SLOVAK HEMOPHILIA SOCIETY

The Royal Family: A Project to Increase Awareness of Hemophilia in Slovakia

Jaroslav Janovec, Slovak Hemophilia Society
Katarina Zvedelova and Lele Zemanova, Bayer Slovakia

Objectives

- The aim was to create a unique piece of artwork; a symbol to remind Slovak people with hemophilia that their individuality is an important aspect of hemophilia
- The artwork also provides a communication strategy for the Slovak Hemophilia Society drawing public attention to this rare disease and inviting debate between professionals and the lay public

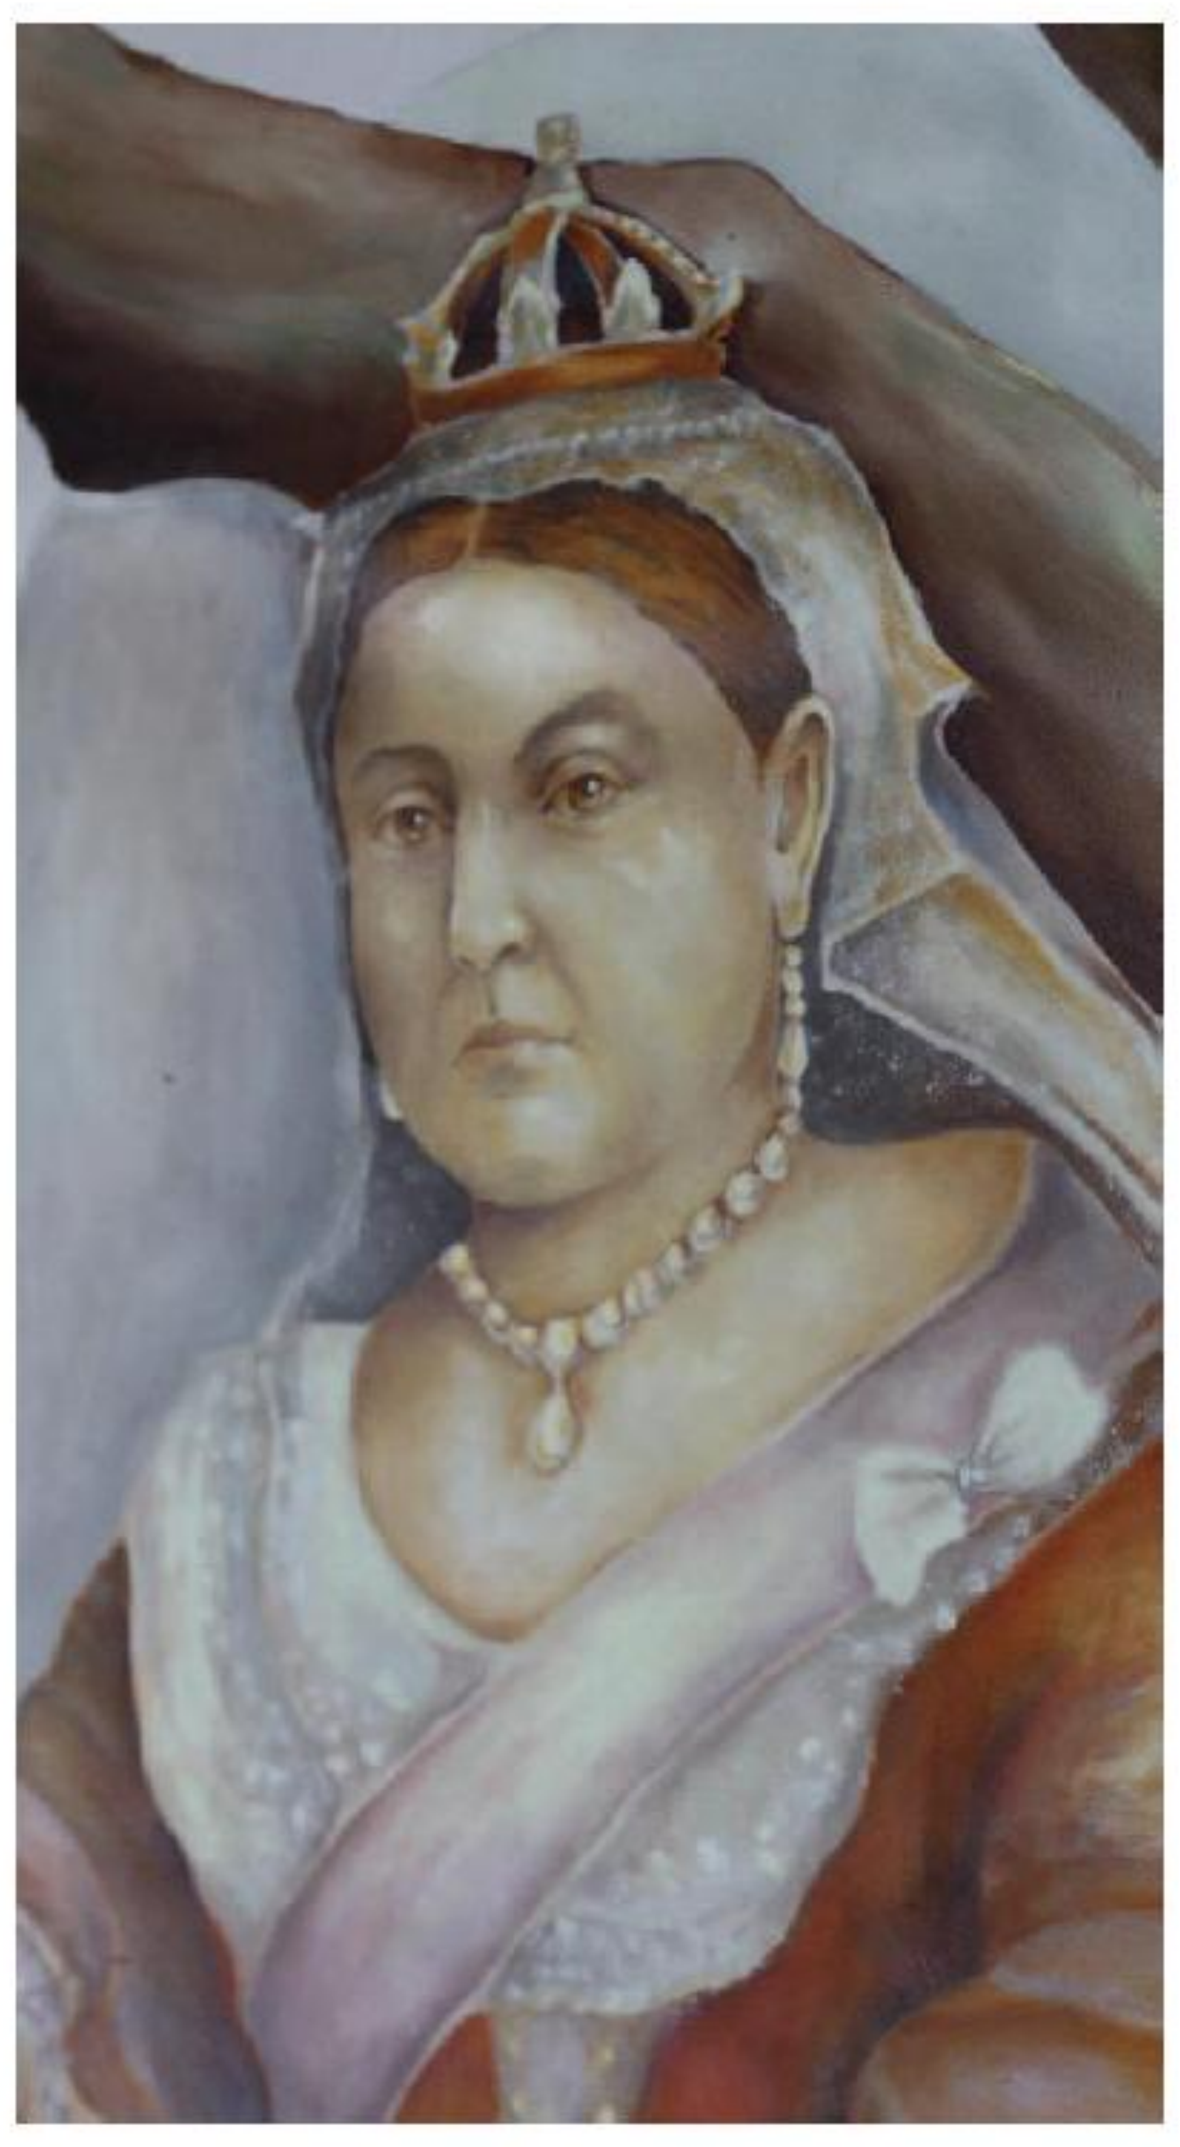

