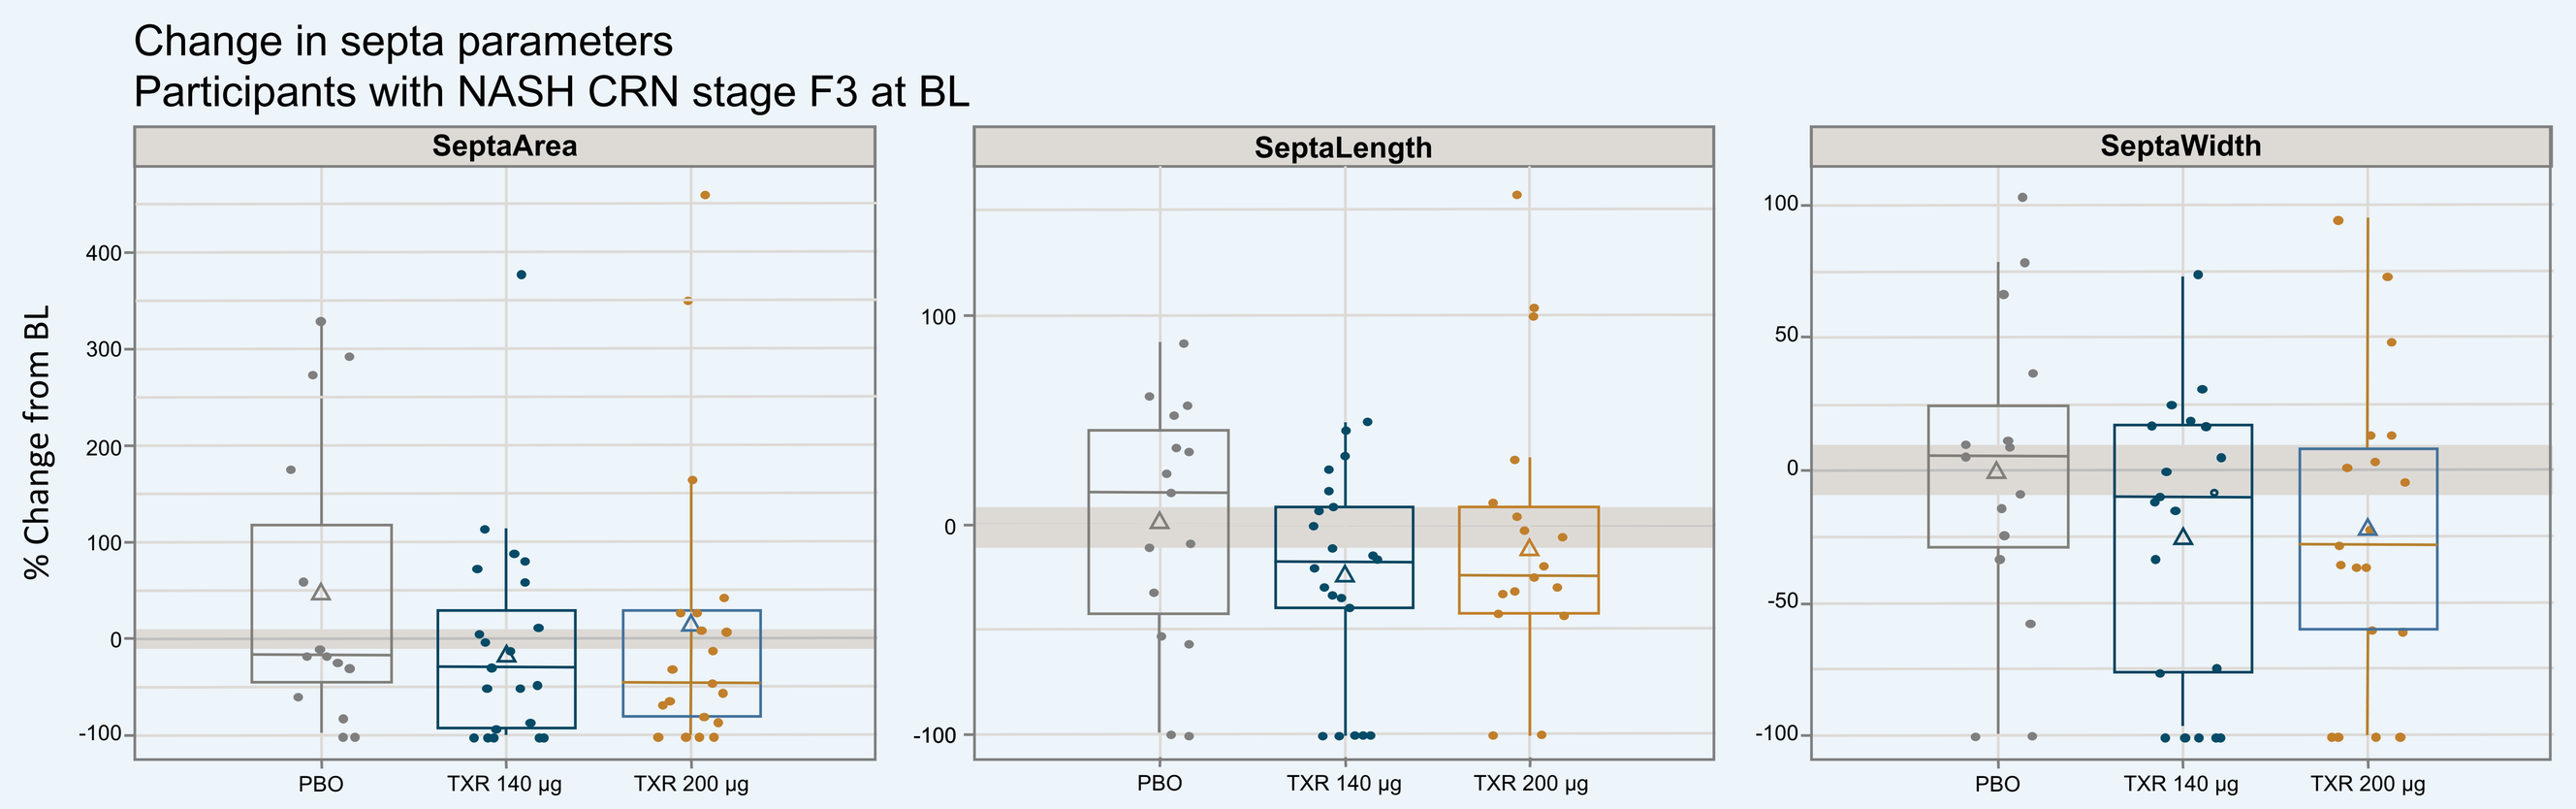
Change in septa parameters
Participants with NASH CRN stage F3 at BL
SeptaArea
SeptaLength
SeptaWidth
% Change from BL
PBO
TXR 140 µg
TXR 200 µg

