RECONVOCC : Can we reconvene chronic hepatitis C patients who were lost to follow-up ?

A ABERGEL[1], F BAILLY[2], J FOUCHER[3], J GOURNAY[4], B CHANTERANNE[1], F FAURE[1], S OUKIL[1], I ROSA[5], C GUILLEMARD[6], M MESZAROS[7], JP BRONOWICKI[8], V CANVA[9], I PORTAL [10], V LOUSTAUD RATTI[11], C SILVAIN[12], S POL [13], G LAMBLIN[1], L MUTI[1], M REYMOND[1] and B BUCHARD[1]

1 CHU Estaing, Clermont-Ferrand, France; 2 Hopital De La Croix Rousse, HCL, Lyon, France ; 3 CHU de Bordeaux, Bordeaux, France ; 4 University Hospital of Nantes, Nantes, France; 5 Centre Hospitalier Intercommunal, Créteil, France; 6 CHU Côte de Nacre, Caen, France ; 7 Hopital Gui De Chauliac, Montpellier, France; 8 CHU Nancy, Nancy, France ; 9 CHU Lille, Lille, France ; 10 Hopital de la Timone, Marseille, France ; 11 Limoges Dupuytren University Hospital Center, Limoges, France; 12 CHU Poitiers, Poitiers, France ; 13 Hopital Cochin, AP-HP, Paris, France

INTRODUCTION

Hepatitis C virus (HCV) elimination by 2030, as targeted by the World Health Organization (WHO), requires that 90% of people with chronic hepatitis C (CHC) be diagnosed and 80% treated. France is one of the countries engaged in this viral elimination strategy.

AIM

In the RECONVOCC study we assessed the effectiveness of a recall strategy. This project was initiated by the "Federation des Pôles de Référence Hépatites" (FPRH) and the main objective was to recall patients with CHC who were lost to follow-up in 15 French academic centers.

METHOD

The study population consists of patients HCV positive seen in 15 hospitals from 2003 to 2018 who were not cured. The patient should be lost to follow up for over 18 months and be born after 1934. RECONVOCC takes place in 2 phases (Figure 1). The first, based on patient databases and medical records identified eligible patients including those tested in the virology department and followed by non hepatologists. To contact them and obtain information on HCV status, several methods were used such as townhall databases, hospital entry offices, health insurance databases, contacting patients or general practitioners or gastroenterologists or other specialists by phone or by letter... The second part corresponds to the inclusion of patients in the care process with data collection. The case report form used is REDCAP.

