LBA-001

Ramucirumab as second-line treatment in patients with advanced hepatocellular carcinoma (HCC) and elevated alpha-fetoprotein (AFP) following first-line sorafenib: Pooled efficacy and safety across two global randomized Phase 3 studies (REACH-2 and REACH)

Andrew X Zhu[1], Richard S Finn[2], Peter R Galle[3], Josep M Llovet[4], Jean-Frederic Blanc[5], Takuji Okusaka[6], Ian Chau[7], Paolo Abada[8], Yanzhi Hsu[9], Masatoshi Kudo[10]

[1]Massachusetts General Hospital Cancer Center, Harvard Medical School, Boston, MA, USA; [2]University of California, Los Angeles Medical Center, Los Angeles, CA, USA; [3]Universitätsmedizin, Mainz, Germany; [4]Mount Sinai School of Medicine, New York, NY, USA; [5]CHU de Bordeaux, Hôpital Haut-Lévêque, Pessac, France; [6]National Cancer Center Hospital, Tokyo, Japan; [7]Royal Marsden Hospital, Sutton, UK; [8]Eli Lilly and Company, Lilly Corporate Center, Indianapolis, IN, USA; [9]Eli Lilly and Company, New York, NY, USA; [10]Kindai University, Osaka-Sayama City, Osaka, Japan

BACKGROUND

- Hepatocellular carcinoma (HCC) is the second most common cause of cancer related mortality globally[1]
- Sorafenib (1L) and regorafenib (2L) are the only globally approved drugs for treatment of HCC[2,3,4] Nivolumab (2L) is approved in US; Lenvatinib (1L) and cabozantinib (2L) have demonstrated benefits in phase 3 studies[5,6,7]
- Ramucirumab, a human IgG1 monoclonal antibody (mAb), inhibits ligand activation of VEGFR2[8]
- REACH-2 and REACH were two global, double-blind, randomized, placebo-controlled multicenter, phase 3 studies of ramucirumab vs placebo in patients with HCC after prior sorafenib[8]
- Primary endpoint of REACH-2 was met, demonstrating an improved overall survival compared to placebo in patients with baseline AFP ≥400 ng/mL, a population of poor prognosis, confirming the findings in REACH[9]

OBJECTIVES & METHODS

- REACH-2 and REACH were both global, randomized, placebo-controlled phase 3 trials with similar study eligibility, protocol procedures, and treatment regimen, resulting in similar efficacy and safety outcomes; pooling of the two study populations to assess the patients with an elevated AFP is acceptable
- The pooled analysis performed with patients from REACH-2 and REACH (AFP ≥400 ng/mL) enables a more precise assessment of the efficacy and safety of ramucirumab treatment in this population provided by the larger dataset
- Pooled analyses of patients from REACH-2 and REACH with baseline AFP ≥400 ng/mL was performed at the individual patient data level. Efficacy analyses were stratified by study
- Pooled analyses were pre-specified prior to REACH-2 database lock

Key Inclusion Criteria

- Advanced HCC (histological or radiological imaging confirmation)
- Child-Pugh A (5-6)
- ECOG PS score of 0 or 1
- Barcelona Clinic Liver Cancer (BCLC) stage C or B refractory or not amenable to locoregional therapy
- Adequate hematologic and biochemical parameters
- Prior sorafenib treatment (discontinued due to progression or intolerance)
- Baseline AFP ≥400 ng/mL (only for REACH-2)

Key Exclusion Criteria

- Esophageal or gastric varices requiring immediate intervention
- Prior anti-vascular endothelial growth factor (VEGF) pathway therapy other than sorafenib
- Uncontrolled hypertension
- History of or current hepatic encephalopathy (any grade) or clinically meaningful ascites
- Arterial thrombotic event within 6 months
- Therapeutic anticoagulation or chronic antiplatelet agents including nonsteroidal anti-inflammatory drugs (NSAIDs)

RESULTS

Table 1. Demographics and Baseline Characteristics

n (%)		REACH-2 Ramucirumab n = 197	REACH-2 Placebo n = 95	REACH (AFP ≥400ng/mL) Ramucirumab n = 119	REACH (AFP ≥400ng/mL) Placebo n = 131	Pooled Ramucirumab n = 316	Pooled Placebo n = 226
Sex	Male	154 (78.2)	79 (83.2)	92 (77.3)	110 (84.0)	246 (77.8)	189 (83.6)
Age (years)	Median	64	64	62	59	64	62
Race	Asian	102 (51.8)	45 (47.4)	66 (55.5)	78 (59.5)	168 (53.2)	123 (54.4)
ECOG PS[a]	0	113 (57.4)	55 (57.9)	60 (50.4)	63 (48.1)	173 (54.7)	118 (52.2)
Geographic region							
Region 1	Americas, Europe, Israel & Australia	101 (51.3)	50 (52.6)	-	-	154 (48.7)	108 (47.8)
Region 2	Asia (excluding Japan)	55 (27.9)	27 (28.4)	-	-	101 (32.0)	78 (34.5)
Region 3	Japan	41 (20.8)	18 (18.9)	-	-	61 (19.3)	40 (17.7)
Child-Pugh Score	A – 5	123 (62.4)	54 (56.8)	67 (56.3)	81 (61.8)	190 (60.1)	135 (59.7)
Baseline BCLC Stage	B	34 (17.3)	20 (21.1)	11 (9.2)	9 (6.9)	45 (14.2)	29 (12.8)
Sorafenib Discontinuation	Progressive Disease	166 (84.3)	76 (80.0)	108 (90.8)	122 (93.1)	274 (86.7)	198 (87.6)
	Intolerance	31 (15.7)	19 (20.0)	11 (9.2)	9 (6.9)	42 (13.3)	28 (12.4)
Etiology	Hepatitis B	71 (36.0)	36 (37.9)	53 (44.5)	66 (50.4)	124 (39.2)	102 (45.1)
	Hepatitis C	48 (24.4)	28 (29.5)	35 (29.4)	28 (21.4)	83 (26.3)	56 (24.8)
	Significant Alcohol Use	48 (24.4)	21 (22.1)	23 (19.3)	22 (16.8)	71 (22.5)	43 (19.0)
Macrovascular Invasion[a]	Present	70 (35.5)	33 (34.7)	43 (36.1)	44 (33.6)	113 (35.8)	77 (34.1)
Extrahepatic Spread	Present	141 (71.6)	70 (73.7)	85 (71.4)	101 (77.1)	226 (71.5)	171 (75.7)
Baseline AFP (ng/mL)[a]	Median	3920.0	2741.0	5293.0	7022.0	4101.6	4047.5
	Min-Max	408.0 – 230500.0	419.0-473163.0	410.8 – 853200.0	429.0 – 628390.0	408.0 – 853200.0	419.0 – 628390.0