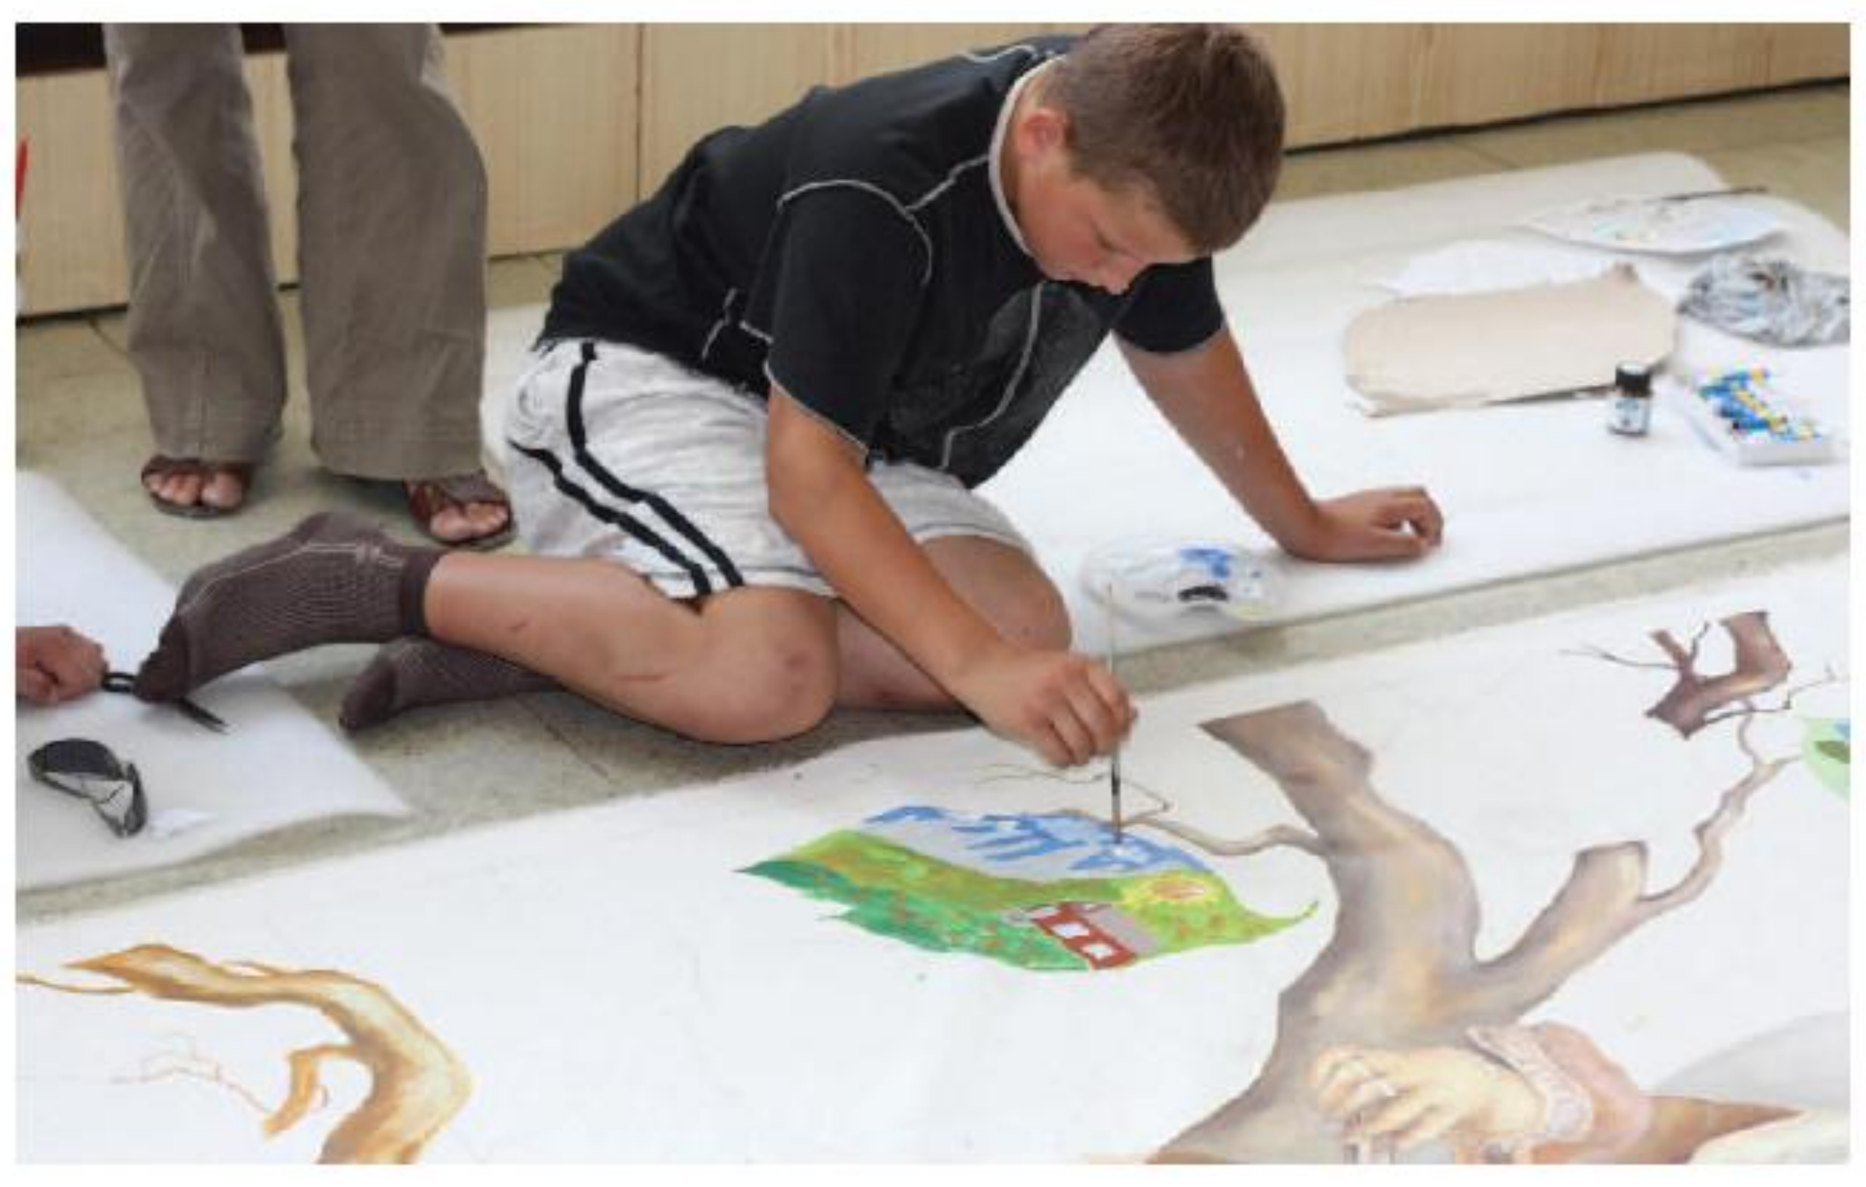

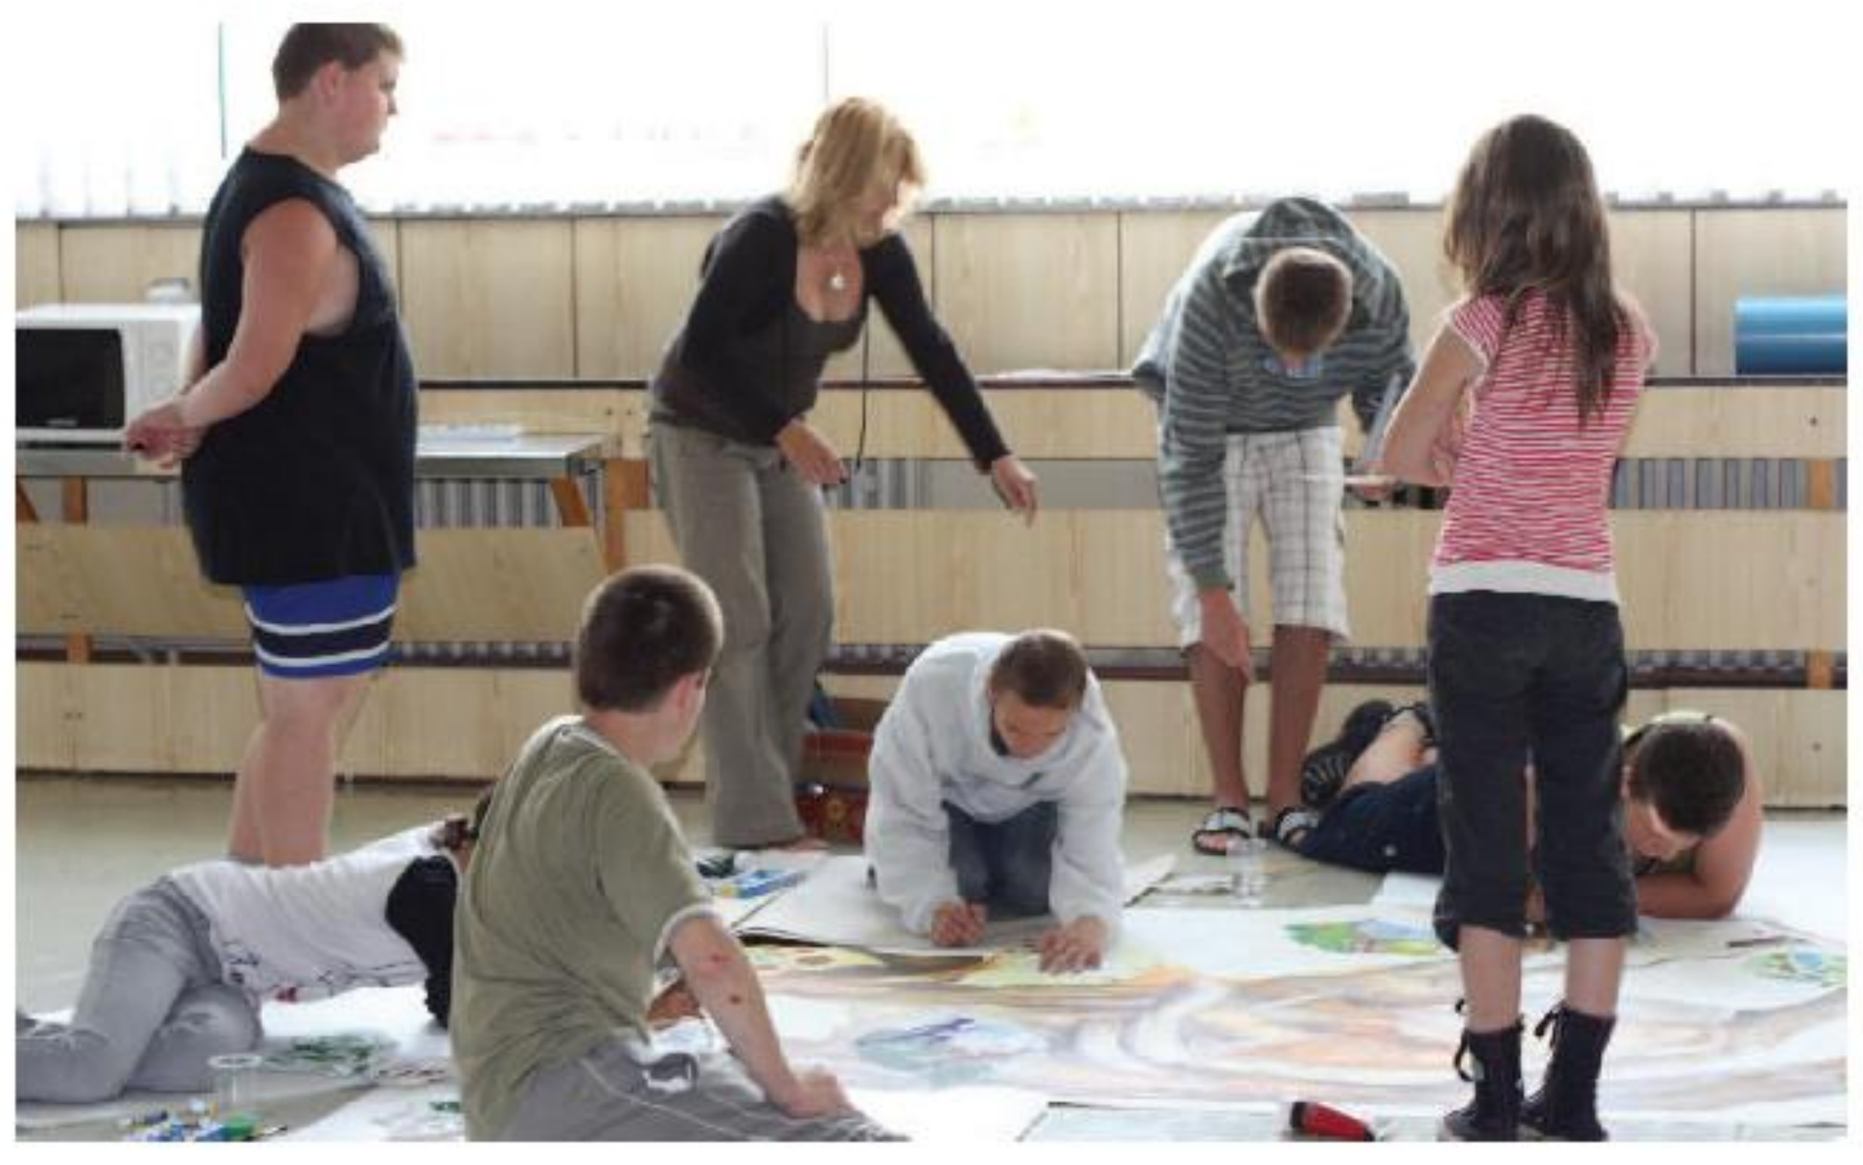

- The project presents the possibility of recombinant coagulation factors as a treatment for hemophilia
- Joint activities with the Slovak Hemophilia Society were arranged throughout the year, showcasing the patient organization

Design Idea

- In August 2011, a group of children with hemophilia and Slovak painter Darina Berkova created a unique piece of artwork
- This image resembles a tapestry describing a family tree from the 18th century
- Through the image the public can learn more about congenital bleeding disorders
- This image is used for various promotional purposes

Exhibitions

- The picture was exhibited in the eight Slovak cities
- The first exhibition took place on 9th September 2010 in a shopping center in Nitra. The travelling exhibition culminated in April 2011 on World Hemophilia Day at the International Hemophilia Congress in the capital cit y Bratislava
- The image was installed in the main foyer of the Slovak National Theater from 14th April until 6th May. Guests (healthcare professionals, people with hemophilia, journalists and Bayer employees) were each given a badge to wear: a symbol of a Royal crown

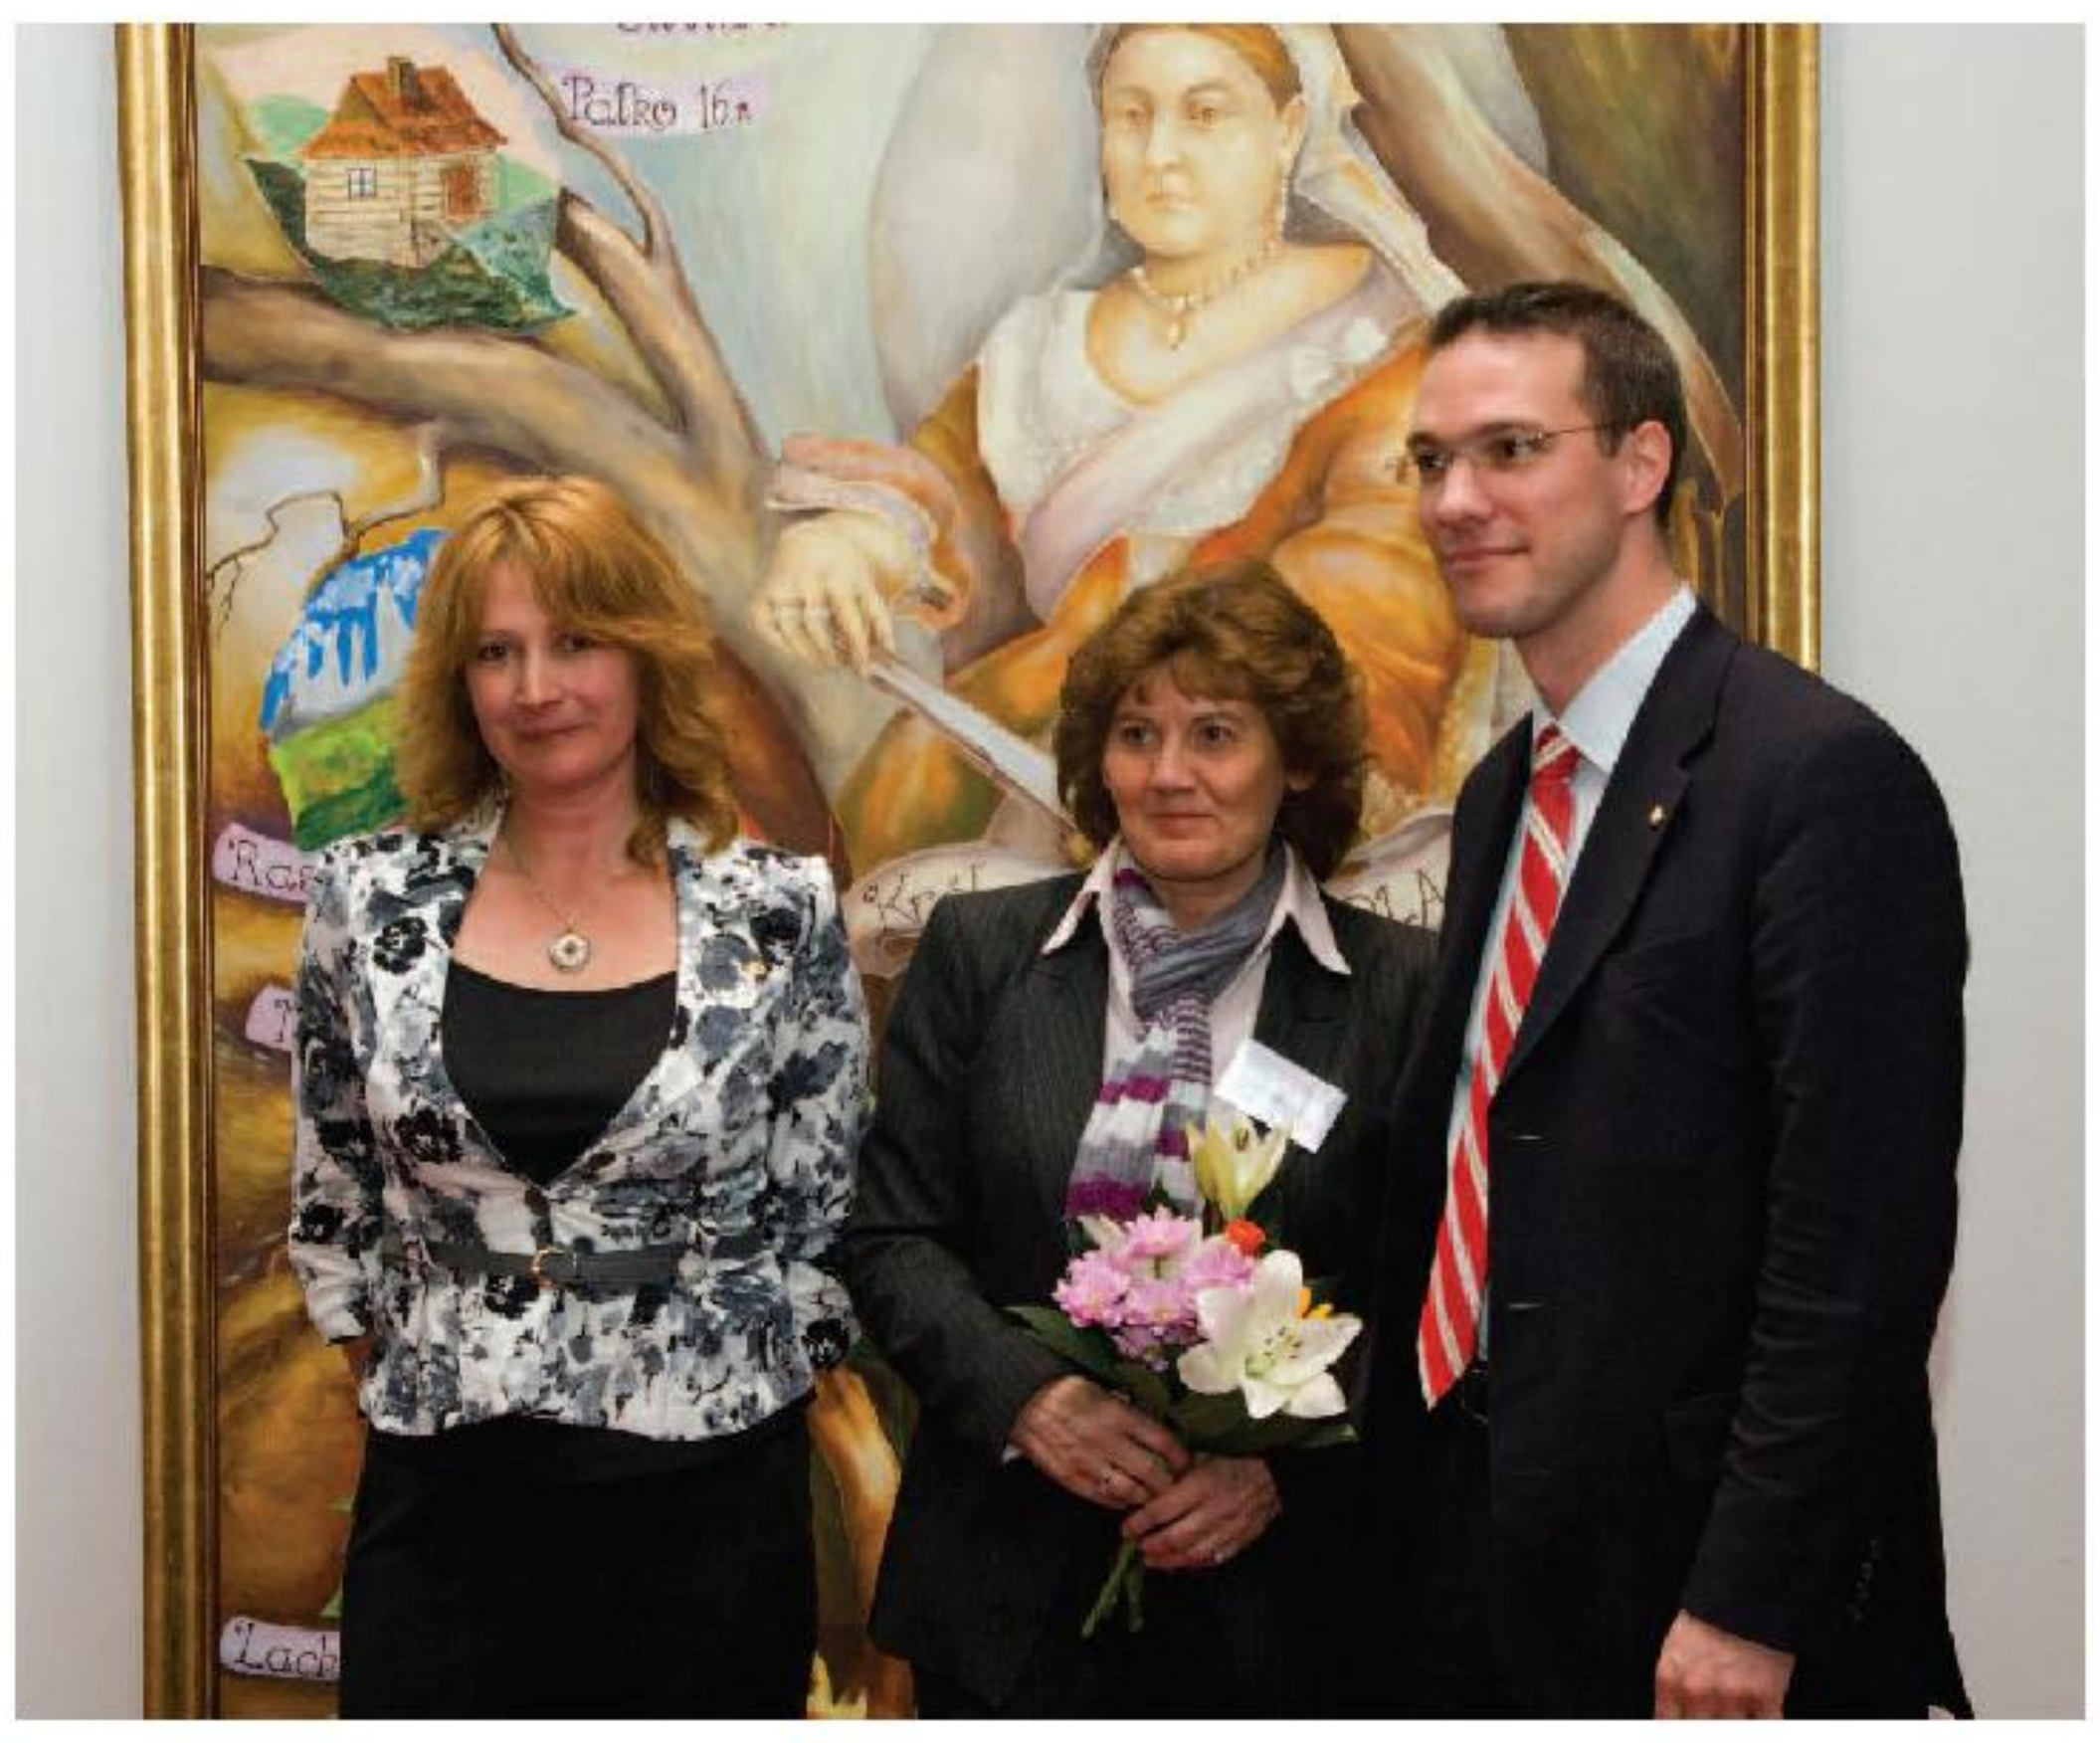

From left: Painter D. Berkova, President of National Hemophilia Center Doc. MUDr. A. Batorova, and Managing Director of Bayer Slovakia Dr. H. Nusser

Method

- Painter Darina Berkova spent two months preparing the concept and design of the image, which features Queen Victoria sitting on a tree trunk: a symbolic Royal throne
- Children drew the tree leaves, which symbolize their feelings and life stories
- 20 children aged 8-18 participated in the project

