BSH 2020 VIRTUAL
9 -14 NOVEMBER

60 1960–2020 British Society for Haematology Listening • Learning • Leading

The Impact of Immune Thrombocytopenia (ITP) on Activities of Daily Living: Findings reported by UK patients and physicians from the ITP World Impact Survey (I-WISh)

N.COOPER[1], D. PROVAN[2], M. MORGAN[3], T. BAILEY[4], B.TAYLOR[5]
1 Hammersmith Hospital, Imperial College, London, UK
2 Barts and The School of Medicine and Dentistry, London, UK
3 ITP Support Association, Bedfordshire, UK
4 Adelphi Real World, Bollington, Cheshire, UK
5 Novartis Pharmaceuticals, London, UK

NOVARTIS | Reimagining Medicine

INTRODUCTION

- Immune Thrombocytopenia is an autoimmune disease which affects up to 4,000 patients in the UK[1] and is characterised by a reduced platelet count (clinically defined as a platelet count of less than 100 x 109/L)[2]
- There are a number of first- and second-line treatments available for ITP. Treatment strategies aim to achieve a platelet count which is associated with preventing spontaneous bleeding[3] and ultimately improve quality of life (QoL)
- The I-WISh study capture patient and physician perceptions on how ITP impacts patients' activities of daily living

AIMS & METHODS

- The ITP World Impact Survey (I-WISh) is a cross-sectional survey of ITP patients and treating haematologists across thirteen countries. Overall, 1507 patients and 472 physicians participated in the study between December 2017 and August 2018
- Patients were recruited via both the ITP Support Association UK and by treating physicians. Eligible patients were aged 18 or over at point of data capture and were diagnosed with ITP
- As part of the survey, patients were asked to complete the ITP Life Quality Index (ILQI) which assessed the burden of their disease on various aspects of their daily life. Physicians were asked similar questions pertaining to their perception of how ITP impacted patients' ability to perform activities of daily living, including impact on physical activity, sex life and travel plans
- A steering committee of expert physicians and patient advocacy ITP specialists designed and endorsed the survey materials
- Here we present data from UK patients and physicians
- Impact statements on emotional burden and activities of daily living were asked using a 7-point Likert scale with high agreement and satisfaction scores = 5–7

RESULTS

Demographics

- 31 physicians and 120 patients participated in the UK. Mean (SD) patient age was 55 (14.17) years and over two thirds of patients were female (68%)
- On average, patients reported to having been diagnosed for 11.4 (10.25) years
- Over two thirds (68%) of patients reported their current health state to be ≥ 5 using a 7-point Likert scale (7 is excellent health)
- Physicians were primarily specialised in either haematology or haematology-oncology and had reported a mean ITP caseload of 47.5 (39.22). The majority of physicians were based in university/teaching hospitals (77%) versus regional/community hospitals (23%)
- Patient and physician demographics are summarised in **table 1**

Table 1. Respondent demographics

Patient Demographics	Patients (n=120)
Age, mean (SD)	55.0 (14.17)
Gender: female, n (%)	
Years since diagnosis, mean (SD)	11.4 (10.25)
Self-reported current health state (7 is excellent health)	
Score of 1–3, n (%)	11 (9)
Score of 4, n (%)	27 (23)
Score of 5–7, n (%)	81 (68)
Physician Demographics	**Physicians (n=31)**
Primary specialty	
Haematology, n (%)	27 (87)
Haematology-Oncology, n (%)	4 (13)
Current ITP caseload, mean (SD)	47.5 (39.22)
Primary setting	
University/Teaching hospital	24 (77)
Regional/Community hospital	7 (23)

Impact on activities of daily living

- **Figure 1a** shows the proportion of patients who indicated ITP at least 'sometimes' impacts energy levels, capacity to exercise, ability to perform daily activities and sexual activity. **Figure 1b** displays the proportion of physicians who perceive high impact of ITP in similar activities
- The majority of patients (84%) reported that ITP at least 'sometimes' impacts their energy levels versus two thirds of physicians (64%) reported a high impact of fatigue
- Nearly three quarters (73%) of patients reported ITP to impact capacity to exercise at least 'sometimes', aligning with 70% of physicians who indicated a high impact
- Patients and physicians agreed on the impact of ITP on activities of daily living. 68% of patients reported that ITP impacted them undertaking everyday tasks while 67% of physicians recognised a similar impact

