P-077

Predictive factors associated with ramucirumab monotherapy or combination therapy among patients with gastric/gastroesophageal junction cancer in the community oncology setting

William Schelman, MD, PhD[1], A. Scott Paulson, MD[2], Kat M. Aguilar, MPH[3], Jamyia Clark, MPH[3], Zhanglin Lin Cui, PhD[1], Astra M. Liepa, PharmD[1], Lisa M. Hess, PhD[1]

[1]Eli Lilly and Company, Indianapolis, IN; [2]Texas Oncology, Dallas, TX; [3]McKesson Specialty Health, The Woodlands, TX

BACKGROUND

The introduction of targeted therapies, such as ramucirumab, have provided patients with advanced or metastatic gastric/gastroesophageal junction (GEJ) cancer with promising treatment options, yet there remains uncertainty as to the optimal treatment for the care of patients.

In the United States (US), ramucirumab is approved as a monotherapy and in combination with paclitaxel for patients with advanced or metastatic gastric GEJ adenocarcinoma after prior fluoropyrimidine- or platinum-containing therapy. The safety and efficacy of ramucirumab were demonstrated in two Phase 3 multicenter, randomized, double-blind, placebo-controlled trials. In the REGARD trial, ramucirumab monotherapy was associated with significantly improved overall survival (OS) compared to best supportive care (5.2 vs. 3.8 months; hazard ratio [HR] 0.776; 95% confidence interval [CI] 0.603-0.998; P=0.047).[1] Similarly, patients randomized to ramucirumab and paclitaxel in the RAINBOW trial had significantly longer OS compared to those that received paclitaxel alone (9.6 vs. 7.4 months; HR 0.807; 95% CI 0.678-0.962; P=0.017).[2]

Using real-world data from a community oncology setting, the aims of this study were to describe patient characteristics and clinical outcomes of gastric/GEJ cancer patients who received ramucirumab and to explore factors associated with monotherapy and combination therapy.

METHODS

Study design:

- Retrospective observational cohort study

Study population:

- Adult patients with gastric/GEJ cancer who initiated ramucirumab at a US Oncology Network (USON) clinic between 21Apr2014 (date of first US approval for ramucirumab) and 30Jun2016
- The USON treats approximately 10% of patients in the community oncology setting using treatment pathways to deliver evidence-based and standardized care
- Patients were classified into ramucirumab monotherapy and combination therapy subgroups based on their initial administration

Data Sources:

- The electronic healthcare record of the USON
- The Social Security Death Index

Follow-up

- Patients were followed from initiation of ramucirumab until 30Jun2016 or date of last contact, whichever occurred first
- For the outcomes analysis, patients were required to have at least 3 months of follow-up time

Statistical analysis:

- Descriptive analyses were performed to assess the study population and ramucirumab treatment by line of therapy (LOT)
- Multivariable logistic regression models including baseline demographic and clinical factors were used to evaluate predictors for the use of ramucirumab monotherapy or combination therapy
- Overall survival was evaluated using the Kaplan-Meier method

Figure 1. Patient attrition

Patients with a documented diagnosis of gastric or esophageal cancer (n=21,563)

↓

Patients seen at USON clinics utilizing the full EHR capacities of iKM (n=19,150)

↓

Patient with ≥ 2 office visits at USON clinics (n=16,751)

↓

Exclusion of patients enrolled in a clinical trial (n=16,637)

↓

Patients who received an order for ramucirumab during the study period and have evidence of that order being administered (n=505)

↓

Patients aged ≥ 18 years at initiation of ramucirumab (n=505)

↓

Final study population (n=505)

↓

Subgroups:

Monotherapy (n=115)
Combination therapy (n=390)