Figure 1 : Differents steps of RECONVOCC Study

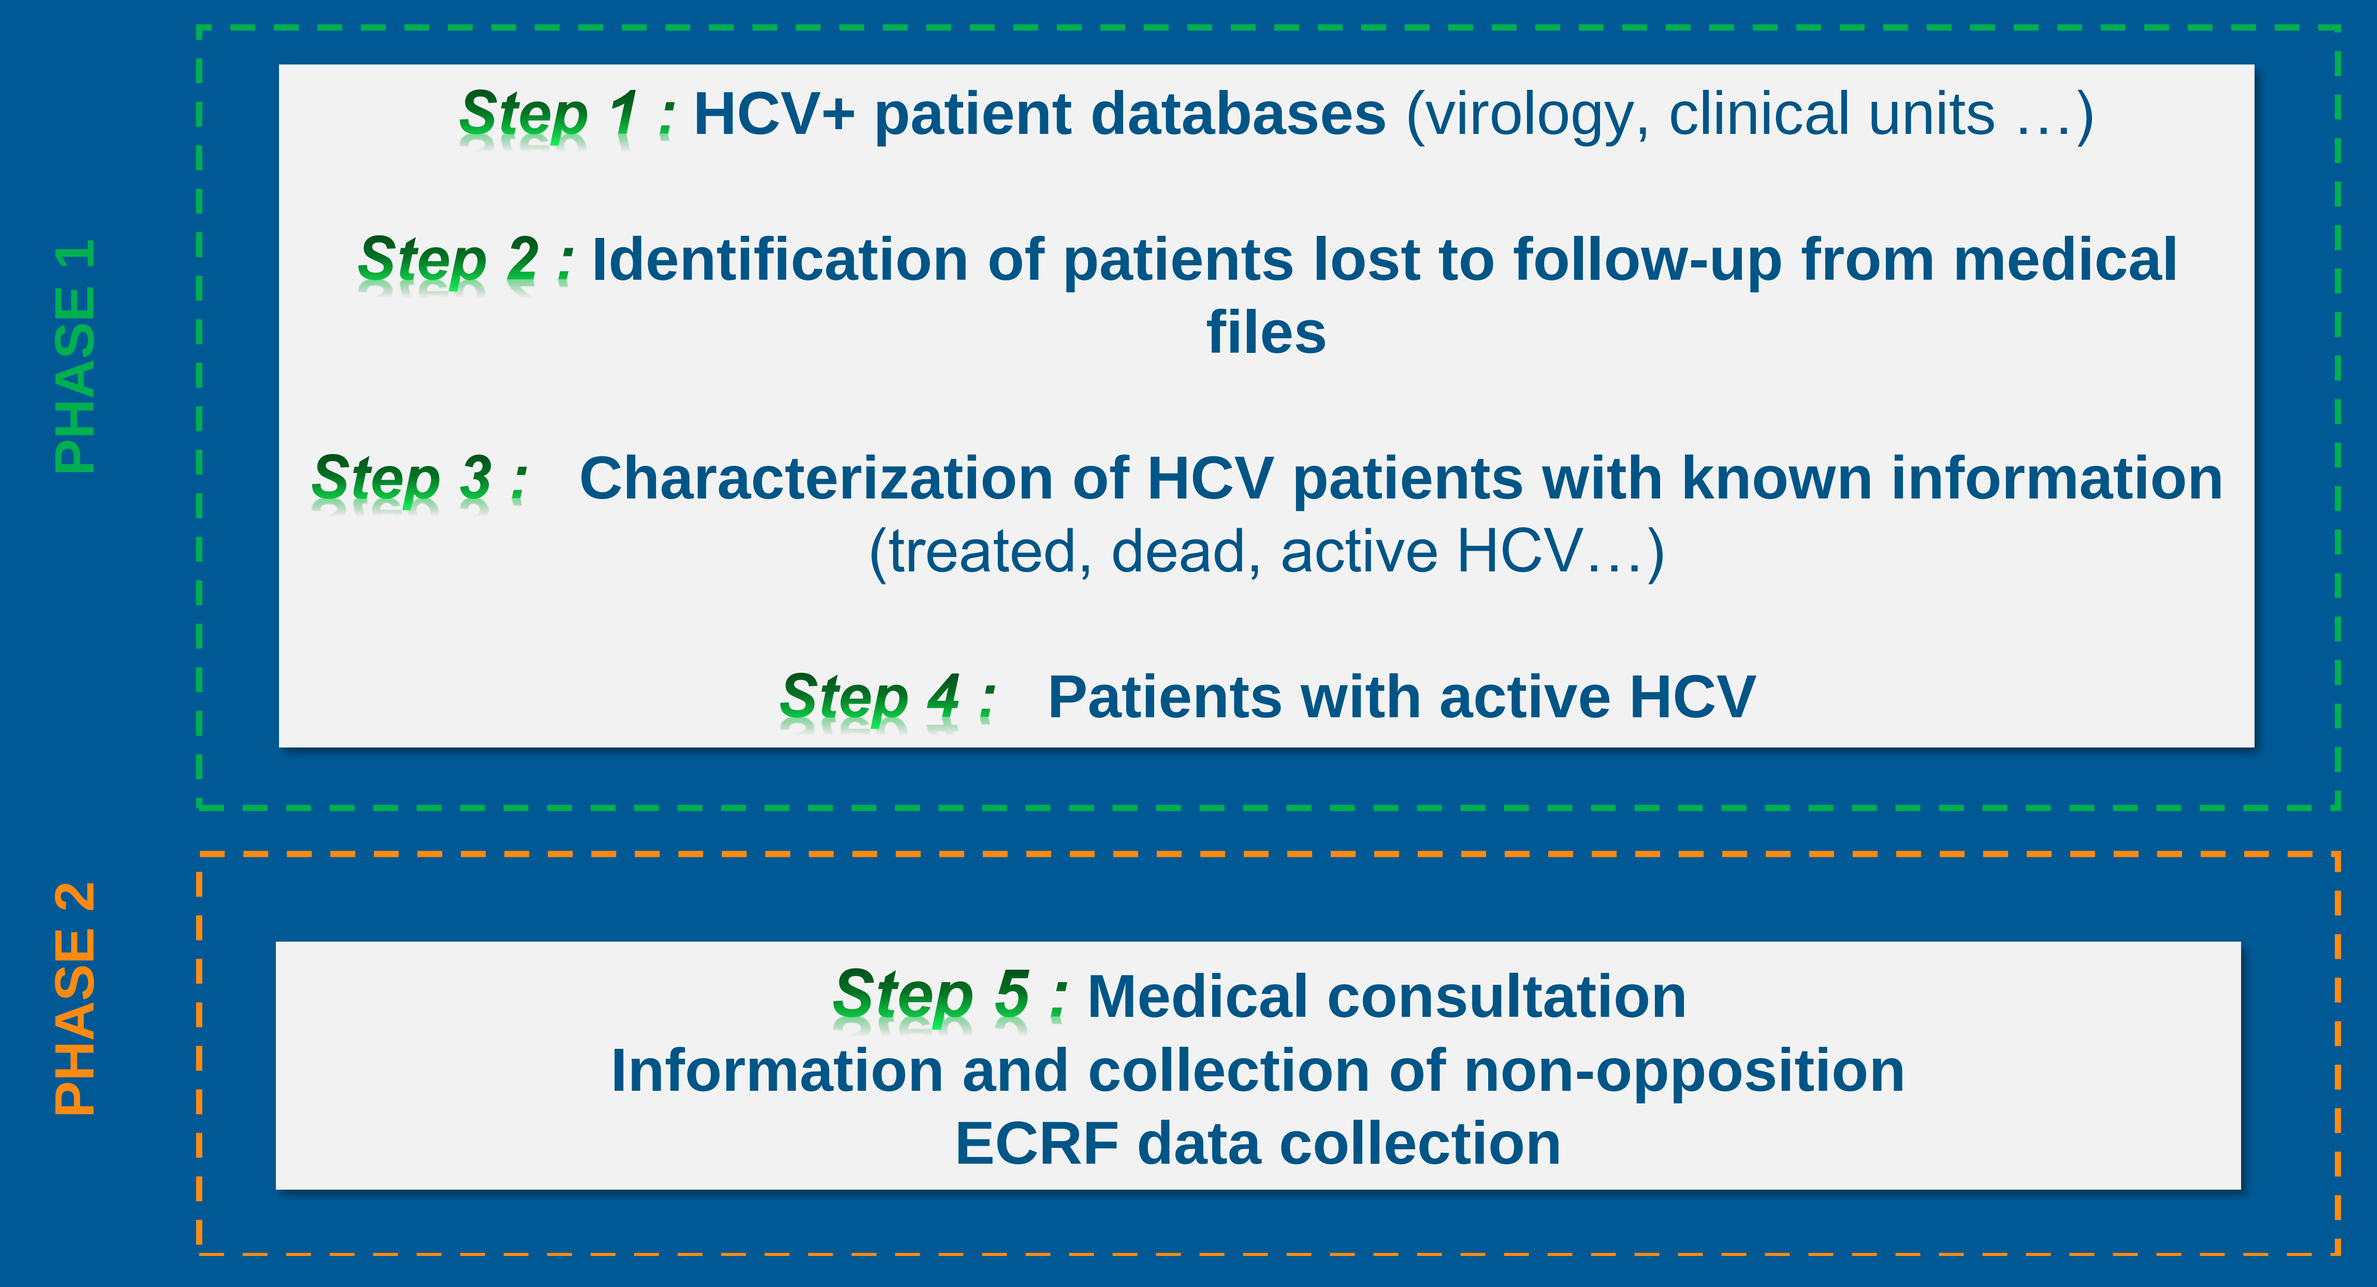
PHASE 1
Step 1 : HCV+ patient databases (virology, clinical units …)
Step 2 : Identification of patients lost to follow-up from medical files
Step 3 : Characterization of HCV patients with known information (treated, dead, active HCV…)
Step 4 : Patients with active HCV
PHASE 2
Step 5 : Medical consultation
Information and collection of non-opposition
ECRF data collection

