Preclinical validation of copper 64 as a translational tool for evaluating the pharmacodynamics of VTX-801 gene therapy in Wilson's disease

EASL
European Association for the Study of the Liver

Bernard Bénichou[1], Maria Collantes[2], Daniel Moreno[3], Margarita Ecay[2], Cristina Gazquez[3], Oihana Murillo[3], Veronica Ferrer[1], Ruben Hernadez-Alcoceba[3], Ivan Peñuelas[2], Gloria Gonzalez-Aseguinolaza[1,3]
[1]Vivet Therapeutics, Paris, France and Pamplona, Spain.
[2]Department of Nuclear Medicine, IdisNA, Clínica Universidad de Navarra, Avda. Pío XII, 31080, Pamplona, Spain.
[3]Gene Therapy and Regulation of Gene Expression Program, IdisNA, FIMA. University of Navarra. Pamplona, Spain.

Vivet THERAPEUTICS

1 INTRODUCTION

Wilson's disease is an autosomal recessive metabolic defect of hepatocyte copper excretion into the bile, caused by absent or reduced ATP7B copper transporter function. The ATP7B transporter has a dual role: it transports copper into the trans-Golgi compartment for incorporation into the plasma protein ceruloplasmin, and into the bile for excretion of excess copper stores [1]. Recently, we have demonstrated that the administration of an adeno associated vector (AAV) encoding a mini version of the human *ATP7B* cDNA (AAV-miniATP7B = **VTX-801**) provides long-term correction of copper metabolism in Wilson's disease (WD) mice [2].

2 AIM

In anticipation of a future gene therapy clinical trial, we considered the value of using biliary copper excretion as a pharmacodynamic biomarker [3,4]. Since direct collection of bile may be invasive, we have evaluated the excretion of radiocopper (^{64}Cu) into the faeces as an alternative but relevant biomarker in AAV-miniATP7B-treated WD mice.

3 METHOD

Male and female WD mice were injected with VTX-801 at 6 weeks of age. Three months later, $^{64}CuCl_2$ was injected intravenously in treated mice as well as in control mice of the same age (untreated 18-week-old *wild-type* (WT), *Atp7b +/-* (HE) and *Atp7b$^{-/-}$* (WD) mice). Abdominal PET analyses were performed 24 and 48h later. 24h faeces, 24h urine and serial blood samples were collected over a period of 72h post radiocopper injection, at which time animals were sacrificed and organs collected (liver, kidney, lung, brain and spleen); the radioactive signal was then measured for each biological sample in a gamma counter.

4 RESULTS

Experimental groups	N=	Treatment
Atp7b$^{+/+}$ (WT)	3M/3F	Saline
Atp7b$^{+/-}$ (HE)	3M/3F	Saline
Atp7b$^{-/-}$ (WD)	3M/3F	Saline
Atp7b$^{-/-}$ (WD)	3M/3F	VTX-801 $5x10^{12}$ vg/kg

