P5150

Real-World Incidence, Characteristics and Management of Cytokine Release Syndrome Induced by Chimeric Antigen Receptor T-Cell Therapies Across Hematological Malignancies

Nayan Chaudhary,[1] Sumi Roy,[1] Chu-Fang Lin,[1] Maneesh Tandon,[2] Antonia Kwan,[1] Peter Kuebler,[1] **Ashwini Shewade**[1]*

*Presenting author. E-mail: shewade.ashwini@gene.com

Summary

This study uniquely **summarizes real world data (RWD) on cytokine release syndrome (CRS)** following the use of commercially available chimeric antigen receptor (CAR) T-cell therapies **across multiple hematologic malignancies**, based on information from clinical notes **abstracted retrospectively from electronic health records (EHRs)**

Incidence of CRS induced from commercial CAR T-cell therapies ranged from **56% in diffuse large B-cell lymphoma (DLBCL) to 72% in mantle cell lymphoma (MCL)**

Tocilizumab and corticosteroids were the most commonly used treatments for CRS, and **37–48%** of patients with **Grade 1 CAR T-cell therapy-induced CRS** received **tocilizumab**

Median time to **CRS onset** following CAR T-cell therapy infusion ranged from **1 day** in **multiple myeloma (MM)** to **4 days** in **follicular lymphoma (FL) and MCL**

[1]Genentech, Inc., South San Francisco, CA, USA;
[2]F. Hoffmann-La Roche Ltd, Basel, Switzerland.

Background

- CRS is a potentially life-threatening and supraphysiologic response following immunotherapy, resulting in activation or engagement of endogenous or infused T cells and/or other immune effector cells.[1]
- CRS occurs with varying frequency and severity, depending on the underlying disease and immunotherapy used, such as CAR T-cell therapy or off-the-shelf T-cell engaging bispecific antibodies.[2]
- This study describes the real-world incidence, severity and clinical management of CRS induced by commercially available CAR T-cell therapies across a range of hematologic malignancies including DLBCL, FL, MCL and MM.

Methods

- This was a retrospective, observational study. The study included patients diagnosed with DLBCL, FL, MCL or MM on or after January 1, 2011, who received commercially available CAR T-cell therapies.
- RWD (clinical cutoff date for follow-up of August 31, 2022) were obtained from the Flatiron Health US nationwide, EHR-derived, de-identified database of patients with cancer within a network comprised primarily of community cancer practices (~280 US cancer clinics, ~800 sites of care)
 - The Flatiron Health network also includes practices designated as academic sites by the National Cancer Institute
 - The longitudinal database comprises patient-level structured and unstructured data, curated via technology-enabled abstraction.[3,4]
- CAR T-cell therapy, clinical characteristics and disease outcomes specific to CAR T-cell treatment, as well as CRS and its clinical management, were abstracted from unstructured documents in EHRs in addition to routinely available structured data
 - EHRs were abstracted for patients treated with the following CAR T-cell therapies: axicabtagene ciloleucel (axi-cel), tisagenlecleucel (tisa-cel), lisocabtagene maraleucel (liso-cel), brexucabtagene autoleucel (brexu-cel), idecabtagene vicleucel (ide-cel) and ciltacabtagene autoleucel (cilta-cel).
- Data pertaining to diagnosis of CRS and resolution were abstracted based on explicit mention of and attribution to CRS in the patients' charts. Additionally, data related to the following treatments and whether the treatment was administered for CRS management were abstracted: tocilizumab, dexamethasone, siltuximab, anakinra and methylprednisolone.

Results: 386 patients were included in the study, mainly diagnosed with DLBCL (N=185)

- While 81–95% of patients received CAR T-cell therapy at an academic center, 12–14% of patients with DLBCL and MCL received CAR T-cell therapy at a community center (**Table 1**).
- With the exception of ide-cel and cilta-cel, the CAR T-cell therapies included in this study have been approved for use after one or two prior lines of therapy.[5–10] In this study, a considerably higher proportion of patients received CAR T-cell therapies in later lines (43% in DLBCL and 88% in MM received CAR-T therapy after four or more lines of therapy; **Figure 1A**).
- CAR T-cell therapy by year of administration across hematologic malignancies is shown in **Figure 1B**.