RESULTS

Out of a total of 45 439 patients identified as HCV positive, approximately 88% (276/315) were followed inthe hepatology department. Up to now RECONVOCC's investigative work identifies 560 patients still carrying HCV. Of the 331 patients included in the eCRF (Figure 2) 315 patients were analysed (Table 1), 59% (186/315) were men. Mean age was 56 +/- 12 years and BMI was 26. 238/315 (76%) were treatment naïve. Twenty five per cent of patients (79/315) had clinically significant fibrosis (F3-F4). 44% (138/315) of patients were intravenous drug abusers (Table 2 repartition men/women). Genotypes 1a (30%) and 1b (16%) were the most frequent. 167/315 (53%) patients had a known co-morbidity. The most commonly cited reason for stopping hepatology monitoring were the lack of information for 36% of patients included and the low severity of liver fibrosis (15%) (Figure3). The average duration of the follow-up break was more than 7 years. The repartition was explained in Table 4. Until now 227 patients have already started treatment and 125 were cured. 64 patients were included but untreated (Table 3)

Figure 2: Inclusions by center

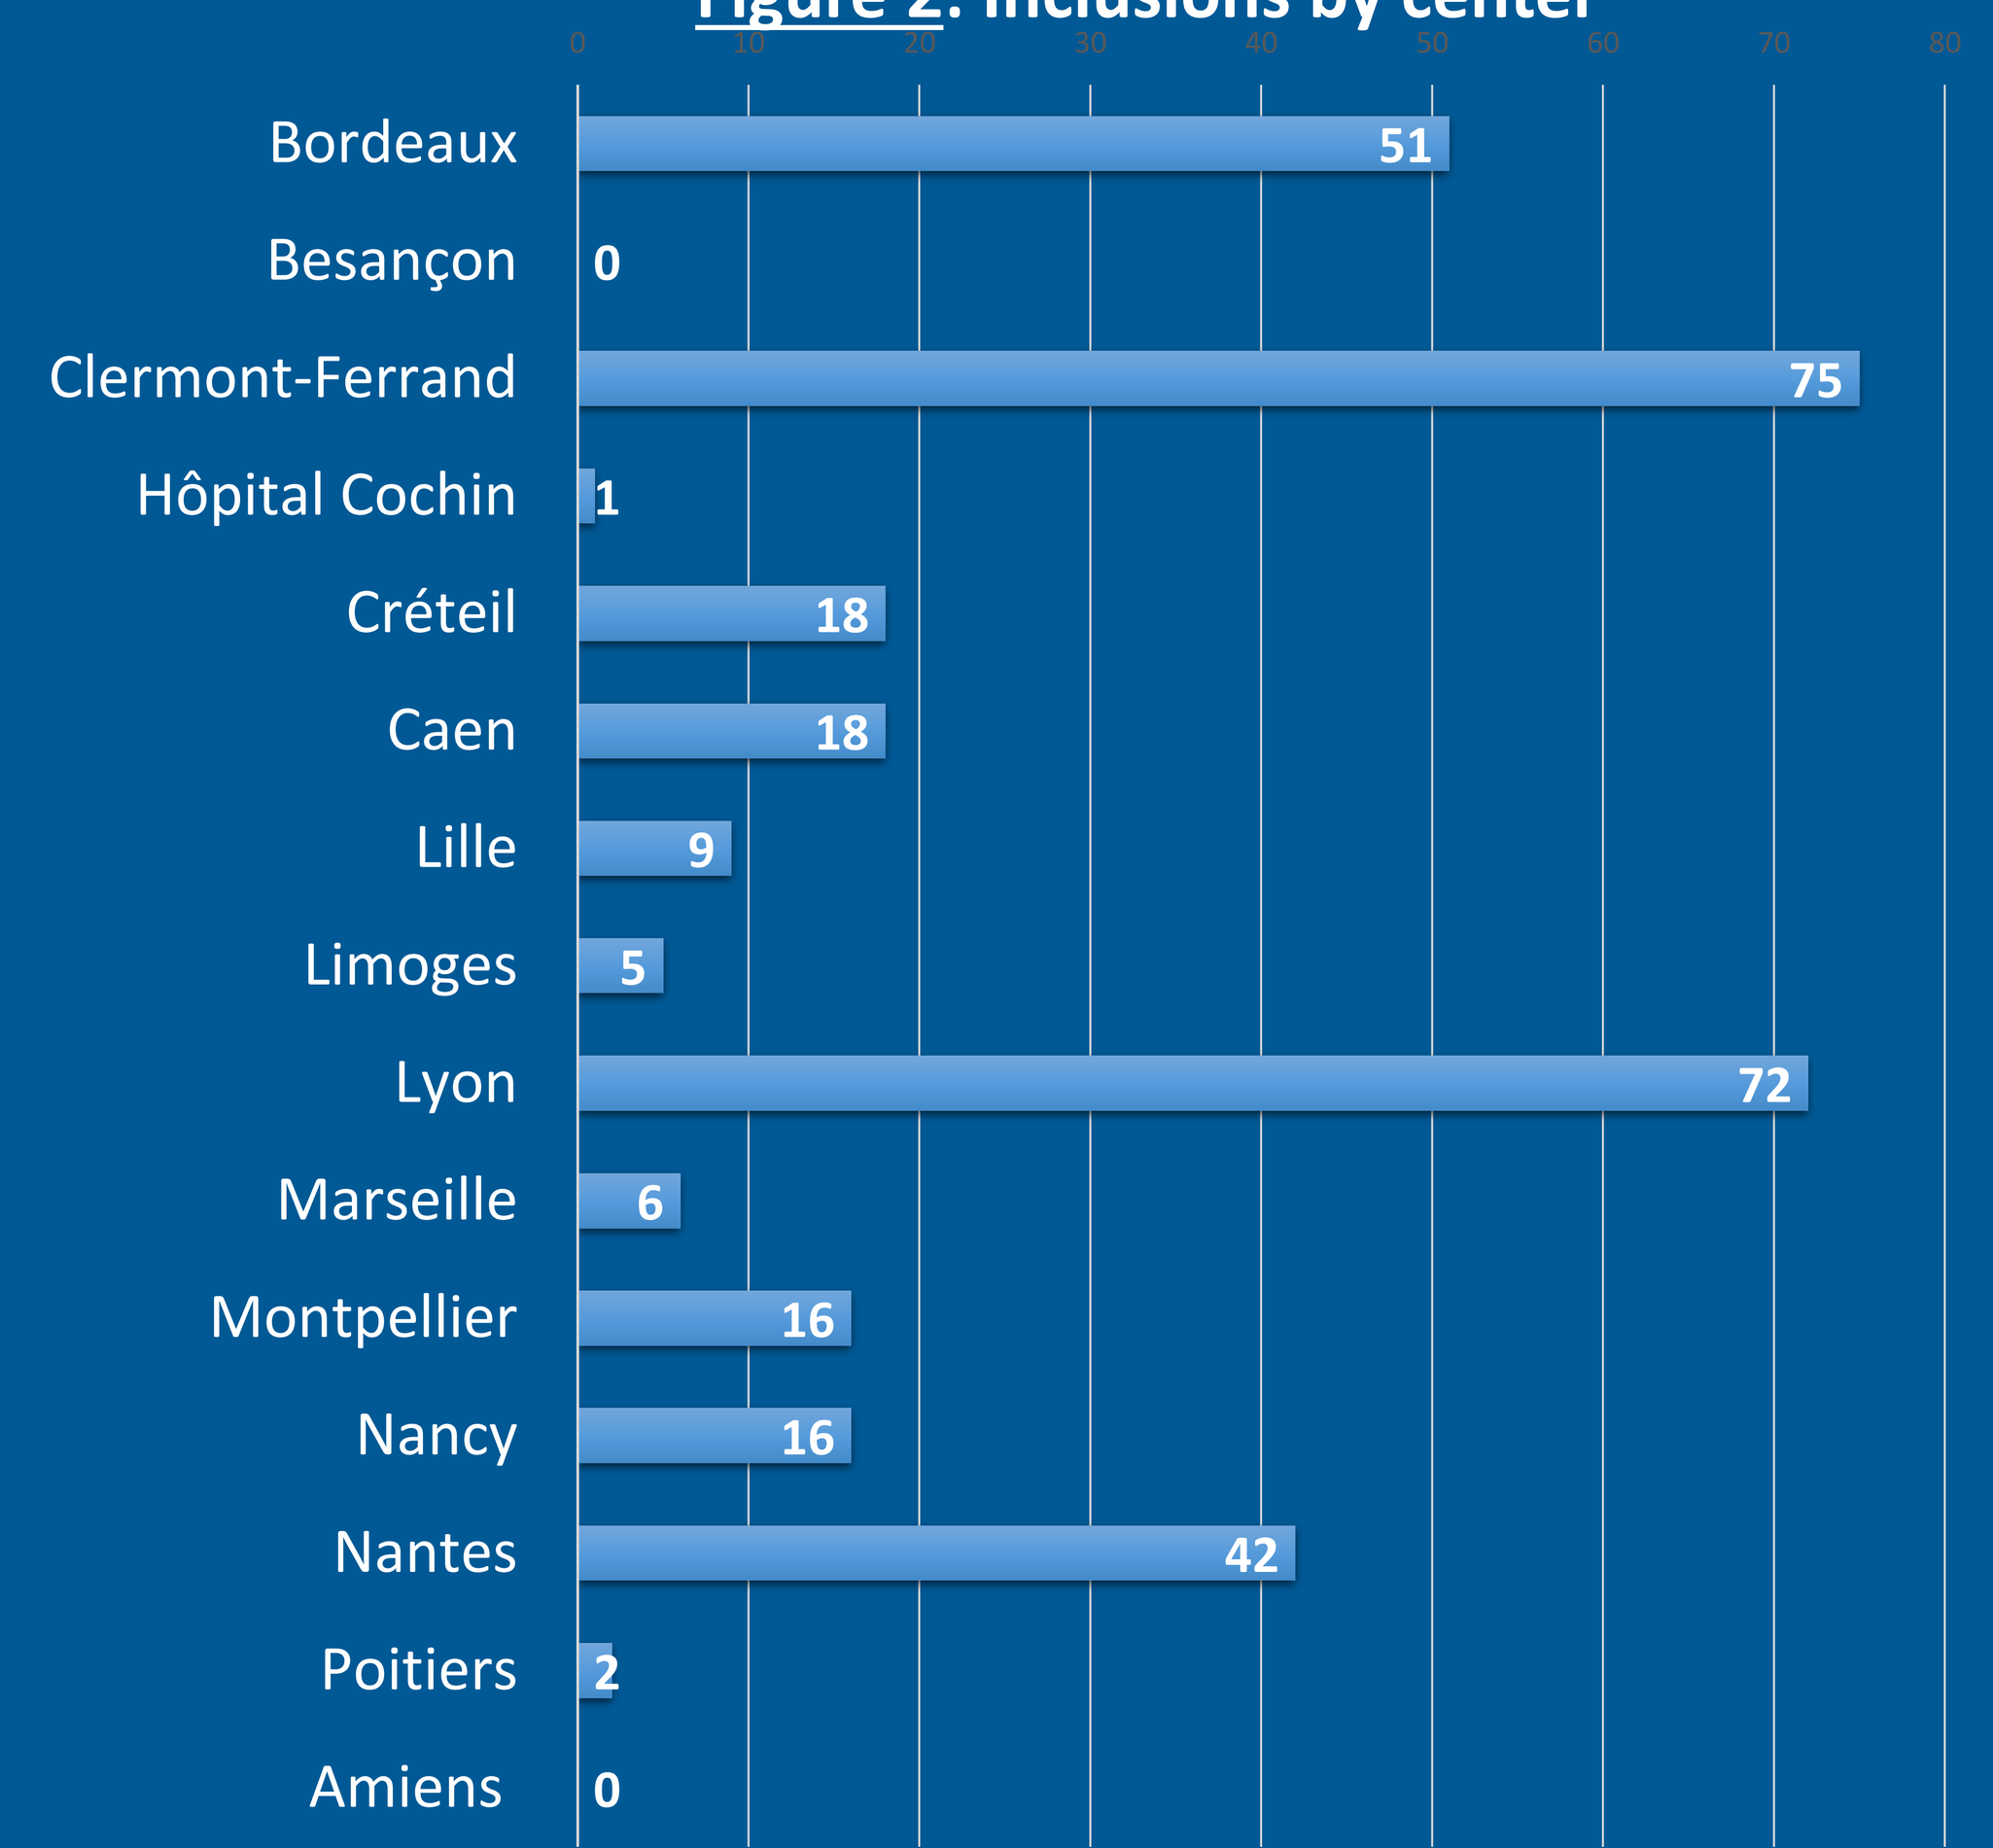
Bordeaux 51
Besançon 0
Clermont-Ferrand 75
Hôpital Cochin 1
Créteil 18
Caen 18
Lille 9
Limoges 5
Lyon 72
Marseille 6
Montpellier 16
Nancy 16
Nantes 42
Poitiers 2
Amiens 0