RESULTS

Table 1. Baseline and clinical demographics

	Total study cohort (n = 505)	Monotherapy subgroup (n = 115)	Combination therapy subgroup (n = 390)	P-value
Median age at index (years; IQR)	65.1(57.4,72.3)	67.5(60.4,75.8)	64.1(57.0,71.0)	0.0006○
Male - n (%)	379(75.1%)	85(73.9%)	294(75.4%)	0.7486▫
Race - n (%)				
Black	32(6.3%)	8(7.0%)	24(6.2%)	0.9558▫
White	407(80.6%)	93(80.9%)	314(80.5%)	
Other	25(5.0%)	6(5.2%)	19(4.9%)	
Not documented	41(8.1%)	8(7.0%)	33(8.5%)	--
Payer information - n (%)				
Medicaid	22(4.4%)	3(2.6%)	19(4.9%)	0.0220◊
Medicare	197(39.0%)	54(47.0%)	143(36.7%)	
Private	174(34.5%)	30(26.1%)	144(36.9%)	
Other	18(3.6%)	1(0.9%)	17(4.4%)	
Not documented	94(18.6%)	27(23.5%)	67(17.2%)	--
Physician patient volume (patients/year) - n (%)				
< 100	374(74.1%)	96(83.5%)	278(71.3%)	0.0205◊
100 – 199	121(24.0%)	17(14.8%)	104(26.7%)	
200+	10(2.0%)	2(1.7%)	8(2.1%)	
Median time since initial diagnosis (months; IQR)	10.1(5.8,18.1)	12.6 (6.6,22.7)	9.7(5.5,17.6)	0.0318○
Median time since prior therapy to start of ramucirumab (months; IQR)	1.0(0.7,2.3)	0.9(0.7,1.7)	1.0(0.7,2.6)	0.1151○
Stage at initial gastric/GEJ cancer diagnosis - n (%)				
I	19(3.8%)	4(3.5%)	15(3.9%)	0.9717◊
II	72(14.3%)	16(13.9%)	56(14.4%)	
III	99(19.6%)	24(20.9%)	75(19.2%)	
IV	285(56.4%)	62(53.9%)	223(57.2%)	
Not documented	30(5.9%)	9(7.8%)	21(5.4%)	--
ECOG PS=0 - n (%)	25(5.0%)	3(2.6%)	22(5.6%)	0.4136◊
Metastatic disease at baseline - n (%)	489(96.8%)	113(98.3%)	376(96.4%)	0.5435◊
Any treatment prior to ramucirumab* - n (%)	433(85.7)	90(78.3%)	343(88.0%)	0.009▫
Prior treatments containing the following agents* - n (%)				
Anti-angiogenic	1(0.2%)	0(0.0%)	1(0.3%)	1.000◊
Antineoplastic	6(1.2%)	0(0.0%)	6(1.5%)	0.1809◊
Irinotecan	48(9.5%)	17(14.8%)	31(8.0%)	0.0281▫
Anthracycline	67(13.3%)	8(7.0%)	59(15.1%)	0.0232▫
Trastuzumab	75(14.9%)	16(13.9%)	59(15.13%)	0.7474▫
Taxane	146(28.9%)	38(33.0%)	108(27.7%)	0.2660▫
Platinum	316(62.6%)	47(40.8%)	269(69.0%)	< 0.0001▫
Fluoropyrimidine	334(66.1%)	49(42.6%)	285(73.1%)	< 0.0001▫

Abbreviations: ECOG PS, Eastern Cooperative Oncology Group performance status; GEJ, gastroesophageal junction; IQR, interquartile range

**Prior treatments were assessed in the 6-month period prior to ramucirumab initiation*

◊ P-value calculated based on a Fisher's Exact test; ○ Kruskal-Wallis test; ▫ Chi-square test

Demographic and clinical characteristics

- The majority of the study population were male (75.1%) with a median age of 65.1 years
- Patients who received monotherapy were older (67.5 vs. 64.1 years, p=0.0006) and started ramucirumab treatment later following gastric/GEJ cancer diagnosis (12.6 vs. 9.7 months, p=0.0318) than those who received combination therapy
- Significantly more patients in the combination therapy subgroup had received prior treatment than those in the monotherapy subgroup (88.0% vs. 78.3%; p=0.009)

Treatment characteristics

- A higher proportion of patients who received combination therapy also received prior treatment with platinum- (69.0% vs. 40.9%; p<0.0001) or fluoropyrimidine-containing regimens (73.1% vs. 42.6%; p<0.0001)
- Most patients started ramucirumab in the second-line setting (N=407, 80.6% overall; 56.5% of monotherapy and 87.7% of combination therapy users)
- When assessing ramucirumab infusions by LOT, the mean number of ramucirumab infusions was higher in the 4th LOT for combination therapy versus monotherapy (6.7 vs. 4.9, p=0.0190); no differences in the number of infusions were observed for other LOTs

Clinical outcomes

- The mean overall follow-up time from the index diagnosis to the last contact date, end of study period or date of death, whichever came first, was 338.7 days (± 195.7)
- Median survival for second-line monotherapy was 5.5 months (95% confidence interval [CI] 4.3, 7.8) and for second-line combination therapy was 7.4 months (95% CI 6.6, 8.8)