Figure 1a. Patient-reported impact of ITP on activities of daily living

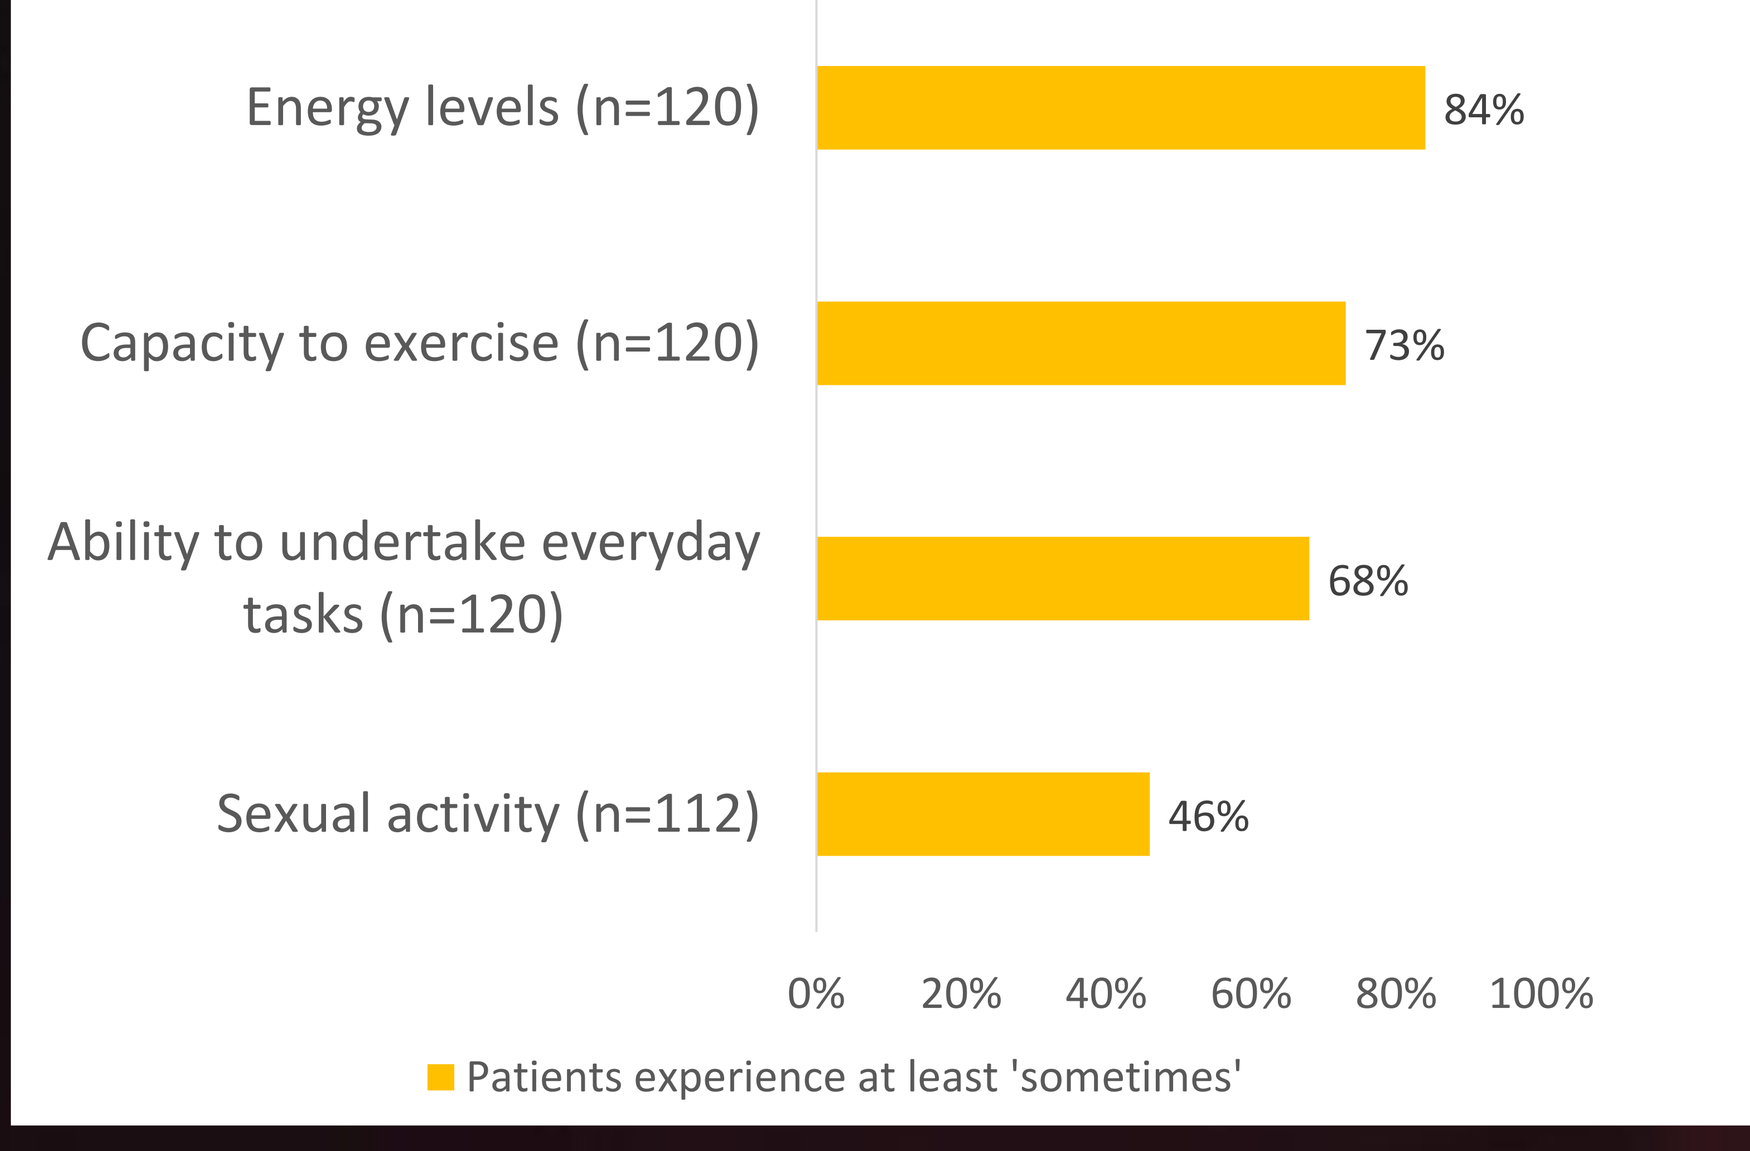
Energy levels (n=120) 84%
Capacity to exercise (n=120) 73%
Ability to undertake everyday tasks (n=120) 68%
Sexual activity (n=112) 46%
0% 20% 40% 60% 80% 100%
Patients experience at least 'sometimes'

