BSH 2020 VIRTUAL
9 -14 NOVEMBER

60 1960–2020 British Society for Haematology Listening • Learning • Leading

Whose choice is it anyway? A multi-national and multi-stakeholder perspective on shared treatment decision making in older adults with AML

Nigel H. Russell,[1] Thomas W. LeBlanc,[2] Loriana Hernandez-Aldama,[3] Charlotte Panter,[4] Timothy J. Bell,[5] Lucy Morgan,[4] Francois Peloquin,[6] Adam Gater,[4] Verna Welch,[5] Louise O'Hara,[4] Ryan Hohman,[7] Diana Merino Vega,[7] Roland B. Walter, [8] Neil Horikoshi,[9] Dawn Maze[10]

[1] Department of Haematology, Nottingham University Hospital, Nottingham, UK; [2] Division of Hematological Malignancies and Cellular Therapy, Department of Medicine, Duke University School of Medicine, Durham, US; [3] ArmorUp for LIFE, Philadelphia, USA; [4] Adelphi Values, Bollington, Cheshire, UK; [5] Pfizer Inc., New York, USA; [6] Pfizer Canada Inc., Kirkland, Canada; [7] Friends of Cancer Research, Washington DC, USA; [8] Fred Hutchinson Cancer Research Center, Seattle, USA; [9] Aplastic Anemia and MDS International Foundation, Maryland, USA; [10] Department of Medical Oncology and Hematology, Princess Margaret Cancer Centre, University Health Network, Ontario, Canada.

A separate electronic version of this poster may be obtained by scanning this QR code with your smartphone app.

Key messages

1. Patients consider physicians to have the most influence in treatment decision-making; however, patient and physicians' priorities do not always align.
2. The shock of diagnosis, as well as patients' level of treatment knowledge and articulacy, may compromise their role in decision making.
3. The limited number of medications suitable for older acute myeloid leukemia (AML) patients has a significant influence on treatment decisions.

Implications of findings

- Given the differing priorities between patients and physicians and the perception of the physician's influence on treatment decision-making, any physician interactions and treatment information provided should be carefully considered and tailored to the individual's needs and goals.
- It is important to treat the patient holistically and to recognise patient's emotional, spiritual and physical needs and goals upon diagnosis.
- Educating patients about the available treatment options will help ensure that patients feel informed and subsequently empowered to contribute to treatment decision making.

Background and objective

- AML is a rare haematologic cancer primarily affecting older people (median 68 years old at diagnosis).[1]
- There are a range of treatment options, including intensive chemotherapy (IC), low intensity therapy (including non-intensive chemotherapy [NIC]), and best supportive care (BSC). Some patients do not receive any treatment.
- Given the diverse treatment options and numerous factors influencing treatment decisions, it is important to understand what trade-offs patients, their families, and physicians are willing to make to ensure that the chosen treatment path is most suited to the patient's needs and circumstances.
- However, there is limited qualitative research exploring this, as such, this study aimed to explore the multi-stakeholder perspective on shared treatment decision making in older adults with AML who are unsuitable for IC.

Methodology

- Fifteen (15) US AML patient-family member dyads, and physicians in the US (n=4), UK (n=3), and Canada (n=3) took part in 60-minute semi-structured qualitative interviews to explore treatment decision-making.
- Participants rated pre-defined treatment characteristics from 0 (not at all important) to 3 (very important) for their importance in AML treatment decision making.
- Verbatim transcripts of audio-recorded interviews were analysed using thematic analysis and Atlas.ti software.[2]

Participant sample

Patients with AML were ≥ 65 years old and deemed unsuitable or unwilling to take IC. Most patients had never received treatment for AML (n=13/15). Note, the sample was demographically diverse (Table 1).

Family members were ≥ 18 years old, demographically diverse, and had direct contact with the patient ≥ once per week.

Physicians were oncologists and/or haematologists who worked in a variety of settings and treated >10 cases of AML per month (Table 2). Physicians did not treat interviewed AML patients.