[a] Prognostic in a stepwise Cox regression

Figure 1. Overall Survival

a

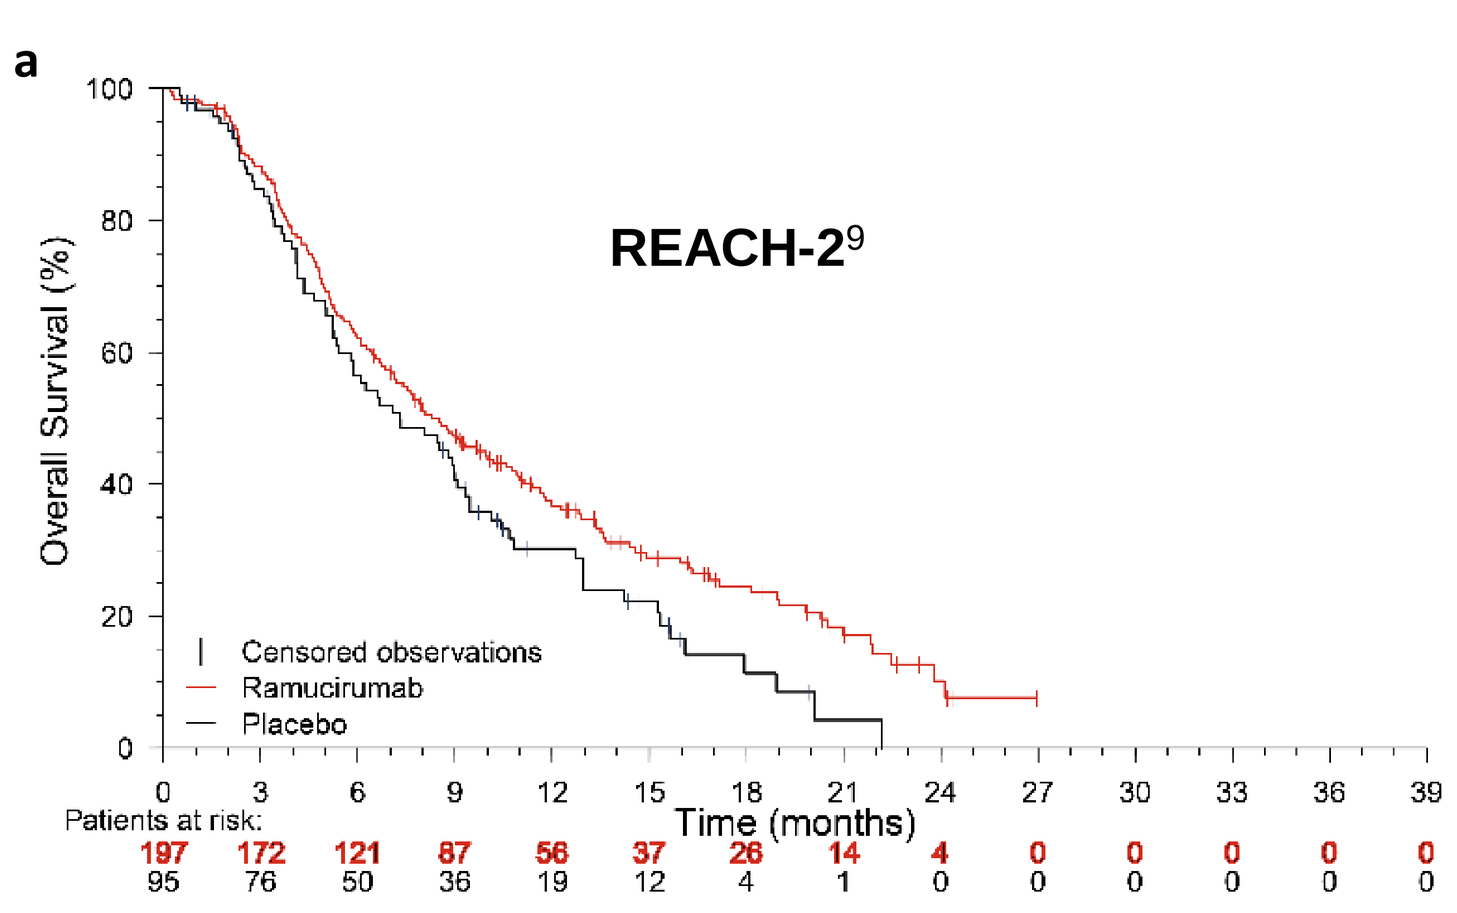

REACH-2[9]
Overall Survival (%)
Censored observations
Ramucirumab
Placebo
Time (months)
Patients at risk:
197 172 121 87 56 37 26 14 4 0 0 0 0 0
95 76 50 36 19 12 4 1 0 0 0 0 0 0

	RAM (n = 197)	Placebo (n = 95)	Δ
Median, months	8.5	7.3	1.2
HR (95% CI); p-value	0.710 (0.531, 0.949); 0.0199		
Censored for PDT: mOS	8.1	6.6	1.5
Censored for PDT: HR (95% CI); p-value	0.647 (0.462, 0.907); 0.0109		

