Physical and psychosocial benefits of exercise for patients with haemophilia

Claude Negrier,[1] Axel Seuser,[2] Angela Forsyth,[3] Sebastien Lobet,[4] Adolfo Llinas,[5] Maria Rosas[6] and Lily Heijnen[7]

1. Centre Régional de Traitement de l'Hémophilie, Lyon, France; 2. Kaiser-Karl-Klinik Bonn, Bonn, Germany; 3. Christiana Care Health System Hemophilia Program, Newark, DE, USA; 4. Université Catholique de Louvain, Brussels, Belgium; 5. University Hospital and Universidad de los Andes, Bogotá, Colombia; 6. National Hemophilia Center, Caracas, Venezuela; 7. Van CreveldKliniek, UMC, Utrecht and Rehabilitation Centre De Trappenberg, Huizen, The Netherlands.

Introduction

- Exercise is believed to be beneficial for people with haemophilia (PWH) and its practice is widely encouraged
- Exercise for PWH is associated with specific challenges, such as the risk of injury, overloading and potential bleeds
- However, the value of physiotherapy, physical activity, exercise and sport as an integral component of a comprehensive treatment regimen in PWH remains to be fully recognised and implemented
- Here we consider evidence from studies assessing physical activity and fitness levels in the haemophilic population to determine the impact of exercise and sport participation on the physical, psychosocial and clinical status of PWH
- Detailed recommendations will be proposed that enable safe participation in a range of physical activities (based on the published evidence and an expert consensus reached by the authors)

Methods

- A systematic literature review of exercise and sport participation and its impact on PWH in terms of clinical, physical and psychosocial outlook was performed
- Inclusion and exclusion criteria were defined as follows:
 - Literature with at least the abstract published in English, indexed by Embase and/or Medline between 2001 and January 2012
 - Congress abstracts were not included unless subsequently published as peer-reviewed articles
 - The search terms used were 'haemophilia' and 'activity', 'exercise', 'fitness', 'sport', 'athletics', 'cycling', 'football', 'soccer', 'swimming', 'joints', 'protection of joints', 'prevention of arthropathy', 'musculoskeletal system', 'physiotherapy', 'strengthening' or 'stretching'
- Results were considered in terms of their relevance to exercise for PWH, and relevant articles were categorised as primarily relating to physical and medical issues, psychosocial aspects, or recommended exercises and sports