Table 2 : Contamination mode: men / women difference

	women (n = 129)	men (n = 186)
Intravenous drug user	32 (25%)	106 (57%)
Transfusion	35(27%)	32 (17%)
Other	31 (24%)	22 (12%)
Not done	31 (24%)	26 (14%)

Table 4 : Duration of lost to follow-up (in months) of the patients included * (n = 315)

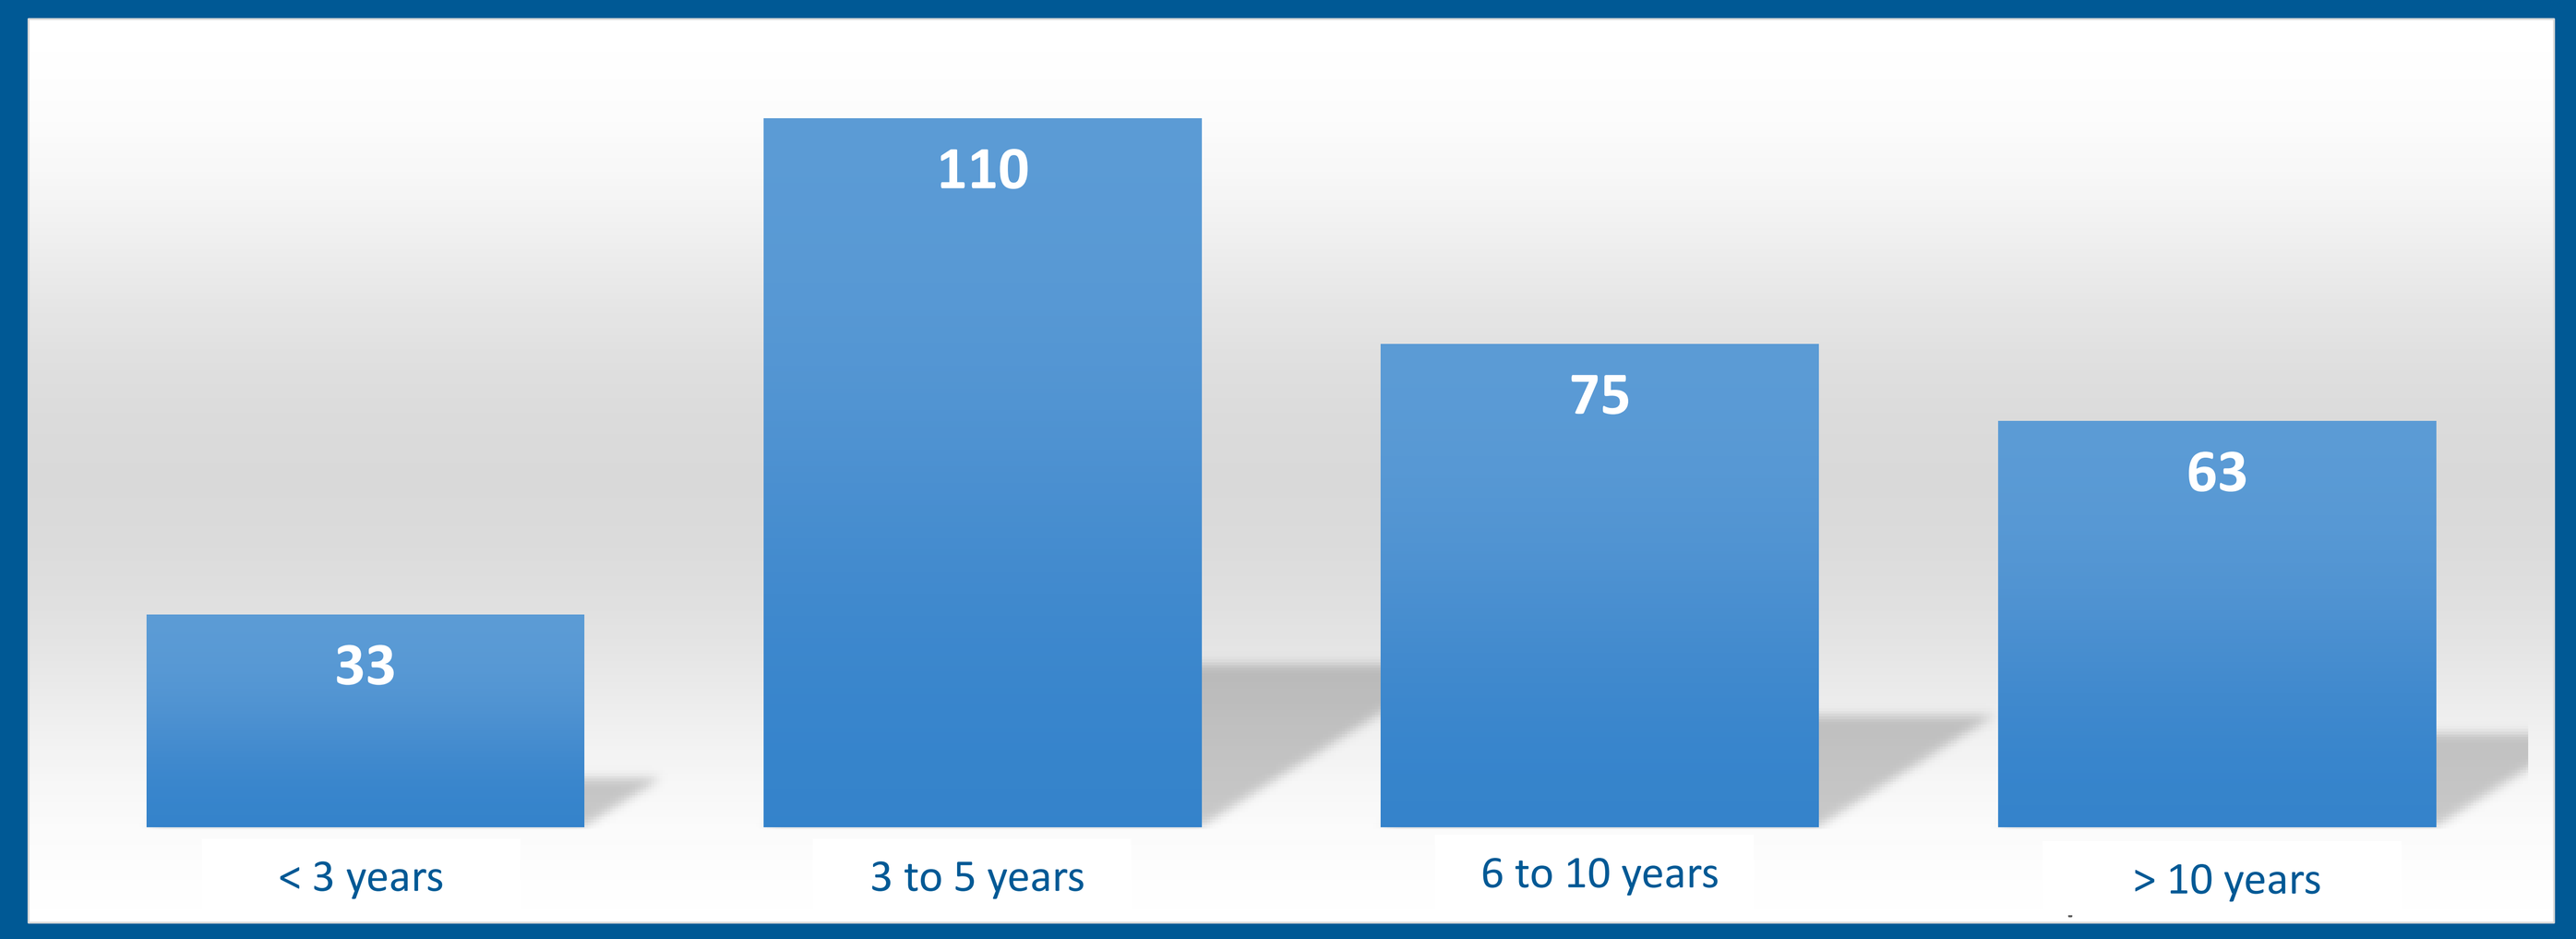
33
110
75
63
< 3 years
3 to 5 years
6 to 10 years
> 10 years

* For 34 patients data not known / not applicable to date

Figure 3: Reasons why patient did not return for a consultation (%)