b

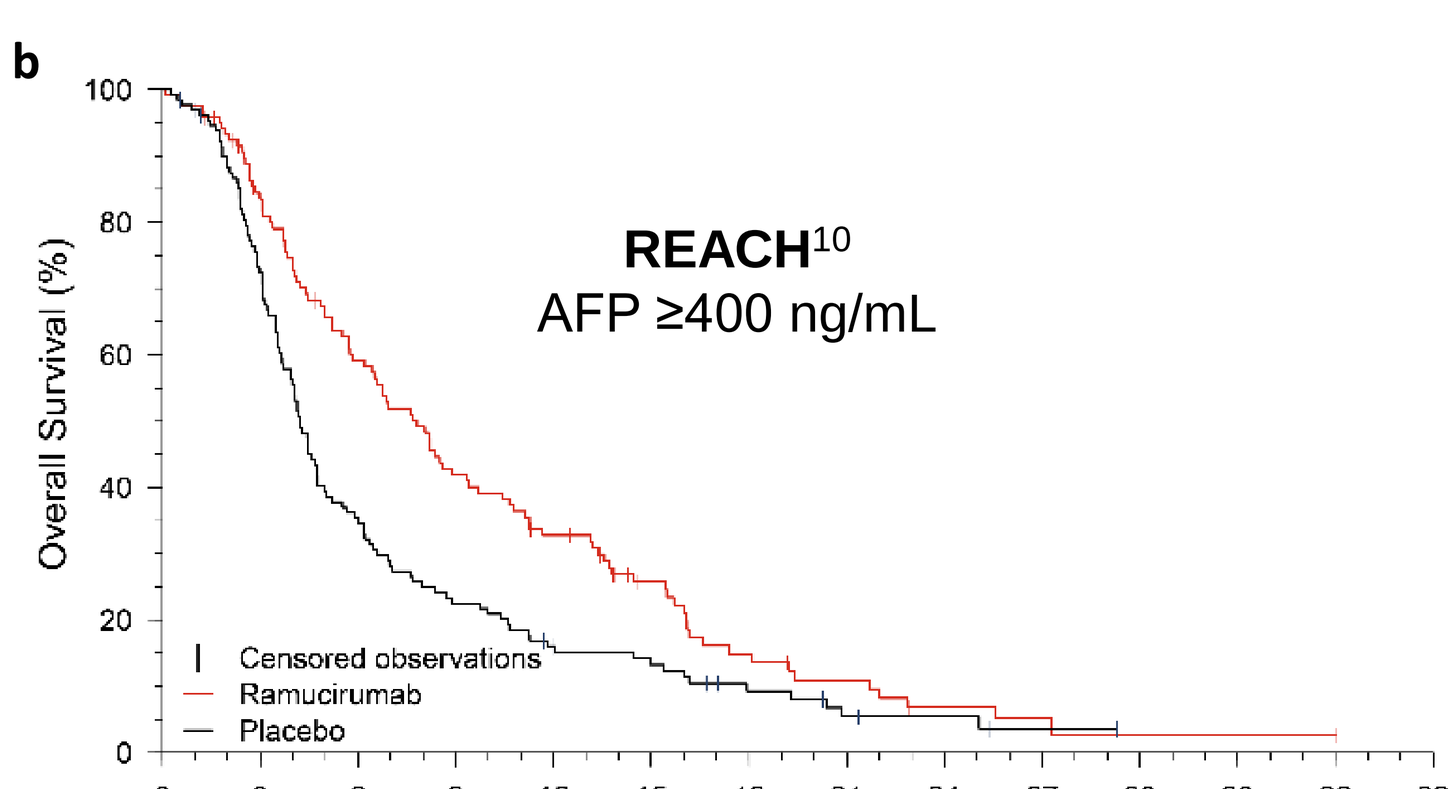

REACH[10]
AFP ≥400 ng/mL
Overall Survival (%)
Censored observations
Ramucirumab
Placebo
Time (months)
Patients at risk:
119 93 65 46 35 22 12 8 4 2 1 1 1 0
131 90 44 28 18 14 8 4 3 1 0 0 0 0

	RAM (n = 119)	Placebo (n = 131)	Δ
Median, months	7.8	4.2	3.6
HR (95% CI); p-value	0.674 (0.508, 0.895); 0.0059		
Censored for PDT: mOS	7.6	4.1	3.5
Censored for PDT: HR (95% CI); p-value	0.635 (0.456, 0.884); 0.0066		