Physical Benefits of Exercise for PWH

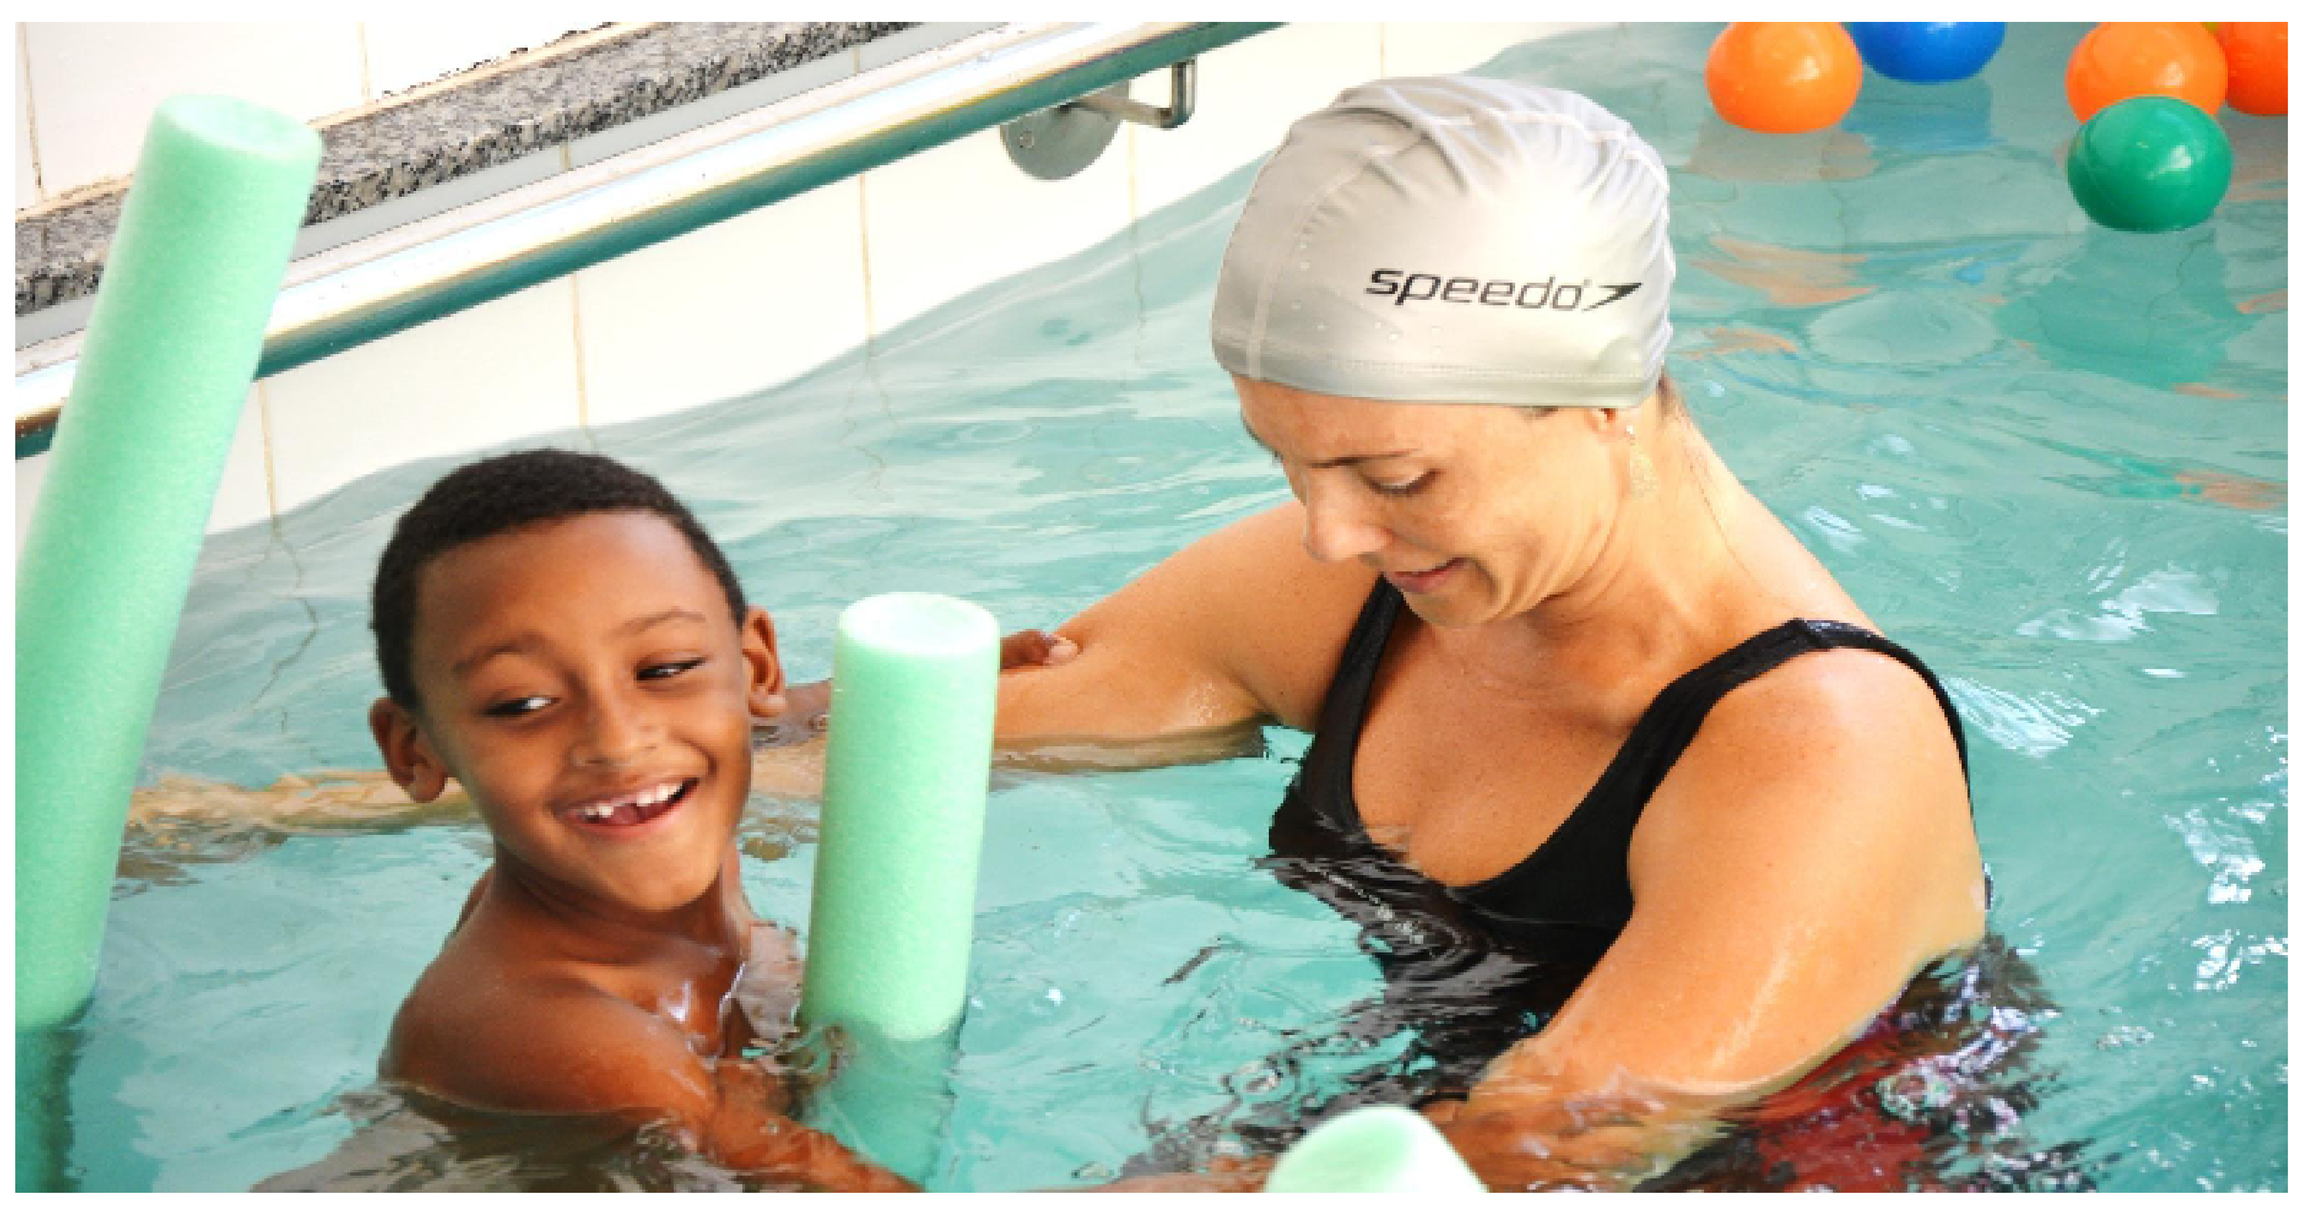
speedo

- Properly managed exercise and participation in appropriate sports are highly beneficial for PWH and can lead to
 - improved muscle strength, joint stability and coordination
 - A reduction in, or prevention of, musculoskeletal haemorrhages[1–4]
- Many forms of exercise have been shown to be of benefit to PWH, including dynamic, isokinetic and isometric exercise, light weight training, proprioceptive and isometric training and electrically-stimulated strength exercises
- It is important for PWH to start physical activities at a young age as
 - The physical signs of sedentary behaviour have been found in some boys with haemophilia[1]
 - Early functional changes in gait and squat are measurable prior to structural changes becoming detectable[5]
- However, there remains a lack of knowledge amongst young haemophilia patients as to the role of exercise in the management of their condition

Psychosocial Benefits of Exercise for PWH

- Improvement of physical performance can lead to increased social inclusion and consequently higher self-esteem, as well as social adaptation[6,7]
- Decreased bleeding frequency by strengthening muscles and reducing the impact on the joints through weight loss could potentially result in reduced factor usage and improved psychological well-being through feeling less dependent on medication

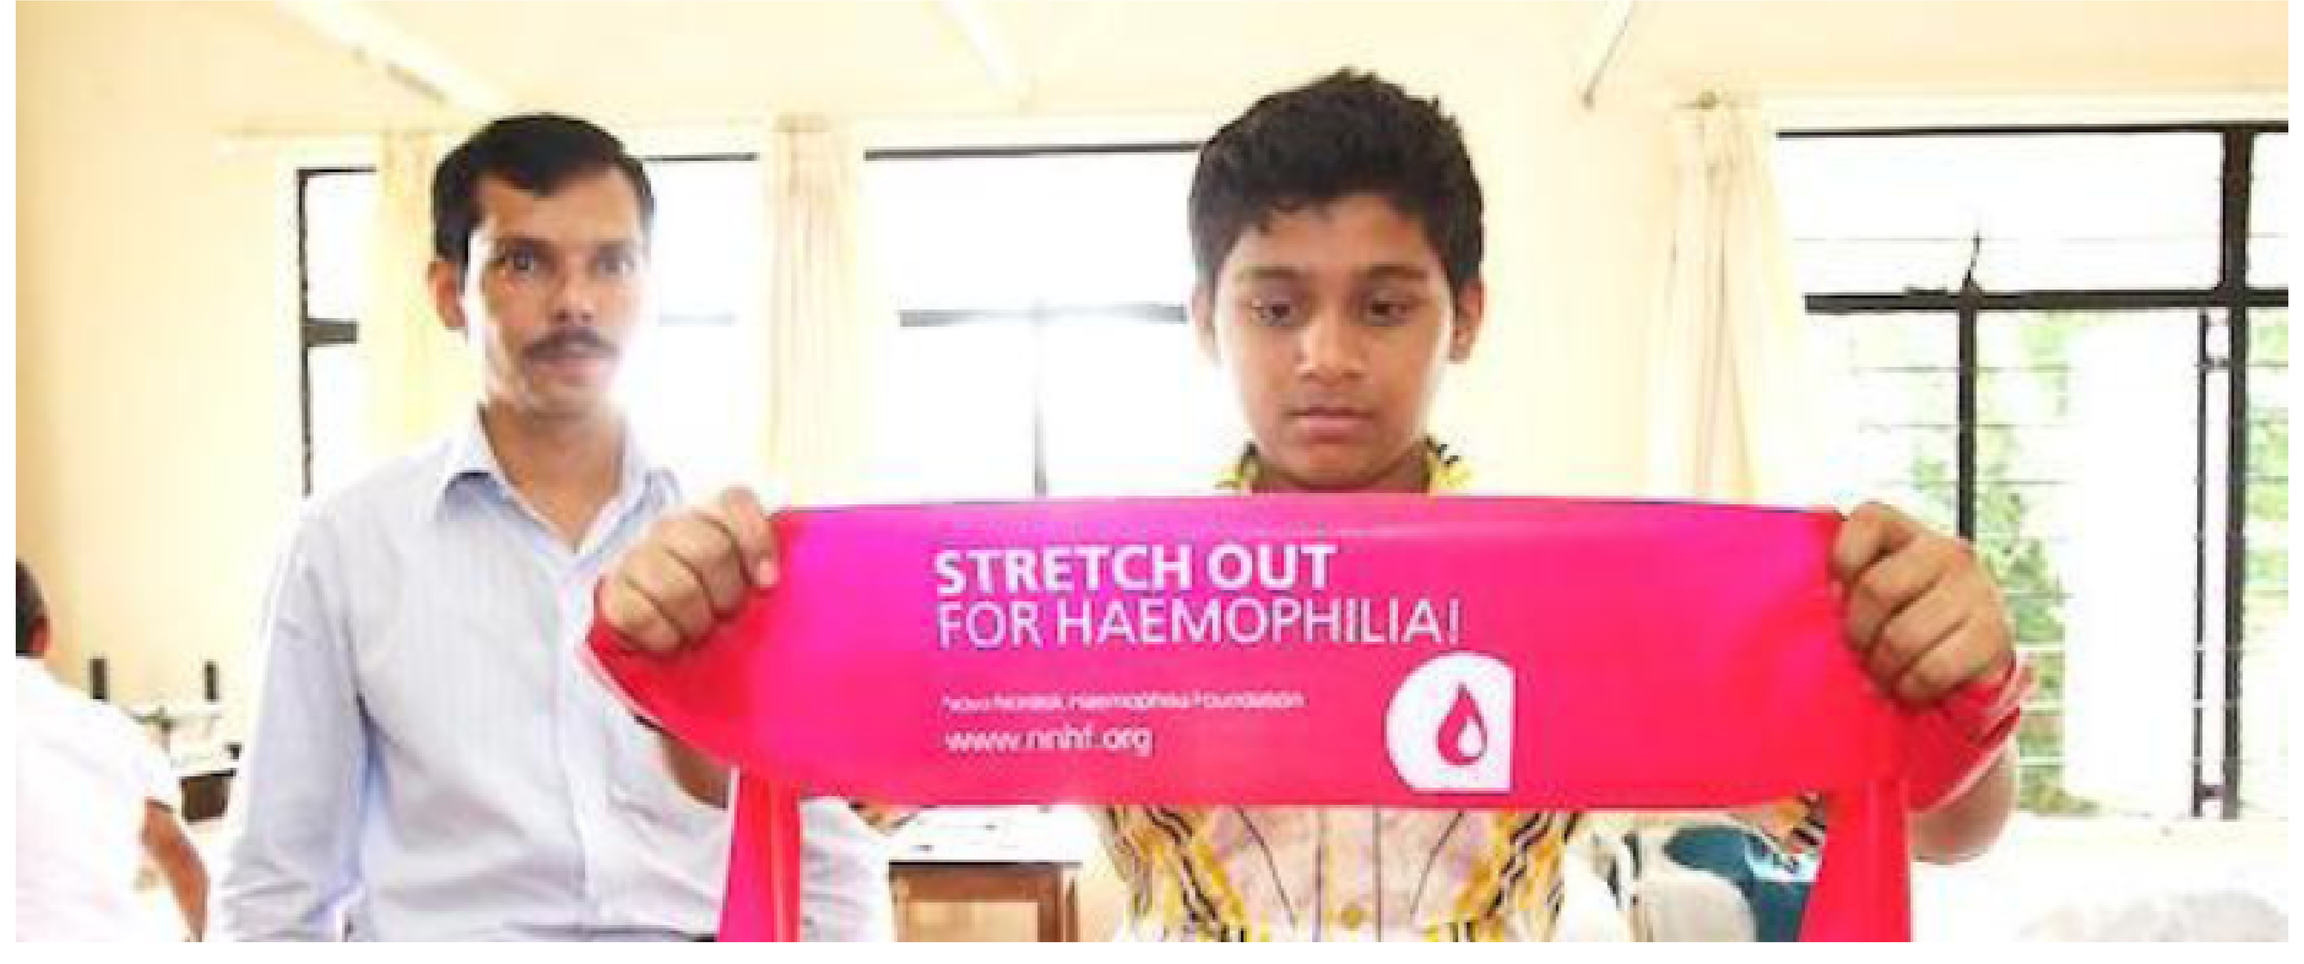
STRETCH OUT
FOR HAEMOPHILIA!