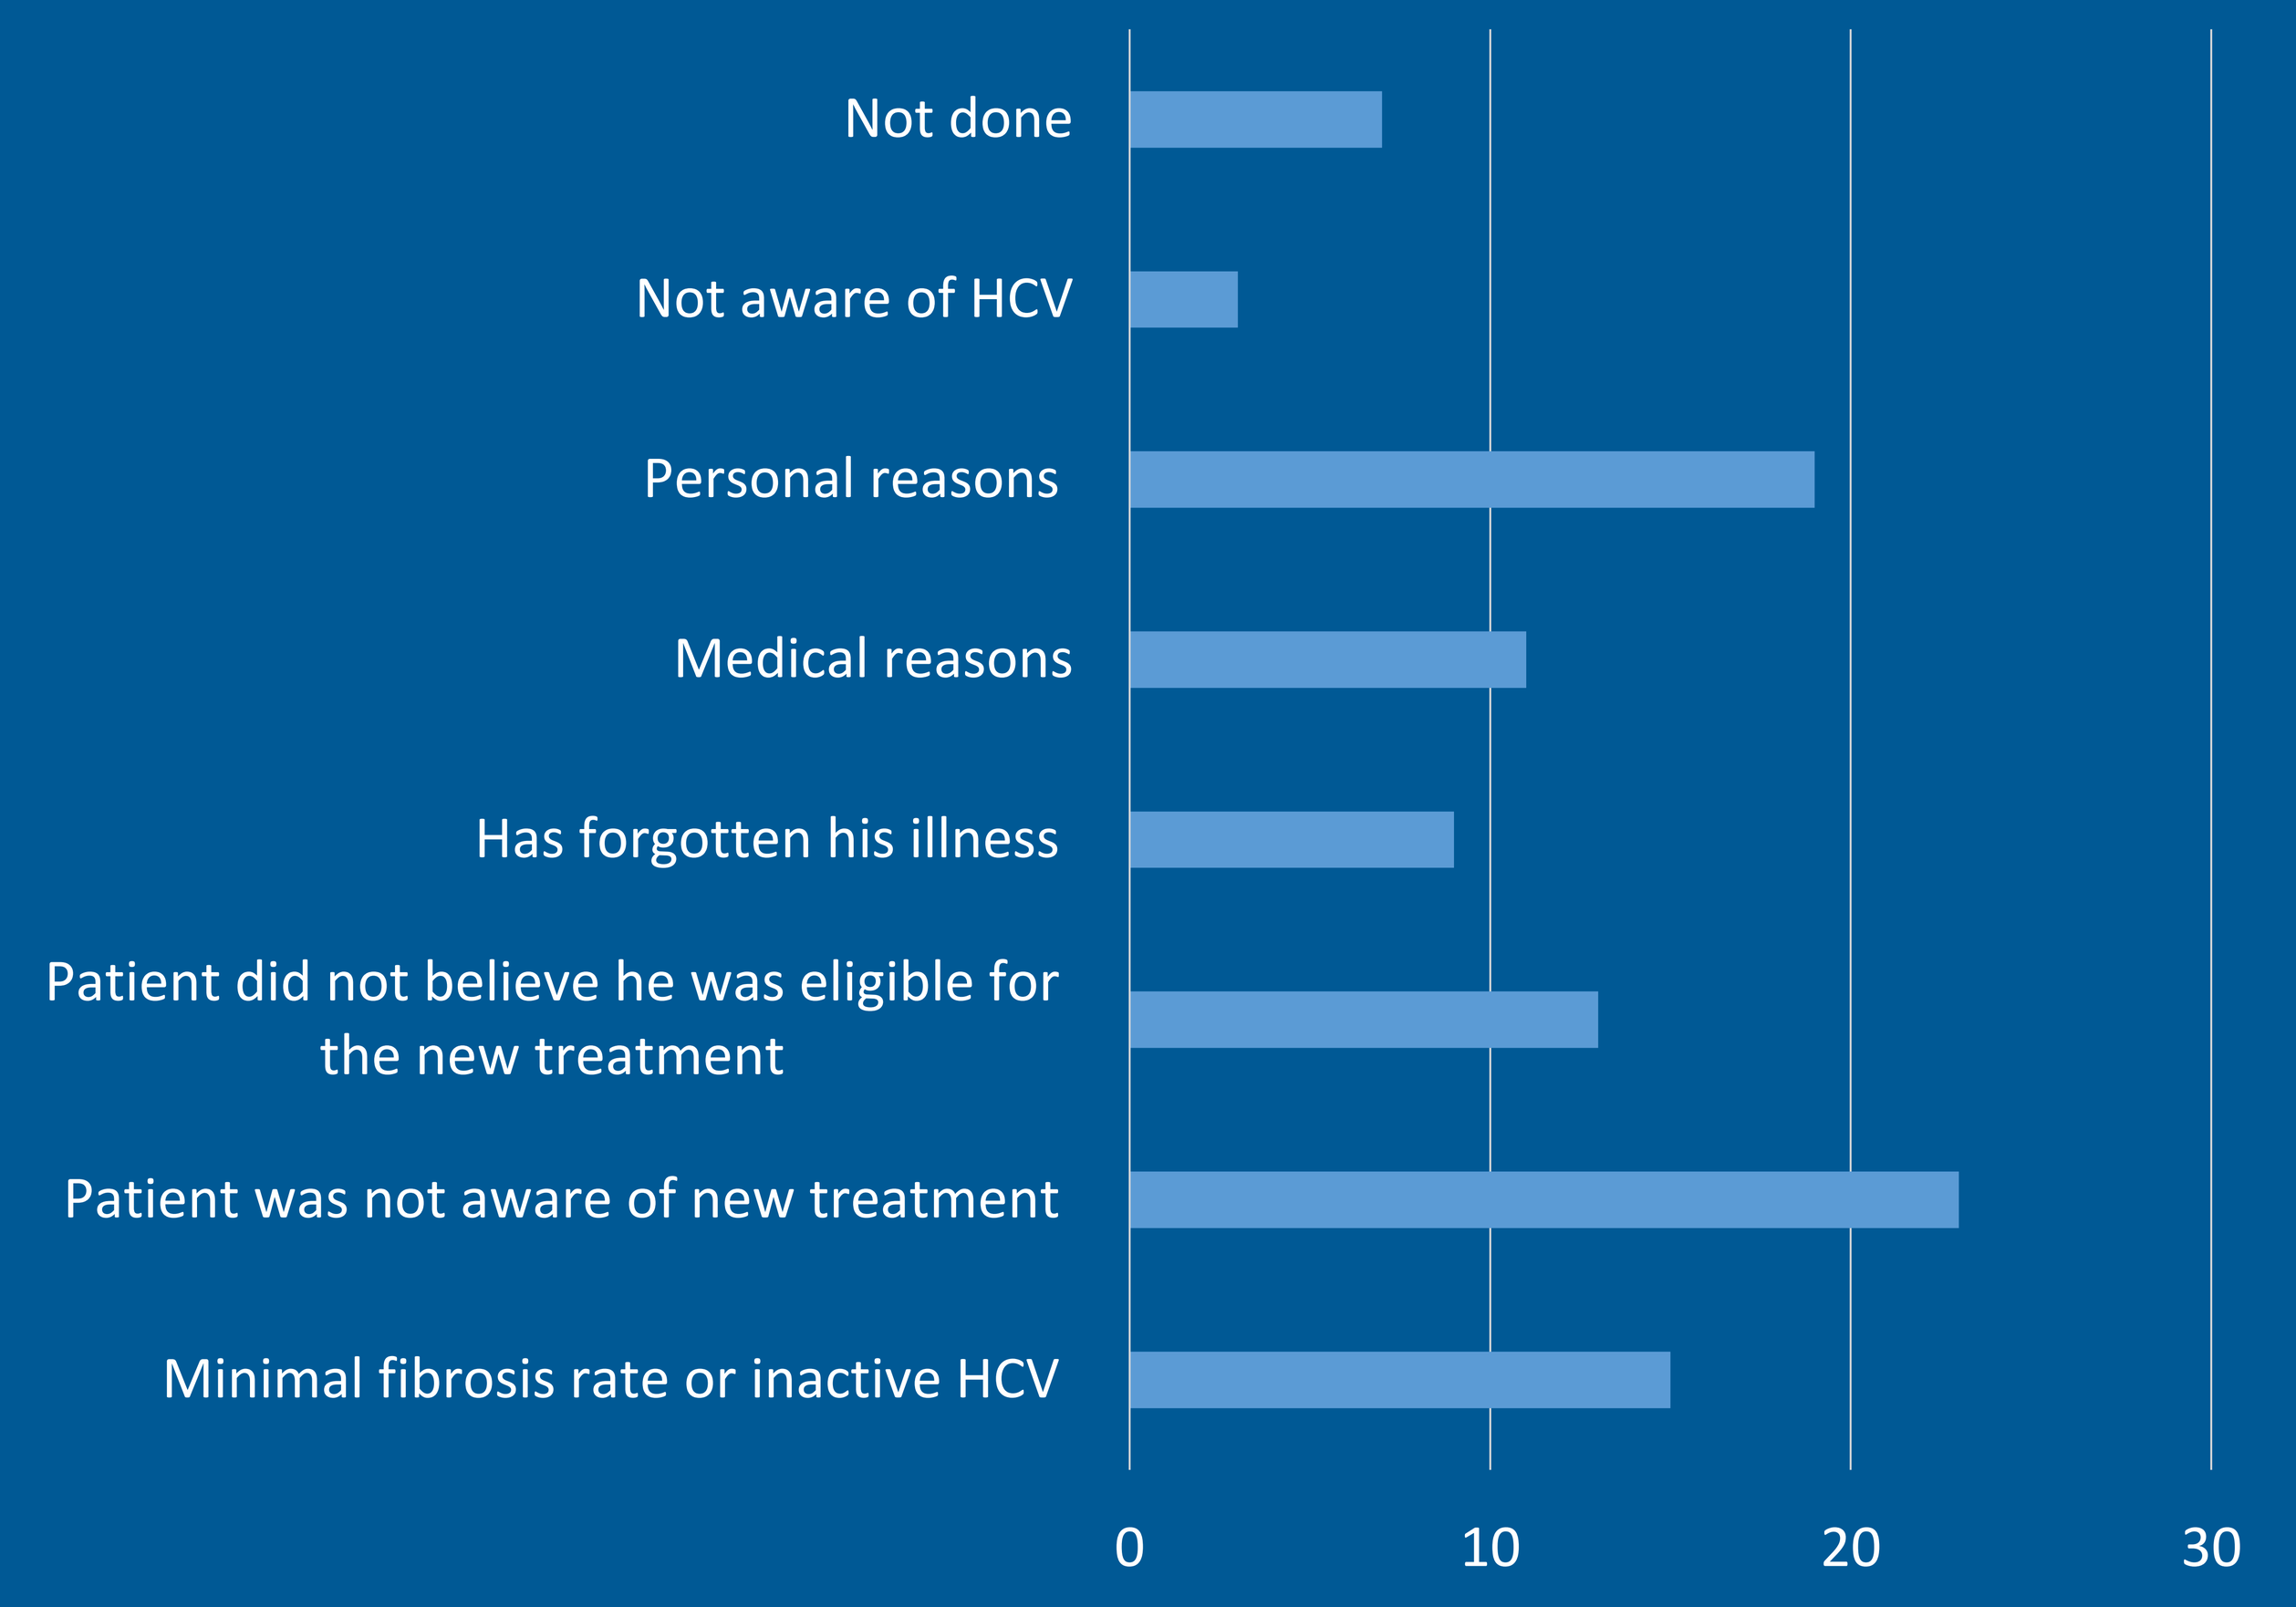
Not done
Not aware of HCV
Personal reasons
Medical reasons
Has forgotten his illness
Patient did not believe he was eligible for the new treatment
Patient was not aware of new treatment
Minimal fibrosis rate or inactive HCV
0
10
20
30