Abbreviation: PDT, Post-Discontinuation Therapy

c

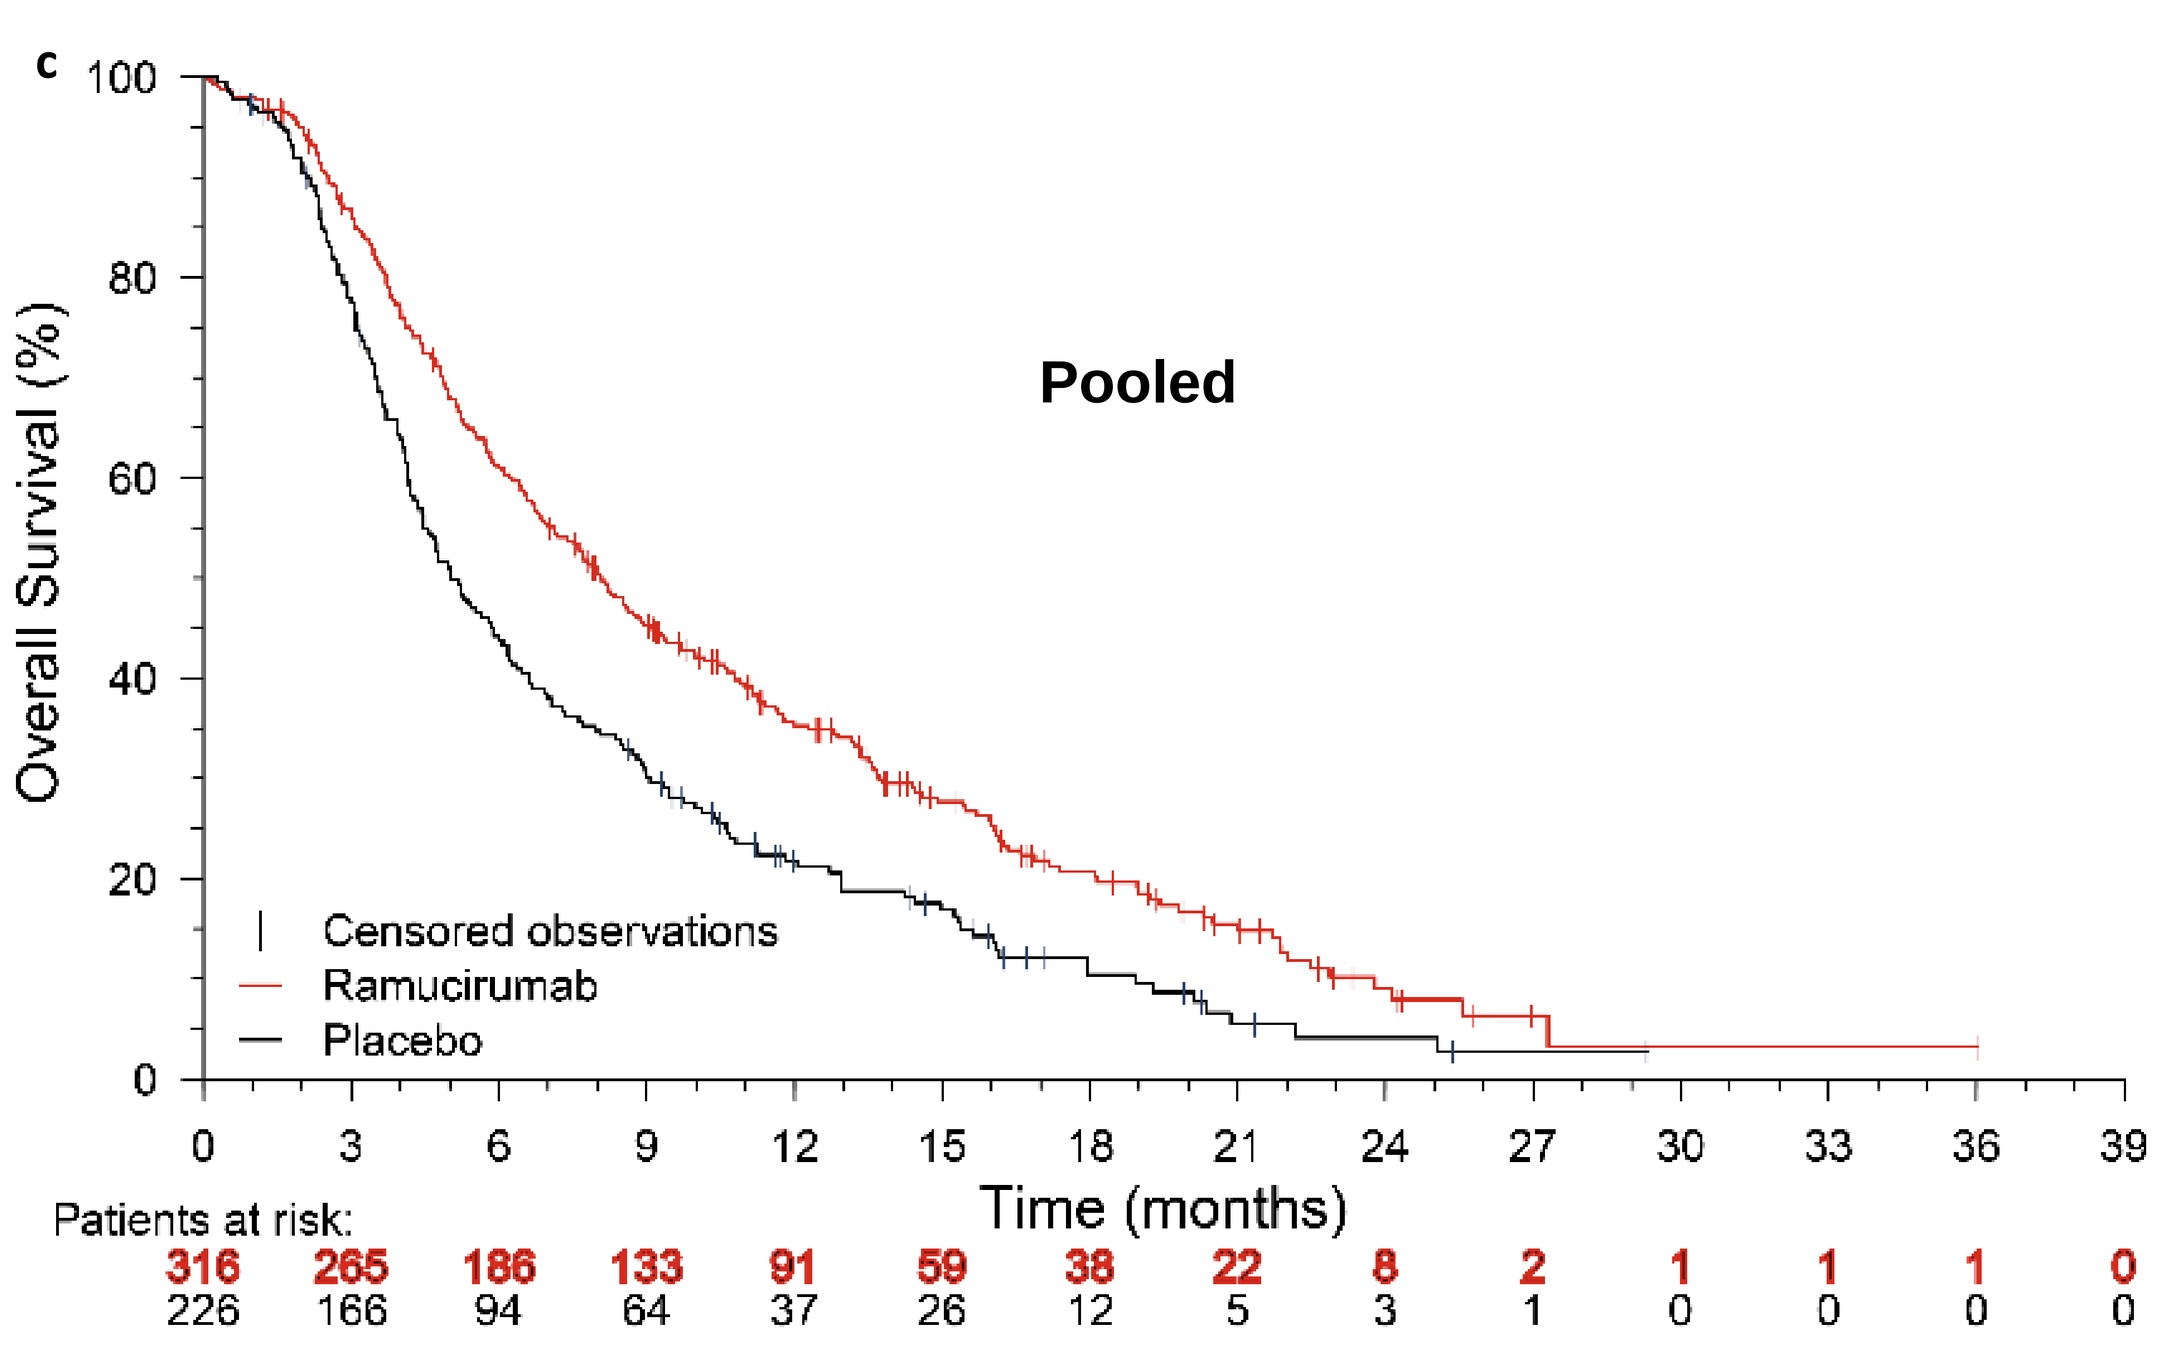

Pooled
Overall Survival (%)
Censored observations
Ramucirumab
Placebo
Time (months)
Patients at risk:
316 265 186 133 91 59 38 22 8 2 1 1 1 0
226 166 94 64 37 26 12 5 3 1 0 0 0 0

	Ramucirumab (n = 119)	Placebo (n = 131)	Difference	P-value
Median, months	8.1	5.0	3.1	
HR (95% CI)	0.694 (0.571, 0.842)			0.0002
Death, n (%)	246 (77.8)	190 (84.1)		

No heterogeneity in treatment effect observed across both studies

A random effect frailty model after adjusting for Study (as random effect) produced a similar treatment result (HR=0.689; p=0.0002)

