Substantial Clinical Benefits With Odevixibat Treatment Across Progressive Familial Intrahepatic Cholestasis Genetic Deficiencies: Subgroup Analysis of Serum Bile Acids, Pruritus, and Safety Using Pooled Data From the PEDFIC 1 and 2 Studies

Richard J. Thompson[1], Patrick Horn[2], Roderick H.J. Houwen[3], Florence Lacaille[4], Quanhong Ni[2], Philip Stein[2], Mary Elizabeth Tessier[5], Carrie Thompson[2], Jennifer M. Vittorio[6], Lise Kjems[2]

[1]Institute of Liver Studies, King's College London, London, UK; [2]Albireo Pharma, Inc., Boston, MA, USA; [3]Wilhelmina Children's Hospital and University Medical Center, Utrecht, Netherlands; [4]Paediatric Gastroenterology-Hepatology-Nutrition Unit, Hôpital Universitaire Necker-Enfants Malades, Paris, France; [5]Department of Pediatrics, Section of Pediatric Gastroenterology, Hepatology, and Nutrition, Baylor College of Medicine/Texas Children's Hospital, Houston, TX, USA; [6]Department of Surgery, Center for Liver Disease and Transplantation, Columbia University Medical Center, New York, NY, USA

INTRODUCTION

- Progressive familial intrahepatic cholestasis (PFIC): a group of rare genetic diseases of hepatocellular origin characterized by cholestasis, fat-soluble vitamin deficiency, and progressive liver disease[1]
 - Mutations in proteins with diverse functions underlie PFIC, including familial intrahepatic cholestasis protein 1 (FIC1; encoded by *ATP8B1*) and bile salt export pump (BSEP; encoded by *ABCB11*), corresponding with PFIC type 1 (PFIC1) and PFIC type 2 (PFIC2), respectively[2]
 - PFIC2 can be further categorized by BSEP subtype[3]
 - BSEP subtype 1: patients with p.E297G and/or p.D482G missense mutations that result in residual BSEP activity
 - BSEP subtype 2: patients with ≥1 missense mutation but not p.D482G and/or p.E297G; this group likely has a range of BSEP function, depending on the type of amino acid substituted
 - BSEP subtype 3: patients with protein-truncating mutations and no residual BSEP function
- Ileal bile acid transporter (IBAT): resorbs intestinal bile acids for recirculation to the liver; potential therapeutic target for pharmacologic inhibition of the enterohepatic circulation[4] (**Figure 1**)

Figure 1. Role of IBAT in the Enterohepatic Circulation of Bile Acids and Effects of IBAT Inhibition