Figure 1b. Physician-reported impact of ITP on activities of daily living

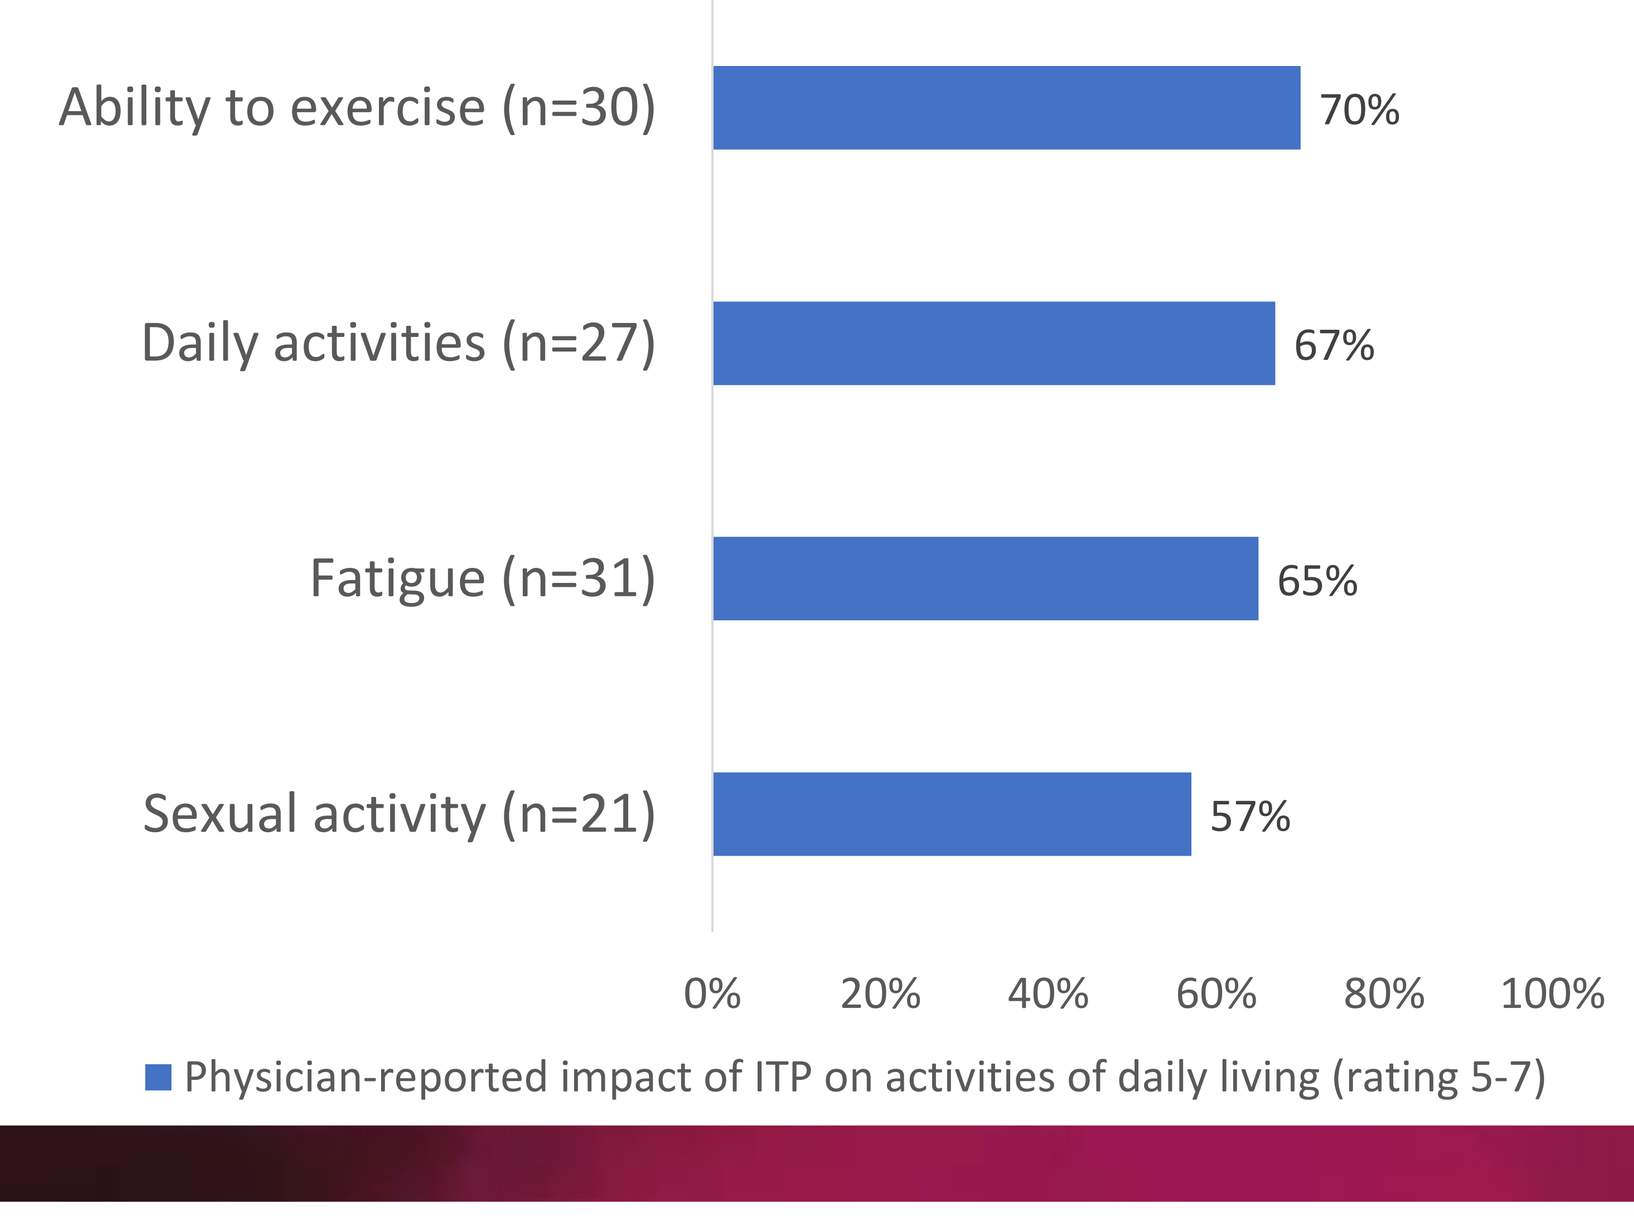
Ability to exercise (n=30) 70%
Daily activities (n=27) 67%
Fatigue (n=31) 65%
Sexual activity (n=21) 57%
0% 20% 40% 60% 80% 100%
Physician-reported impact of ITP on activities of daily living (rating 5-7)

Impact on travel plans

- Overall, at least two thirds of physicians reported ITP to have a high impact (rating 5–7) on each aspect of travel shown in **figure 2**. The leading travel-related impacts reported by physicians were concerns around the need for travel insurance (84%) and concerns around increased risk of bleeding (81%)

Impact on ability to work

- Overall, just over half (59%) of the patient sample were in work at the point of data capture, either full-time (33%) or part-time (26%) **(figure 3)**
- Of those patients in employment, patients reported to have worked an average of 28.5 hours in the past 7 days, missing 2.2 hours on average due to their ITP
- 16% of those patients 'currently' working reported ITP to have a high impact (score ≥ 5) on their productivity at work, while 24% of patients reported ITP to have a high impact on their productivity for regular activities **(figure 4)**
- **Figure 5** shows the direct effect of ITP on patients' working life. The top three impacts reported by patients were 'seriously considered reducing hours at work' (46%), 'reducing hours at work' (34%) and 'considered terminating job' (31%)

Figure 2. Physician-reported impact of ITP on travel plans (n=31)

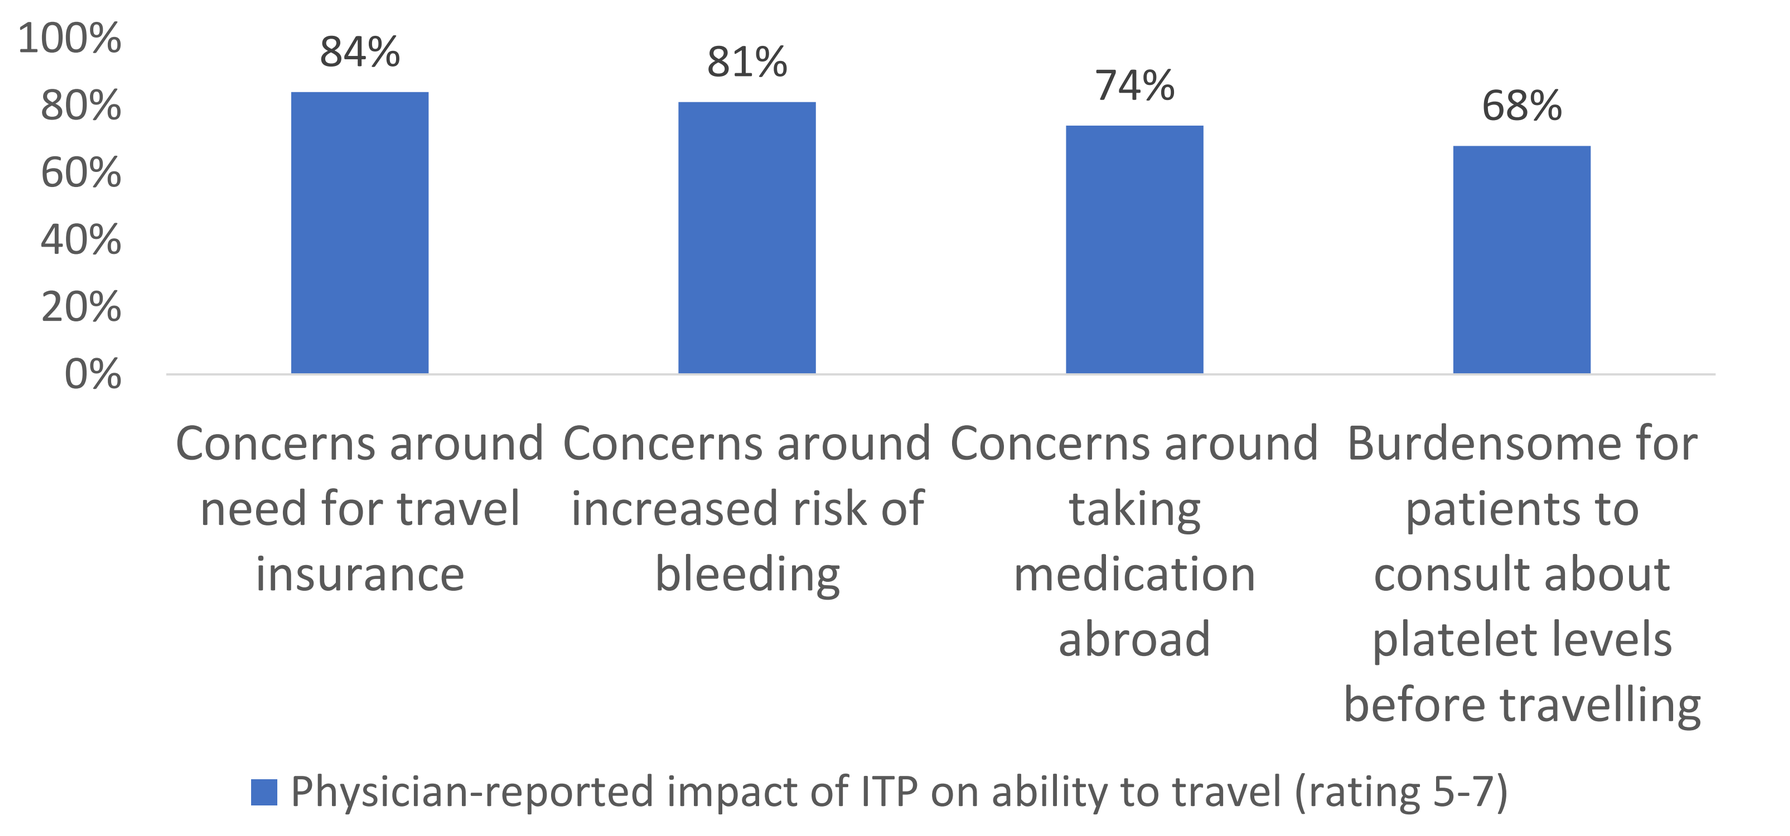
100% 80% 60% 40% 20% 0%
84% 81% 74% 68%
Concerns around need for travel insurance
Concerns around increased risk of bleeding
Concerns around taking medication abroad
Burdensome for patients to consult about platelet levels before travelling
Physician-reported impact of ITP on ability to travel (rating 5-7)

Figure 4. Patient-reported impact of ITP on productivity

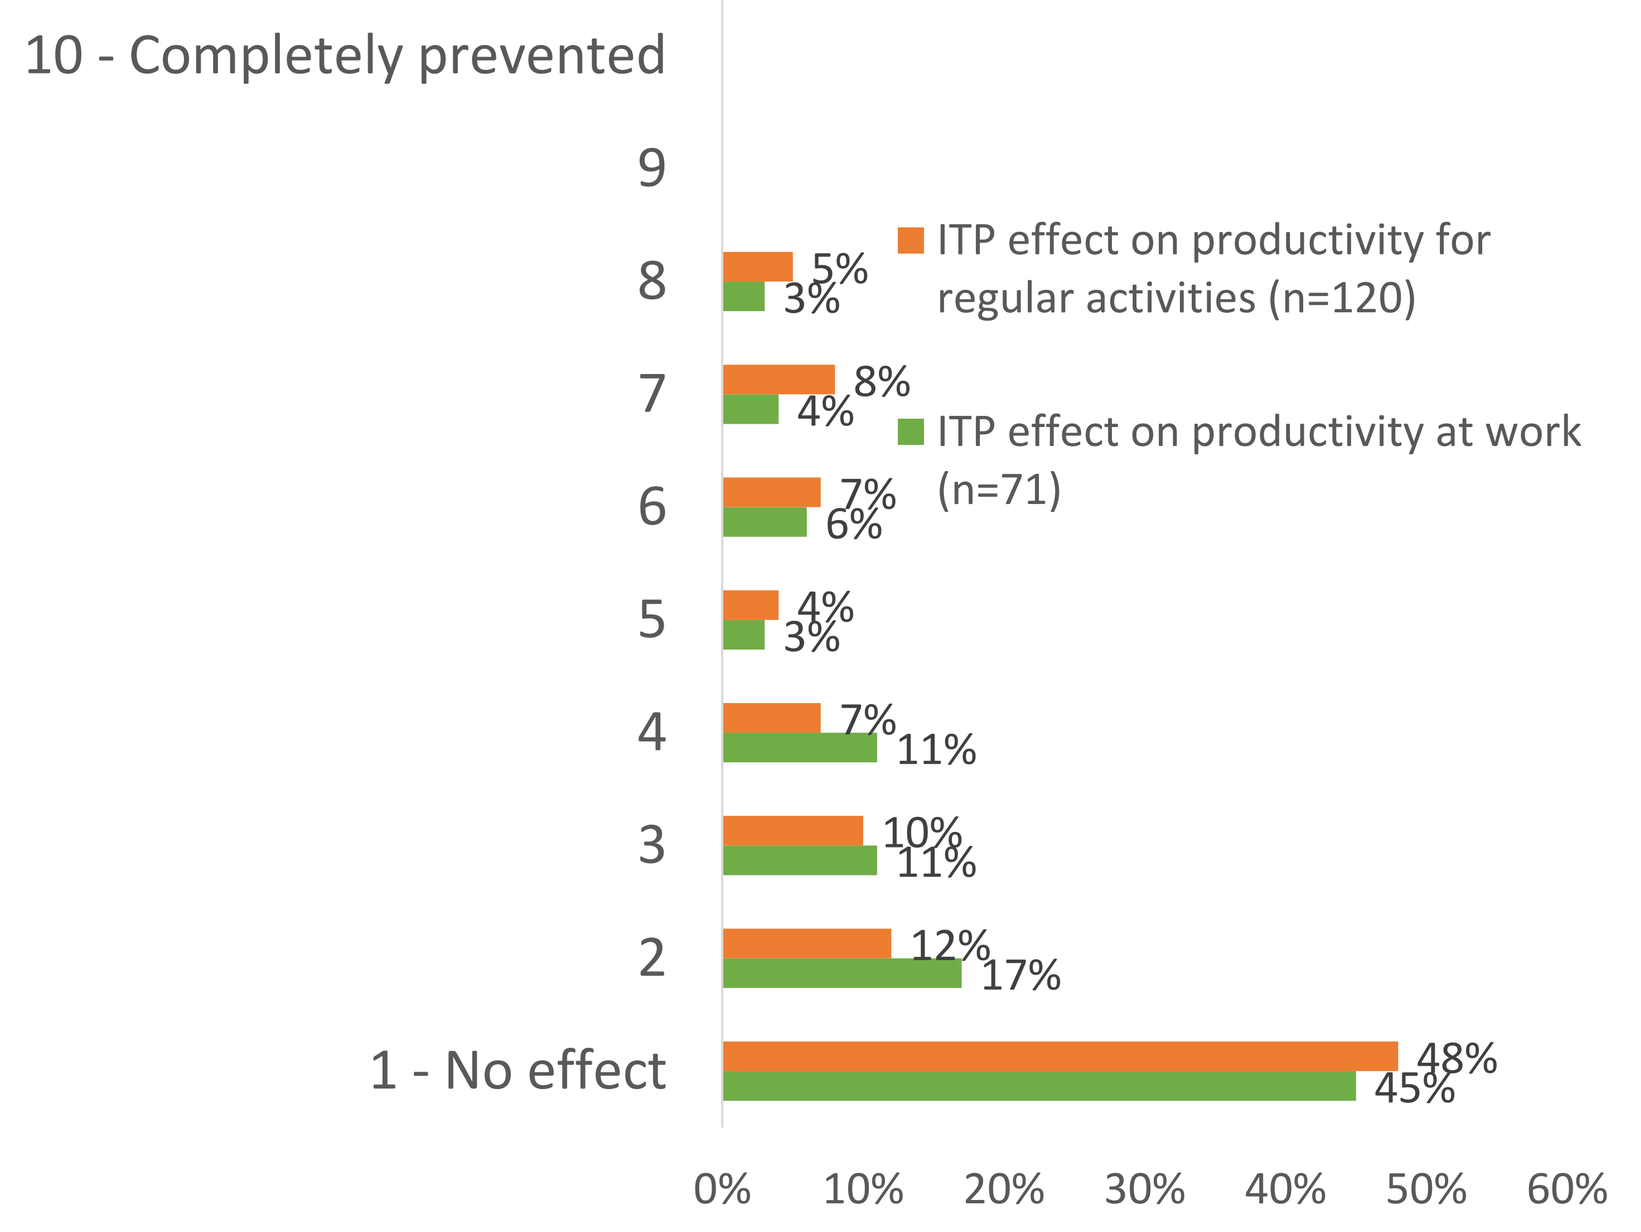
10 - Completely prevented
9
8 5% 3%
7 8% 4%
6 7% 6%
5 4% 3%
4 7% 11%
3 10% 11%
2 12% 17%
1 - No effect 48% 45%
0% 10% 20% 30% 40% 50% 60%
ITP effect on productivity for regular activities (n=120)
ITP effect on productivity at work (n=71)

Figure 3. Patient employment status at point of data capture

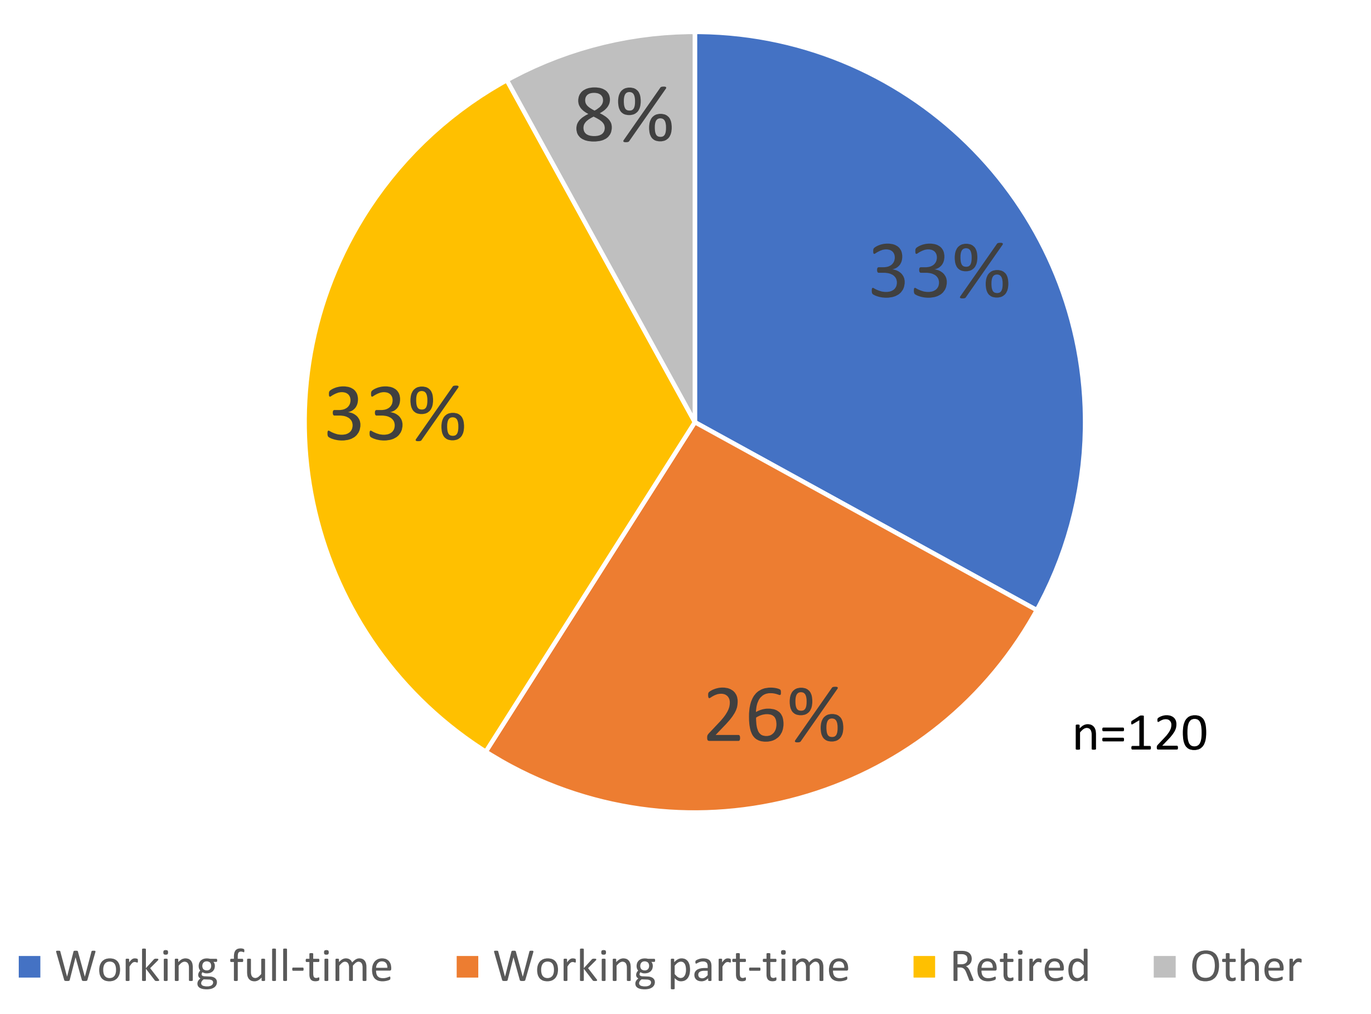
33%
26%
33%
8%
n=120
Working full-time
Working part-time
Retired
Other