Table 2. Forest Plot for Pooled Overall Survival

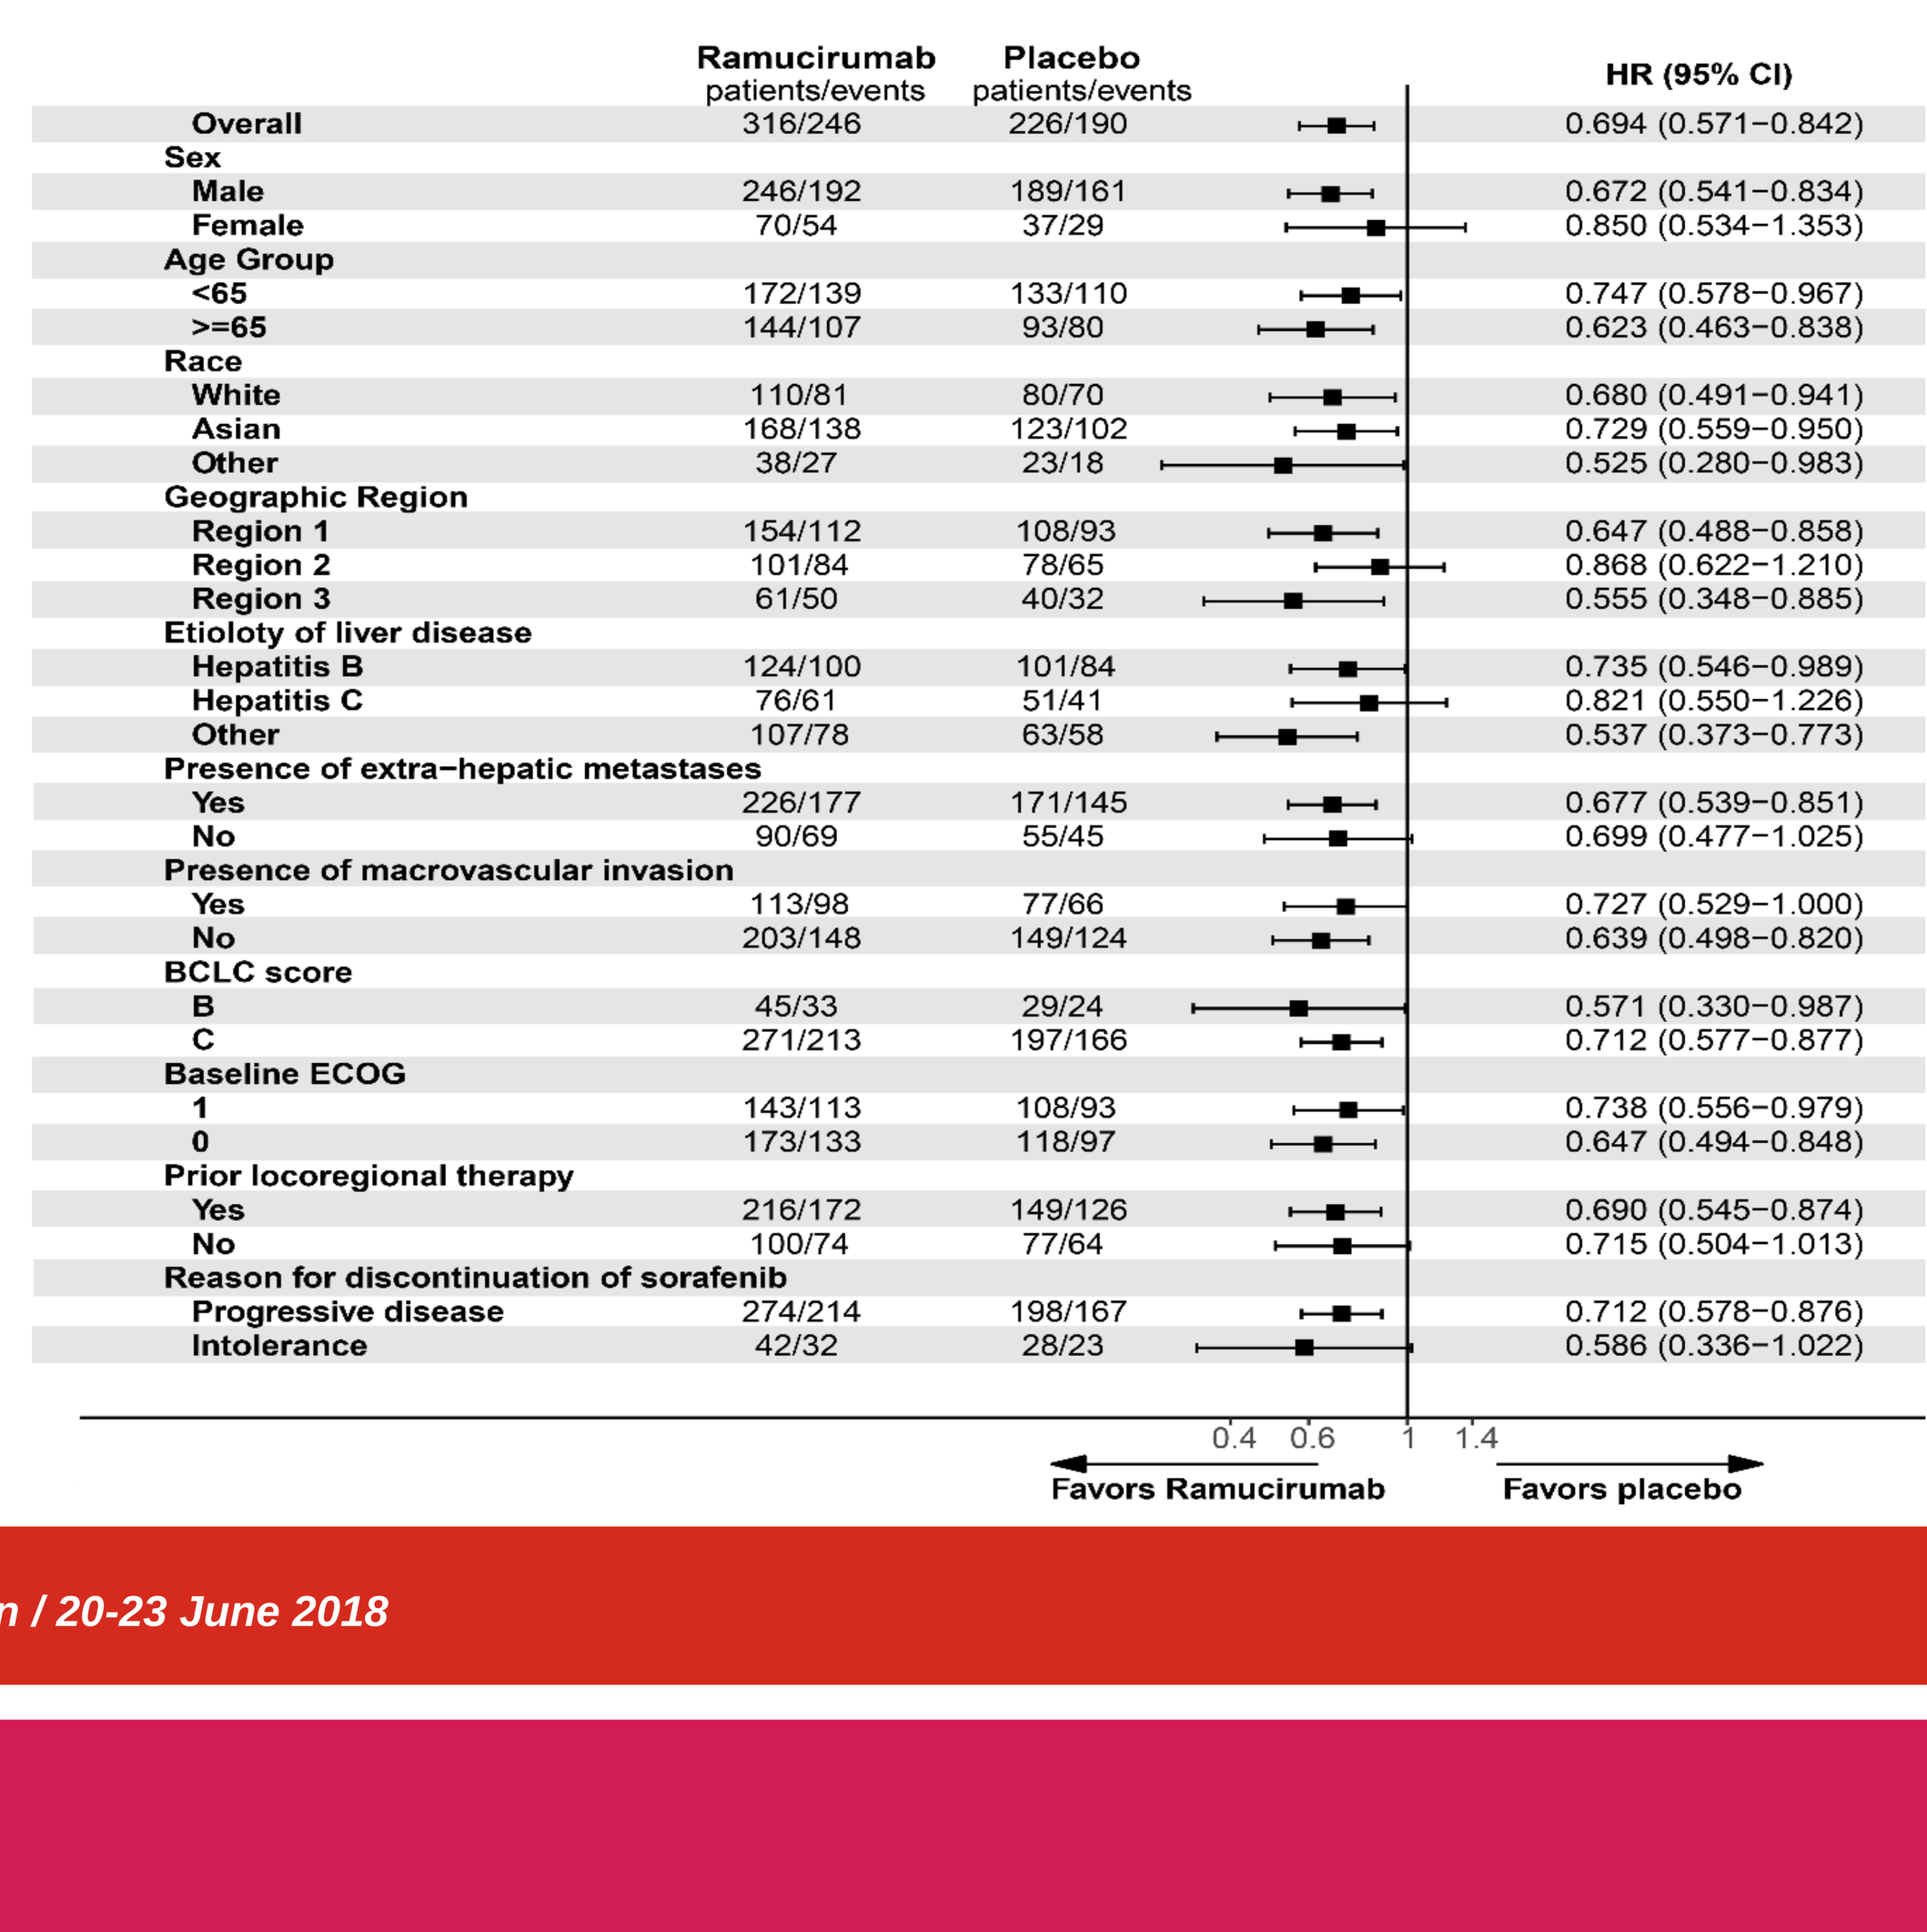

0.4 0.6 1 1.4
Favors Ramucirumab
Favors placebo

	Ramucirumab patients/events	Placebo patients/events	HR (95% CI)
Overall	316/246	226/190	0.694 (0.571−0.842)
Sex			
Male	246/192	189/161	0.672 (0.541−0.834)
Female	70/54	37/29	0.850 (0.534−1.353)
Age Group			
<65	172/139	133/110	0.747 (0.578−0.967)
>=65	144/107	93/80	0.623 (0.463−0.838)
Race			
White	110/81	80/70	0.680 (0.491−0.941)
Asian	168/138	123/102	0.729 (0.559−0.950)
Other	38/27	23/18	0.525 (0.280−0.983)
Geographic Region			
Region 1	154/112	108/93	0.647 (0.488−0.858)
Region 2	101/84	78/65	0.868 (0.622−1.210)
Region 3	61/50	40/32	0.555 (0.348−0.885)
Etioloty of liver disease			
Hepatitis B	124/100	101/84	0.735 (0.546−0.989)
Hepatitis C	76/61	51/41	0.821 (0.550−1.226)
Other	107/78	63/58	0.537 (0.373−0.773)
Presence of extra−hepatic metastases			
Yes	226/177	171/145	0.677 (0.539−0.851)
No	90/69	55/45	0.699 (0.477−1.025)
Presence of macrovascular invasion			
Yes	113/98	77/66	0.727 (0.529−1.000)
No	203/148	149/124	0.639 (0.498−0.820)
BCLC score			
B	45/33	29/24	0.571 (0.330−0.987)
C	271/213	197/166	0.712 (0.577−0.877)
Baseline ECOG			
1	143/113	108/93	0.738 (0.556−0.979)
0	173/133	118/97	0.647 (0.494−0.848)
Prior locoregional therapy			
Yes	216/172	149/126	0.690 (0.545−0.874)
No	100/74	77/64	0.715 (0.504−1.013)
Reason for discontinuation of sorafenib			
Progressive disease	274/214	198/167	0.712 (0.578−0.876)
Intolerance	42/32	28/23	0.586 (0.336−1.022)

Region 1 – Americas, Europe, Israel and Australia
Region 2 – Asia (excluding Japan)
Region 3 – Japan

Table 3. PFS & Best Overall Response

	REACH-2 Ramucirumab n = 197	REACH-2 Placebo n = 95	REACH (AFP ≥400ng/mL) Ramucirumab n = 119	REACH (AFP ≥400ng/mL) Placebo n = 131	Pooled Ramucirumab n = 316	Pooled Placebo n = 226
Progression-free Survival						
Median, months	2.8	1.6	2.7	1.5	2.8	1.5
HR (95% CI); p-value	0.452 (0.339, 0.603); <0.0001		0.699 (0.531, 0.920); 0.0106		0.572 (0.472, 0.694); <0.0001	
Overall Response Rate ORR [CR+PR] (95% CI)	9 (4.6) (1.7, 7.5)	1 (1.1) (0.0, 3.1)	8 (6.7) (3.4, 12.7)	1 (0.8) (0.1, 4.2)	17 (5.4) (2.9, 7.9)	2 (0.9) (0.0, 2.1)
Disease Control Rate DCR [CR+PR+SD] (95% CI)	118 (59.9) (53.1, 66.7)	37 (38.9) (29.1, 48.8)	60 (50.4) (41.6, 59.2)	47 (35.9) (28.2, 44.4)	178 (56.3) (50.9, 61.8)	84 (37.2) (30.9, 43.5)

Abbreviations: HR, Hazard Ratio; CR, Complete Response; PR, Partial Response; SD, Stable Disease
RECIST 1.1 (Response Evaluation Criteria in Solid Tumors)

Table 4. Dose Exposure, Relative Intensity, Discontinuation, and Dose Adjustments Due to AEs

n (%)	REACH-2 Ramucirumab n = 197	REACH-2 Placebo n = 95	REACH (AFP ≥400ng/mL) Ramucirumab n = 119	REACH (AFP ≥400ng/mL) Placebo n = 128	Pooled Ramucirumab n = 316	Pooled Placebo n = 223
Relative Dose Intensity (%)						
Median	97.9	99.8	98.6	99.5	98.3	99.6
Cycles received per patient						
Median	6.0	4.0	4.0	3.0	5.0	3.0
Mean	9.5	5.1	7.6	5.4	8.8	5.3
Discontinued due to treatment related AE	21 (10.7)	3 (3.2)	8 (6.7)	4 (3.1)	30 (9.5)	8 (3.6)
Dose adjustment[a] due to treatment related AE	33 (16.8)	6 (6.3)	19 (16.0)	6 (4.7)	52 (16.5)	12 (5.4)
Patients with ≥ 1 treatment related TEAE, Grade ≥ 3	40 (20.3)	7 (7.4)	32 (26.9)	21 (16.4)	72 (22.8)	28 (12.6)
Death due to treatment related AE on study treatment	3 (1.5)	0	1 (0.8)	3 (2.3)	4 (1.3)	1 (0.4)

Abbreviation: AE, Adverse Event
Treatment relatedness was determined by the investigator
[a] Dose adjustment = dose delay/reduction/omission in REACH-2; dose delay/omission In REACH

Table 5. TEAEs Occurring in ≥15% in Ramucirumab Arm and Higher Rate than Control Arm

MedDRA Preferred Term	REACH-2 Ramucirumab n = 197 n (%) Any Grade	REACH-2 Ramucirumab Grade ≥3	REACH-2 Placebo n = 95 n (%) Any Grade	REACH-2 Placebo Grade ≥3	REACH (AFP ≥400ng/mL) Ramucirumab n = 119 n (%) Any Grade	REACH Ramucirumab Grade ≥3	REACH Placebo n = 128 n (%) Any Grade	REACH Placebo Grade ≥3	Pooled Ramucirumab n = 316 n (%) Any Grade	Pooled Ramucirumab Grade ≥3	Pooled Placebo n = 223 n (%) Any Grade	Pooled Placebo Grade ≥3
Peripheral edema	50 (25.4)	3 (1.5)	13 (13.7)	0	41 (34.5)	0	20 (15.6)	0	92 (29.1)	3 (0.9)	38 (17.0)	0
Fatigue	54 (27.4)	7 (3.6)	16 (16.8)	3 (3.2)	22 (18.5)	1 (0.8)	22 (17.2)	3 (2.3)	76 (24.1)	8 (2.5)	39 (17.5)	6 (2.7)
Decreased appetite	46 (23.4)	3 (1.5)	19 (20.0)	1 (1.1)	23 (19.3)	1 (0.8)	25 (19.5)	0	70 (22.2)	4 (1.3)	46 (20.6)	1 (0.4)
Ascites	35 (17.8)	8 (4.1)	7 (7.4)	2 (2.1)	33 (27.7)	7 (5.9)	25 (19.5)	7 (5.5)	66 (20.9)	15 (4.7)	33 (14.8)	9 (4.0)
Hypertension	48 (24.4)	24 (12.2)	12 (12.6)	5 (5.3)	20 (16.8)	14 (11.8)	8 (6.3)	3 (2.3)	66 (20.9)	38 (12.0)	20 (9.0)	8 (3.6)
Nausea	37 (18.8)	0	11 (11.6)	0	24 (20.2)	0	26 (20.3)	0	62 (19.6)	0	36 (16.1)	0
Abdominal pain	39 (19.8)	3 (1.5)	12 (12.6)	2 (2.1)	22 (18.5)	2 (1.7)	30 (23.4)	6 (4.7)	61 (19.3)	5 (1.6)	41 (18.4)	9 (4.0)
Diarrhea	32 (16.2)	0	14 (14.7)	1 (1.1)	26 (21.8)	1 (0.8)	12 (9.4)	0	58 (18.4)	1 (0.3)	26 (11.7)	1 (0.4)
Proteinuria	40 (20.3)	4 (2.0)	4 (4.2)	0	14 (11.8)	0	8 (6.3)	0	56 (17.7)	4 (1.3)	12 (5.4)	0
Headache	28 (14.2)	0	5 (5.3)	1 (1.1)	25 (21.0)	1 (0.8)	9 (7.0)	0	53 (16.8)	1 (0.3)	14 (6.3)	1 (0.4)