Table 1 : Patients caracteristics

	N=315 patients
Age, average (SD)	56 (12)
Men, n (%)	186(59%)
BMI, average (SD)	26 (3,7)
ALAT>N, n(%)	142 (45%)
Hepatology, n (%)	276 (88%)
Naive treatment, n (%)	238 (76%)
Fibrosis, n (%)	
F0-F2	225 (71%)
F3-F4	79 (25%)
Not done	11 (4%)
Genotype, n (%)	
1a	93 (30%)
1b	50(16%)
Comorbidity, n (%)	167 (53%)
Patients who started a treatment, n (%)	227 (72%)
Cured patients, n (%)	125 (40%)

Table 3 : Included but untreated patients at the moment

N = 64	Included untreated patients	%
23	Non-HCV + carriers (cured / false positives)	36%
8	Fear of adverse events/ Refusal of treatment	13%
11	Therapeutic abstention / medical reason	17%
16	Awaiting Examinations / Refletion	25%
6	Lost to follow up	9%

* For 24 patients information on start of treatment missing to date

CONCLUSIONS

This work shows that there is still a high number of patients infected with the HCV who have not yet benefited from treatment with direct antivirals agents. Patients who were lost to follow-up should be reconvened to achieve HCV elimination in 2030. The study is still underway.

CONTACT INFORMATION

aabergel@chu-clermontferand.fr

ACKNOWLEDGEMENTS

This study was supported by ABBVIE

Scan to download the poster
General hepatology
Armand Abergel
PO-785
ILC2021