Figure 5. Patient-reported impact of ITP on working life

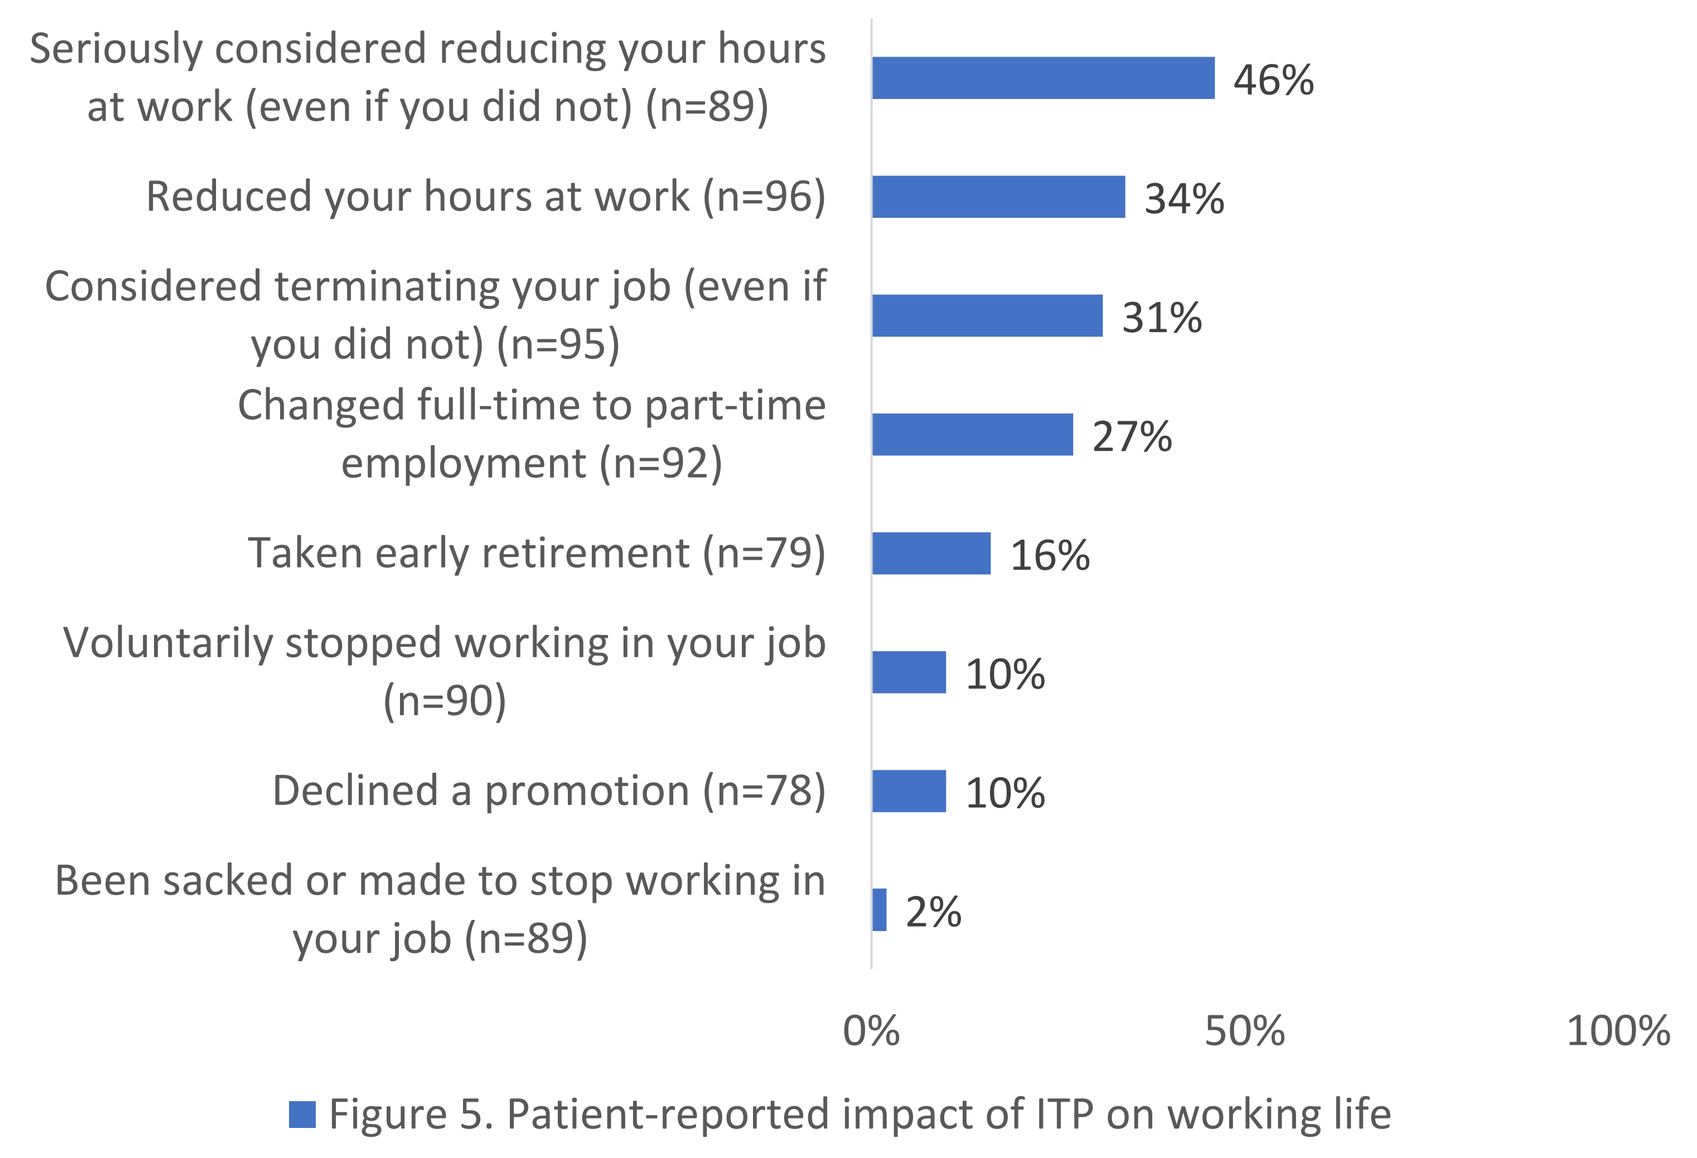
Seriously considered reducing your hours at work (even if you did not) (n=89) 46%
Reduced your hours at work (n=96) 34%
Considered terminating your job (even if you did not) (n=95) 31%
Changed full-time to part-time employment (n=92) 27%
Taken early retirement (n=79) 16%
Voluntarily stopped working in your job (n=90) 10%
Declined a promotion (n=78) 10%
Been sacked or made to stop working in your job (n=89) 2%
0% 50% 100%
Figure 5. Patient-reported impact of ITP on working life

CONCLUSIONS

- Overall, there is alignment between patients and physicians on the impact of ITP on patients' ability to perform activities of daily living, with reduced energy levels being reported by the vast majority of patients
- Patients also reported a considerable impact of ITP on their productivity, with a third of patients reporting to have reduced work hours due to their disease and over a quarter changing from a full-time to a part-time role
- These data highlight the daily burden of living with ITP which impact patients' ability to perform routine activities and work, and demonstrate the need for greater disease control

ACKNOWLEDGEMENT