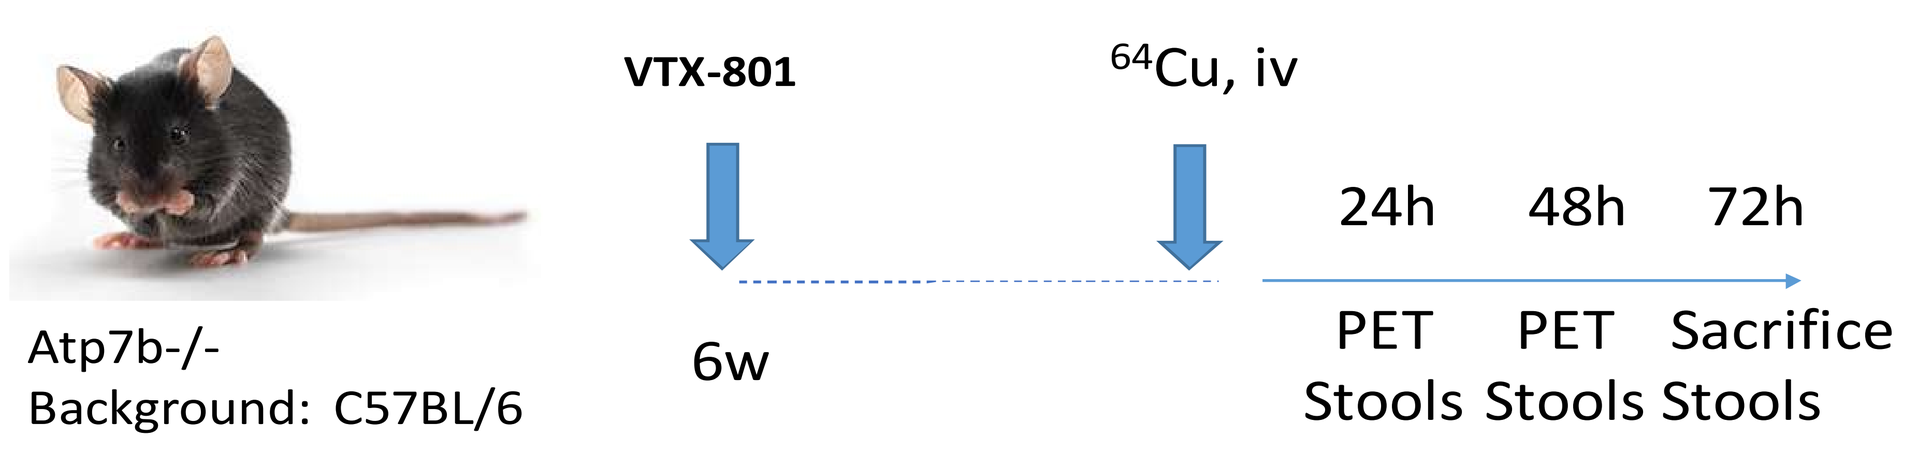
Atp7b-/-
Background: C57BL/6
VTX-801
6w
^{64}Cu, iv
24h
48h
72h
PET
PET
Sacrifice
Stools
Stools
Stools

Figure 1: Experimental design. Animals received an intravenous (i.v.) dose of $5x10^{12}$ vg/kg of VTX-801 (or an equivalent volume of saline solution) at 6 weeks of age. Three months later, animals were injected (i.v.) with ^{64}Cu and subjected to the following analyses: at 24, 48 and 72 hours post ^{64}Cu injections, stools were harvested and radioactivity was measured. At 24 and 48 hours post ^{64}Cu injections, biodistribution was analysed by PET and at 72 hours after ^{64}Cu administration, animals were sacrificed and radioactivity was measured in different organs. As controls, untreated 18-week old WT, HE and WD mice received a ^{64}Cu iv injection and the same procedure was followed.