Medical Aspects of Exercise for PWH

- Adequate weight-bearing physical activity in adolescence, especially around puberty, is associated with the acquisition and maintenance of normal bone mineral density
 - A recent meta-analysis of seven case-controlled studies reported significantly reduced lumbar bone mineral density in children and adults with severe haemophilia versus controls[8]
 - However, it has also been reported that adult PWH on long-term prophylaxis do not experience a loss of bone mass irrespective of their degree of physical activity[9]
- Exercise for PWH with inhibitors remains a challenge
 - It is not possible for these patients to receive prophylaxis with factor VIII or IX replacement therapy
 - Long-term bypassing agent prophylaxis is not routinely employed
 - Physical activity is often limited due to repeated joint bleeds and arthropathy
 - Consequently, high-risk sports are not recommended for PWH with inhibitors[10,11]

Recommendations

1. PWH should be encouraged to regularly participate in physical activity and sports appropriate to their individual circumstances
2. Physical activity should be encouraged from as young an age as practical to help establish a positive attitude and develop a lifelong habit of participation
3. Before starting exercise, PWH should work with their haemophilia treatment centre (HTC) physiotherapist and other staff
4. Guidelines for treatment with factor replacement should be set by the HTC
5. In situations of limited or no factor availability it is still beneficial for PWH to participate in physical activity appropriate to their individual circumstances
6. When participating in an organised sport or physical activity, PWH should reduce the likelihood of injury by being properly prepared
7. To be comprehensive, an exercise, sporting or physical activity programme should include a number of different components
8. Injury or bleeding occuring after physical activity, sport or exercise require individual management
9. Physical activity should be encouraged for elderly PWH

- It is intended that the recommendations are applicable worldwide and, in combination with the reported evidence, highlight the physical, medical, social and economic benefit that can be gained by the appropriate participation of PWH in physical activity, exercise and sport
- Any recommendations made should be used as a guide only and it is vital that a full consultation with an appropriate healthcare professional is completed before starting any exercise programme or engaging in sporting activity

Summary

- Participation in physical activity, exercise and sports realises numerous physical benefits, as well as supporting the emotional and social well-being of PWH
- Involvement in appropriate physical activity should be encouraged as part of the global approach to the management of haemophilia
- Because each person presenting with haemophilia is unique, the choice of the most suitable sport or exercise programme should
 - be made collaboratively between patient, doctor and physiotherapist
 - take into account the patient's physical status, interests and social requirements
- A full analysis of the literature and detailed guidelines for the implementation of the above recommendations is in preparation[12]