Table 6. Adverse Events of Special Interest

Consolidated Category	REACH-2 Ramucirumab n = 197 n (%) Any Grade	REACH-2 Ramucirumab Grade ≥3	REACH-2 Placebo n = 95 n (%) Any Grade	REACH-2 Placebo Grade ≥3	REACH (AFP ≥400ng/mL) Ramucirumab n = 119 n (%) Any Grade	REACH Ramucirumab Grade ≥3	REACH Placebo n = 131 n (%) Any Grade	REACH Placebo Grade ≥3	Pooled Ramucirumab n = 316 n (%) Any Grade	Pooled Ramucirumab Grade ≥3	Pooled Placebo n = 223 n (%) Any Grade	Pooled Placebo Grade ≥3
Liver Injury / liver failure	78 (39.6)	36 (18.3)	28 (29.5)	15 (15.8)	63 (52.9)	26 (21.8)	57 (44.5)	39 (30.5)	140 (44.3)	63 (19.9)	87 (39.0)	59 (26.5)
Ascites[a]	35 (17.8)	8 (4.1)	7 (7.4)	2 (2.1)	33 (27.7)	7 (5.9)	25 (19.5)	7 (5.5)	66 (20.9)	15 (4.7)	33 (14.8)	9 (4.0)
Hepatic Encephalopathy[a]	8 (4.1)	6 (3.0)	0	0	5 (4.2)	3 (2.5)	1 (0.8)	1 (0.8)	13 (4.1)	9 (2.8)	2 (0.9)	1 (0.4)
Bleeding / Hemorrhage Events	48 (24.4)	10 (5.1)	12 (12.6)	3 (3.2)	30 (25.2)	5 (4.2)	29 (22.7)	12 (9.4)	79 (25.0)	15 (4.7)	40 (17.9)	15 (6.7)
Epistaxis[a]	27 (13.7)	1 (0.5)	3 (3.2)	0	12 (10.1)	0	9 (7.0)	0	39 (12.3)	1 (0.3)	12 (5.4)	0
GI Hemorrhage Events	12 (6.1)	7 (3.6)	5 (5.3)	2 (2.1)	11 (9.2)	4 (3.4)	13 (10.2)	9 (7.0)	24 (7.6)	11 (3.5)	19 (8.5)	12 (5.4)
Hypertension	49 (24.9)	25 (12.7)	12 (12.6)	5 (5.3)	21 (17.6)	15 (12.6)	8 (6.3)	3 (2.3)	68 (21.5)	40 (12.7)	20 (9.0)	8 (3.6)
Proteinuria	40 (20.3)	4 (2.0)	4 (4.2)	0	17 (14.3)	0	8 (6.3)	0	59 (18.7)	4 (1.3)	12 (5.4)	0
Infusion Related Reactions[b]	17 (8.6)	0	3 (3.2)	0	5 (4.2)	1 (0.8)	1 (0.8)	0	30 (9.5)	1 (0.3)	7 (3.1)	0
Arterial Thromboembolic Events	5 (2.5)	3 (1.5)	1 (1.1)	1 (1.10)	0	0	2 (1.6)	1 (0.8)	5 (1.6)	3 (0.9)	3 (1.3)	2 (0.9)
Venous Thromboembolic Events	2 (1.0)	0	2 (2.1)	1 (1.1)	1 (0.8)	1 (0.8)	4 (3.1)	4 (3.1)	3 (0.9)	1 (0.3)	6 (2.7)	5 (2.2)
Gastrointestinal Perforation	2 (1.0)	2 (1.0)	2 (2.1)	2 (2.1)	0	0	0	0	2 (0.6)	2 (0.6)	2 (0.9)	2 (0.9)
Congestive Heart Failure	1 (0.5)	1 (0.5)	1 (1.1)	1 (1.10)	0	0	0	0	1 (0.3)	1 (0.3)	1 (0.4)	1 (0.4)
Fistula	1 (0.5)	0	0	0	0	0	0	0	1 (0.3)	0	0	0

[a] These are preferred terms
[b] Occurred within 24 hours of infusion

CONCLUSIONS

- REACH-2 (N=292) confirmed the ramucirumab treatment benefit observed in the REACH subgroup of patients with AFP ≥400 ng/mL (n=250)
- The confidence and precision of efficacy and safety assessments were increased by performing pooled analyses of REACH-2 and REACH (AFP ≥400 ng/mL) (N=542)
- In the pooled population, ramucirumab improved median survival by 3.1 months (OS HR: 0.694 [0.571, 0.842]; p = 0.0002; 5.0 placebo to 8.1 ramucirumab months)
- Ramucirumab OS treatment benefit was consistent and robust across all subgroups. Sensitivity analyses, including random effect models were consistent
- Ramucirumab was well tolerated, with a safety profile consistent with monotherapy ramucirumab treatment
- Ramucirumab represents an important new potential treatment option for patients with HCC and an elevated AFP after prior sorafenib treatment, a population associated with poor prognosis and aggressive disease

References

1. WHO Cancer Fact sheet No. 297, updated February 2015; Available at: http://www.who.int/mediacentre/factsheets/fs297/en/. Accessed on March 15, 2015.
2. Llovet JM et al. *N Engl J Med* 2008; 359: 378–390
3. Cheng AL et al. *Lancet* Oncol 2009; 10:25-34
4. Bruix J et al. Lancet 2017; 389: 56-66
5. El-Khoueiry AB et al. Lancet 2017. 389:2492-502
6. Kudo M et al. *Lancet* 2018, 391:1163-73
7. Abou-Alfa GKM et al *J Clin Oncol* 2018, 36(suppl 4S; abstr 207)
8. Spratin JL et al. *J Clin* Oncol. 2010; 28(5):780-7
9. Zhu et al. *J Clin Oncol* 36, 2018 (suppl; abstr 4003)
10. Zhu et al. *Lancet Oncol.* 2015; 16(7):859-70

Acknowledgements

- We thank all the patients and their caregivers
- We thank all the investigators and their support staff for participating in the REACH and REACH-2 clinical trials
- Mia Chen, Eli Lilly and Company, provided medical writing support

Privacy Notice Regarding the Collection of Personal Information
By scanning this QR code, you are consenting to have your IP address and, if you choose, email address temporarily retained in a secured computer system and used only for counting purposes, performing file download, and sending you an email. Your information will not be shared for any other purpose, unless required by law. You will not receive any future communications from Eli Lilly and Company based on the system-retained information. Contact information at: http://www.lilly.com/Pages/contact.aspx

WCGC 2018_Zhu

World Congress on Gastrointestinal Cancer 2018 / Barcelona, Spain / 20-23 June 2018

Sponsored by Eli Lilly and Company

20WCGIC 1--LB

Clinical Hepatocellular

Andrew Zhu

DOI: 10.3252/pso.eu.20wcgic.2018

Poster presented at: ESMO World Congress on Gastrointestinal Cancer 20–23 June, 2018 Barcelona, Spain

PosterSessionOnline.com