Table 1. Patient characteristics.

	DLBCL N=185	FL N=67	MCL N=92	MM N=42
Mean age at initiation of CAR T-cell therapy treatment, years (SD)	61.4 (12.3)	61.8 (8.7)	65.7 (8.8)	65.0 (6.6)
Gender, n (%)				
Female	73 (39.5)	25 (37.3)	24 (26.1)	19 (45.2)
Male	112 (60.5)	42 (62.7)	68 (73.9)	23 (54.8)
Race/ethnicity, n (%)				
Asian	<5	<5	<5	<5
Black or African American	13 (7.0)	<5	<5	<5
Hispanic, any race	13 (7.0)	8 (11.9)	<5	<5
Other race	15 (8.1)	<5	11 (12.0)	<5
White	129 (69.7)	47 (70.1)	69 (75.0)	32 (76.2)
CAR T-cell therapy treatment setting, n (%)				
Academic	150 (81.1)	60 (89.6)	76 (82.6)	40 (95.2)
Community	26 (14.1)	<5	11 (12.0)	<5

Column percentages may not add up to 100% due to the exclusion of missing data categories. Numbers less than five were supressed due to concerns with re-identification. SD, standard deviation.

Figure 1A. CAR T-cell therapy by line of therapy (LoT) across malignancies.