Conclusion

- Successfully promoting the benefits of exercise may result in PWH becoming more active, leading to improved physical performance and quality of life

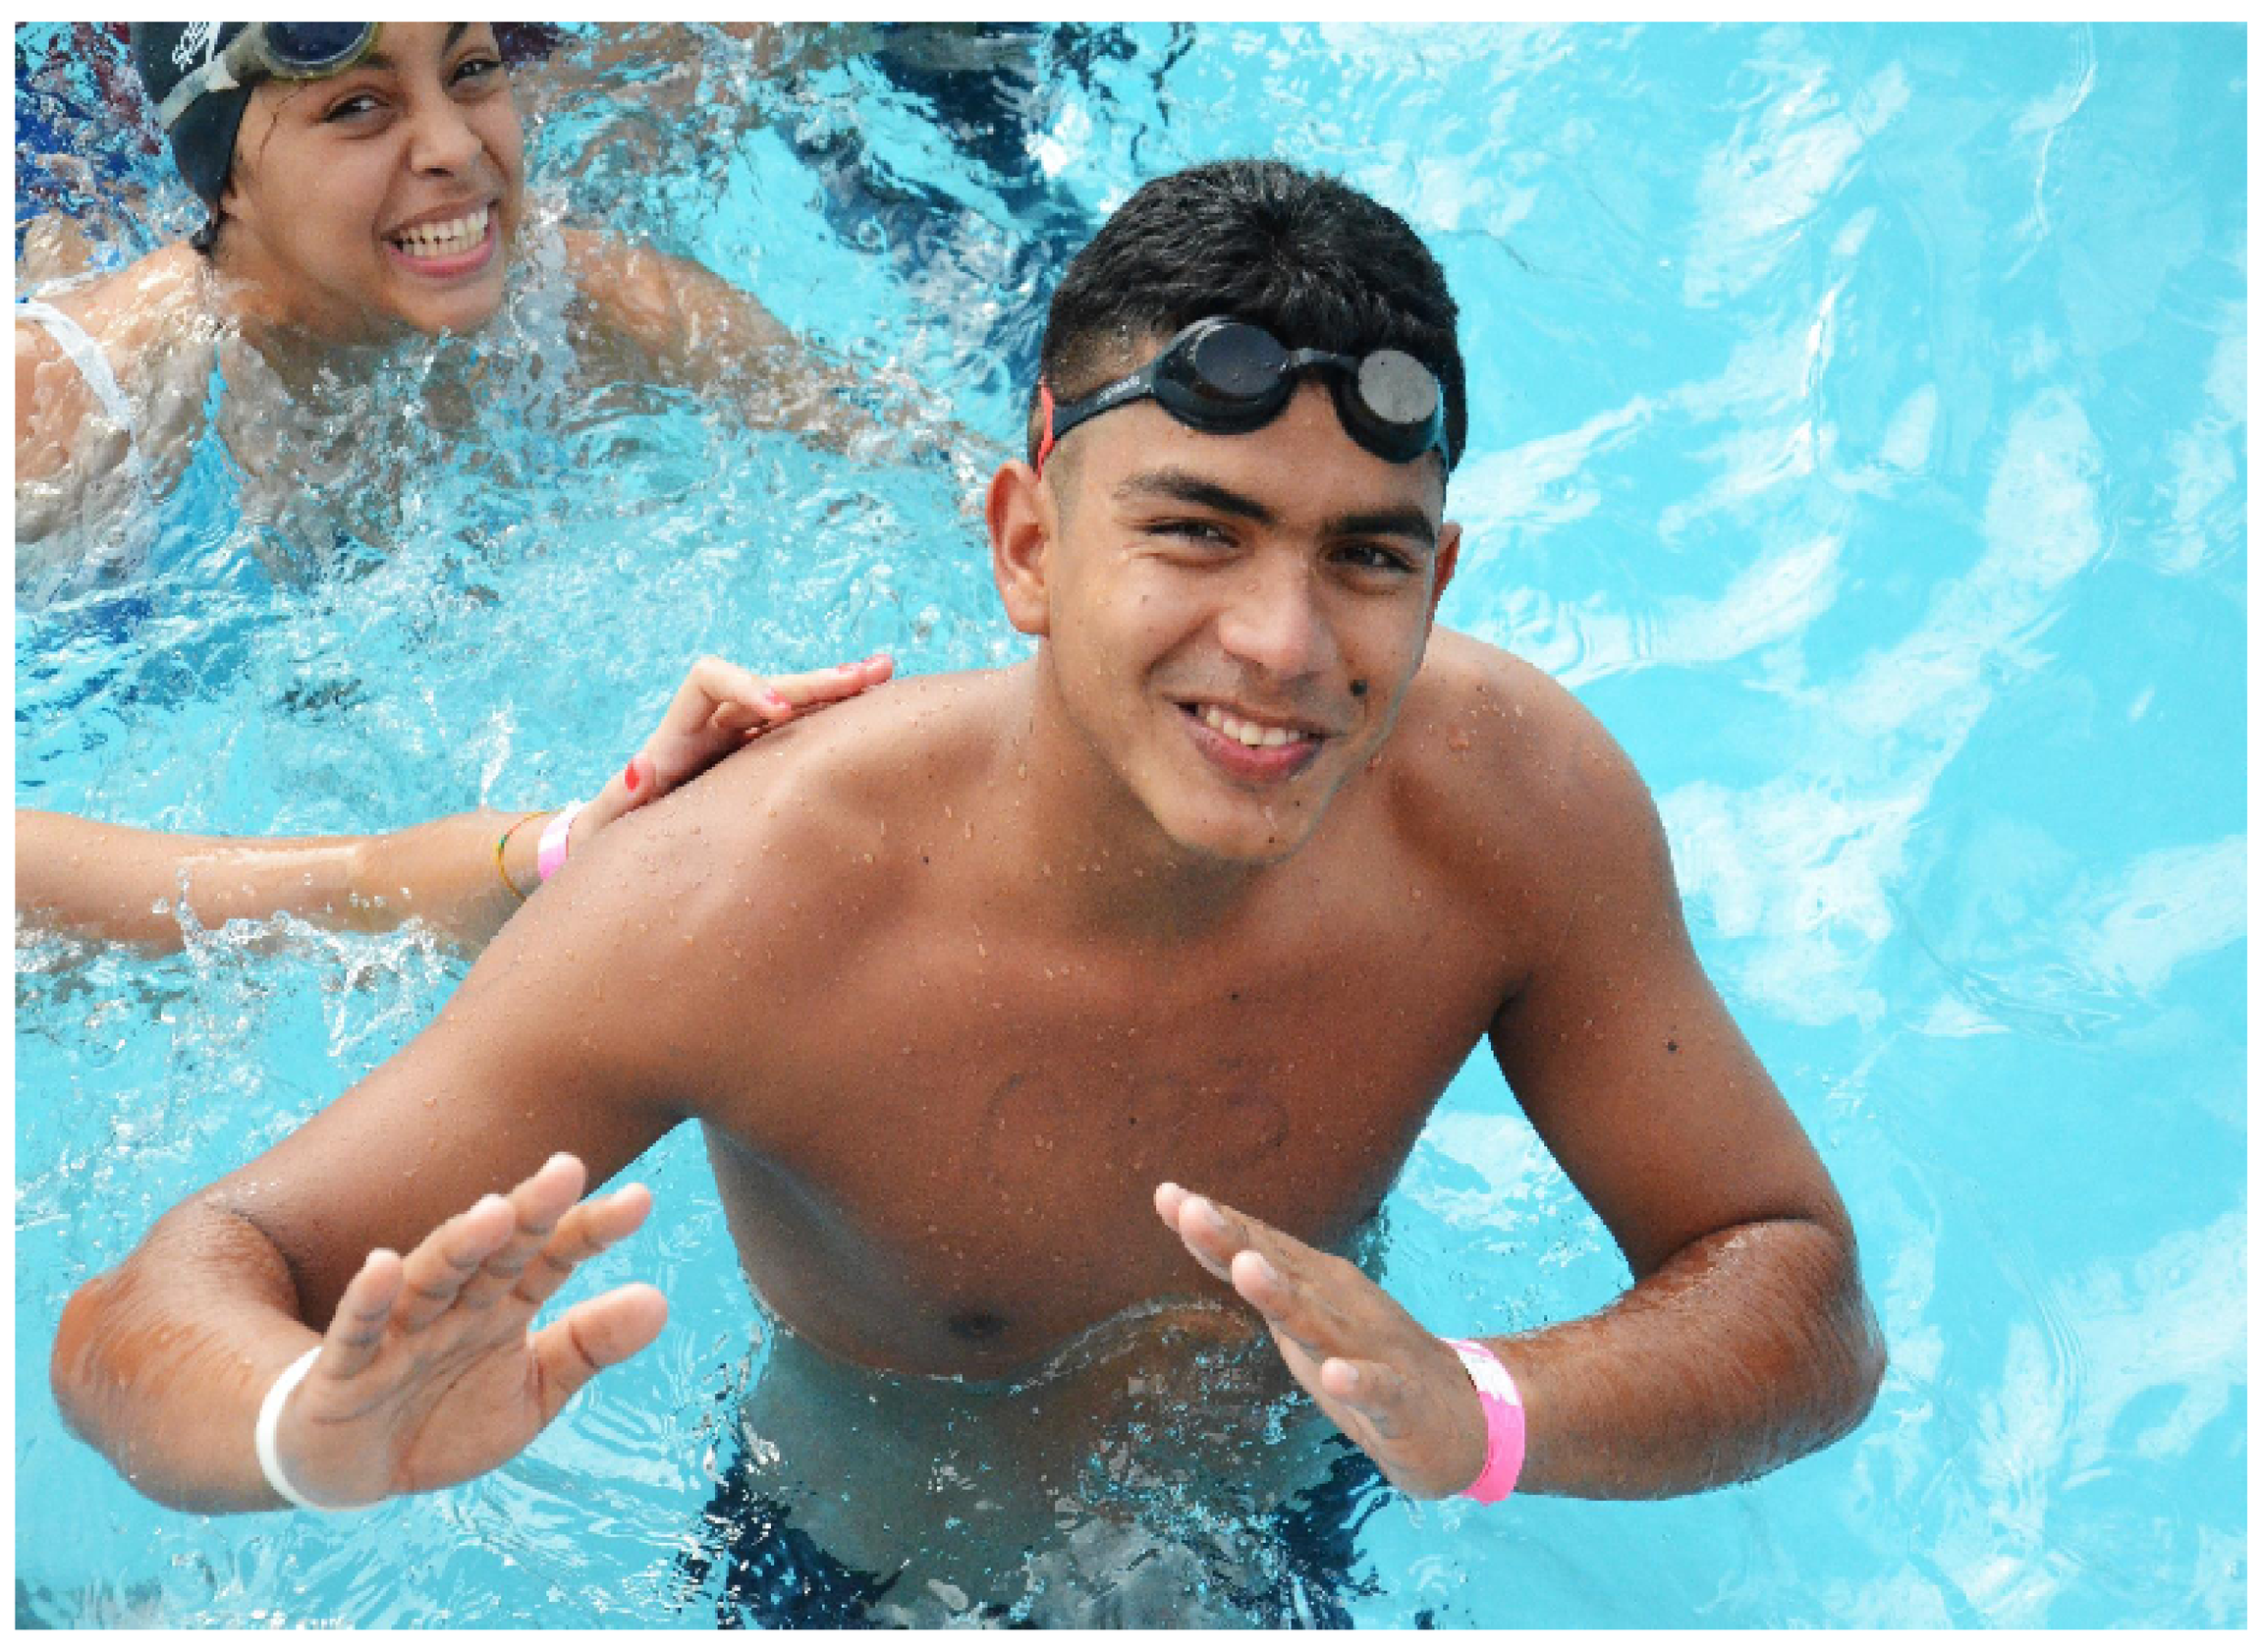

References

1. Tiktinsky R, et al. Haemophilia 2002; 8: 22
2. Pierstorff K, et al. Klin Padiatr 2011; 223: 189
3. Hilberg T, et al. Haemophilia 2003; 9: 86
4. Harris S, Boggio LN. Haemophilia 2006; 12: 237
5. Seuser A, et al. Hämostaseologie 2009 (1), 69
6. Von Mackensen S. Haemophilia 2007; 13 (Suppl. 2): 38
7. Buxbaum NP, et al. Haemophilia 2010; 16: 656
8. Iorio A, et al. Thromb Haemost 2010; 103: 596
9. Khawaji M, et al. Haemophilia 2010; 16: 495
10. Heijnen L. Haemophilia 2008; 14 (Suppl. 6): 45
11. Kurme A, Seuser A. Hamburg: Omnimed, 2004: 33
12. Negrier C, et al. Manuscript submitted for publication

Acknowledgement

- Assistance with poster development (funded by the Novo Nordisk Haemophilia Foundation) was provided by Matthew deSchoolmeester of Bioscript Stirling Ltd

WFH2012

265--We

Sports and Hemophilia

Claude Negrier

DOI: 10.3252/pso.eu.WFH2012.2012

Poster presented at: WFH 2012 WORLD CONGRESS PARIS, FRANCE

PosterSessionOnline.com