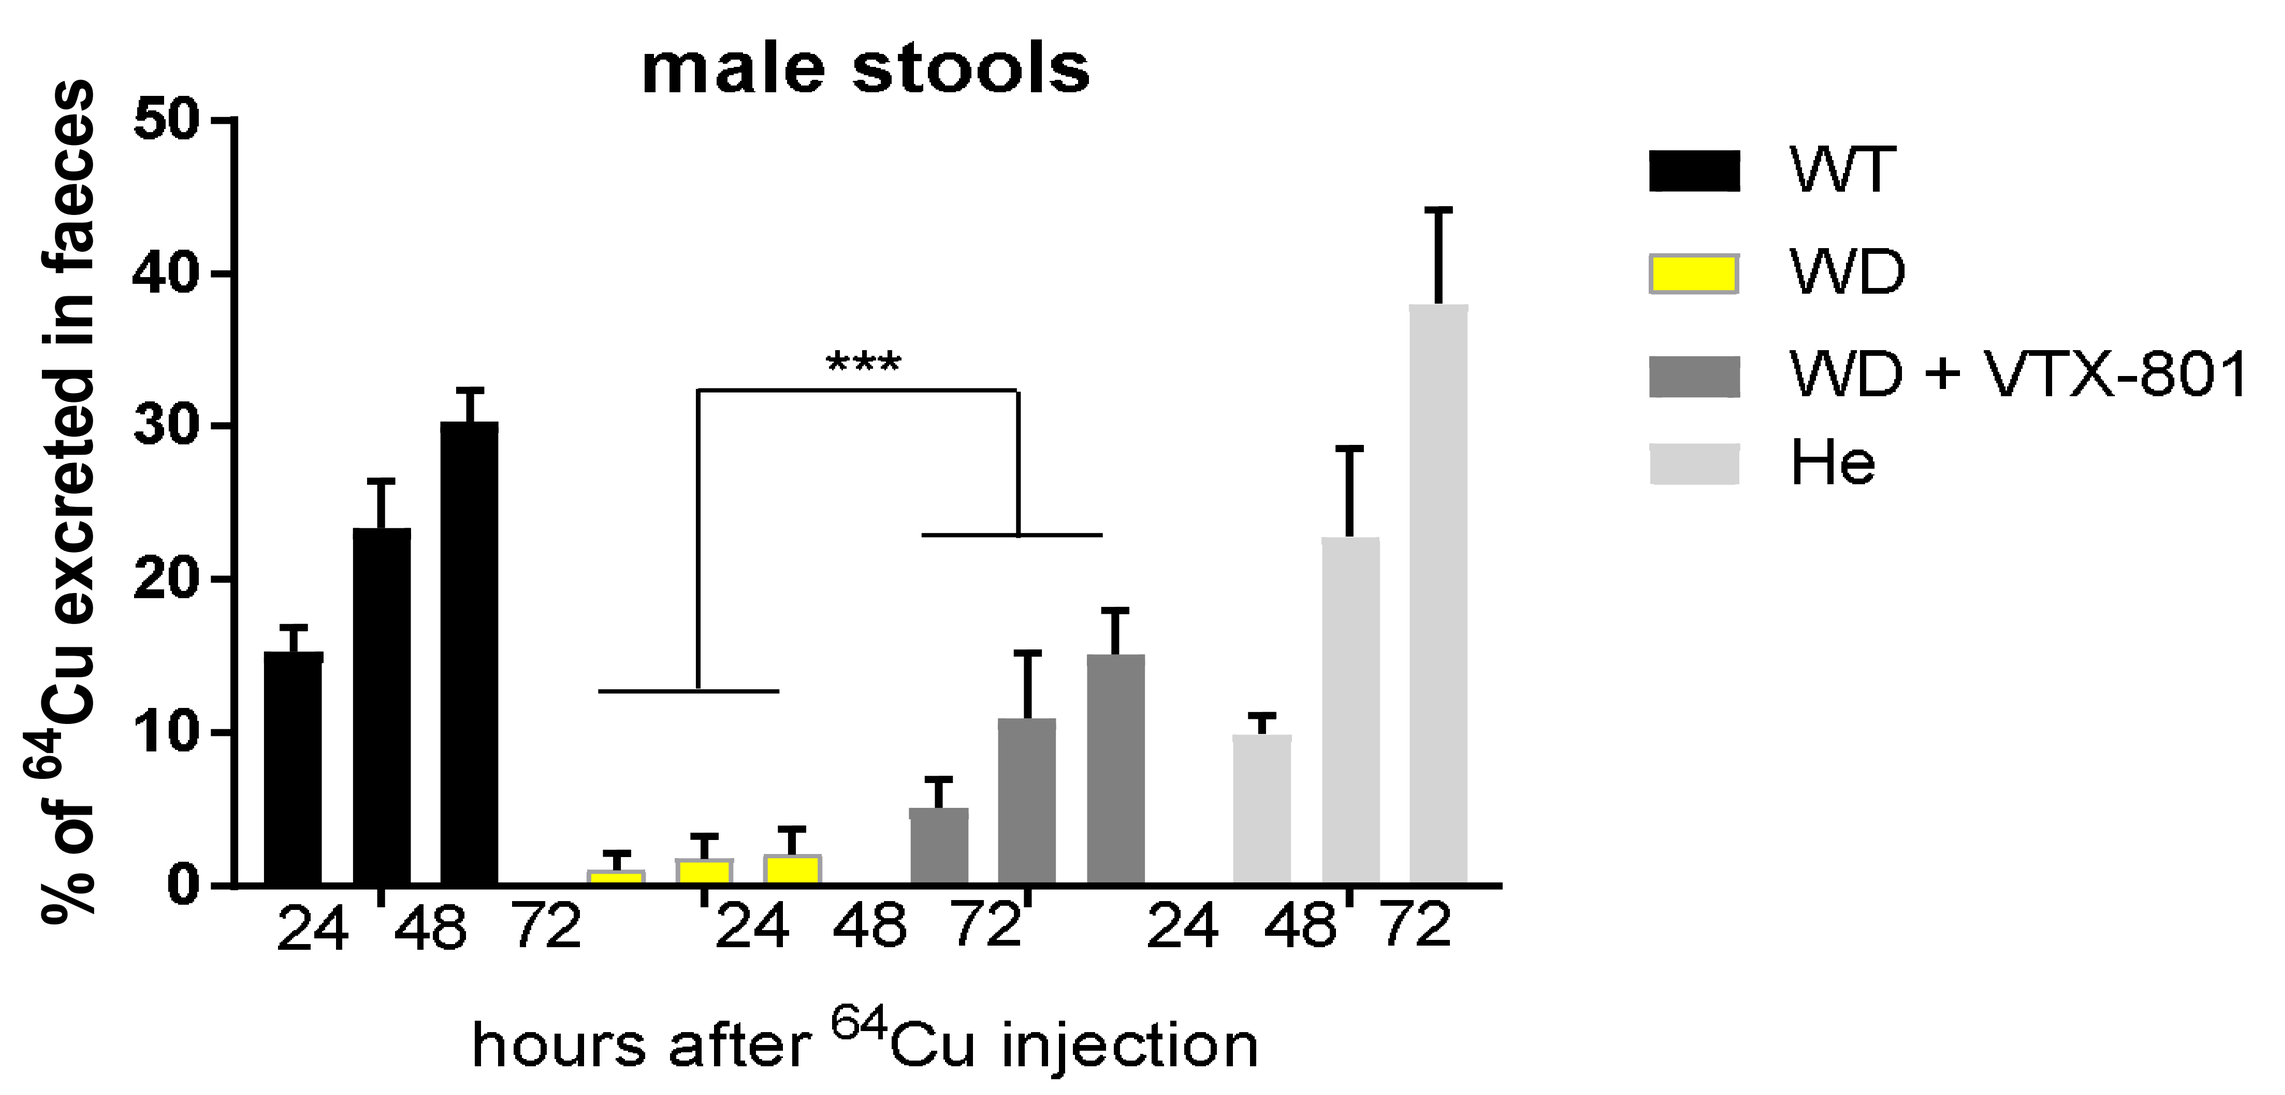
male stools
% of ^{64}Cu excreted in faeces
50
40
30
20
10
0