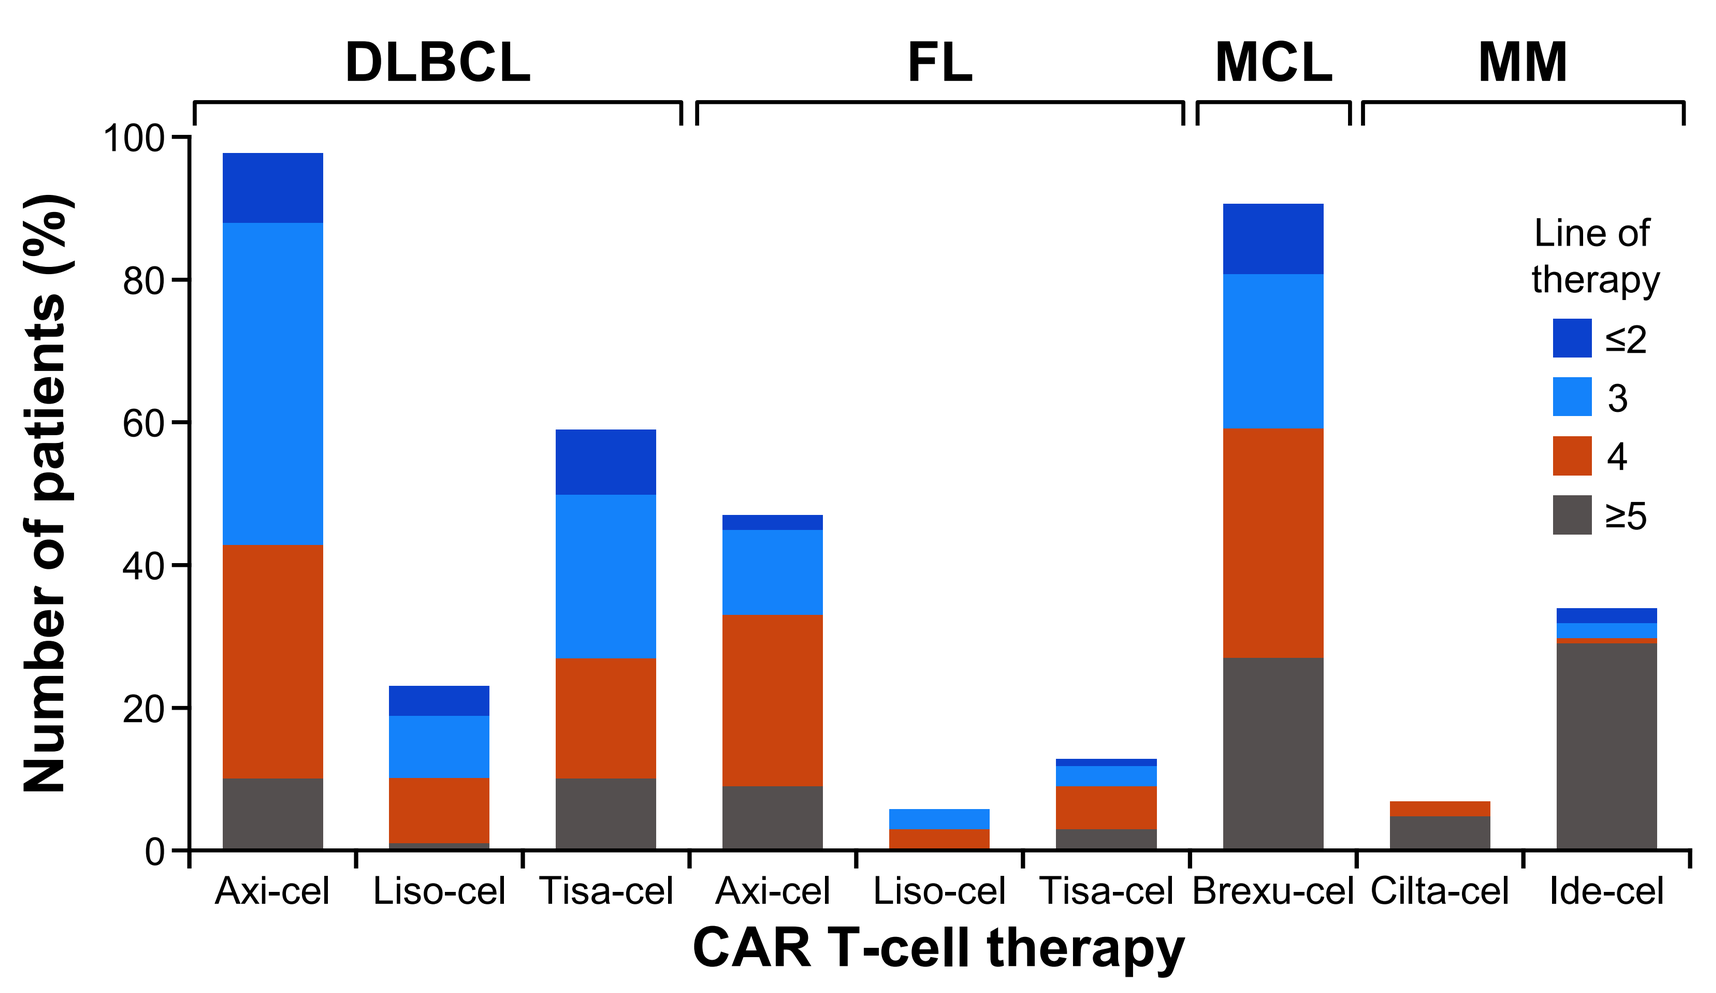

DLBCL
FL
MCL
MM
Number of patients (%)
100
80
60
40
20
0
Line of therapy
≤2
3
4
≥5
Axi-cel
Liso-cel
Tisa-cel
Axi-cel
Liso-cel
Tisa-cel
Brexu-cel
Cilta-cel
Ide-cel
CAR T-cell therapy

Figure 1B. CAR T-cell therapy by year of administration across malignancies.