Table 1. Demographic and clinical characteristics of patients and family members

Description		Patient (N=15)	Family member (N=15)
Age, in years, mean (range)		72.2 (65-80)	49.9 (22-71)
Sex, n (%)			
	Male	5 (33.3%)	8 (53.3%)
	Female	10 (66.7%)	7 (46.7%)
Race, n (%)			
	White	7 (46.7%)	8 (53.3%)
	Black/African	6 (40.0%)	4 (26.7%)
	Multi-racial	1 (6.7%)	2 (13.3%)
	Other	-	1 (6.7%)
	Missing	1 (6.7%)	-
Time since diagnosis in months, mean (range)		5.1 (2-9)	N/A
Current/previous treatments, n (%)			N/A
	None	13 (86.7%)	
	Currently on BSC	1 (6.7%)	
	Discontinued NIC	1 (6.7%)	

Table 2. Demographic characteristics of physicians

Demographics	US (n=4)	UK (n=3)	Canada (n=3)	Total (N=10)
Work settings, n (%)*				
Hospital	1 (25.0%)	3 (100.0%)	2 (66.7%)	6 (60.0%)
Private practice	2 (50.0%)	1 (33.3%)	-	3 (30.0%)
Outpatient	2 (50.0%)	1 (33.3%)	2 (66.7%)	1 (10.0%)
How long have you been treating patients with leukaemia's? (years)				
Mean (range)	15.3 (7-27)	15 (5-20)	22.3 (17-25)	17.3 (5-27)
How many patients with AML do you see per month?				
Mean (range)	23 (12-40)	21.7 (15-30)	21.7 (10-40)	22.2 (10-40)
How many patients with AML do you see per month who are unsuitable for IC?				
Mean (range)	11.3 (7-15)	9.3 (8-10)	7.7 (3-15)	9.6 (3-15)

*Physicians could select multiple options

Results

1 Patients consider physicians to have the most influence in treatment decision-making; however, patient and physicians' priorities do not always align

- Stakeholders reported that patients (n=40/40), physicians (n=31/40) and family members (n=26/40) were involved in treatment decision-making.
- The majority of patients (n=9/15), and all physicians asked (n=6/10), reported that the physician had the most influence on treatment decisions. In contrast, most family members reported that patients had the most influence (n=9/15; Figure 1).

Interviewer: *"Who, who would you say ultimately had the most influence in your decision?"* *"…the doctor . Yeah. I had some input, but he told me all about it."* (US AML patient not receiving treatment)

- Some family members (n=3/15) felt excluded from the process, and as such may not have been aware of the balance between patient and physician input.

"I was told what was going to happen. And it was like, wow. The decision was made without me... I'm just going to deal with it. You know, because they decided not to take any treatment." (Wife of US AML patient not receiving treatment)

Figure 1. Participant report of who had the most influence on treatment decision making

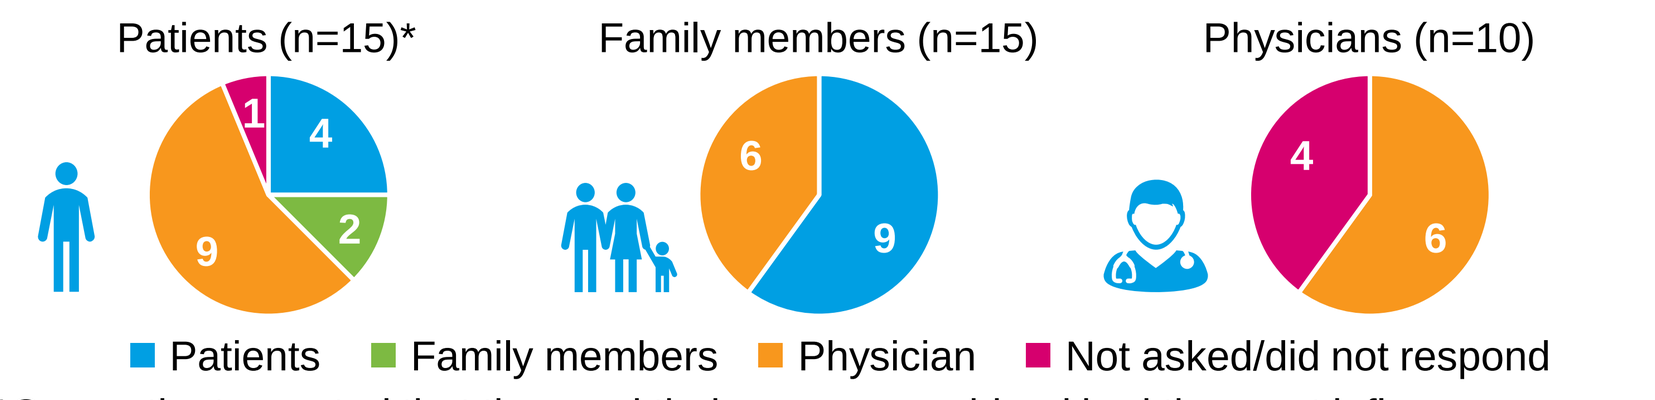
Patients (n=15)*
Family members (n=15)
Physicians (n=10)
1
4
2
9
6
9
4
6
Patients
Family members
Physician
Not asked/did not respond

*One patient reported that they and their spouse combined had the most influence.

Stakeholder priorities in treatment decision making

- Across stakeholder groups, better quality of life, longer survival and improvement of AML symptoms were most commonly considered very important factors influencing treatment decisions (Figure 2). However, there was some misalignment for several other treatment characteristics:

Possibility of being hospitalised

- While most family members (n=13/15) and patients (n=9/15) rated the possibility of being hospitalised as 'very important', only one physician did (n=1/10).
- Patients most frequently explained this was because they preferred to be at home in a familiar environment (n=3/9) and that they might become more ill in hospital (n=3/9), but family members most commonly reported a fear or dislike of hospitals (n=3/13) and not wanting to leave patients alone (n=3/13).

"…there's no place I'd rather be than home. Um, I don't want to languish someplace, um, in a hospital or hospice." (US AML patient not receiving treatment)

- Physicians who rated this consideration as 'not important' or 'slightly important' (n=5/10) believed that going to hospital was an accepted and necessary part of treatment (n=3/5).

"I have a lot of patients with like venetoclax that have the neutropenic fever. They get in the hospital for a few days and then they're gone…So I don't think it's a great concern." (US Physician)

Treatment side effects

- A greater proportion of patients (n=11/15) and family members (n=14/15) than physicians (n=4/10) felt that the treatment side effects were 'very important' in treatment decision making.
- Patients (n=6/11) and family members (n=5/14) believed that the side effects (e.g. vomiting, hair loss) would be worse than AML symptoms. Specifically, most patients not on treatment (n=9/13) reported a fear of side effects as the primary reason for opting not to take treatment.

"…the toll it would take physically, I mean I'm sick regardless… it's just piling more on to what I already have…" (US AML patient not receiving treatment)

- Physicians who considered side effects 'slightly important' or 'somewhat important' (n=4/6) felt that it was necessary to balance the drawbacks of side effects against the benefits of treatment.

Risk of Infection

- Unlike other stakeholders (patients [n=7/15]; physicians [n=3/10]), most family members (n=13/15) rated the risk of infection as 'very important' because it could compromise patient health (n=11/13).
- In contrast, most physicians reasoned that infections were an unavoidable, but manageable part of treatment (n=6/10).

"Because you could die from infections and you don't want infections." (Son of US AML patient not receiving treatment)

"…we tell the patient that you will have a infection. That can happen, so don't worry. And we will treat you." (Canadian Physician)

Figure 2. Treatment factors considered 'very important' by patients, family members, and physicians when considering taking an AML treatment

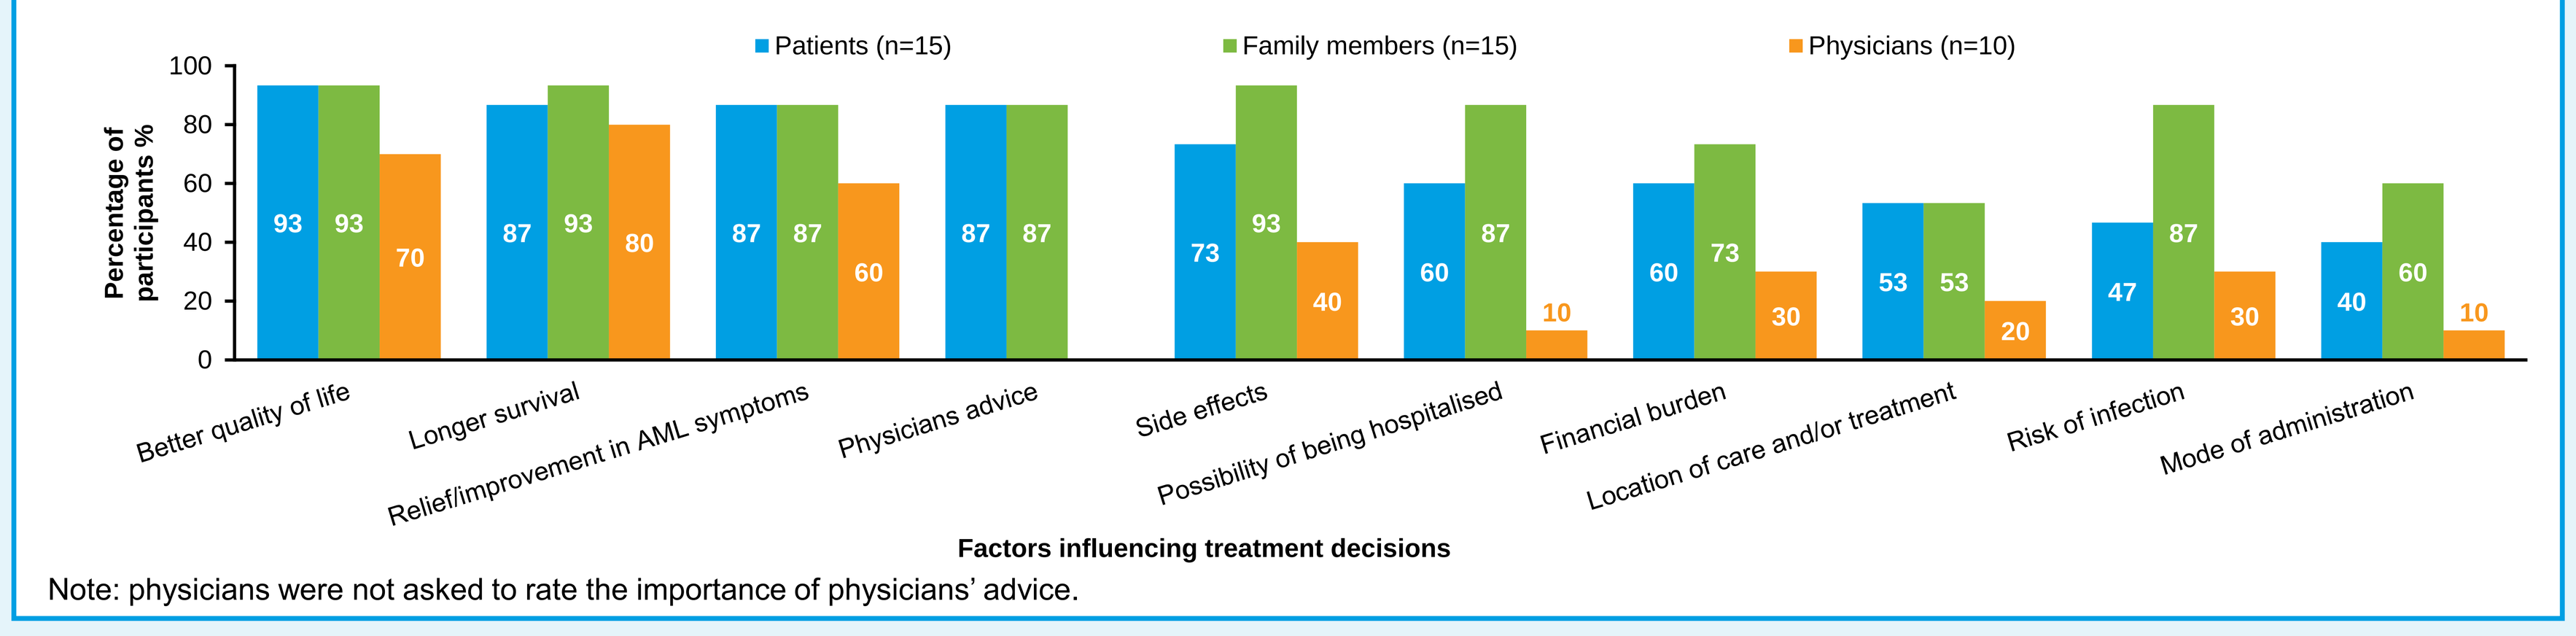
Patients (n=15)
Family members (n=15)
Physicians (n=10)
Percentage of participants %
100
80
60
40
20
0
93 93 70
87 93 80
87 87 60
87 87
73 93 40
60 87 10
60 73 30
53 53 20
47 87 30
40 60 10
Better quality of life
Longer survival
Relief/improvement in AML symptoms
Physicians advice
Side effects
Possibility of being hospitalised
Financial burden
Location of care and/or treatment
Risk of infection
Mode of administration
Factors influencing treatment decisions

Note: physicians were not asked to rate the importance of physicians' advice.

2 The shock of diagnosis, as well as patients' level of treatment knowledge and articulacy, may compromise their role in decision making

Emotional trauma

- Patients (n=9/15) and their family members (n=7/15) experienced shock upon diagnosis as well as other negative emotions such as depression (patients [n=8/15], family members [n=7/15]).
- The suddenness of diagnosis and associated emotional trauma left some patients (n=2/15) unable to process information and make decisions.

"It was very scary. It was very shocking. It was very saddening. And, uh, really we didn't know anything about AML and just like…we really didn't know how to navigate it." (Son of US AML patient not receiving treatment)

"…to be very honest, honestly I was probably too depressed to understand anything. I was in shock." (US AML patient not receiving treatment)

Information gaps

- Most patients, (n=11/15) reported discussing treatments with their physician. However, throughout the interviews, no patients articulated the difference between IC and NIC.
- Even patients who had been on NIC or BSC only discussed 'chemotherapy' (n=2).

Interviewer: *"What are the treatments ..were you offered or were you considering at that time?"* *"It was either.. the chemo or the, um, none treatment."* (US AML patient not receiving treatment)

"They soon there put me on treatment, uh, Vidaza I think it was, the chemo." (US AML patient with experience of NIC)

Interviewer: *"So are you taking any treatment at the moment?"* *"Yes. Chemotherapy."* (US AML patient on BSC)

- One physician commented on the gaps in patients' knowledge of treatment and the need for patient education (n=1).

"Education is quite important… a kind of realistic education on what they can expect with treatment and what they shouldn't expect with treatment, I think is sometimes very helpful." (UK Physician)

3 The limited number of medications suitable for older AML patients has a significant influence on treatment decisions

- All stakeholders agreed that patients' health (including age, comorbidities, and being "unwell") played a large role in determining which treatment options could be considered (n=10/10 physicians, n=11/15 patients and n=11/15 family members). Two patients and a physician specifically commented that there were a lack of available treatment options for older, unwell AML patients.

"…there wasn't going to be an option for me to do chemo and there was no treatment options." (US AML patient not receiving treatment)

"A lot of times these patients cannot have any treatment. So it's just – it's very challenging." (US Physician)

- When asked specifically about first-line NIC prescribed to AML patients, there were international differences in the range of drugs that were prescribed (Figure 3): eight drugs were reportedly prescribed as first line treatments in the US, while only four drugs were prescribed in the UK and/or Canada. This may be associated with the treatments that have been approved for use in each country.
- Half the physicians reported that the availability of treatments would influence treatment decisions (n=5/10).

"…today we're doing a lot more treatments than we used to. I think we used to send a lot of patients to best supportive care before we had, uh, the things I mentioned available." (US Physician)

"The success of the treatment [Ara-C] is not good, but it is a treatment which is available. It gives a bit of freedom to the patient because the patient can give himself or herself the injections at home…" (UK Physician)

Figure 3. First line NIC treatments prescribed by physicians

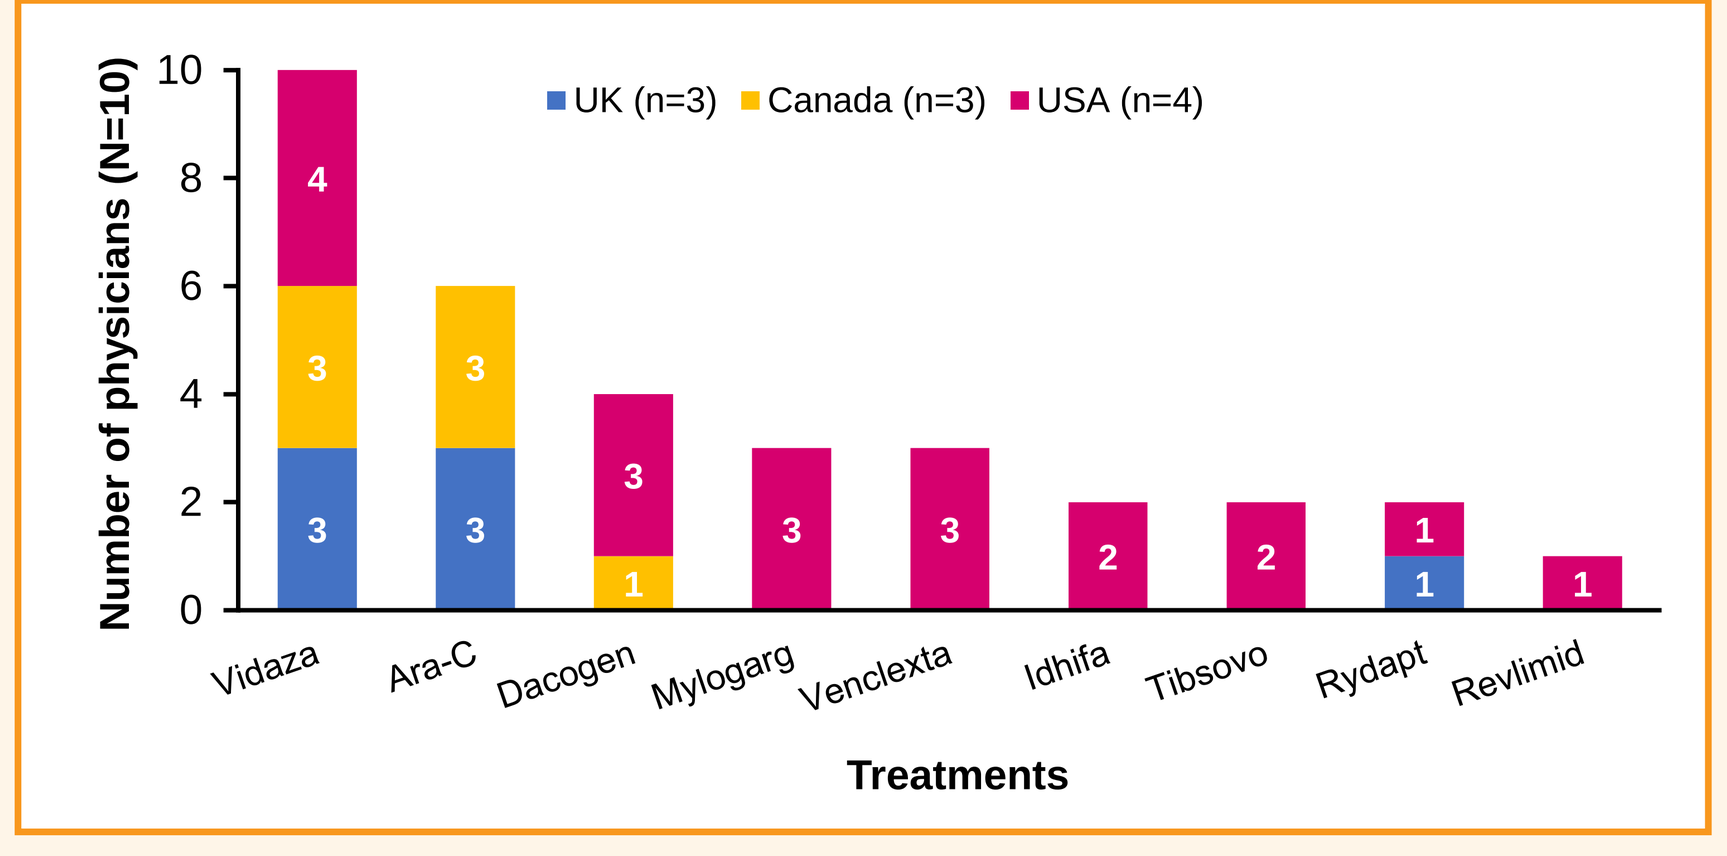
UK (n=3)
Canada (n=3)
USA (n=4)
Number of physicians (N=10)
10
8
6
4
2
0
Vidaza 3 3 4
Ara-C 3 3
Dacogen 1 3
Mylogarg 3
Venclexta 3
Idhifa 2
Tibsovo 2
Rydapt 1 1
Revlimid 1
Treatments

References

1. American Cancer Society. Cancer Facts & Figures 2020. Atlanta, GA: American Cancer Society; 2020.
2. Joffe H, Yardley L. Content and Thematic Analysis. In: Marks DF, Yardley L, eds. Research Methods for Clinical and Health Psychology. London: Sage Publications; 2004:56-68.

Acknowledgements

This research was funded by Pfizer. NR, TWL, LHA, RH, DMV, RBW, NH and DM are contracted by Pfizer to advise on the conduct of this study and the interpretation of the study findings. CP, LM, AG and LOH are employees of Adelphi Values, a health outcomes research agency commissioned by Pfizer to conduct the research described in this poster. The authors of this paper have no other conflicts of interest to disclose.

Contacts:

Contact email: Lucy.Morgan@adelphivalues.com

BSH2020 PO-017-A
ALL and AML
Nicola Moss
Poster presented at: British Society for Haematology Listening • Learning • Leading
PosterSessionOnline.com