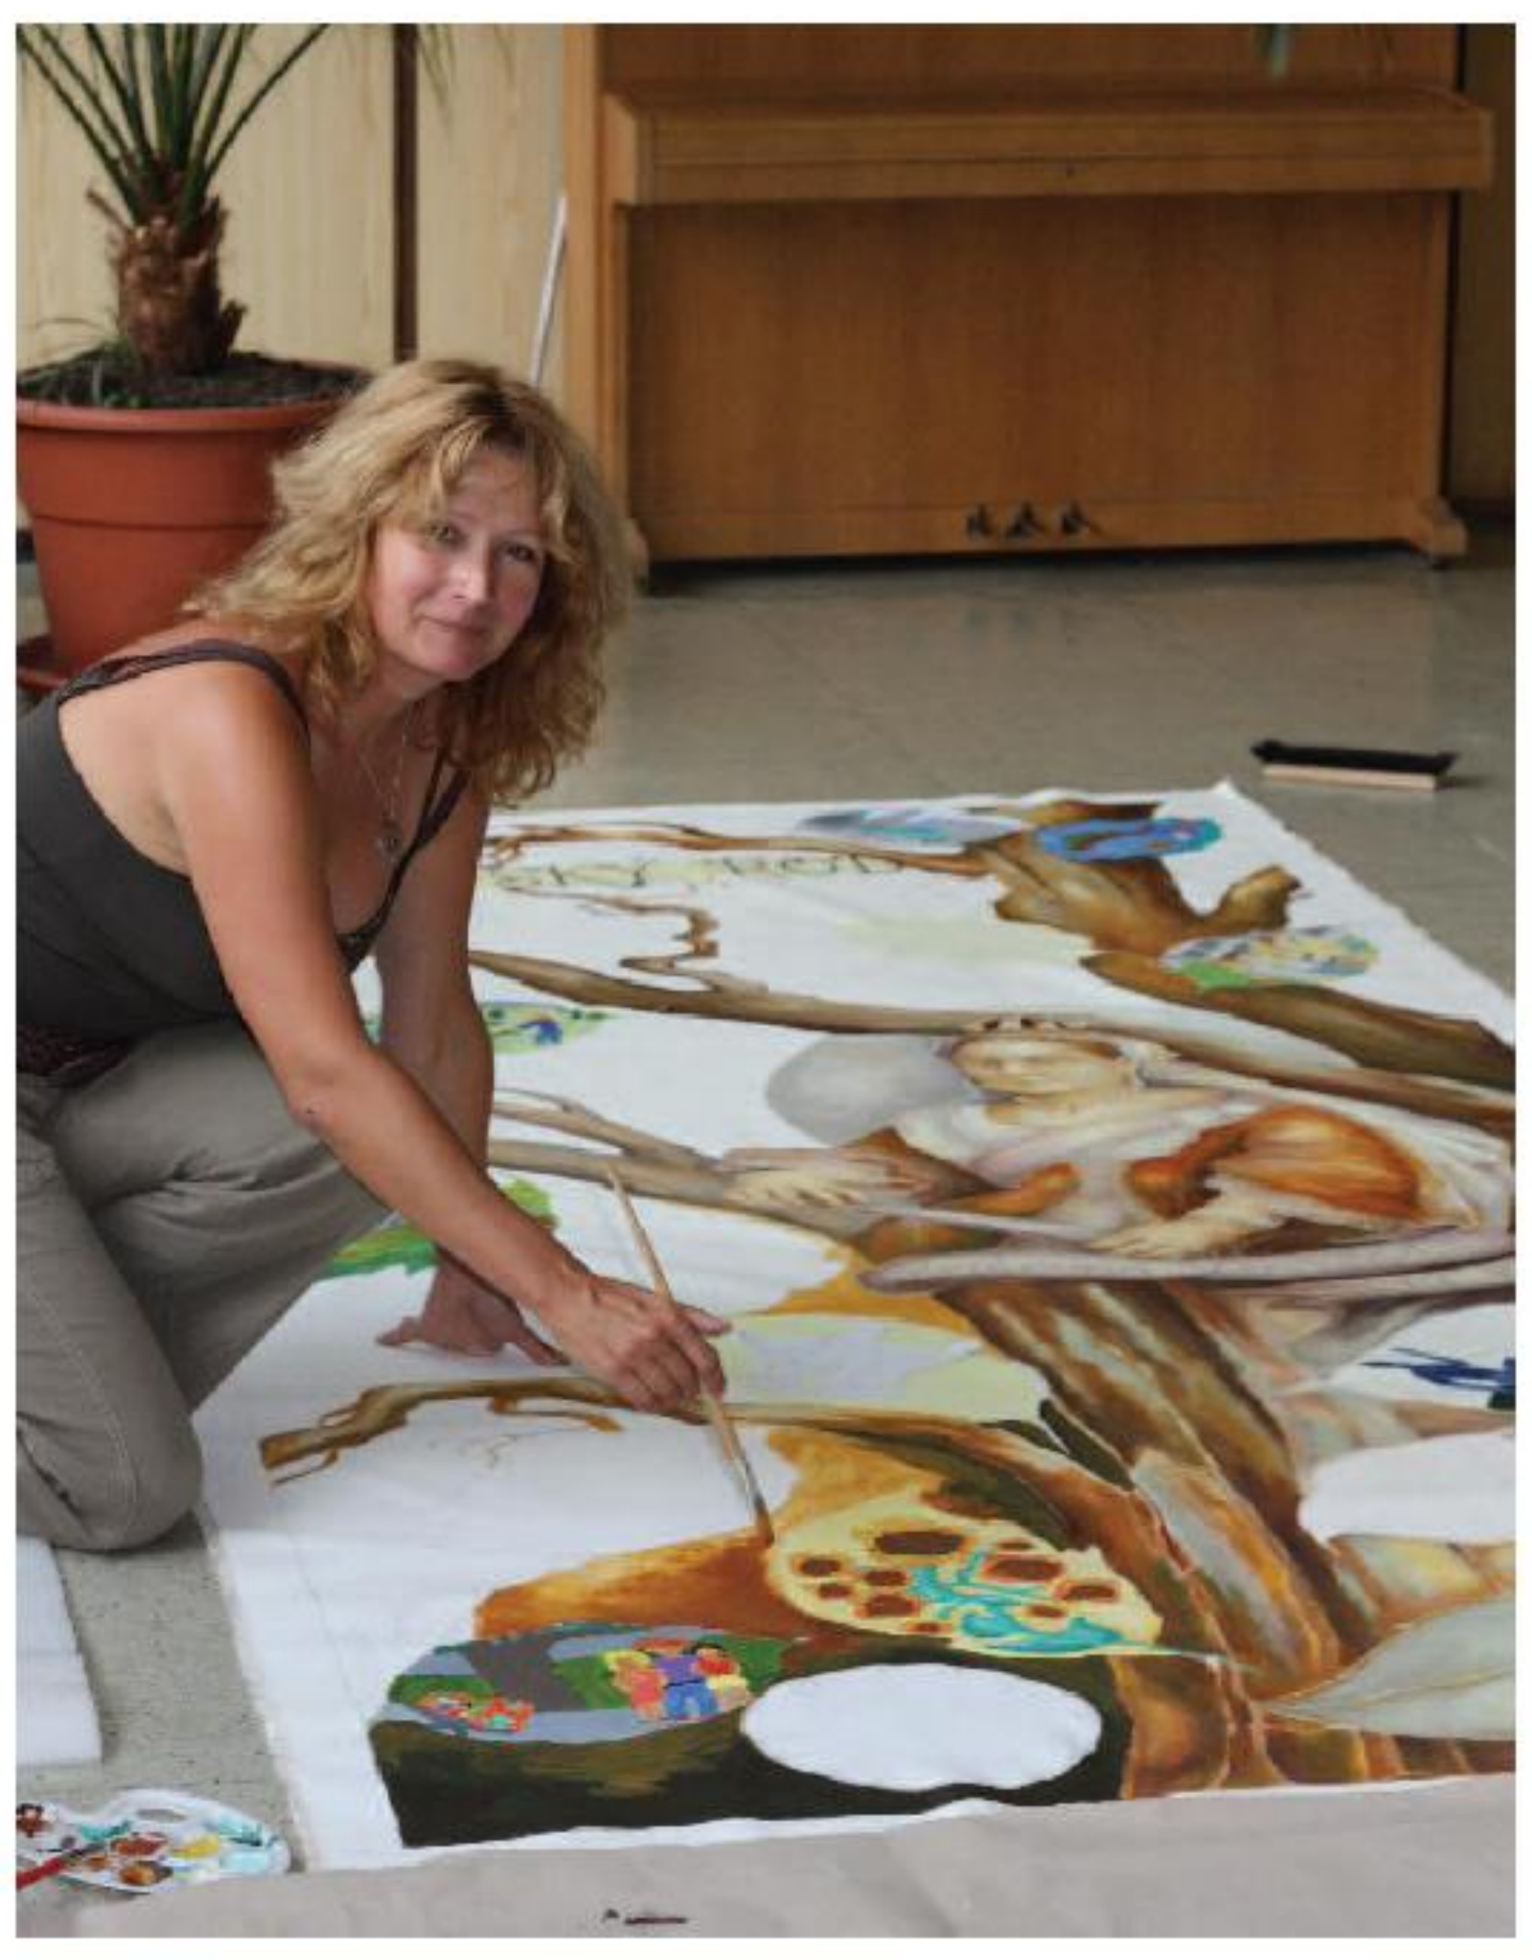

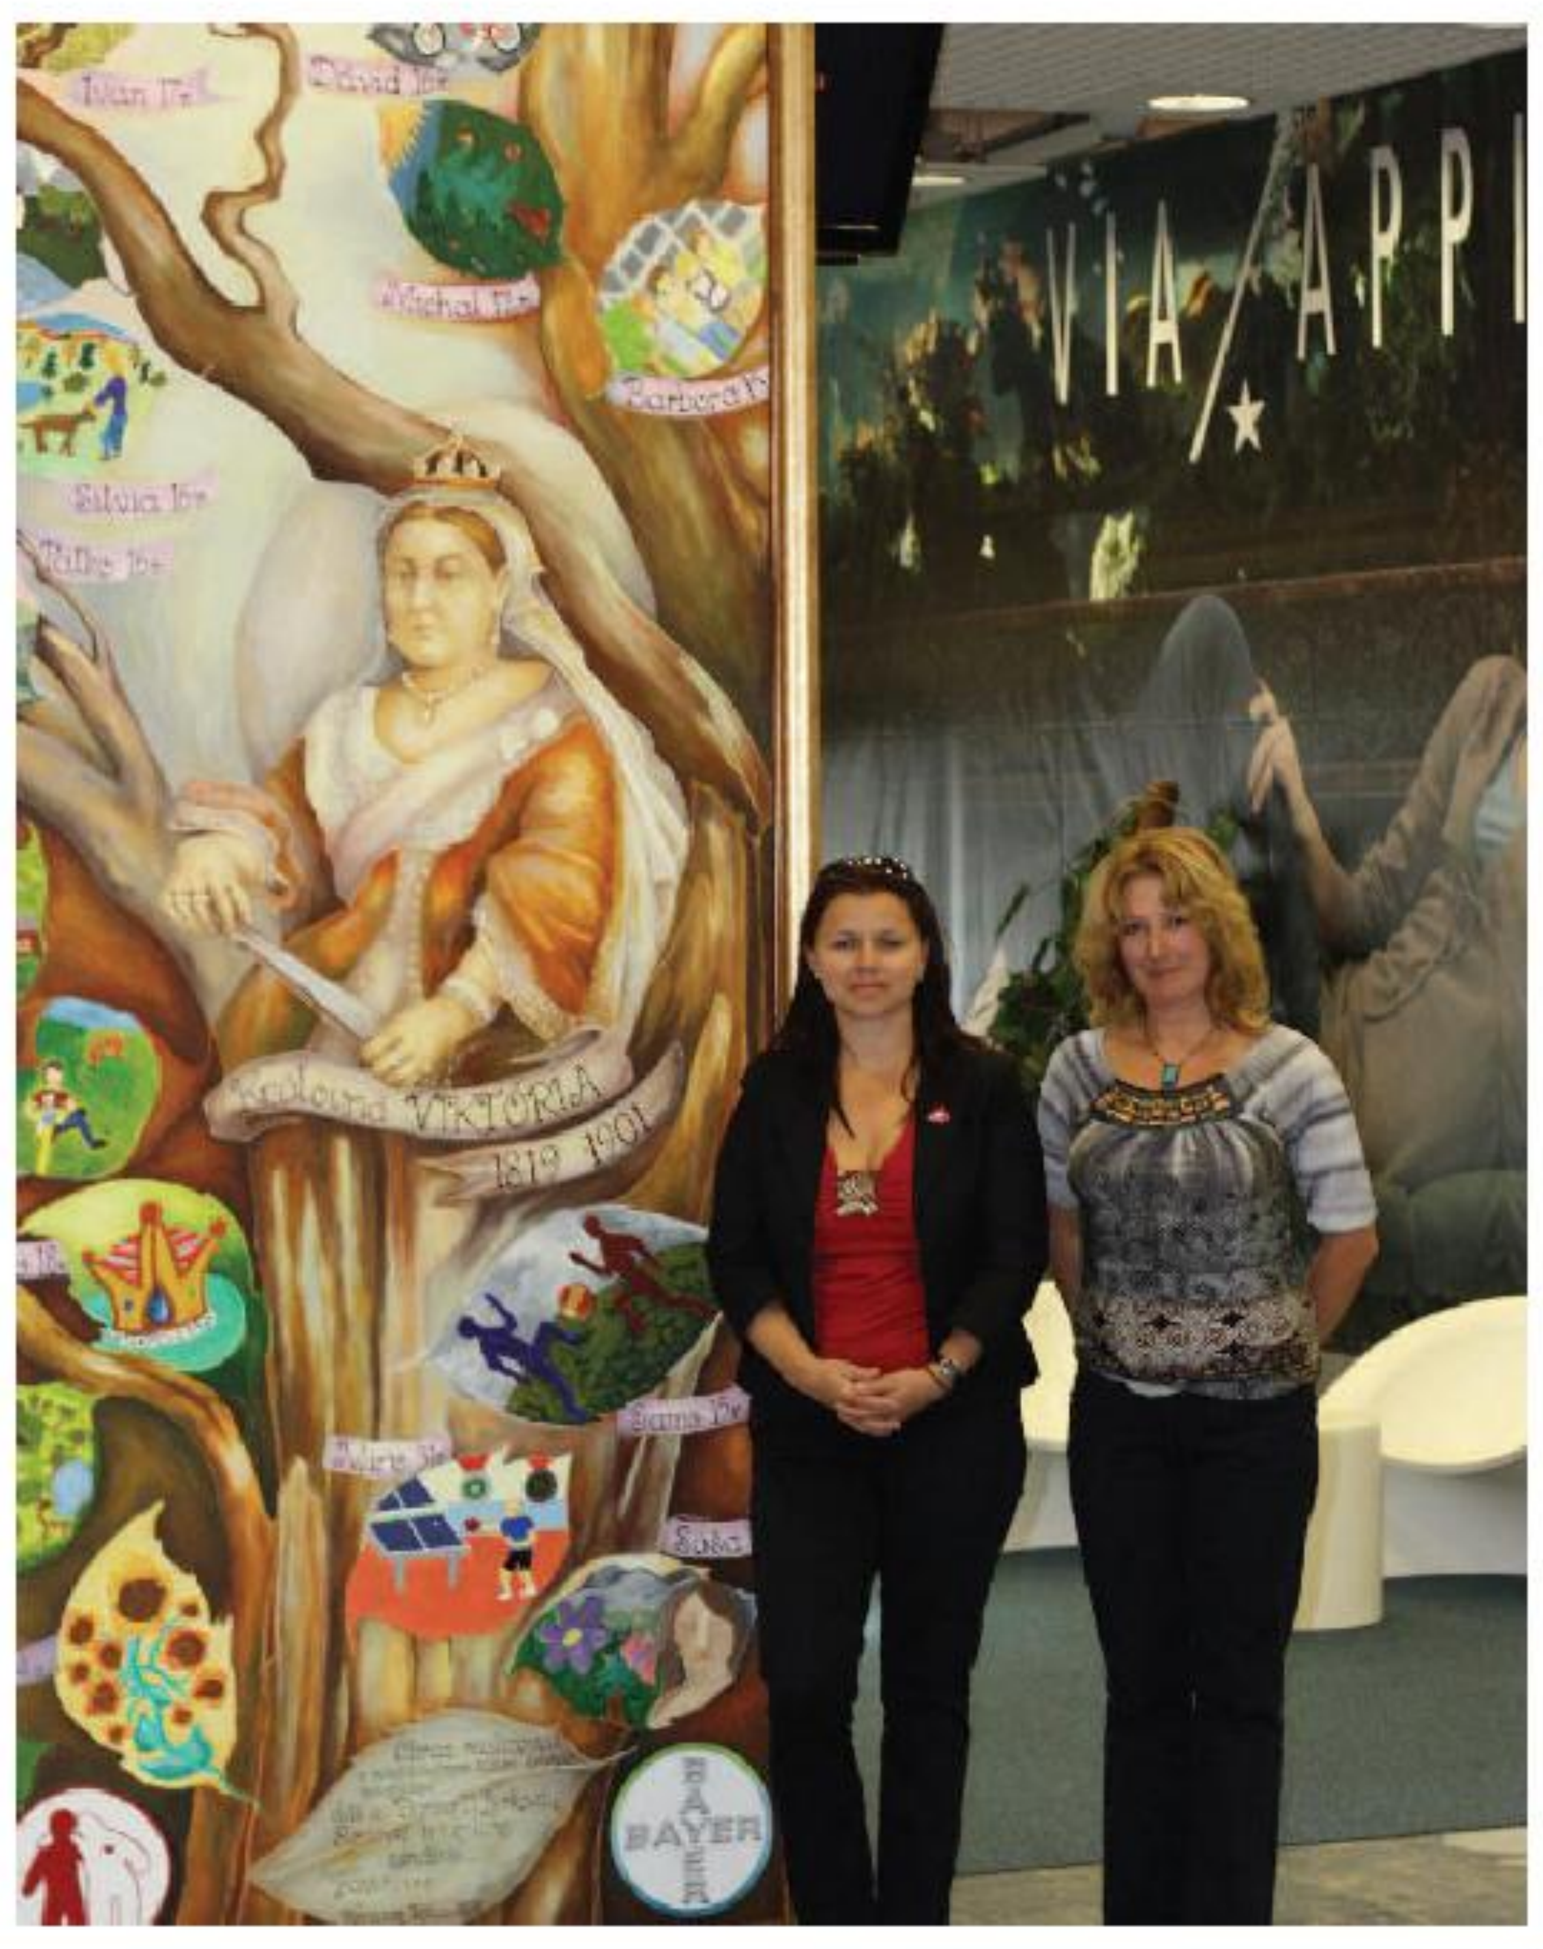

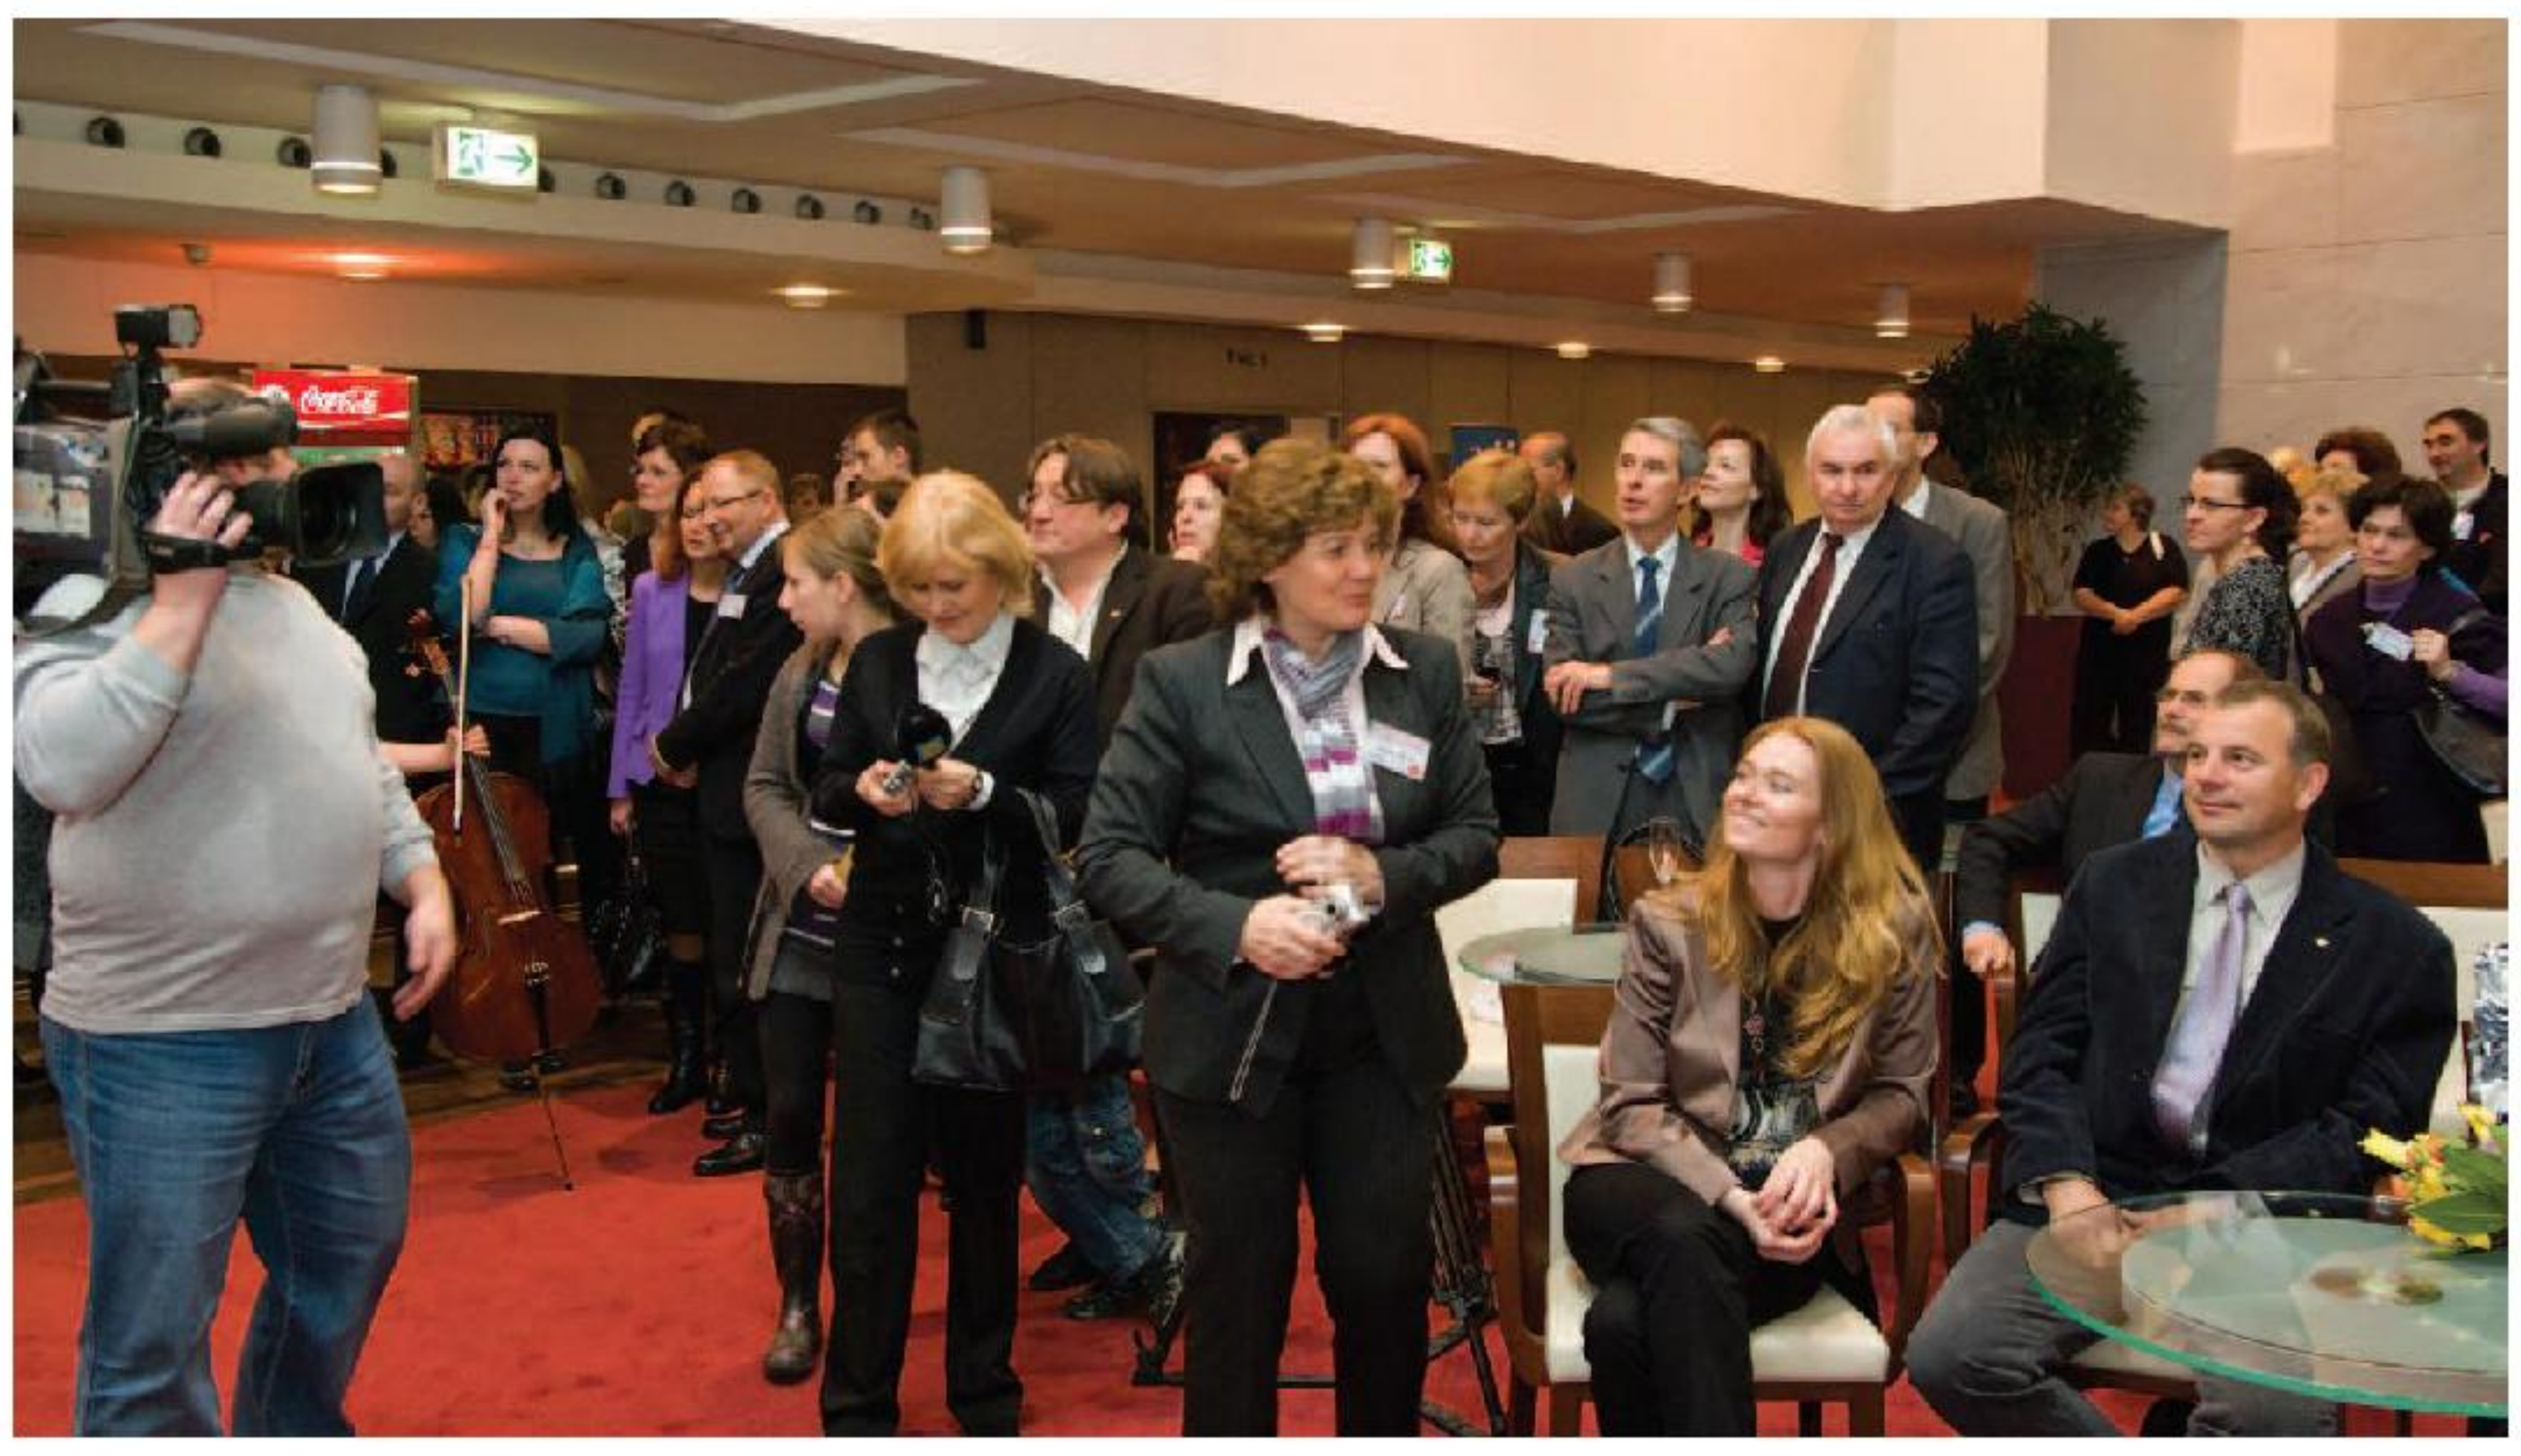

Conclusions

- The creation of this unique image created much interest in the Slovak media. Slovak TV broadcast the project on the main news, and there were many interviews on the radio and articles published
- This project was followed by a theater "Best Practice Award" at the Bayer EU PR Summit 2011 in Vienna
- The Royal Family project was supported by the National Hemophilia Center, in particular the Center's President, Doc. MUDr. Angelika Batorova. The project increases awareness of hemophilia throughout Slovakia and demon strates the possibility of treatment with recombinant coagulation factors