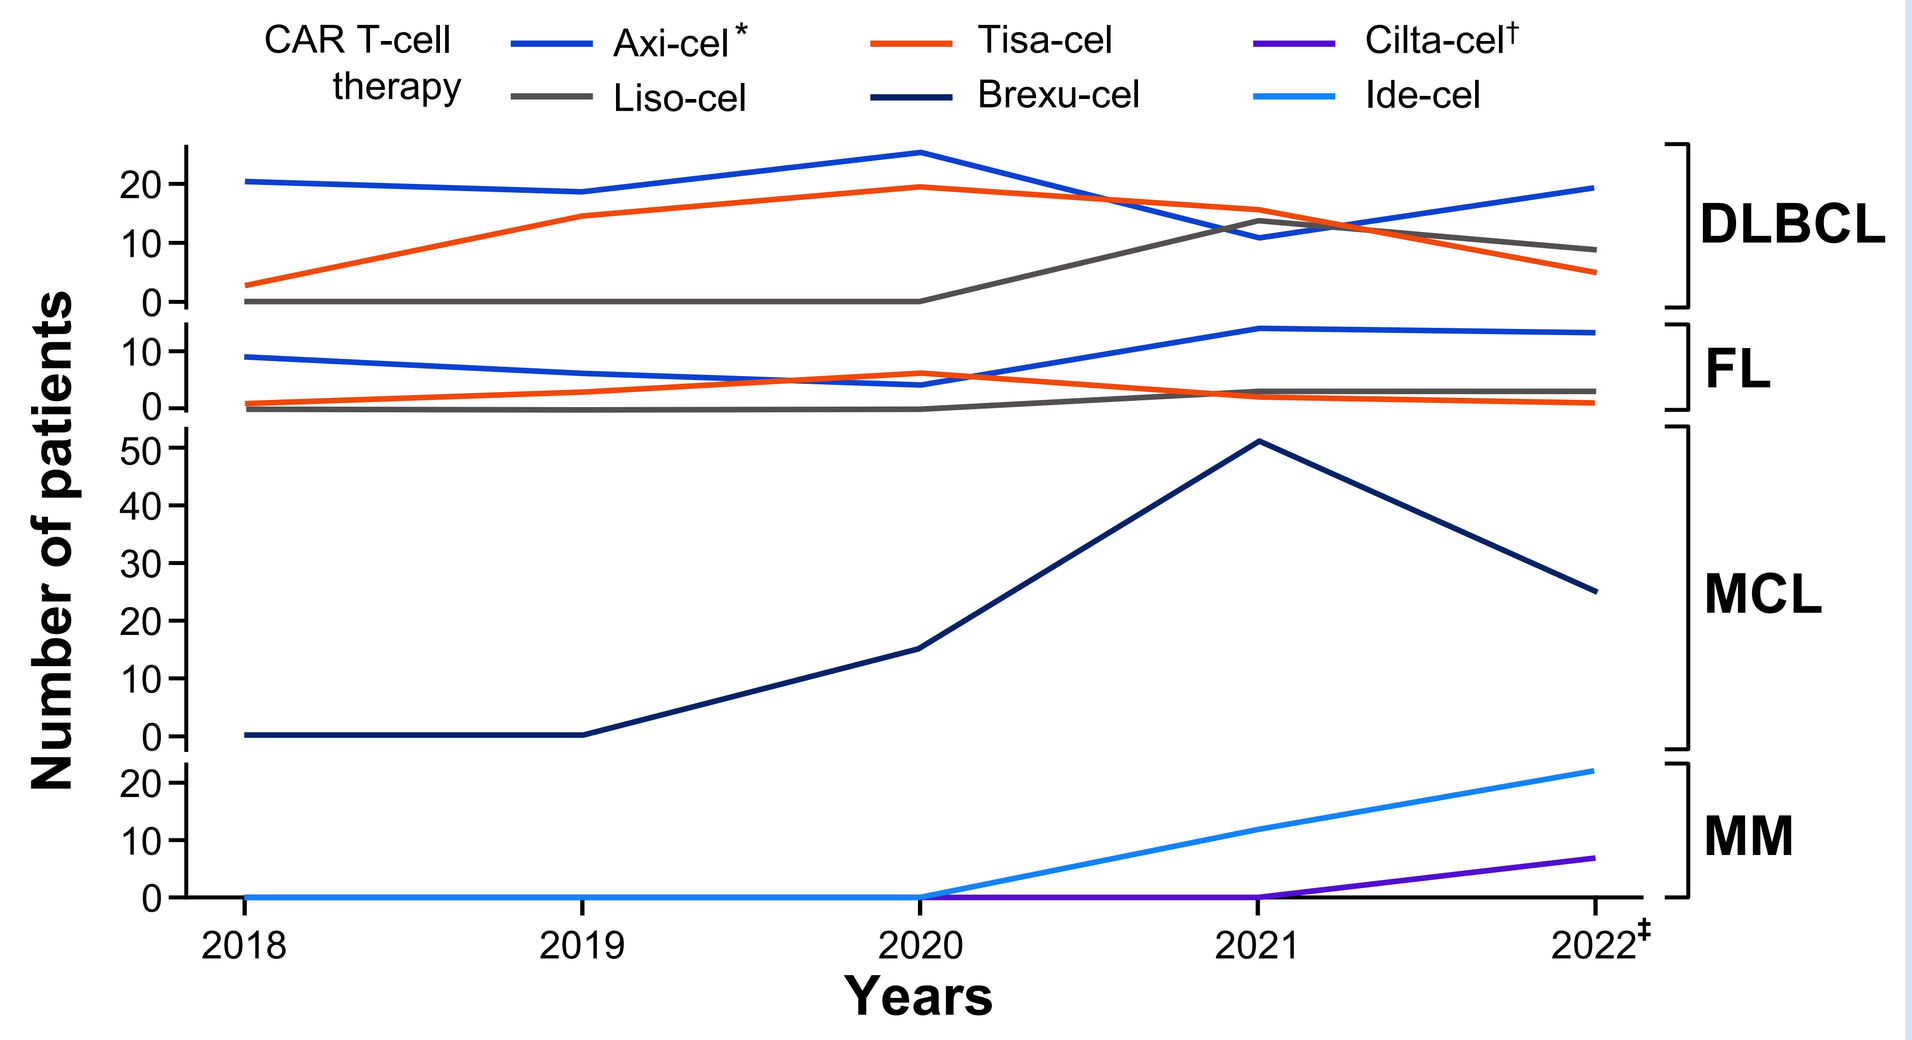

CAR T-cell therapy
Axi-cel*
Liso-cel
Tisa-cel
Brexu-cel
Cilta-cel†
Ide-cel
Number of patients
DLBCL
FL
MCL
MM
2018
2019
2020
2021
2022‡
Years

Axi-cel and tisa-cel were first approved for commercial availability in 2017, brexu-cel in 2020, liso-cel and ide-cel in 2021, and cilta-cel in 2022. *One patient each in the DLBCL and FL cohorts received axi-cel in 2017. †Seven patients in the MM cohort received cilta-cel in 2022. ‡Data for 2022 were only included up to August 31, 2022.

Results: more than half of patients across all malignancies experienced CAR T-cell therapy-induced CRS

- Incidence of CRS ranged from 56% (104/185) in DLBCL to 72% (66/92) in MCL (**Table 2**).
- Excluding categories with small numbers of cases (<15), CRS incidence also varied by individual CAR T-cell therapy and underlying malignancies, from 39% with tisa-cel in DLBCL to 73% with brexu-cel in MCL (**Figure 2**).
- Highest incidence of Grade ≥2 CRS was observed among patients with MCL (38%), whereas Grade ≥3 CRS was most commonly observed among patients with FL (4%).