Table 2. Treatment patterns

	Total study cohort (n = 505)	Monotherapy subgroup (n = 115)	Combination therapy subgroup (n = 390)	P-value
First LOT with ramucirumab - n(%)				
LOT1	15(3.0%)	4(3.5%)	11(2.8%)	**< 0.0001◊**
LOT2	407(80.6%)	65(56.5%)	342(87.7%)	
LOT3	58(11.5%)	31(27.0%)	27(6.9%)	
LOT4+	18(3.6%)	13(11.3%)	5(1.3%)	
LOT unknown	7(1.4%)	2(1.7%)	5(1.3%)	--
Median ramucirumab starting dose (mg/kg; IQR)	8.0(8.0,8.0)	8.0(8.0,8.0)	8.0(8.0,8.0)	1.000○
Treatment immediately following ramucirumab discontinuation - n (%)				
Any subsequent treatment	145(28.7%)	35(30.4%)	110(28.2%)	0.6423▫
Treatments containing the following agents				
Anthracycline	6(1.2%)	1(0.9%)	5(1.3%)	1.000◊
Antineoplastic	9(1.8%)	4(3.5%)	5(1.3%)	0.1247◊
Trastuzumab	17(3.4%)	7(6.1%)	10(2.6%)	0.0657▫
Platinum	23(4.6%)	8(7.0%)	15(3.9%)	0.1598▫
Taxane	23(4.6%)	7(6.1%)	16(4.1%)	0.3697▫
Irinotecan	41(8.1%)	6(5.2%)	35(9.0%)	0.1949▫
Fluoropyrimidine	51(10.1%)	13(11.3%)	38(9.7%)	0.6254▫
Mean number of ramucirumab infusions (SD)				
Across all LOTs	6.9 (6.1)	6.8(6.7)	6.9(5.9)	0.6329○
LOT1	5.5(4.2)	5.5(2.1)	5.6(4.8)	0.5109○
LOT2	7.1(6.3)	7.7(7.6)	7.0(6.1)	0.4358○
LOT3	6.0(5.0)	6.1(4.7)	5.9(5.4)	0.6567○
LOT4+	5.5(6.1)	4.9(7.1)	6.7(3.1)	**0.0190○**
LOT Unknown	7.0(5.3)	2.5(0.7)	8.5(5.3)	0.0651○

Abbreviations: kg, kilograms; LOT, line of therapy; mg, milligrams; SD, standard deviation; IQR, interquartile range

◊ P-value calculated based on a Fisher's Exact test; ○ Kruskal-Wallis test; ▫ Chi-square test