Ribbon indicates range between 10 and -10% change. Triangle represents arithmetic mean.
BL, baseline; CRN, clinical research network; NASH, nonalcoholic steatohepatitis; TXR, tropifexor

- Regression of fibrous septa with a decrease in septa area, septa length and septa width was observed in a greater proportion of patients treated with TXR in comparison with those receiving PBO.

CONCLUSIONS

- SHG/TPEF microscopy is a promising tool for quantitative evaluation of liver fibrosis with new parameters that can not be detected by human eye. This approach provides new insights into the dynamics of fibrosis regression and treatment response, which are not captured by conventional staging systems.
- AI digital pathology reveals details of TXR anti-fibrotic activity in NASH including overall fibrosis reduction (qFibrosis), decrease in the perisinusoidal fibrosis, fine collagen fibres, as well as the septa parameters.
- Spatial correlation between qFibrosis and qSteatosis reduction suggests that anti-metabolic therapies that reduce hepatic lipid load and lipotoxicity drive initially fibrosis regression in the perisinusoidal regions, around the areas of steatosis reduction.

ACKNOWLEDGEMENTS

All authors participated in the development of this poster and approved the final poster for presentation.
The authors thank the patients who participated in this study, as well as the investigators and the study coordinators.
The input of Sandeep Subramaniam (Novartis) for the art work and the poster layout is gratefully acknowledged.

REFERENCES

1 **Angulo P, et al.** *Gastroenterology* 2015;149:389–97
2 **Kleiner DE, et al.** *Hepatology* 2005;41:1313–21
3 **Pai RK, et al.** *Aliment Pharmacol Ther* 2019;50:1100–11
4 **Liu F, et al.** *Hepatology* 2020;71:1953–66
5 **Soon G, et al.** *Clin Mol Hepatology* 2021;27:44–57
6 **Sun Y, et al.** *Hepatology* 2017;65:1438–50

DISCLOSURES

N.V. Naoumov, D. Brees, J. Loeffler, P. Lopez, S. Lamle: Employee, Novartis. **E. Chng, Y. Ren, D. Tai:** Employee, Histoindex. **A. J. Sanyal:** Stockholder; Genfit, Tiziana, Indalo, Hemoshear, Exhalenz, Durect; Collaborations; OWL, Karius, Histoindex, PathAI, Biocellvia; Consultant; Novartis, Novo Nordisk, Eli Llly, Pfizer, Merck, Bristol Myers Squibb, Boehringer Ingelhiem, Intercept, Gilead, Malinckrodt, Salix, Akero, NGM bio, Amgen, Astra Zeneca, Genentech, Rivus, Inventiva, Blade, 89 Bio, Siemens, PathAI, Histoindex, terns, Fractyl, Samsara, Zydus; Intercept, Madrigal, NGM, Novartis, Gilead, Salix, Malinckrodt, Merck, Genentech, Inventiva, Boehringer Ingelhiem, Bristol Myers Squibb, Echosense, Zydus.

CONTACT INFORMATION

Nikolai V. Naoumov: nikolai.naoumov@novartis.com

Poster presented at the International Liver Congress™ 2021 (ILC 2021), Virtual Congress, 23–26 June.

PosterSessionOnline.com
Scan to download the poster
THE INTERNATIONAL LIVER CONGRESS 23-26 JUNE 2021 BEATING LIVER DISEASE together
Metabolism, alcohol and toxicity
Nikolai V. Naoumov
PO-889
ILC2021