24 48 72 24 48 72 24 48 72
hours after ^{64}Cu injection
WT
WD
WD + VTX-801
He

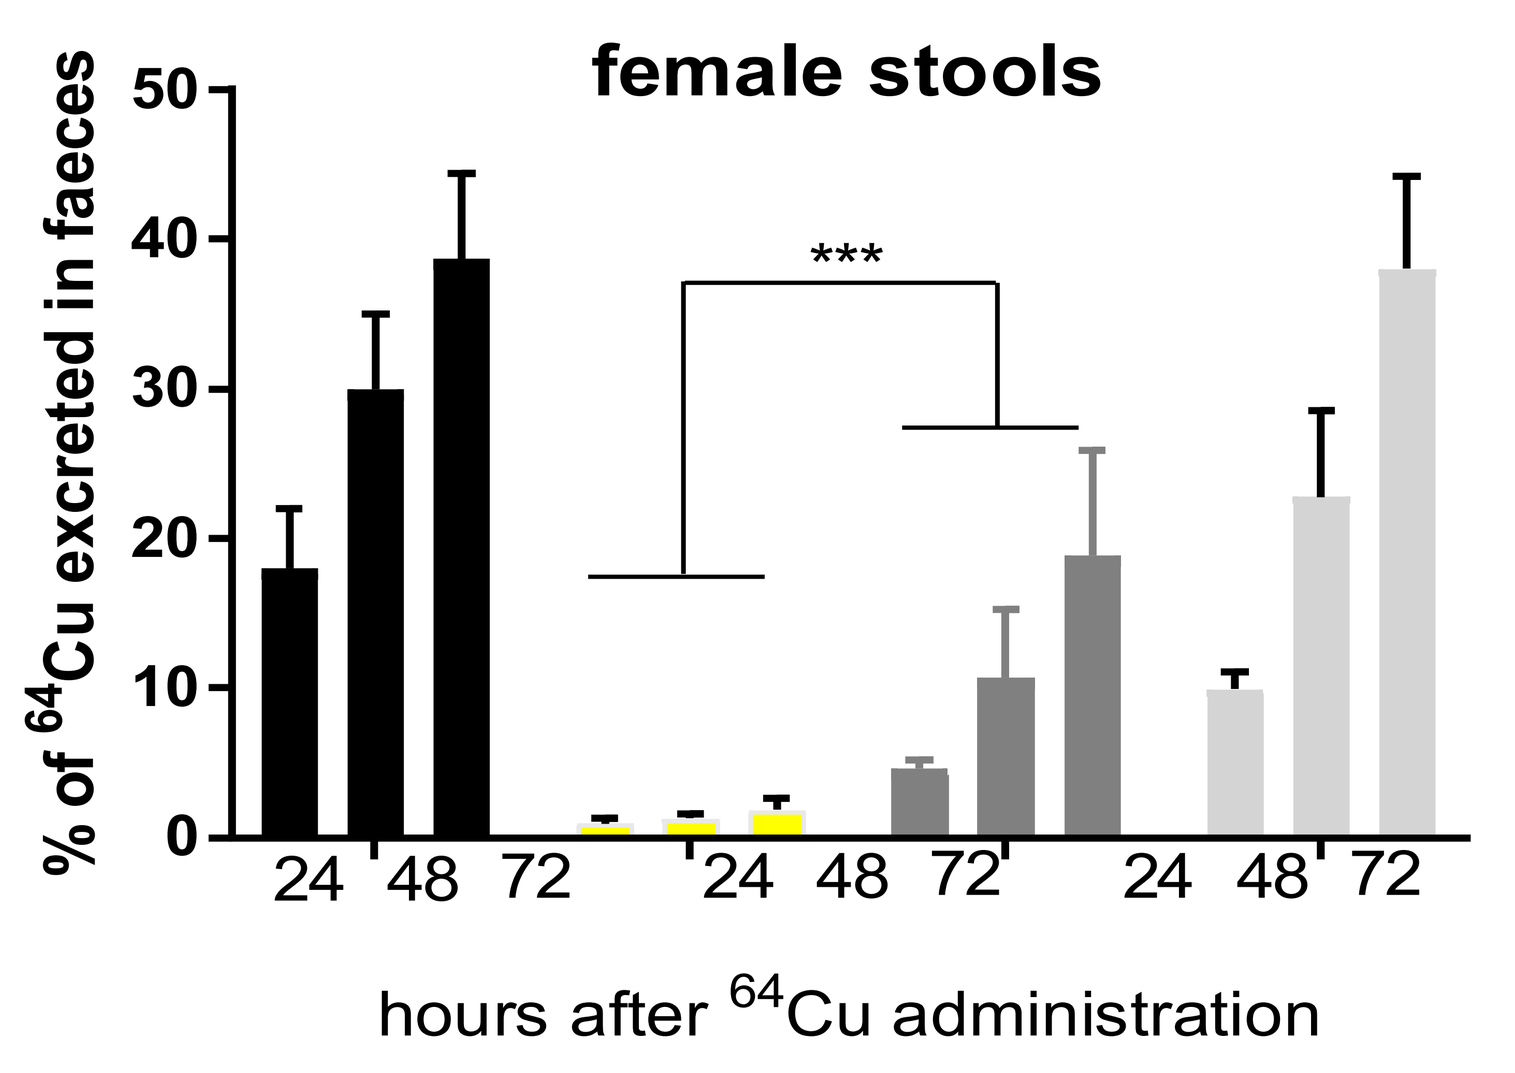
female stools
% of ^{64}Cu excreted in faeces
50
40
30
20
10
0

24 48 72 24 48 72 24 48 72
hours after ^{64}Cu administration

Figure 2: Cumulative percentage of injected ^{64}Cu recovered in the stools of animals 3 months post-VTX-801 injection. Faecal ^{64}Cu excretion rates in animals treated with $5x10^{12}$ vg/kg - a dose that has been demonstrated to be therapeutic and to increase survival in WD mice – were 50% of the values in healthy WT and heterozygous animals.

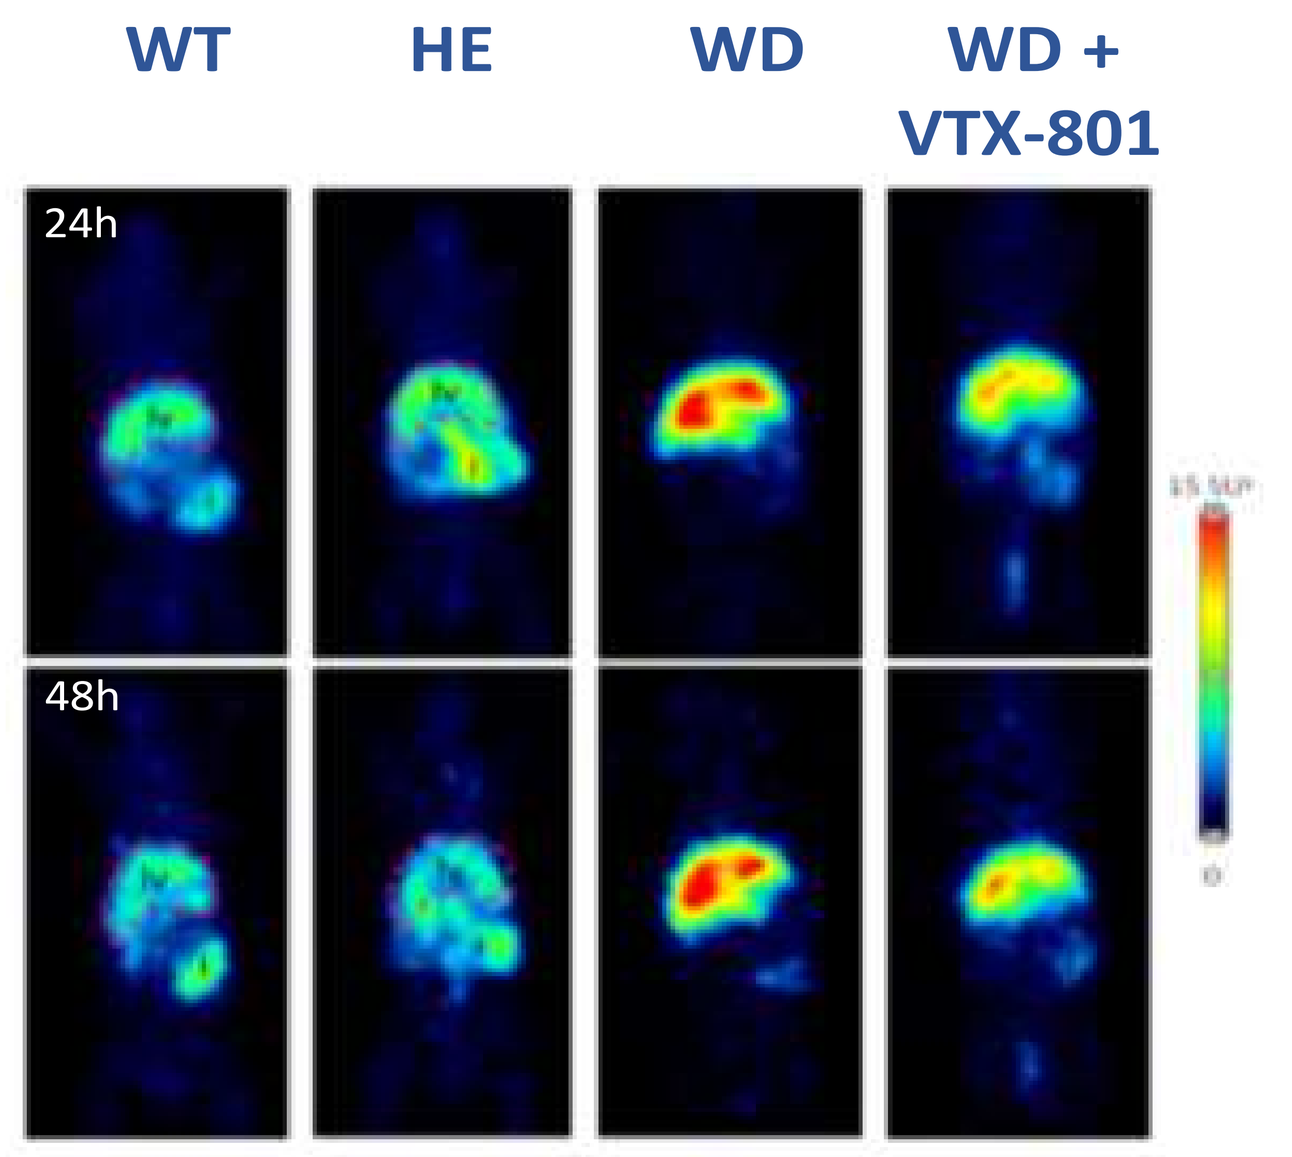
WT
HE
WD
WD + VTX-801
24h
48h

Figure 3: Abdominal PET analysis 24 and 48 hours after ^{64}Cu injection. In WT and HE animals, ^{64}Cu was detected both in the liver and intestine, while in WD animals ^{64}Cu was clearly retained in the liver. In animals treated with a therapeutic dose of VTX-801, radioactivity was detectable in the intestine, consistent with the restoration of fecal copper excretion. Lv:liver, i:intestine.

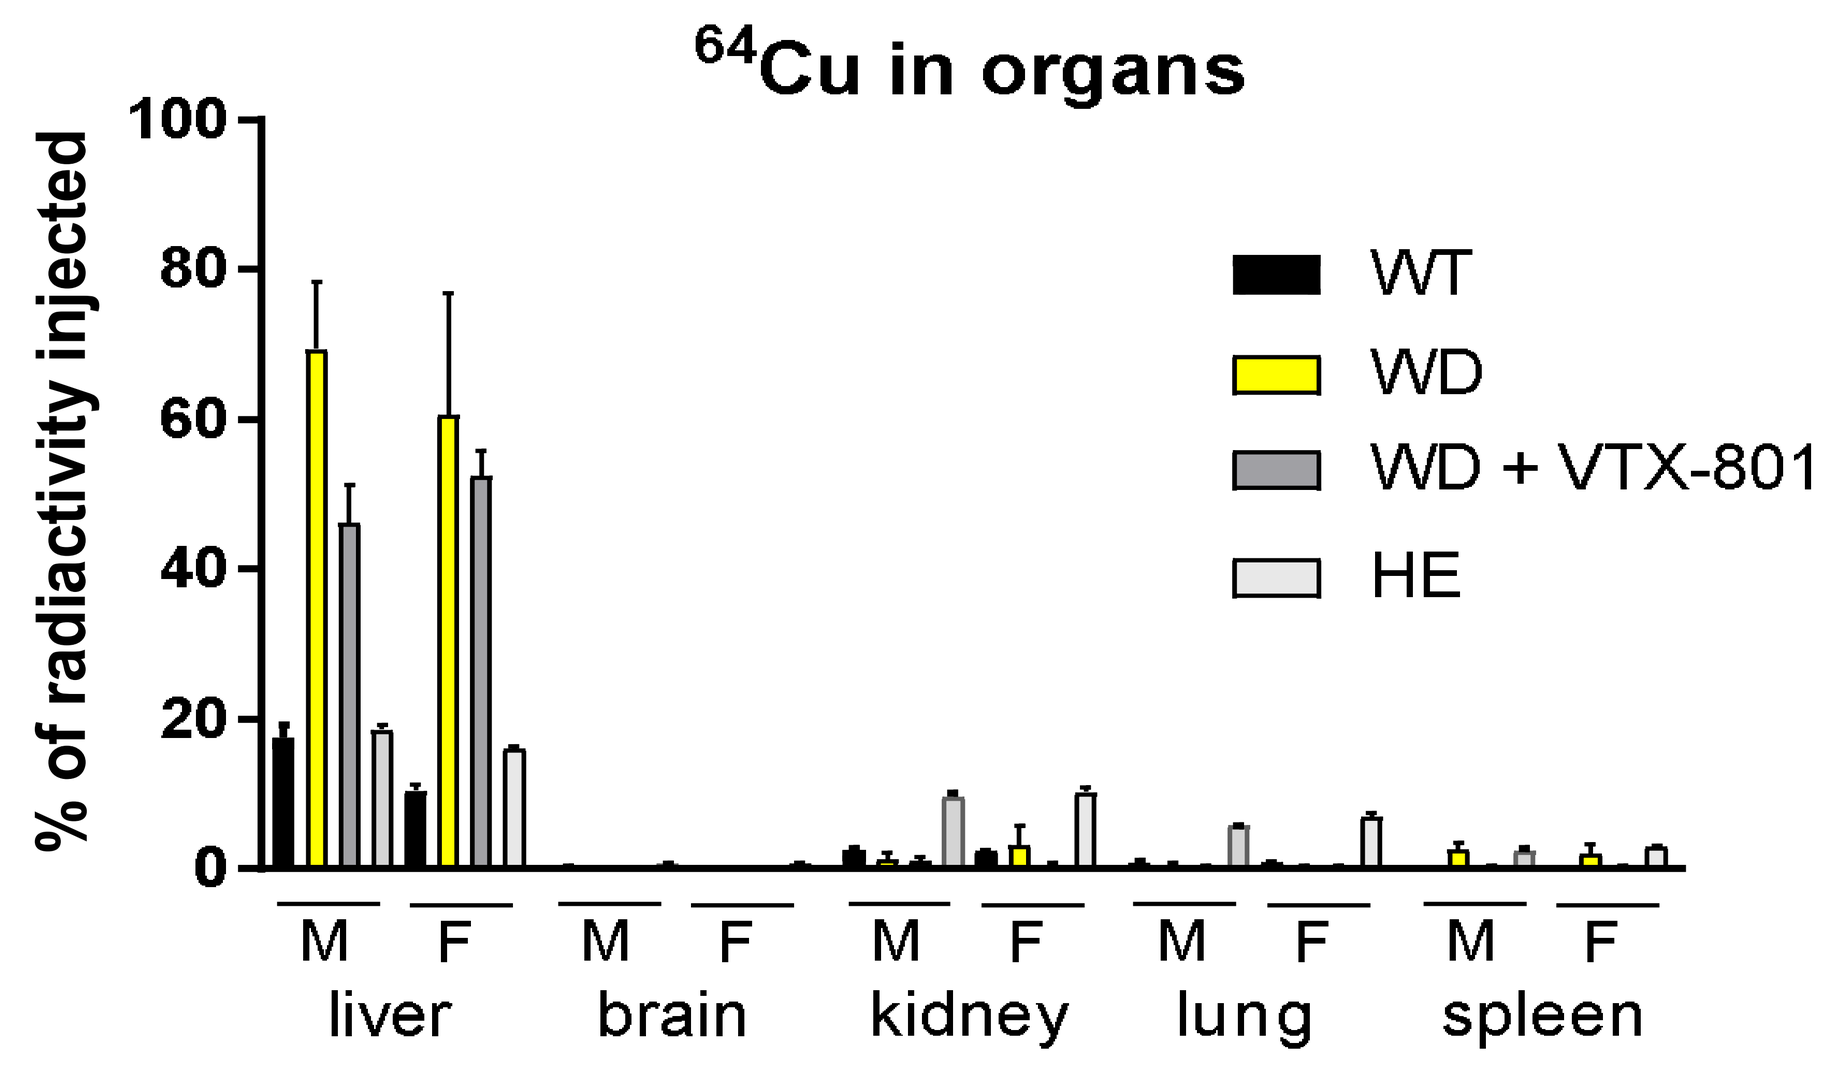
^{64}Cu in organs
% of radiactivity injected
100
80
60
40
20
0
M F
liver
M F
brain
M F
kidney
M F
lung
M F
spleen
WT
WD
WD + VTX-801
HE