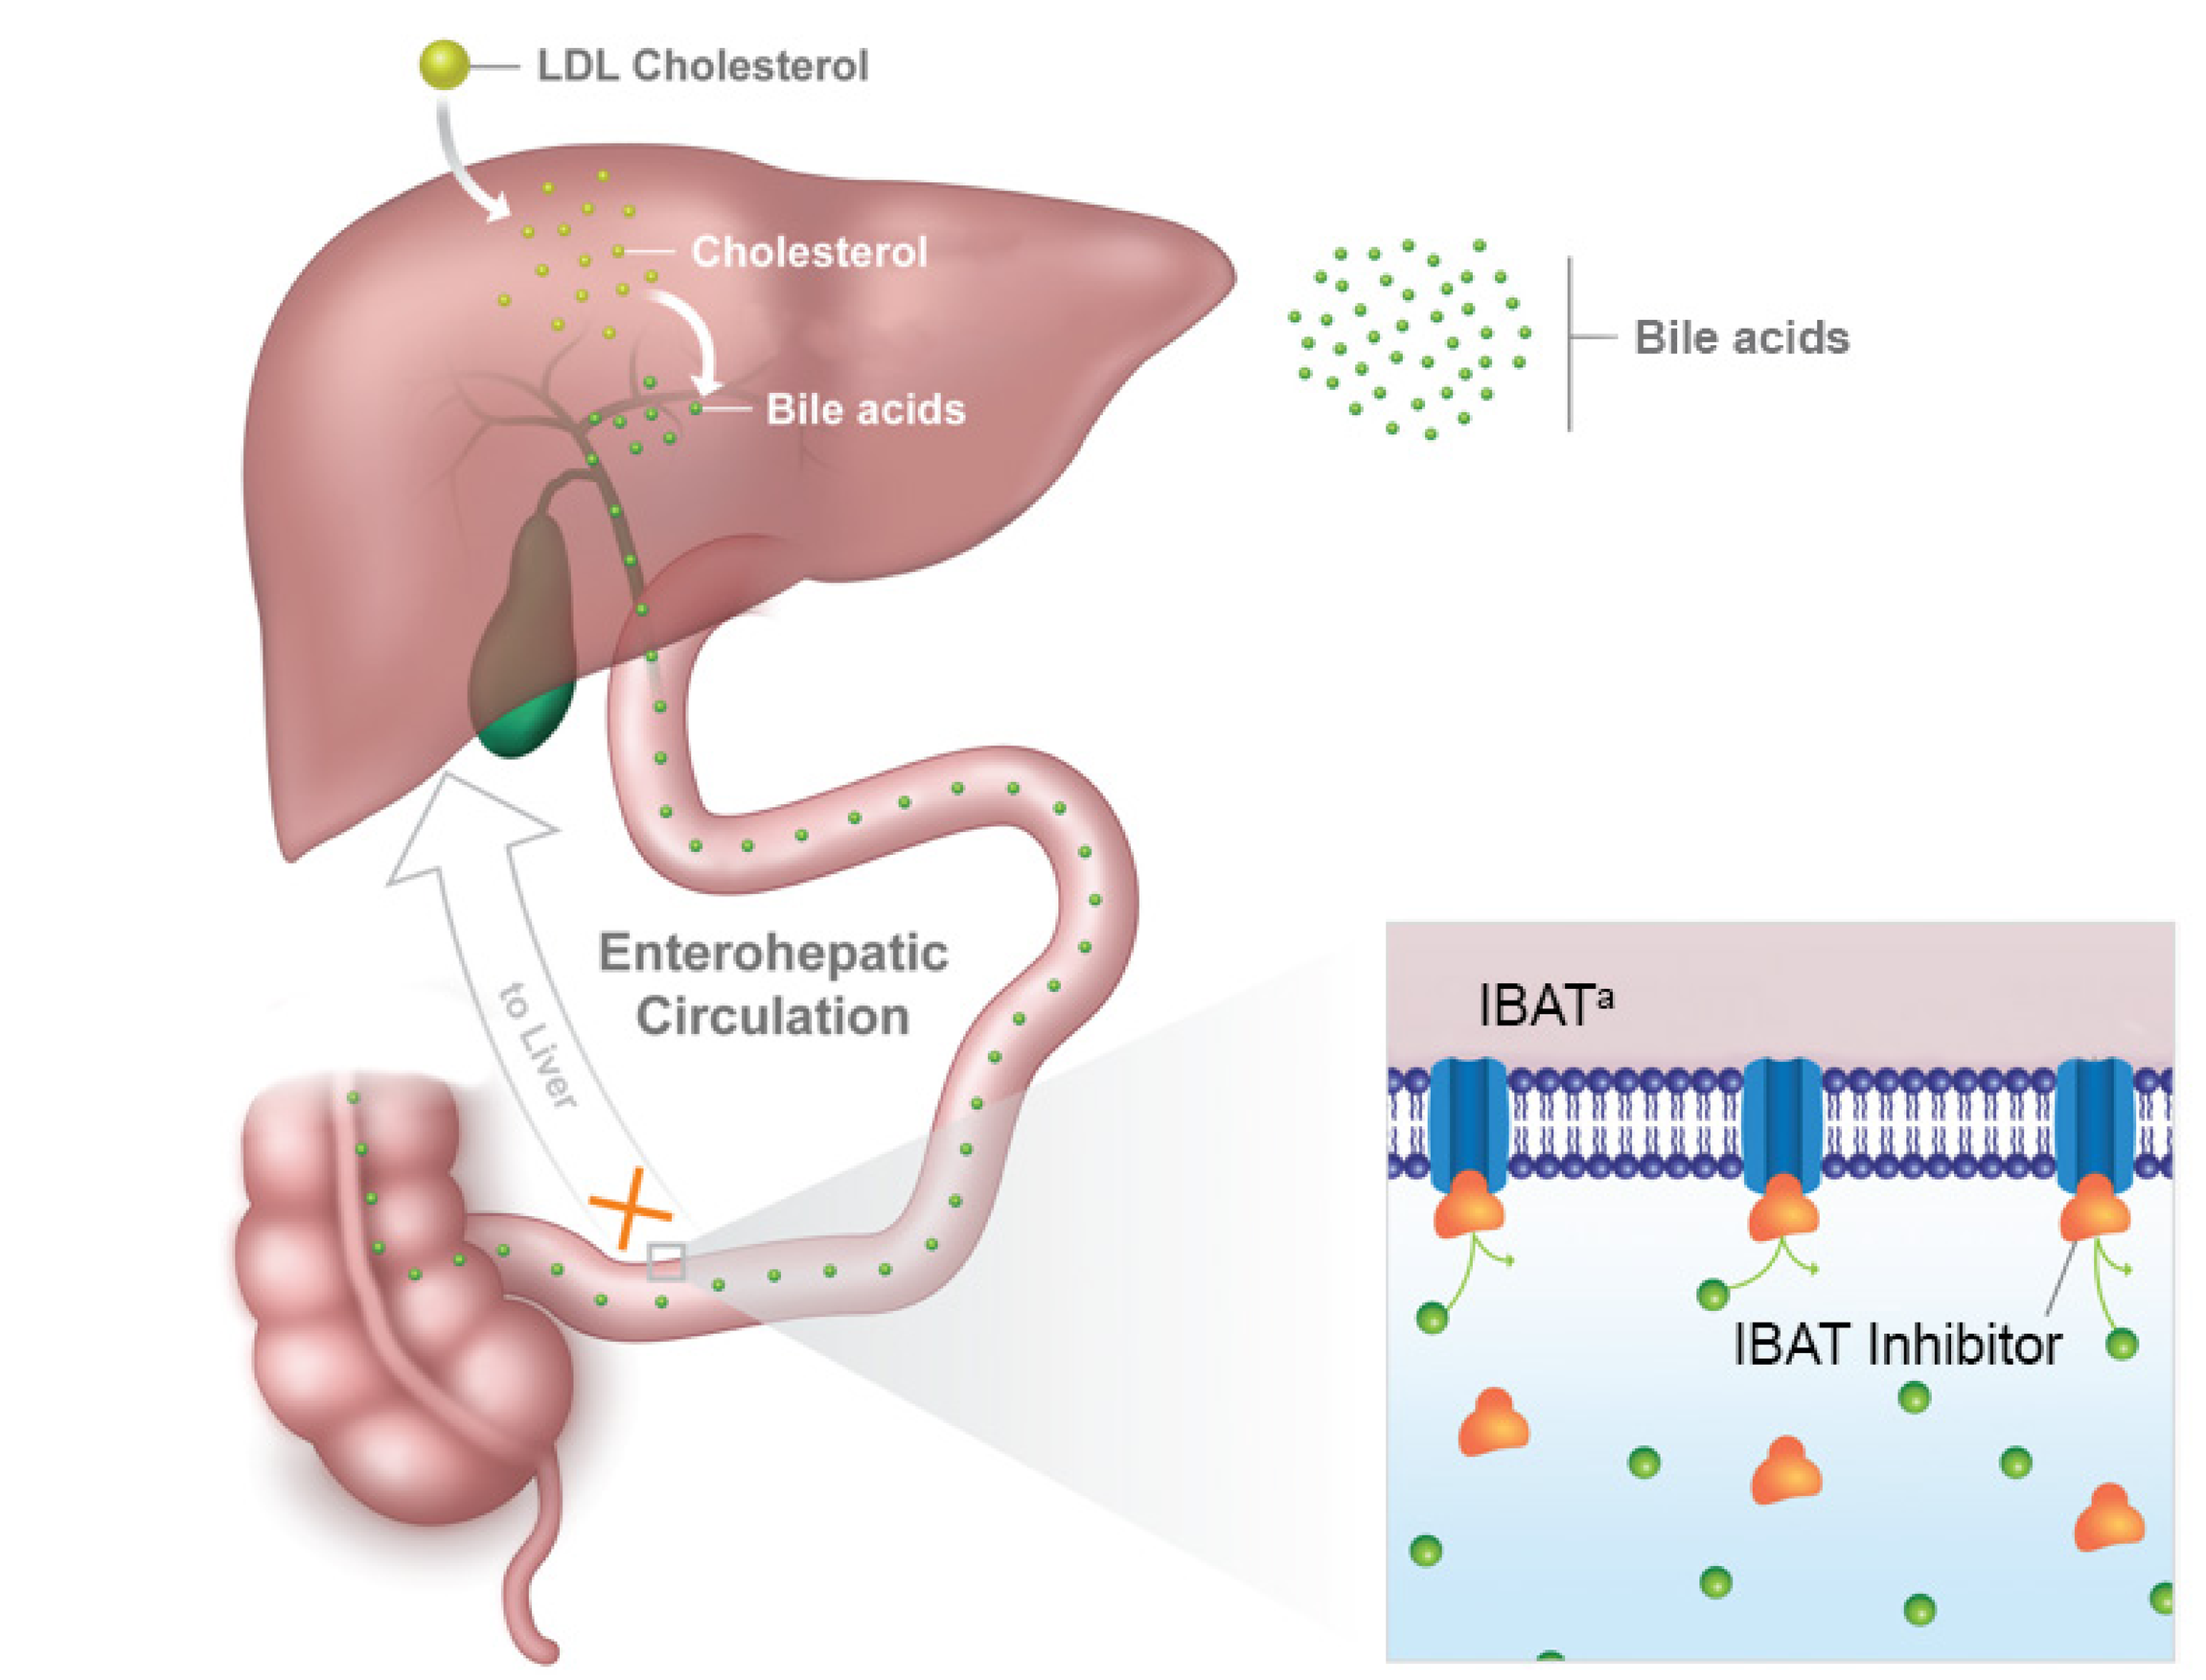

LDL Cholesterol
Cholesterol
Bile acids
Bile acids
Enterohepatic Circulation
to Liver
IBAT[a]
IBAT Inhibitor

[a]Also known as the apical sodium-dependent bile acid transporter.
IBAT, ileal bile acid transporter; LDL, low-density lipoprotein.

- Odevixibat: orally administered, reversible, potent, and selective IBAT inhibitor that decreases bile acid reuptake and increases bile acid clearance through the colon[5]
- Odevixibat investigated as treatment for PFIC in two phase 3 studies: PEDFIC 1 and PEDFIC 2 (**Figure 2**)[6,7]
 - PEDFIC 1 enrolled patients with PFIC1 and PFIC2
 - PEDFIC 2 enrolled patients with any PFIC subtype

Figure 2. PEDFIC 1 and PEDFIC 2 Study Designs and Key Efficacy Outcomes[6,7]