Royal crown badge distributed at the Slovak National Theater

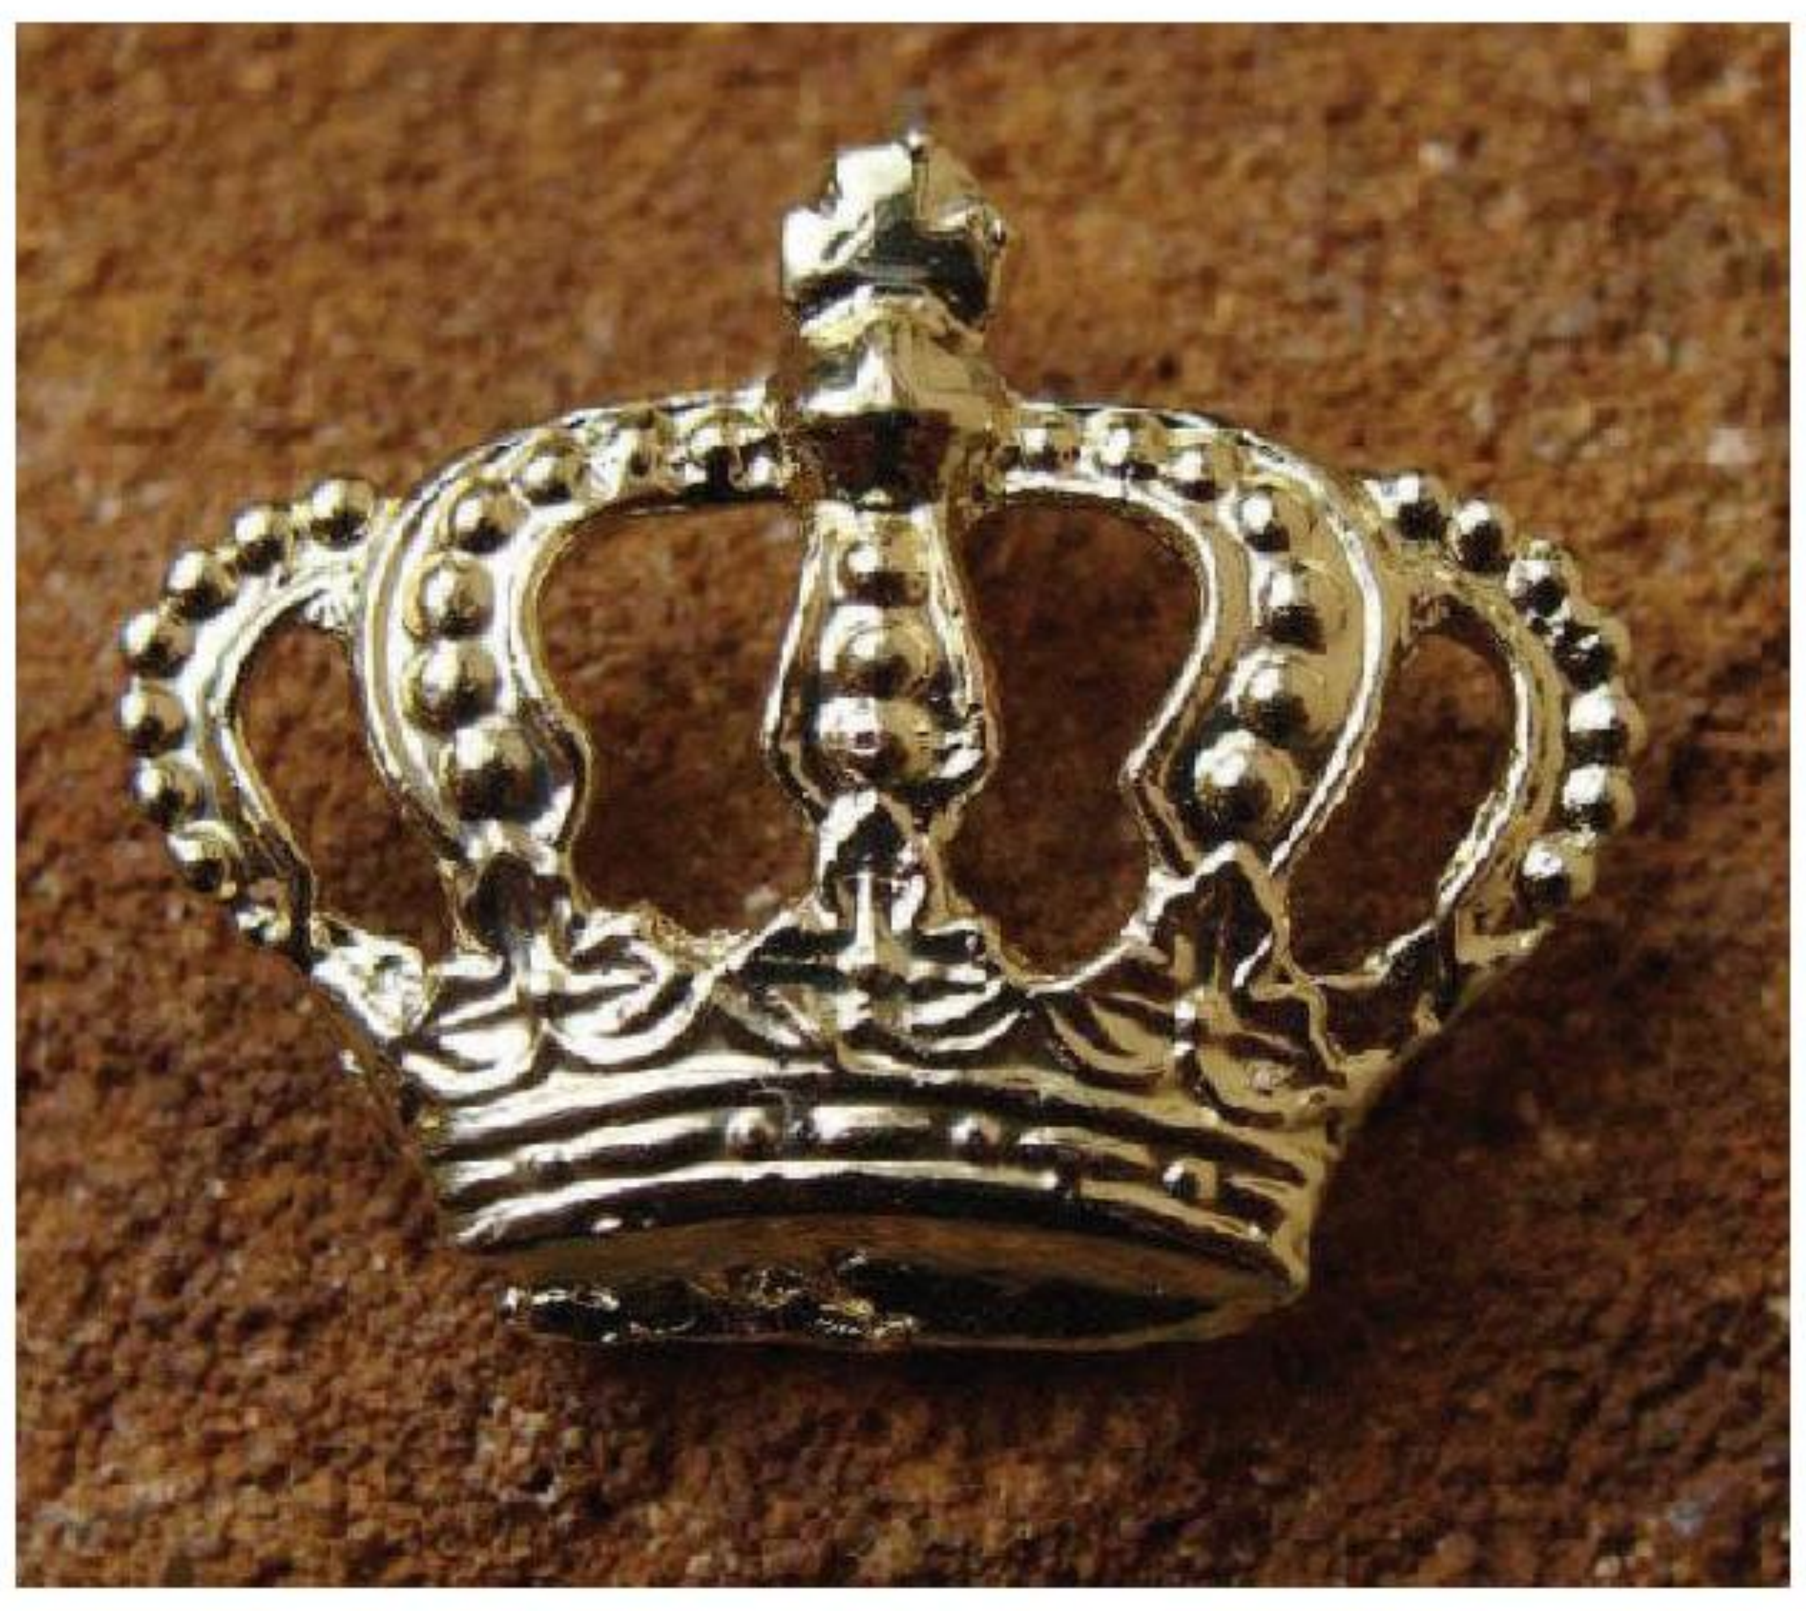

WFH2012 125--Tu

Other

Jaroslav Janovec

DOI: 10.3252/pso.eu.WFH2012.2012

Poster presented at: WFH 2012 World Congress

PosterSessionOnline.com