Figure 4: Percentage of total radioactivity present in liver, brain, kidney, lung and spleen of 18 week-old mice at 72 hours post- ^{64}Cu injection.

5 CONCLUSIONS

Faecal radiocopper excretion was significantly higher in control mice in comparison to WD mice, in which a signal was barely detectable. In VTX-801 treated WD animals, faecal ^{64}Cu elimination was restored. Finally, the radioactive signal in liver was much higher in untreated WD mice compared to controls, and it was reduced in VTX-801 treated WD mice.

In conclusion, faecal excretion of radiocopper represents a very promising pharmacodynamic endpoint to evaluate the therapeutic efficacy of VTX-801 gene supplementation in WD patients.

6 ACKNOWLEDGEMENTS

Infrastructure and technical support was provided by the morphology unit and the animal facility staff at CIMA. The authors acknowledge Africa Vales and Cristina Olagüe for excellent technical support.

7 REFERENCES

[1] Bandmann O, Weiss KH, Kaler SG. Wilson's disease and other neurological copper disorders. Lancet Neurol. 2015 Jan;14(1):103-13.
[2] Murillo O, Moreno D, Gazquez C, et al. Liver expression of a miniATP7B gene results in long-term restoration of copper homeostasis in a Wilson's disease model. Hepatology. 2019
[3] Peng F, Lutsenko S, Sun X, Muzik O. Positron emission tomography of copper metabolism in the Atp7b-/- knock-out mouse model of Wilson's disease. Mol Imaging Biol. 2012 Feb;14(1):70-8.
[4] Członkowska A, Rodo M, Wierzchowska-Ciok A, Smolinski L, Litwin T. Accuracy of the radioactive copper incorporation test in the diagnosis of Wilson disease. Liver Int. 2018 Oct;38(10):1860-1866.

8 CONTACT

Vivet Therapeutics, Rue Tronchet 29, Paris.
ggonzalez@vivet-therapeutics.com
bbenichou@vivet-therapeutics.com

PosterSessionOnline.com
Poster presented at: EASL The Home of Hepatology | THE INTERNATIONAL LIVER CONGRESS™ 2019
DOI: 10.3252/pso.eu.ILC2019.2019
General hepatology
Gloria González-Aseguinolaza
FRI-430
ILC2019