- Time to onset of CRS ranged from 0–16 days, with median onset between 3 and 4 days among patients with lymphoma and earlier median onset of 1 day for patients with MM.
- Median time to CRS resolution ranged from 2 to 6 days across malignancies.

Table 2. CRS incidence and characteristics.

	DLBCL N=185	FL N=67	MCL N=92	MM N=42
CRS incidence,* n (%)				
Any CRS	104 (56.2)	40 (59.7)	66 (71.7)	24 (57.1)
Grade 1	49 (26.5)	20 (29.9)	25 (27.2)	18 (42.9)
Grade 2	42 (22.7)	15 (22.4)	32 (34.8)	6 (14.3)
Grade ≥3	5 (2.7)	3 (4.5)	3 (3.3)	0 (0.0)
Not documented	8 (4.3)	2 (3.0)	6 (6.5)	0 (0.0)
Median time to onset, Days† [min, max]	3 [0, 15]	4 [0, 15]	4 [0, 12]	1 [0, 16]
Median duration, Days†‡ [min, max]	4 [1, 33]	5.5 [1, 20]	5 [1, 8]	2 [0, 8]

*Only reported data have been summarized and therefore percentages may not add up to 100% due to missing data. Data pertaining to CRS were abstracted from patients' EHRs with explicit mention of CRS. †Estimates were based on cases with more than one instance of CRS assessment documented in patients' EHRs. ‡Estimates were calculated using data for those patients where CRS event dates were available with day-level granularity.

Figure 2. CRS incidence by CAR T-cell therapy and malignancy.

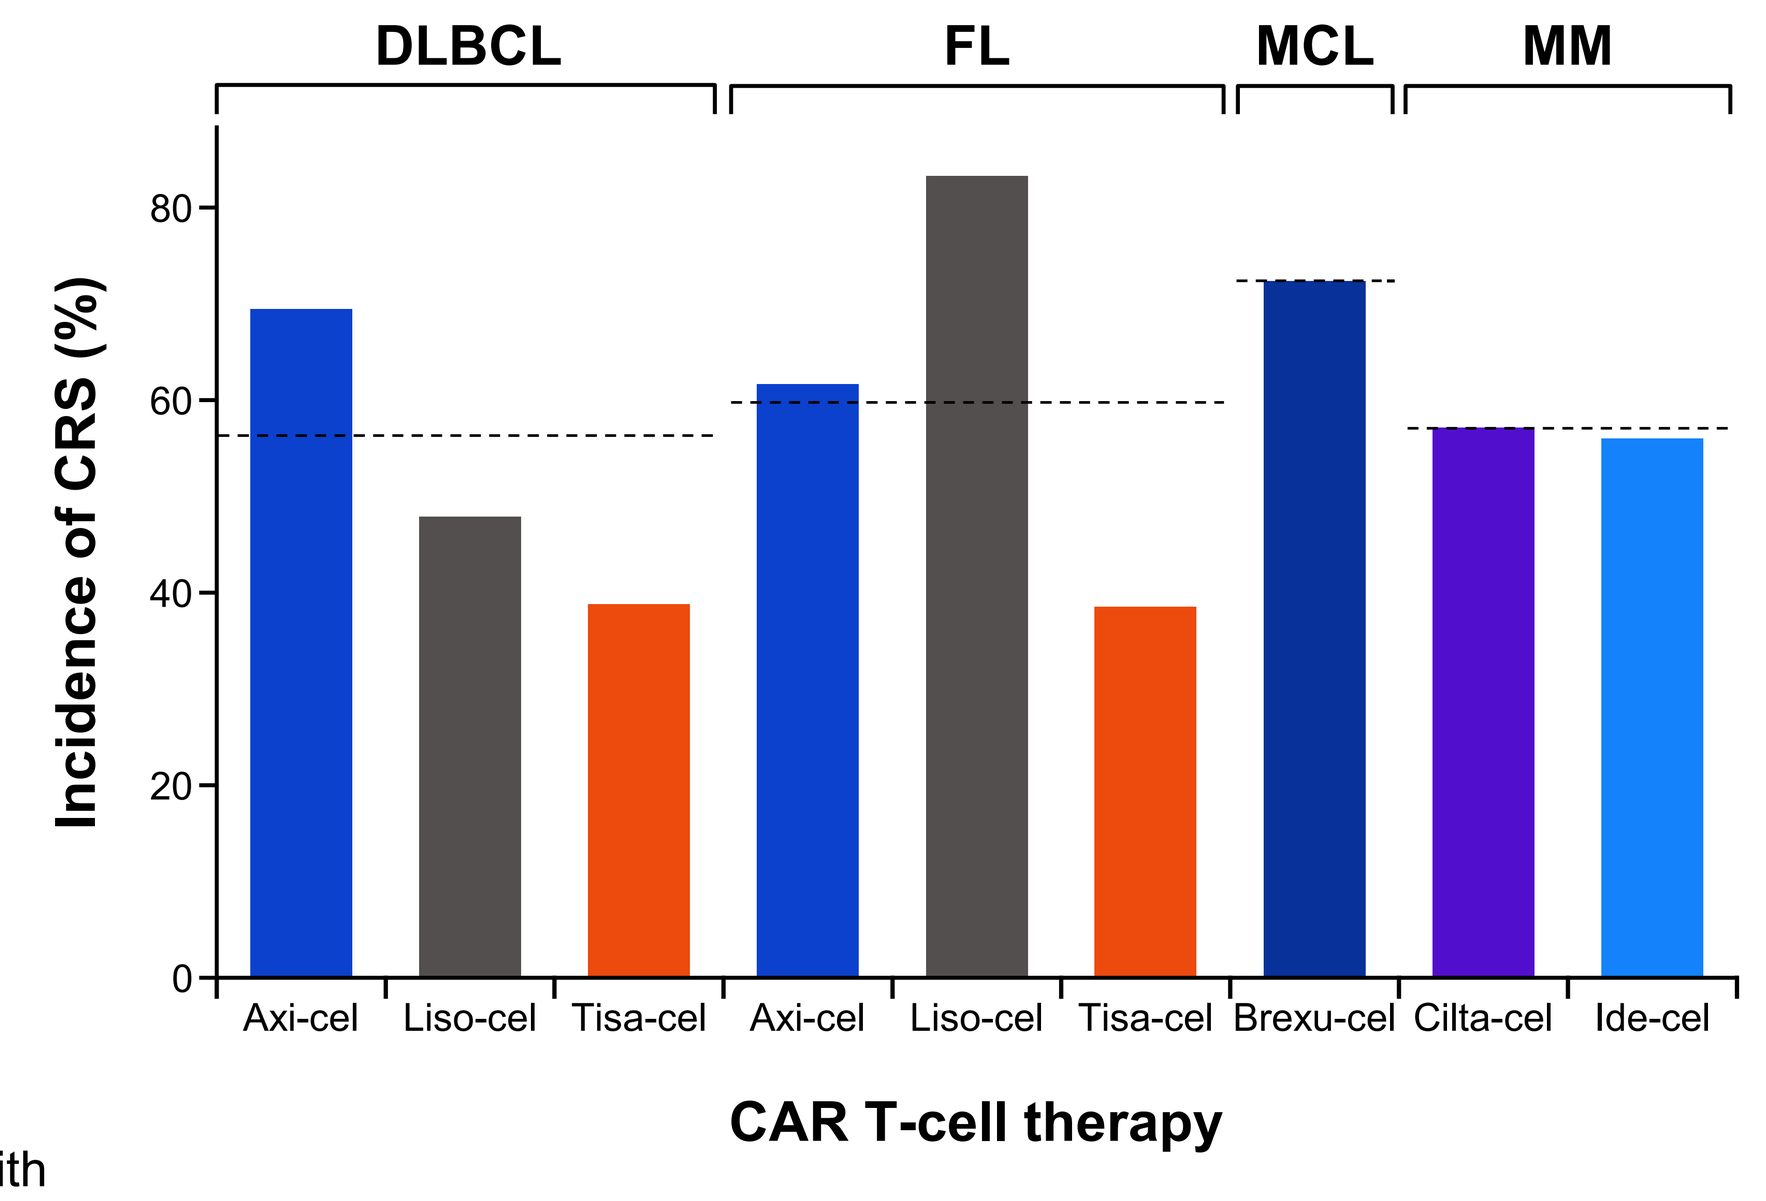

DLBCL
FL
MCL
MM
Incidence of CRS (%)
80
60
40
20
0
Axi-cel
Liso-cel
Tisa-cel
Axi-cel
Liso-cel
Tisa-cel
Brexu-cel
Cilta-cel
Ide-cel
CAR T-cell therapy

Dashed lines indicate overall CRS incidence for each malignancy.

Results: tocilizumab and corticosteroids were the most commonly used treatments for CRS

- Pharmacologic management of CAR T-cell therapy-induced CRS is depicted in **Table 3**.
- Tocilizumab was used for the management of CRS in 37–48% of patients with a maximum reported CRS Grade of 1.
- Anakinra and siltuximab treatment for CRS were administered in less than five patients across all cohorts.

Table 3. Use of tocilizumab and corticosteroids to manage CRS.

	DLBCL N=185	FL N=67	MCL N=92	MM N=42
Grade 1 CRS,* n	**49**	**20**	**25**	**18**
Tocilizumab treatment,† n (%)	18 (36.7)	8 (40.0)	12 (48.0)	7 (38.9)
Corticosteroid treatment,† n (%)	8 (16.3)	5 (25.0)	7 (28.0)	1 (5.6)
Grade 2 CRS,* n	**42**	**15**	**32**	**6**
Tocilizumab treatment,† n (%)	31 (73.8)	10 (66.7)	27 (84.4)	6 (100.0)
Corticosteroid treatment,† n (%)	18 (42.9)	7 (46.7)	20 (62.5)	3 (50.0)
Grade ≥3 CRS treatment,* n	**5**	**3**	**3**	**0**
Tocilizumab treatment,† n (%)	4 (80.0)	2 (66.7)	3 (100)	–
Corticosteroid treatment,† n (%)	3 (60.0)	1 (33.3)	2 (66.7)	–

*Only reported data have been summarized and therefore percentages may not add up to 100% due to missing data. Data pertaining to CRS were abstracted from patients' EHRs with explicit mention of CRS. †Data pertaining to treatments where available and distinctly attributable to CRS in patients' EHRs. Treatment data were unavailable in varying proportions across CRS grades, suggesting either lack of use of treatments or missing documentation.

Discussion

- CAR T-cell therapies were predominantly administered in academic centers; limited use in community practices was also observed.
- Data also showed considerable use of tocilizumab to treat low-grade CRS, suggesting the need for additional evidence on its role to treat CRS beyond the current regulatory approval.
- Increased evidence generation, specifically from usual clinical practice, can contribute to increased awareness and knowledge about CRS diagnosis and management, and in turn realize the maximum potential of T-cell engaging therapeutics.

Limitations

- Further information to accurately characterize the size and capabilities of community centers was not available in this database.
- Consistent with the timing of approval of commercially available CAR T-cell therapies, the sample size for MM was limited compared with other malignancies. For similar reasons, the sample sizes for liso-cel for DLBCL and FL were also limited.
- Safety events are typically under-reported in RWD sources, specifically for CRS when relying on information documented in patients' EHRs, compared with other published literature.
- In accordance with other RWD on CRS, grading criteria for CRS as well as vasopressor and oxygen use were not consistently reported in patients' EHRs.

Presented at the 2023 American Society of Hematology Annual Meeting | December 9–12, 2023

References

1. Lee D, et al. Biol Blood Marrow Transplant 2018;25:625–38.
2. Leclercq G, et al. Oncoimmunology 2022;11:e2083479.
3. Ma X, et al. medRxiv 2020. Available at: https://doi.org/10.1101/2020.03.16.20037143.
4. Birnbaum B, et al. arXiv 2020. Available at: https://arxiv.org/abs/2001.09765.
5. ABECMA® US PI 2021. Available at: https://www.fda.gov/media/147055/download.
6. BREYANZI® US PI 2022. Available at: https://www.fda.gov/media/145711/download.
7. CARVYKTI® US PI 2023. Available at: https://www.fda.gov/media/156560/download.
8. KYMRIAH® US PI 2022. Available at: https://www.fda.gov/media/107296/download.
9. TECARTUS® US PI 2021. Available at: https://www.fda.gov/media/140409/download.
10. YESCARTA® US PI 2022. Available at: https://www.fda.gov/media/108377/download.

Acknowledgments

This study was sponsored by F. Hoffmann-La Roche Ltd. Third party editorial assistance, under the direction of all authors, was provided by Molly Heitz, PhD, and Tayna Carlisle, MSc, of Ashfield MedComms, an Inizio company, and was funded by Genentech, Inc.

Disclosures

NC: employment (Genentech, Inc.), equity (F. Hoffmann-La Roche Ltd); **SR**: employment and equity (Genentech, Inc.); **C-FL**: employment (Genentech, Inc.); **MT**: employment and stock options (F. Hoffmann-La Roche Ltd); **AK**: employment (Genentech, Inc.), equity (F. Hoffmann-La Roche Ltd); **PK**: employment (Genentech, Inc.), equity (F. Hoffmann-La Roche Ltd); **AS**: employment (Genentech, Inc.), stock options (F. Hoffmann-La Roche Ltd).

PosterSessionOnline.com

American Society of Hematology
Helping hematologists conquer blood diseases worldwide

905. Outcomes Research-Lymphoid Malignancies: Poster III
Nayan Chaudhary

MON-5150

ASH2023