Figure 2. Overall survival

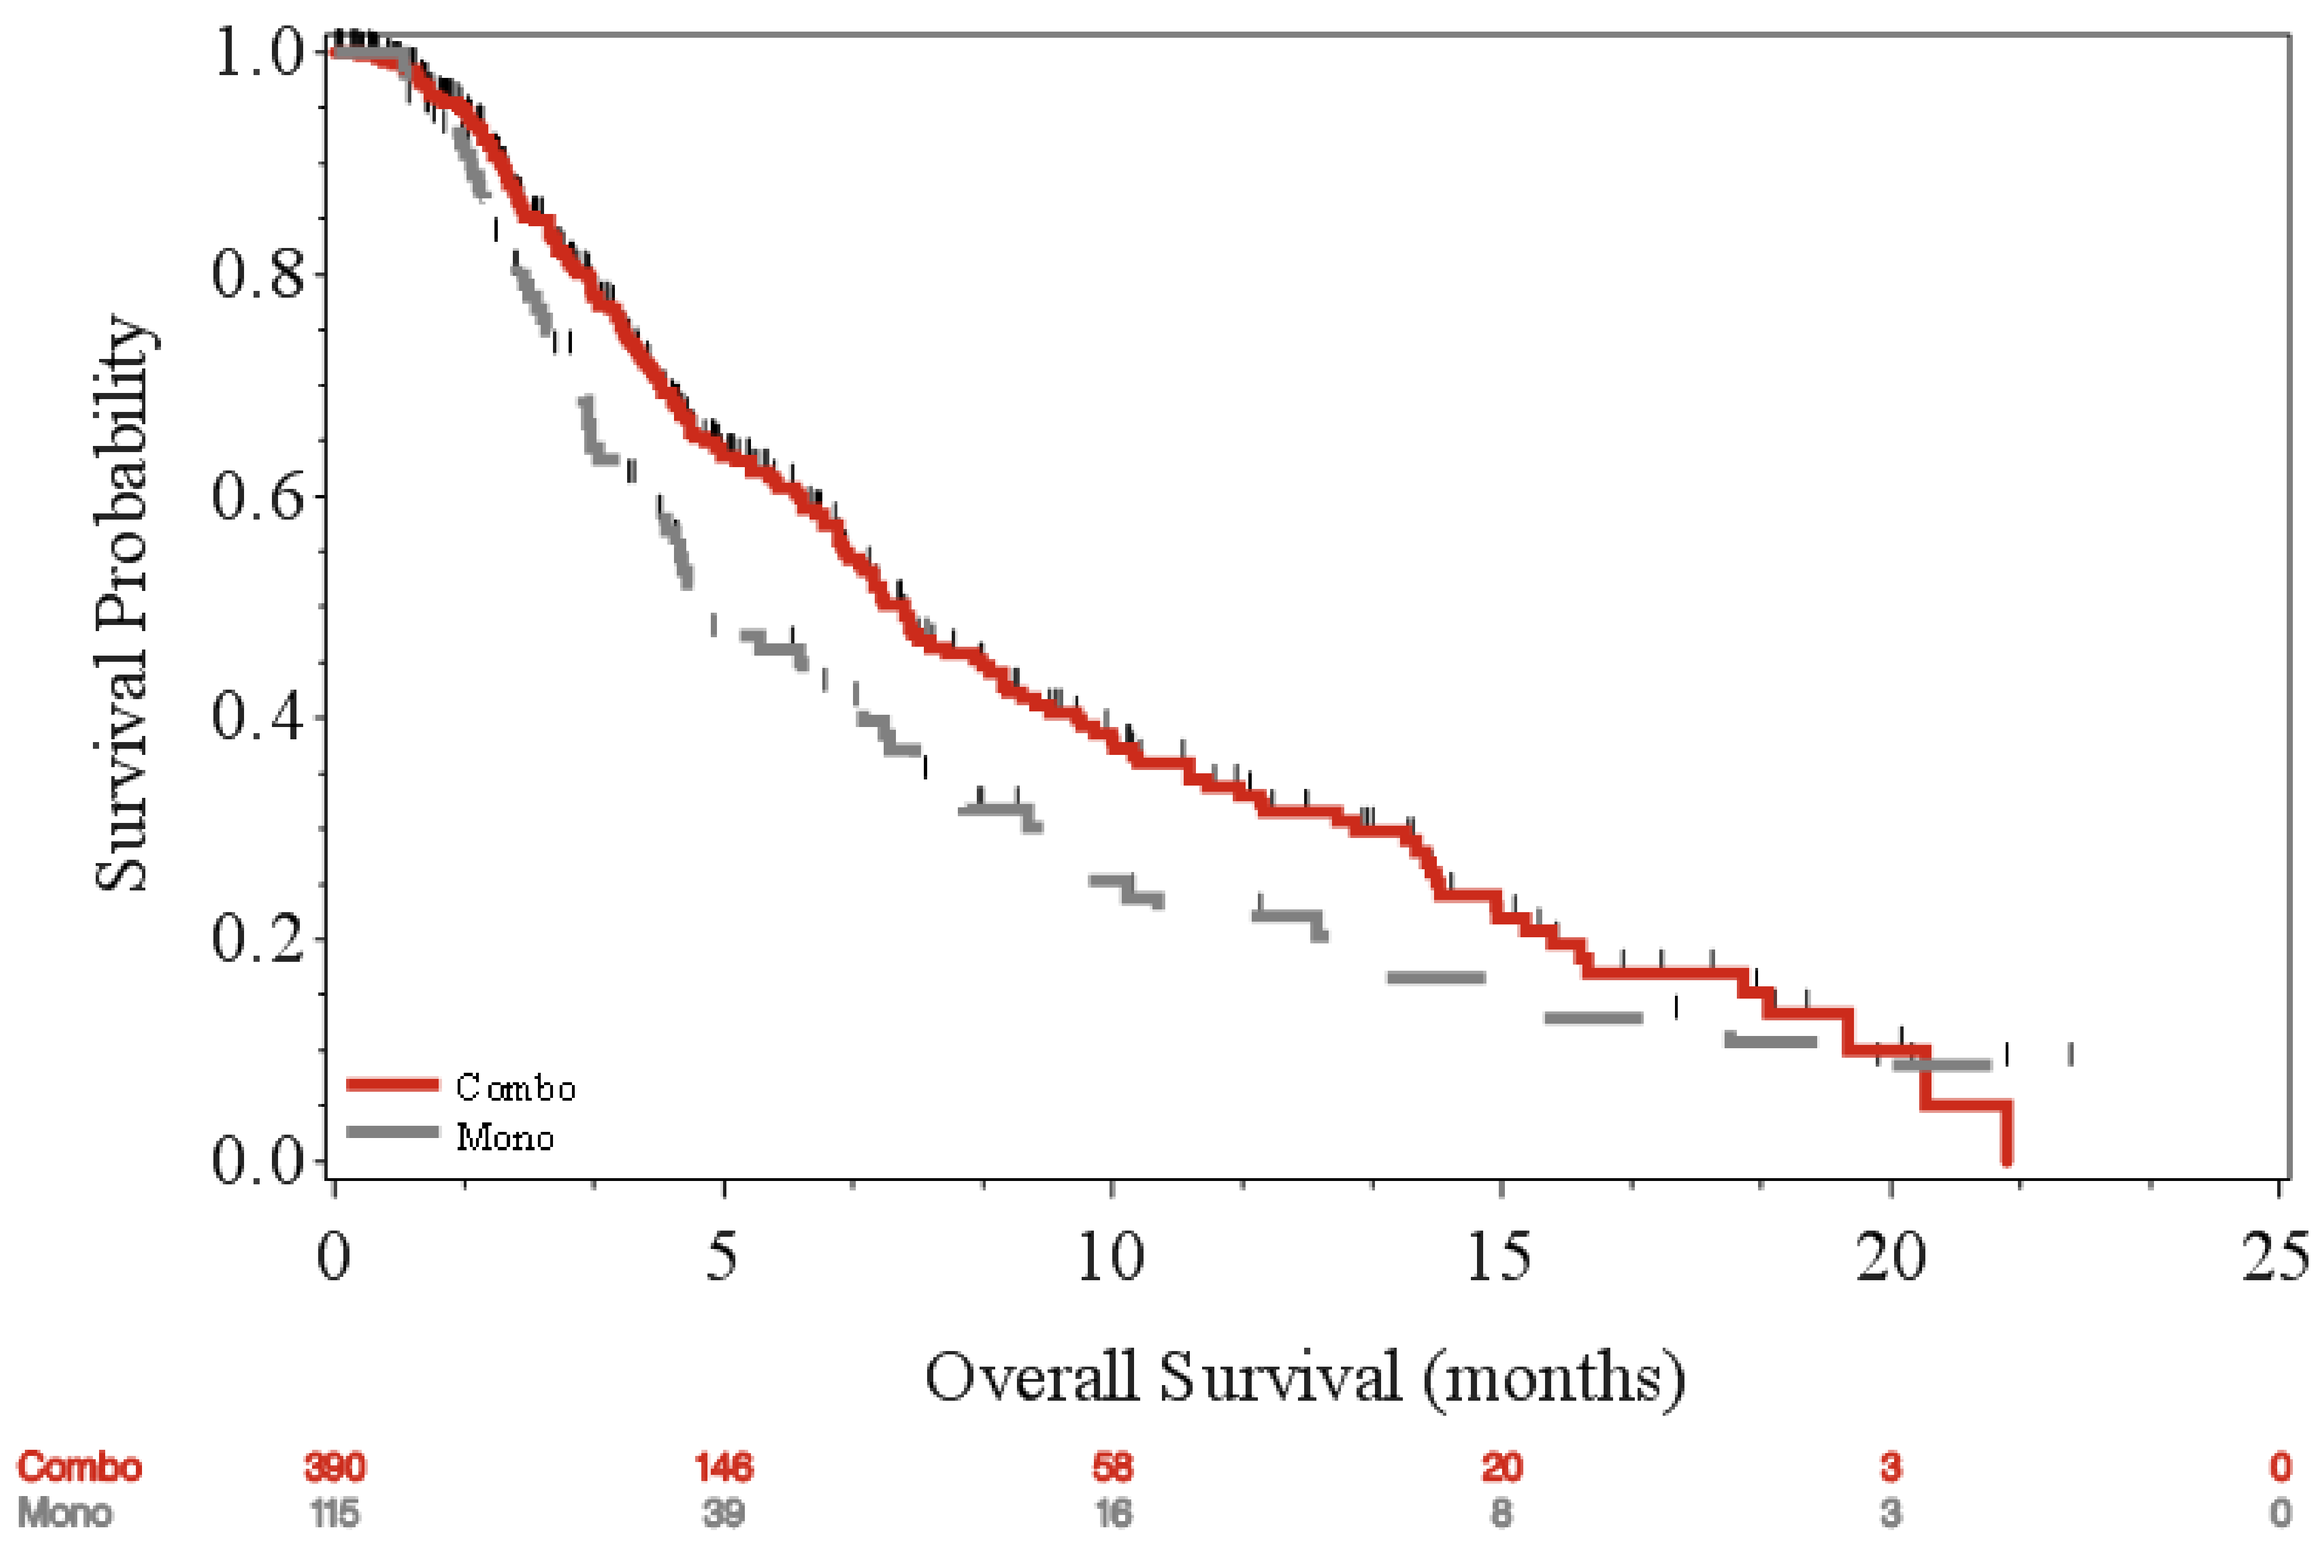
1.0
0.8
0.6
0.4
0.2
0.0
Survival Probability
0
5
10
15
20
25
Overall Survival (months)
Combo
Mono
Combo 390 146 58 20 3 0
Mono 115 39 16 8 3 0