- We thank the patients, caregivers, and physicians who have participated in I-WISh, and Adelphi Real World for executing the survey, collating data and running analyses.
- We thank the ITP Support Association UK for supporting with patient recruitment
- We thank our I-WISh Steering Committee.
- This study was sponsored by Novartis Pharmaceuticals Corporation. Medical writing assistance was provided by Tom Bailey from Adelphi Real World and funded by Novartis Pharmaceuticals Corporation.

REFERENCES

1. Immune Thrombocytopenia (2016). [PDF] Guy's and St Thomas' NHS Foundation Trust, p.2–4. Available at: https://www.guysandstthomas.nhs.uk/resources/patient-information/haematology/Immune-thrombocytopenia-web-friendly.pdf [Accessed 18 Dec. 2019].
2. Trotter P, Hill AQ. Immune thrombocytopenia: improving quality of life and patient outcomes. *Patient Relat Outcome Meas 2018; 9: 369–384*
3. Nomura S. Advances in Diagnosis and Treatments for Immune Thrombocytopenia. *Clin Med Insights Blood Disord 2016; 9: 15-22*

CONTACT INFORMATION

Please contact Barbara Taylor:
barbara-1.taylor@novartis.com

BSH2020 PO-035-B
General Haematology
Tom Bailey
Poster presented at: British Society for Haematology Listening • Learning • Leading
PosterSessionOnline.com