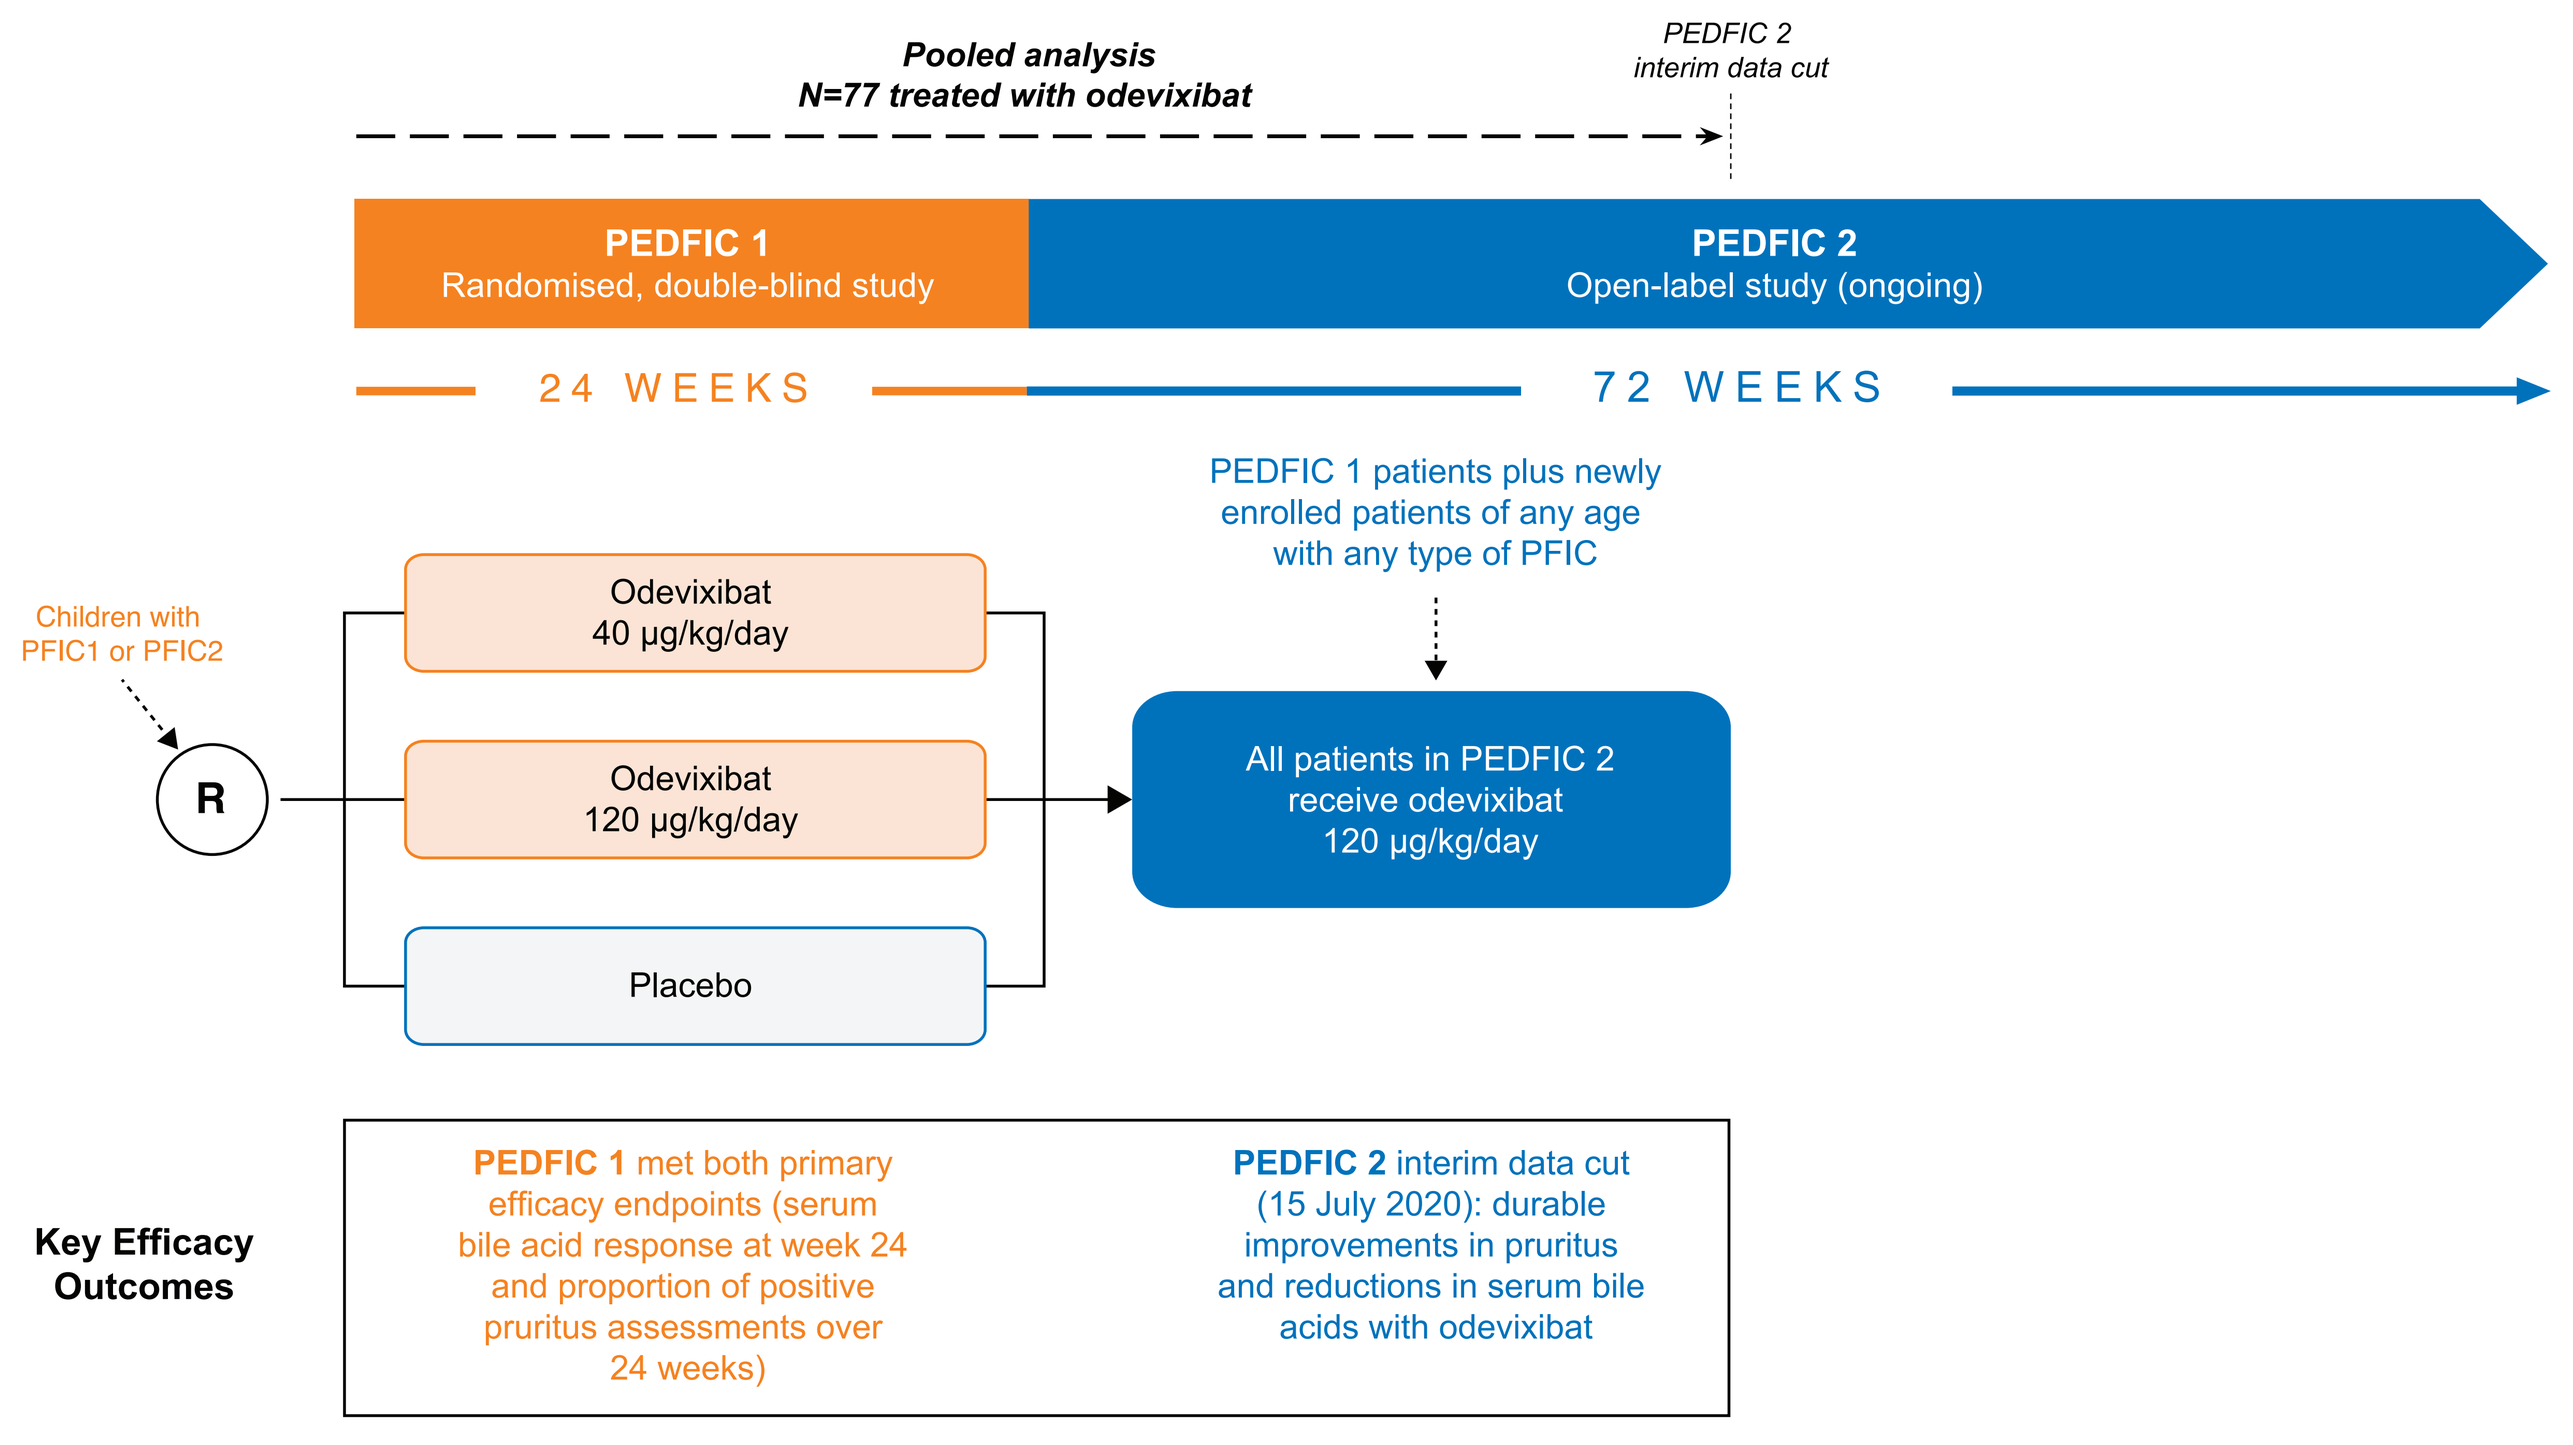

Pooled analysis
N=77 treated with odevixibat
PEDFIC 2 interim data cut
PEDFIC 1
Randomised, double-blind study
PEDFIC 2
Open-label study (ongoing)
24 WEEKS
72 WEEKS
PEDFIC 1 patients plus newly enrolled patients of any age with any type of PFIC
Children with PFIC1 or PFIC2
R
Odevixibat 40 µg/kg/day
Odevixibat 120 µg/kg/day
Placebo
All patients in PEDFIC 2 receive odevixibat 120 µg/kg/day
Key Efficacy Outcomes
PEDFIC 1 met both primary efficacy endpoints (serum bile acid response at week 24 and proportion of positive pruritus assessments over 24 weeks)
PEDFIC 2 interim data cut (15 July 2020): durable improvements in pruritus and reductions in serum bile acids with odevixibat

R, randomised; PFIC, progressive familial intrahepatic cholestasis.

- Objective: describe outcomes by genotype subgroup using pooled data from these studies

METHODS

Patients and Pooling of Data

- Patients eligible for PEDFIC 1 and PEDFIC 2 had PFIC, elevated serum bile acids, and significant pruritus
- Pooled analysis period: from first-ever odevixibat dose in PEDFIC 1 or PEDFIC 2 through interim data cut of PEDFIC 2 (15 July 2020)
 - Main focus: efficacy data through week 24 of PEDFIC 2
 - Includes up to 48 weeks of cumulative odevixibat exposure for patients who initiated odevixibat in PEDFIC 1
 - Includes less than 48 weeks of exposure for most patients who initiated odevixibat in PEDFIC 2
 - Analysis population: patients with PFIC1 or PFIC2 (BSEP subtypes 1 and 2[3]); patients with PFIC2 with BSEP subtype 3 not included in the analysis

Assessments

- Assessments presented by patient genotype group: serum bile acid response, change in pruritus, and safety
 - Serum bile acid response defined as a ≥70% reduction from baseline or a serum bile acid concentration ≤70 µmol/L
 - Blinded serum bile acid measurements taken at all study visits
 - Change in pruritus based on proportion of positive pruritus assessments (PPAs; ie, scratching score ≤1 or a ≥1-point drop from baseline on the PRUCISION instrument) at the patient level
 - Pruritus scored from 0 to 4 by caregivers using a scale validated by blinded, psychometric analysis conducted by an independent group; higher scores indicate worse symptoms, and a decrease of ≥1 point from baseline is clinically meaningful
 - Safety monitoring included assessment of treatment-emergent adverse events (TEAEs)

RESULTS

Patients

- Of 77 patients who received odevixibat during the pooled analysis period, 20 had PFIC1 (26%), 51 (66%) had PFIC2, and 6 (8%) had other PFIC subtypes (**Table 1**)
 - Of those with PFIC2, BSEP subtype 1 or 2 present in 13 (25%) and 36 (71%) patients, respectively
 - Data for 69 patients with PFIC1 and PFIC2 (BSEP subtype 1 and 2) included in these subgroup analyses
- Patient demographics and baseline characteristics, including mean age and proportions of patients using ursodeoxycholic acid or rifampicin at baseline, were similar among patients with different genotypes (**Table 1**)

Table 1. Patient Demographics and Baseline Characteristics

	Overall, N=77	Patients with PFIC1, n=20	Patients with PFIC2, n=51	Patients with PFIC2, BSEP subtype 1, n=13	Patients with PFIC2, BSEP subtype 2, n=36
Age, mean (SD), years	5.2 (4.3)	4.8 (3.4)	4.9 (4.5)	5.1 (5.1)	4.9 (4.5)
Female, n (%)	39 (51)	7 (35)	26 (51)	6 (46)	20 (56)
PFIC type, n (%)					
PFIC1 (FIC1 deficiency)	20 (26)	20 (100)	–	–	–
PFIC2 (BSEP deficiency)	51 (66)	–	51 (100)	13 (100)	36 (100)
Other PFIC types	6 (8)	–	–	–	–
UDCA at baseline, n (%)	62 (81)	15 (75)	42 (82)	11 (85)	29 (81)
Rifampicin at baseline, n (%)	48 (62)	12 (60)	35 (69)	11 (85)	22 (61)
Significant pruritus present per investigator report, n (%)	74 (96)	19 (95)	49 (96)	12 (92)	35 (97)
Serum bile acid level >100 µmol/L within 6 months prior to screening, n (%)	52 (68)	15 (75)	35 (69)	8 (62)	25 (69)

BSEP, bile salt export pump; FIC, familial intrahepatic cholestasis; PFIC, progressive familial intrahepatic cholestasis; SD, standard deviation; UDCA, ursodeoxycholic acid.

Efficacy

Serum Bile Acid Response

- At weeks 46–48 of odevixibat treatment, ≥50% of patients met serum bile acid response criteria, regardless of genotype (**Figure 3A**)
- Percentages of patients who met serum bile acid response increased over time (**Figure 3B**)

Figure 3. Percentage of Patients Who Met Serum Bile Acid Response Criteria After 48 Weeks of Treatment (A) and Over Time (B) With Odevixibat: Subgroup Analysis by Genotype

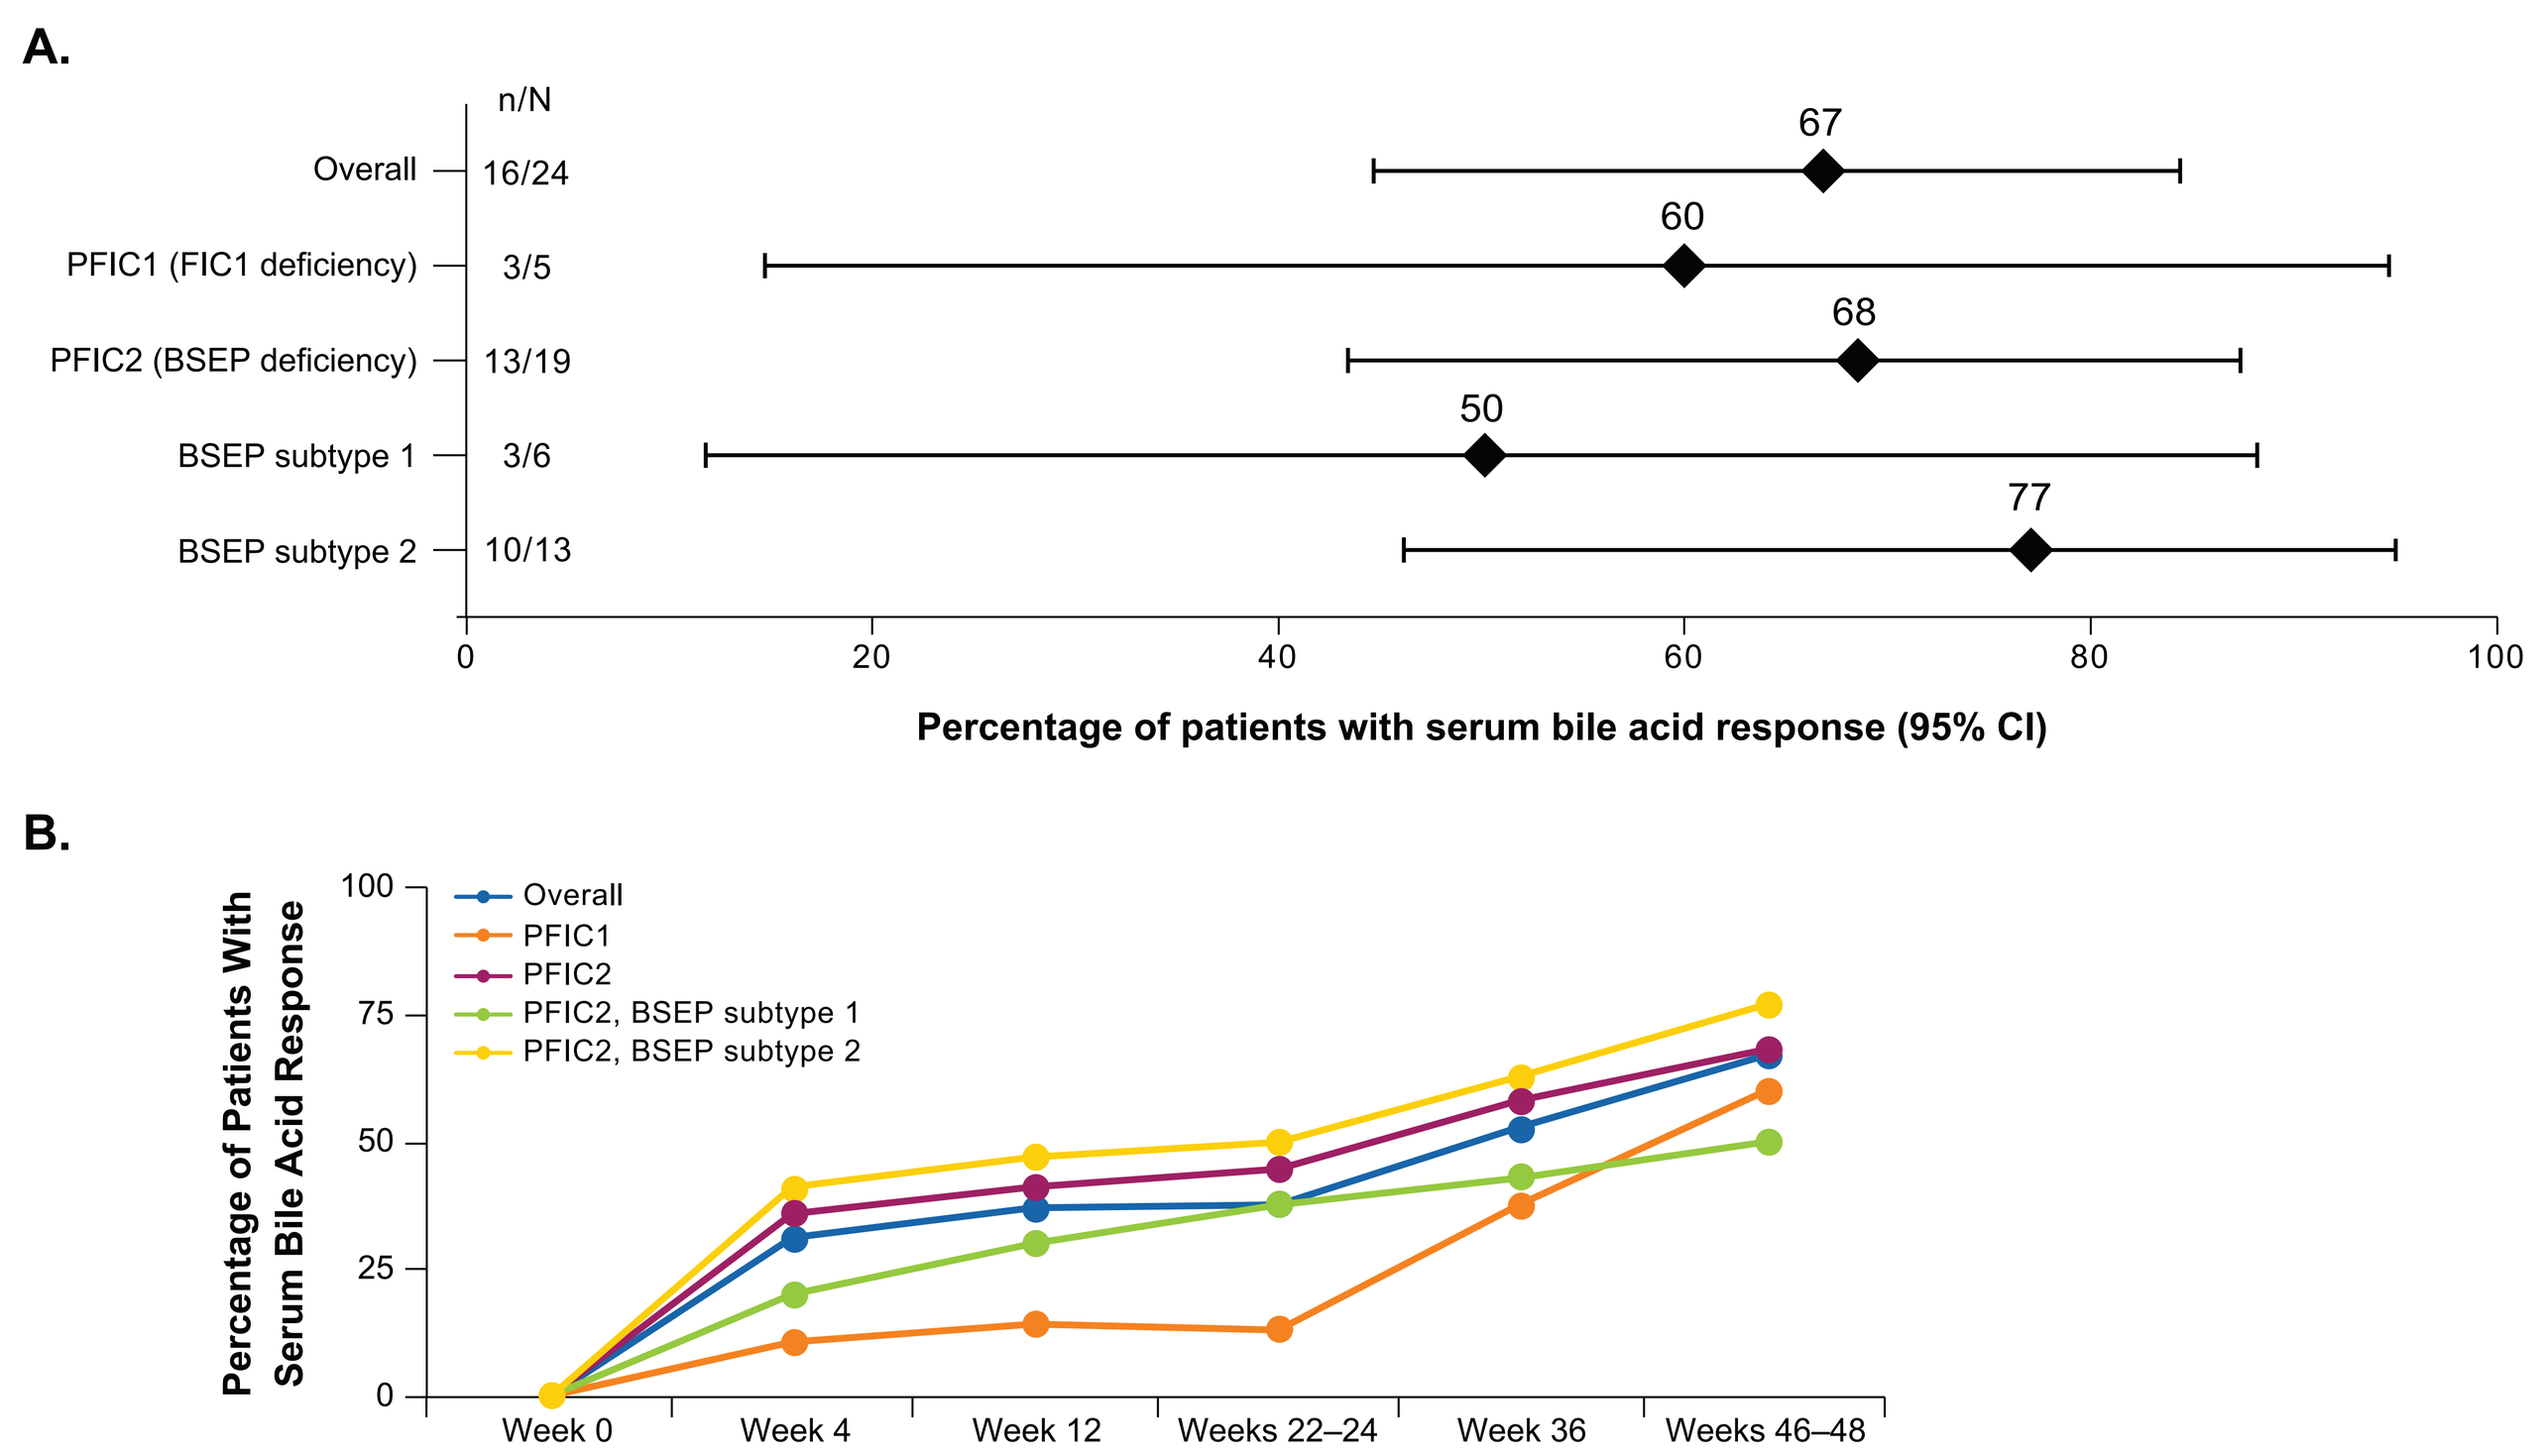

A.
n/N
Overall 16/24 67
PFIC1 (FIC1 deficiency) 3/5 60
PFIC2 (BSEP deficiency) 13/19 68
BSEP subtype 1 3/6 50
BSEP subtype 2 10/13 77
0 20 40 60 80 100
Percentage of patients with serum bile acid response (95% CI)
B.
Percentage of Patients With Serum Bile Acid Response
100 75 50 25 0
Overall
PFIC1
PFIC2
PFIC2, BSEP subtype 1
PFIC2, BSEP subtype 2
Week 0 Week 4 Week 12 Weeks 22–24 Week 36 Weeks 46–48
Patients at each time point, N

Patients at each time point, N	Week 0	Week 4	Week 12	Weeks 22–24	Week 36	Weeks 46–48
Overall	69	68	63	56	34	24
PFIC1	20	18	14	15	8	5
PFIC2	49	45	44	40	26	19
PFIC2, BSEP subtype 1	13	10	10	8	7	6
PFIC2, BSEP subtype 2	36	34	32	30	19	13

BSEP, bile salt export pump; CI, confidence interval; FIC, familial intrahepatic cholestasis; n/N, patients meeting criteria out of all patients with available data at week 48; N, patients with available data at indicated time point; PFIC, progressive familial intrahepatic cholestasis.

Pruritus

- Proportion of PPAs at the patient level was comparable between PFIC2 and PFIC1 patients, although the PFIC1 group was smaller than the PFIC2 group (**Figure 4A**)
 - Over the interval of 0–48 weeks, mean proportions of PPAs were above 60% for patients with PFIC2 (including BSEP subtypes 1 and 2) and mean proportion of PPAs for patients with PFIC1 were 48%
 - Results for patients with PFIC2 by BSEP subgroup (BSEP1, milder disease presentation vs BSEP2, moderate to severe disease presentation[3]) did not demonstrate a clear difference on treatment effect given differences in sample size
- Patients had rapid improvements in pruritus with odevixibat that were sustained with treatment (**Figure 4B**)

Figure 4. Mean Proportion of PPAs Over 48 Weeks of Treatment (A) and Over Time (B) With Odevixibat: Subgroup Analysis by Genotype

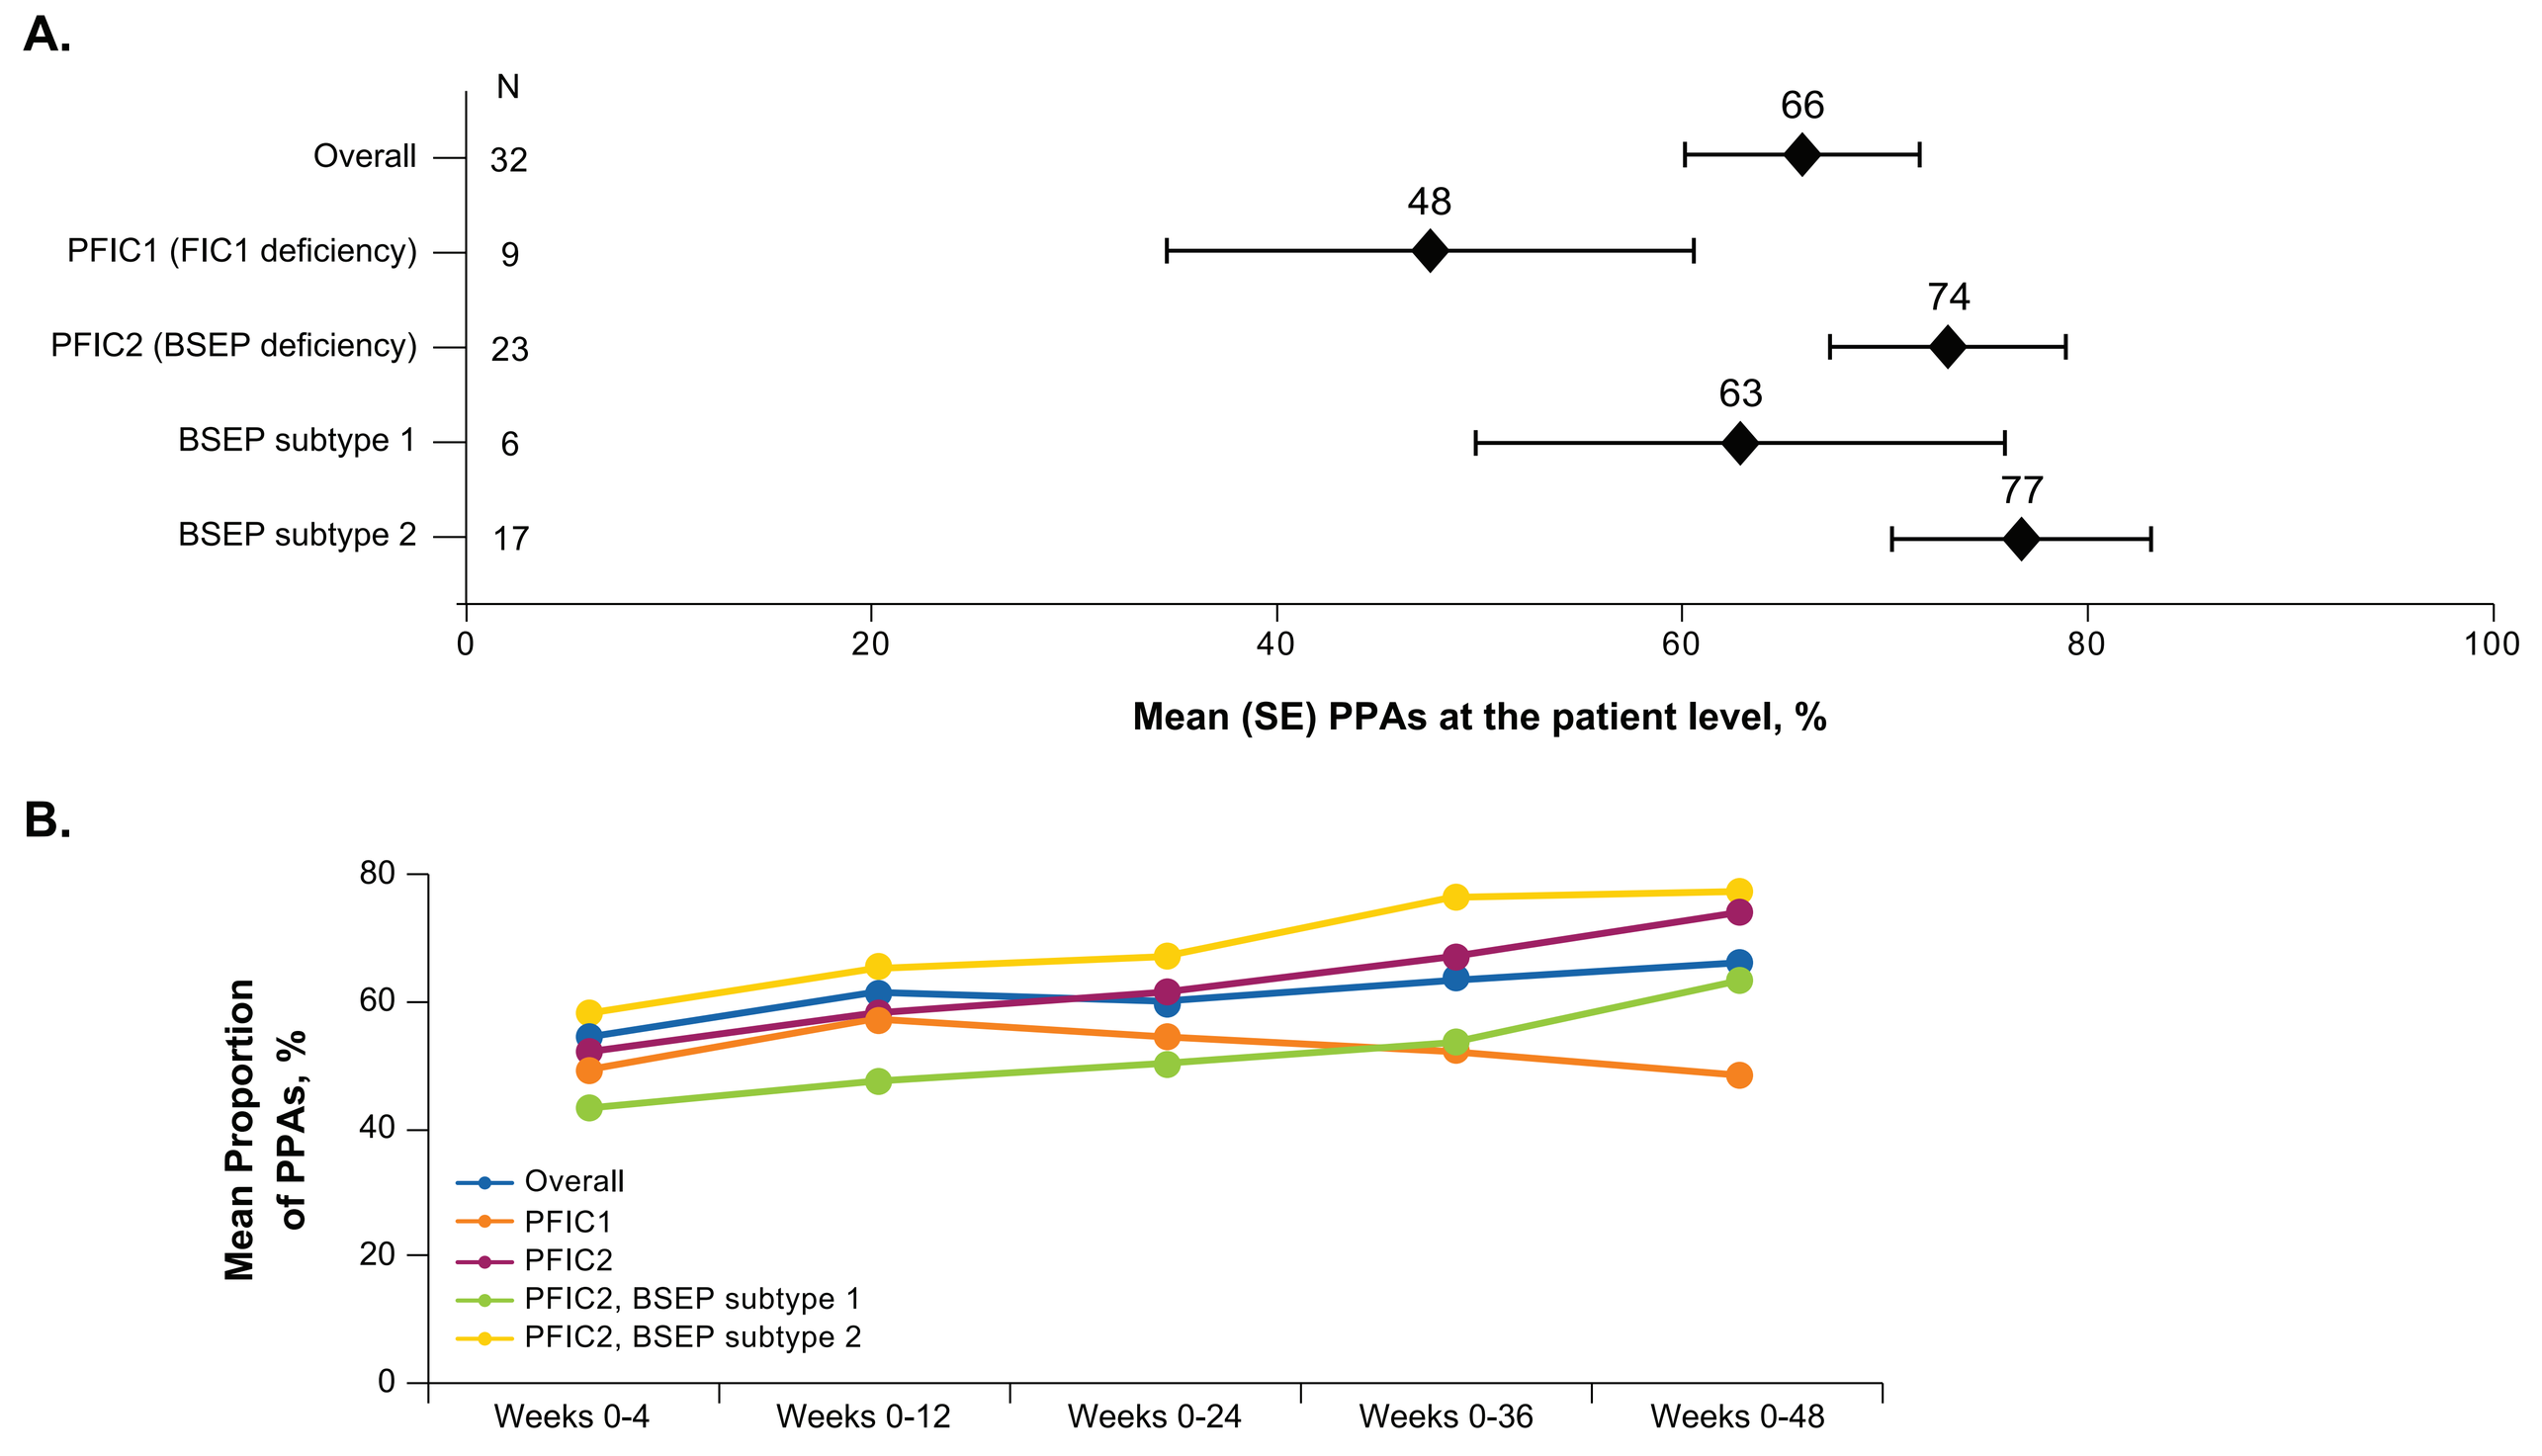

A.
N
Overall 32 66
PFIC1 (FIC1 deficiency) 9 48
PFIC2 (BSEP deficiency) 23 74
BSEP subtype 1 6 63
BSEP subtype 2 17 77
0 20 40 60 80 100
Mean (SE) PPAs at the patient level, %
B.
Mean Proportion of PPAs, %
80 60 40 20 0
Overall
PFIC1
PFIC2
PFIC2, BSEP subtype 1
PFIC2, BSEP subtype 2
Weeks 0-4 Weeks 0-12 Weeks 0-24 Weeks 0-36 Weeks 0-48
Patients at each time point, N

Patients at each time point, N	Weeks 0-4	Weeks 0-12	Weeks 0-24	Weeks 0-36	Weeks 0-48
Overall	75	70	56	46	32
PFIC1	20	17	15	13	9
PFIC2	49	47	39	33	23
PFIC2, BSEP subtype 1	13	12	8	8	6
PFIC2, BSEP subtype 2	36	33	29	23	17

BSEP, bile salt export pump; FIC, familial intrahepatic cholestasis; N, patients with available data at indicated time point; PFIC, progressive familial intrahepatic cholestasis; PPA, positive pruritus assessment; SE, standard error.

Safety

- Incidence of TEAEs in odevixibat-treated patients with PFIC1 or PFIC2 (80% each) and BSEP subtype 1 and 2 (77% and 86%, respectively; **Table 4**) was comparable to that of placebo-treated patients in PEDFIC 1 (85%)[7]
 - Most TEAEs mild or moderate, self-limiting, and considered by the investigator as not related to study drug
 - None of the reported serious TEAEs were considered related to odevixibat treatment

Table 4. Summary of TEAEs and Serious TEAEs by Patient Genotype Group

	Overall, N=77	Patients with PFIC1, n=20	Patients with PFIC2, n=51	Patients with PFIC2, BSEP subtype 1, n=13	Patients with PFIC2, BSEP subtype 2, n=36
Any TEAE, n (%)	61 (79)	16 (80)	41 (80)	10 (77)	31 (86)
TEAEs by preferred term occurring in ≥10% of patients overall, n (%)					
ALT increased	10 (13)	2 (10)	6 (12)	1 (8)	5 (14)
Blood bilirubin increased	12 (16)	4 (20)	6 (12)	2 (15)	4 (11)
Cough	12 (16)	4 (20)	8 (16)	2 (15)	6 (17)
Diarrhoea	15 (19)	5 (25)	10 (20)	1 (8)	10 (28)
Pruritus	9 (12)	3 (15)	6 (12)	3 (23)	3 (8)
Pyrexia	20 (26)	8 (40)	12 (24)	4 (31)	8 (22)
Upper respiratory tract infection	19 (25)	8 (40)	11 (22)	3 (23)	8 (22)
Vomiting	11 (14)	6 (30)	5 (10)	2 (15)	3 (8)
Any serious TEAEs, n (%)	7 (9)	1 (5)	6 (12)	1 (8)	5 (14)
Serious TEAEs by preferred term, n (%)					
Cholestasis	1 (1)	0	1 (2)	1 (8)	0
Dehydration[a]	1 (1)	1 (5)	0	0	0
Hyperbilirubinaemia	1 (1)	0	1 (2)	0	1 (3)
Liver function test increased[a]	1 (1)	1 (5)	0	0	0
Pancreatitis acute	1 (1)	0	1 (2)	0	1 (3)
Stoma complication	1 (1)	0	1 (2)	0	1 (3)
Supraventricular tachycardia	1 (1)	0	1 (2)	0	1 (3)
Urinary tract infection	1 (1)	0	1 (2)	0	1 (3)

[a]Occurred in the same patient.
ALT, alanine aminotransferase; BSEP, bile salt export pump; PFIC, progressive familial intrahepatic cholestasis; TEAE, treatment-emergent adverse event.

CONCLUSIONS

- Patients with PFIC1 or PFIC2 had substantial benefits with odevixibat when treated for up to 48 weeks
- Overall, at least 50% of patients treated with odevixibat in each subgroup achieved ≥70% reduction in serum bile acids from baseline or reached a serum bile acid level ≤70 µmol/L at week 48
- Mean proportions of PPAs, which were based on observer-reported outcome (ObsRO) instrument pruritus scores, were high across subgroups, ranging from 48% to 77% at week 48
- Long-term treatment with odevixibat was well tolerated, regardless of PFIC classification or BSEP subtype

REFERENCES

1. Henkel SA, et al. *World J Hepatol.* 2019;11:450-63. **2.** Bull LN, Thompson RJ. *Clin Liver Dis.* 2018;22:657-69. **3.** van Wessel DBE, et al. *J Hepatol.* 2020;73:84-93. **4.** Kamath BM, et al. *Liver Int.* 2020;40:1812-22. **5.** Graffner H, et al. *Aliment Pharmacol Ther.* 2016;43:303-10. **6.** Thompson RJ, et al. Presented at: Annual Meeting of the American Association for the Study of Liver Diseases; November 13-16, 2020. **7.** Thompson RJ, et al. Presented at: Annual Meeting of the American Association for the Study of Liver Diseases; November 13-16, 2020.

AUTHOR DISCLOSURES

R.J. Thompson: Albireo, Alnylam, Evox Therapeutics, Generation Bio, Mirum Pharma, Rectify Therapeutics, Retrophin, Qing Therapeutics, and Sana Biotechnology – Consultant
P. Horn, Q. Ni, P. Stein, C. Thompson, and **L. Kjems:** – Employed by Albireo Pharma, Inc.
R.H.J Houwen: GMP-Orphan and Univar – Consultant
F. Lacaille: Alexion – Consultant
M.E. Tessier: None
J. M. Vittorio: Mirum Pharma – Consultant

ACKNOWLEDGMENTS

This study was sponsored by Albireo. Medical writing and editorial assistance were provided by Peloton Advantage, LLC, an OPEN Health company, and was funded by Albireo Pharma, Inc.

Presented at the International Liver Congress; June 23–26, 2021 (Virtual)

Scan to download the poster
General hepatology
Richard Thompson
PO-1665
ILC2021