Predictive factors associated with ramucirumab use

- In the multivariable analysis, two factors were significantly associated with monotherapy:
- Lower use of a prior fluoropyrimidine-containing therapy (odds ratio [OR]=0.33; p<0.0001)
- Later LOT of ramucirumab initiation (OR=9.82 for LOT4 vs. LOT1; p=0.0047 and OR=4.39 for LOT3 vs. LOT1; p=0.0244)

Variable	Level	N (monotherapy subgroup)	OR	95% Lower CI	95% Upper CI	Individual effect P-value	Overall effect P-value
Prior fluoropyrimidine-containing treatment	No (ref)	171 (66)	--	--	--	--	**<0.0001**
	Yes	334 (49)	0.332	0.207	0.532	**<0.0001**	
LOT of ramucirumab initiation	1 (ref)	15 (4)	--	--	--	--	**<0.0001**
	2	407 (65)	0.894	0.267	2.997	0.8565	
	3	58 (31)	4.385	1.210	15.883	**0.0244**	
	4+	18 (13)	9.820	2.015	47.846	**0.0047**	

Abbreviations: CI, confidence interval; LOT, line of therapy; ref, reference

CONCLUSIONS

- The majority of the patients in this community oncology setting were treated with combination therapy
- Demographic and patient characteristics of gastric/GEJ cancer patients were similar to clinical trial populations[1,2]
- Overall survival estimates were consistent with phase III registration clinical trials, which demonstrated a median OS of 5.2 (95% CI:4.4-5.6) and 8.6 (95% CI: 7.4-9.8) months for monotherapy and combination therapy, respectively, in the North American/European populations[1,2]
- As expected, patients previously treated with fluoropyrimidine- and platinum-containing regimens were more likely to receive ramucirumab monotherapy; no unexpected predictors of monotherapy or combination therapy use were found
- Based on these findings, it appears that community oncology practices are demonstrating the similar clinical outcomes as observed in clinical trials or academic medical centers
- Future research can expand upon these finding to explore other factors associated with use of ramucirumab and associated clinical outcomes

References:

[1]Fuchs CS, Tomasek J, Yong CJ, Dumitru F, Passalacqua R, Goswami C, et al. Ramucirumab monotherapy for previously treated advanced gastric or gastro-oesophageal junction adenocarcinoma (REGARD): an international, randomised, multicentre, placebo-controlled, phase 3 trial. Lancet (London, England). 2014;383(9911):31-9. Epub 2013/10/08. doi: 10.1016/s0140-6736(13)61719-5. PubMed PMID: 24094768.
[2]Wilke H, Muro K, Van Cutsem E, Oh SC, Bodoky G, Shimada Y, et al. Ramucirumab plus paclitaxel versus placebo plus paclitaxel in patients with previously treated advanced gastric or gastro-oesophageal junction adenocarcinoma (RAINBOW): a double-blind, randomised phase 3 trial. The Lancet Oncology. 2014;15(11):1224-35. Epub 2014/09/23. doi: 10.1016/s1470-2045(14)70420-6. PubMed PMID: 25240821

Presented ESMO World Congress on Gastrointestinal Cancer 2017; Barcelona, Spain - 28 June - 01 July 2017

Sponsored by Eli Lilly and Company and/or one of its subsidiaries

19WCGIC 77--P

Clinical Gastric Cancer

William Schelman

DOI: 10.3252/pso.eu.19wcgic.2017

Poster presented at: