THE HAEMOPHILIA SOCIETY

Collaborative Working: **The Newcastle Initiative**

A multidisciplinary Quality, Innovation, Productivity and Prevention (QIPP) workshop was convened in Newcastle under the auspices of the Specialised Healthcare Alliance and North East Specialised Commissioning Group. Participation included clinicians, nurse specialists, commissioners, patient representatives and industry. The purpose of the day was to explore how haemophilia services could be developed to combine high quality care with improved efficiency in a cash-constrained NHS.

The workshop's recommendations addressed a number of clinical and non-clinical issues affecting haemophilia patients, including the extension of adult prophylaxis on a targeted basis; prioritising guidance on orthopaedic surgery to inform more consistent decision making; and developing haemophilia networks across the country as a means of ensuring equitable access to comprehensive care. The workshop's recommendations were echoed by the Haemophilia Society's 'Fit for the Future' patient survey and have been incorporated in what is now the Newcastle Initiative, which aims to coordinate multidisciplinary collaboration to achieve the following objectives;

- *Widen the focus of haemophilia-related policy communications, whilst leaving no room for repetition of past mistakes*
- *Develop a broader commitment to supporting the highest standards of care*
- *Ensure that patients and their families are at the heart of decision-making on services and treatment*

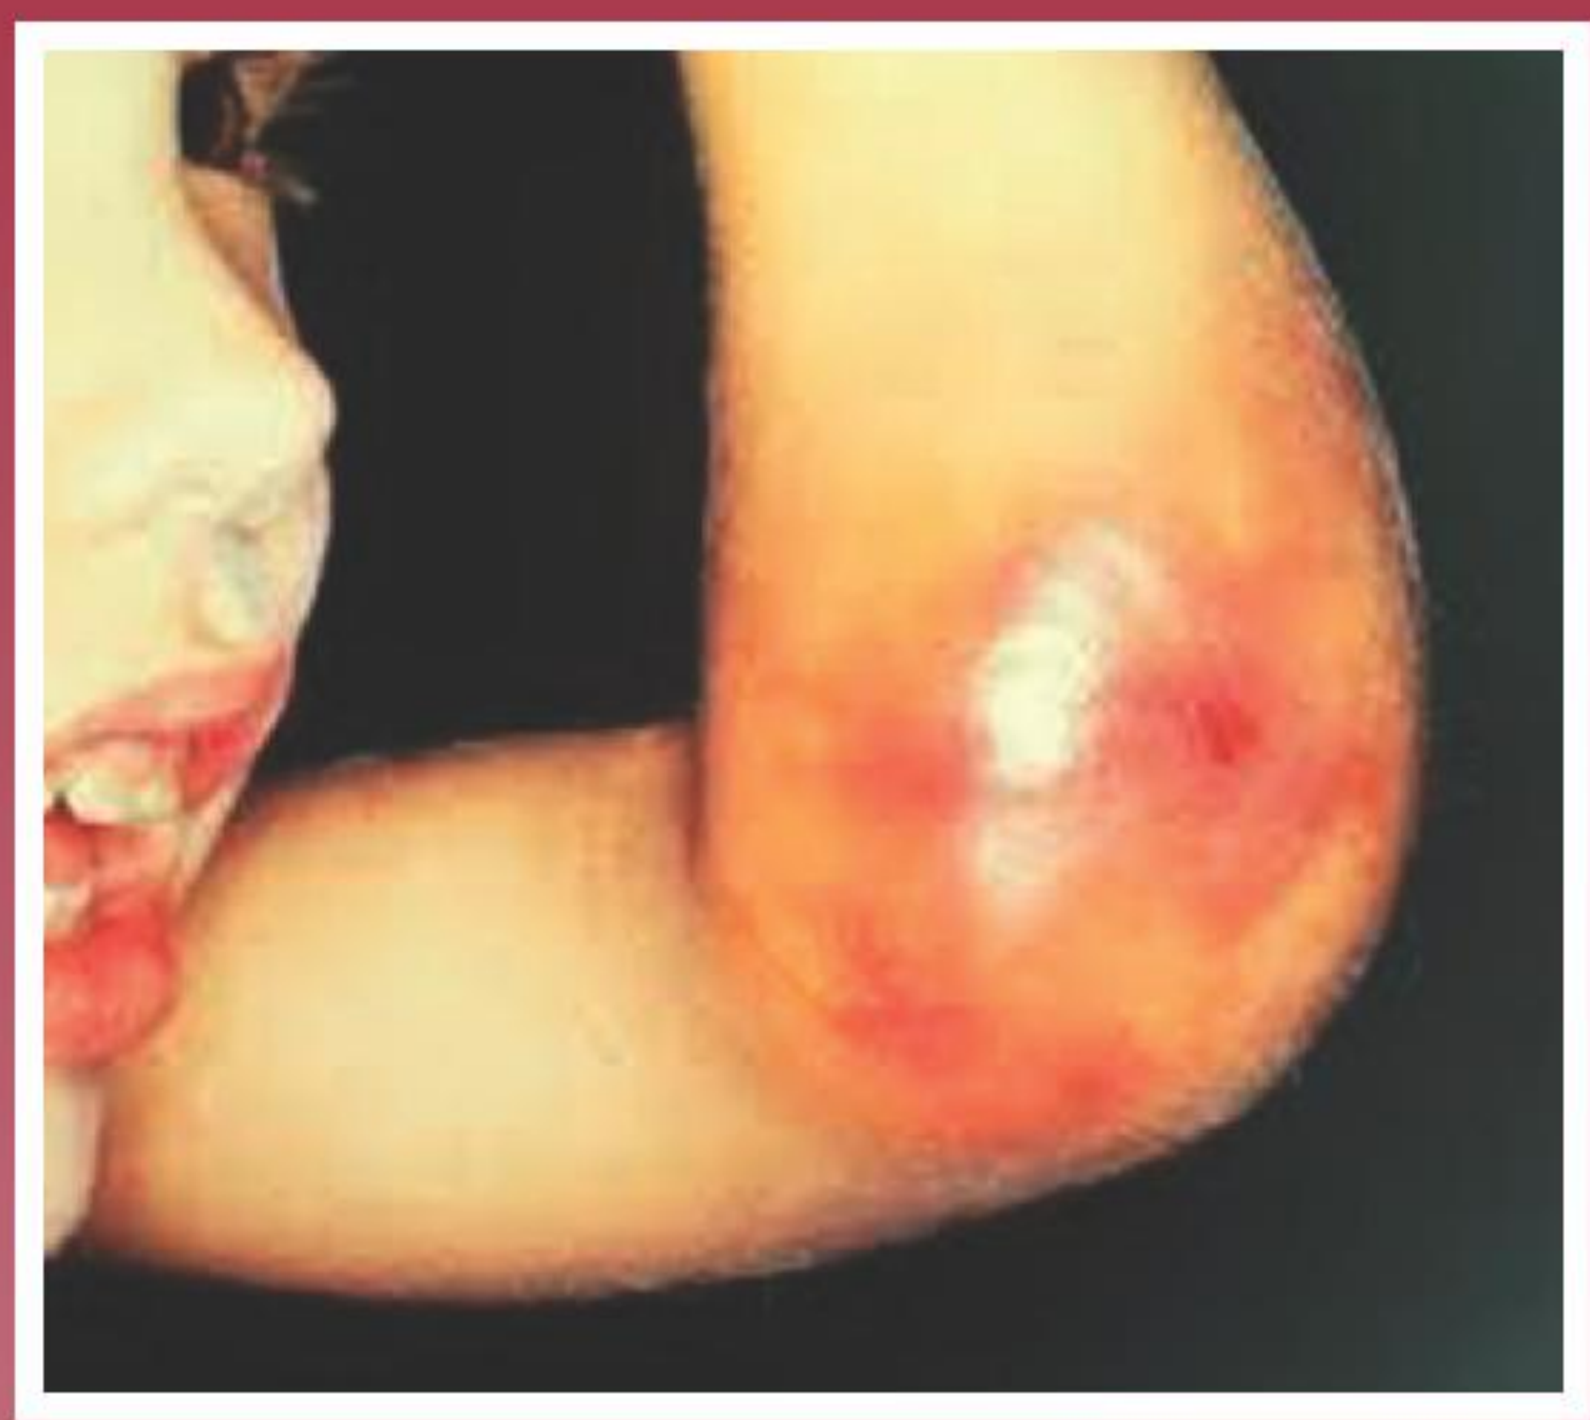

Joint bleeds - UKHCDO to progress multidisciplinary review of orthopaedic surgery for people with haemophilia

The Newcastle Initiative has an annual programme of work aimed at achieving these objectives and implementing the workshop recommendations. Regular meetings are supplemented with targeted campaigning on specific issues. Initial success includes the decision of the United Kingdom Haemophilia Centre Doctors' Organisation (UKHCDO) to take forward a multidisciplinary review of orthopaedic surgery for people with haemophilia.

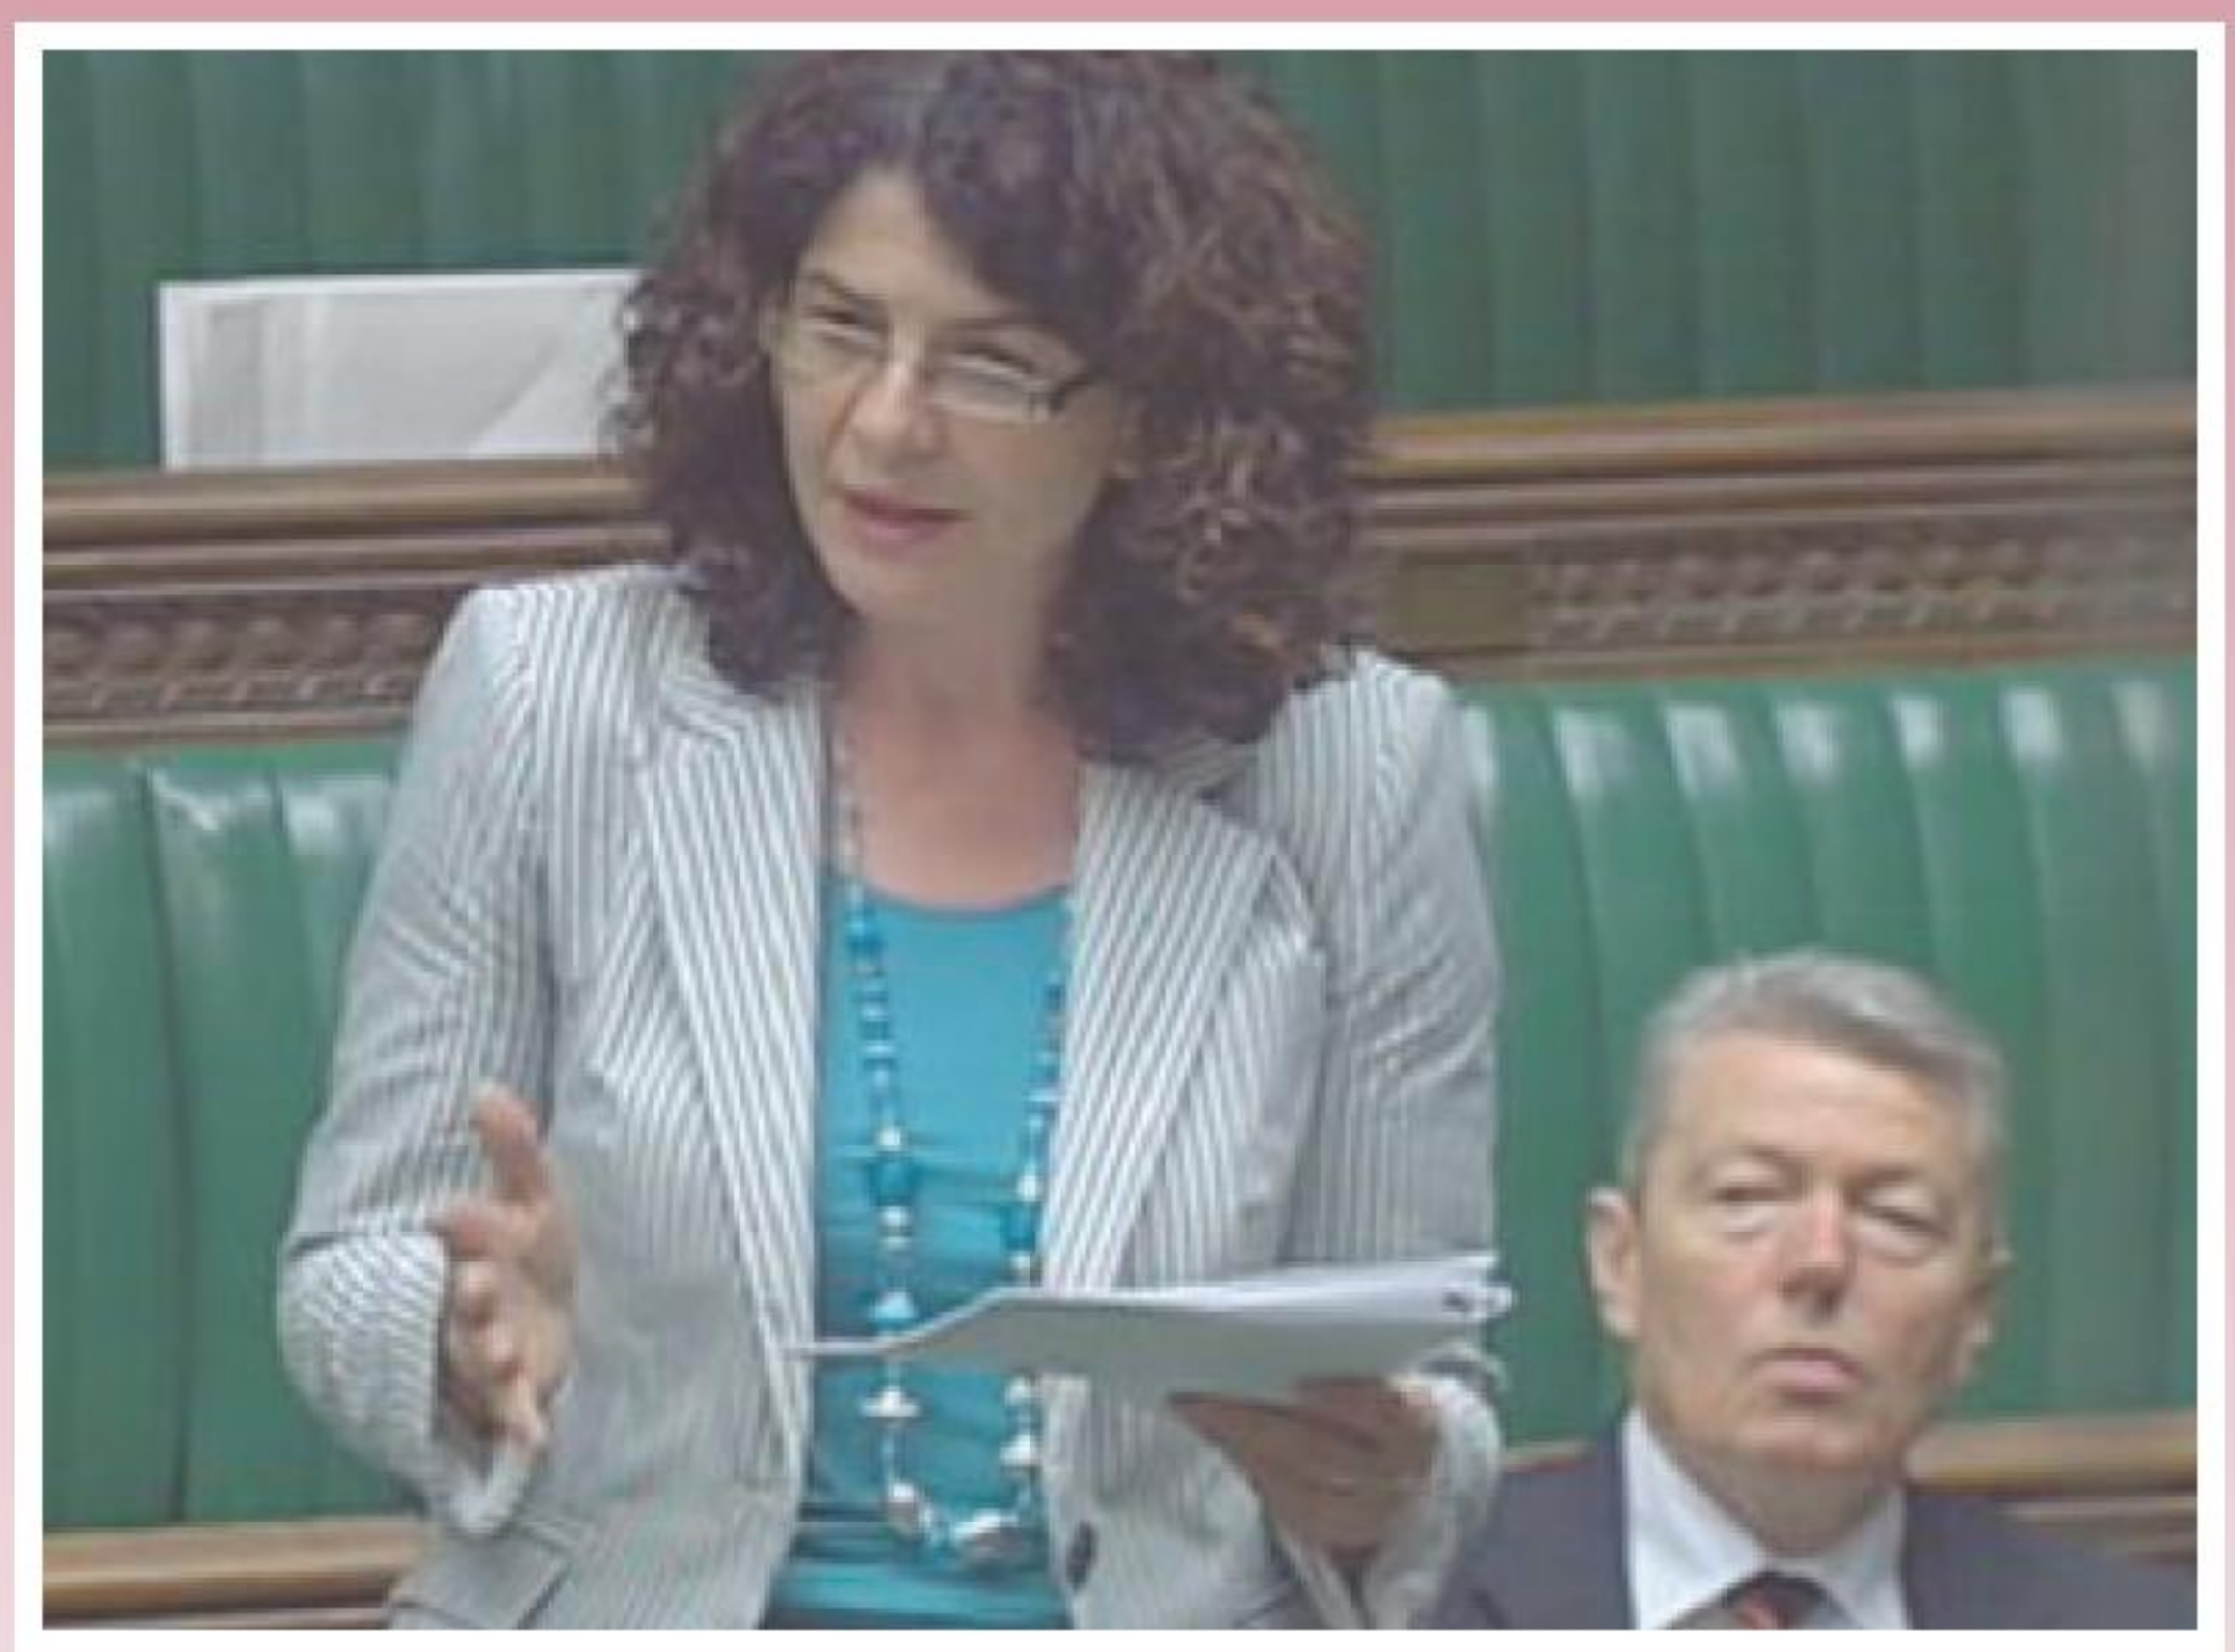

***Diana Johnson MP**, chaired the Private Members Debate in the House of Commons on Haemophilia Care attracting strong support from MPs.*

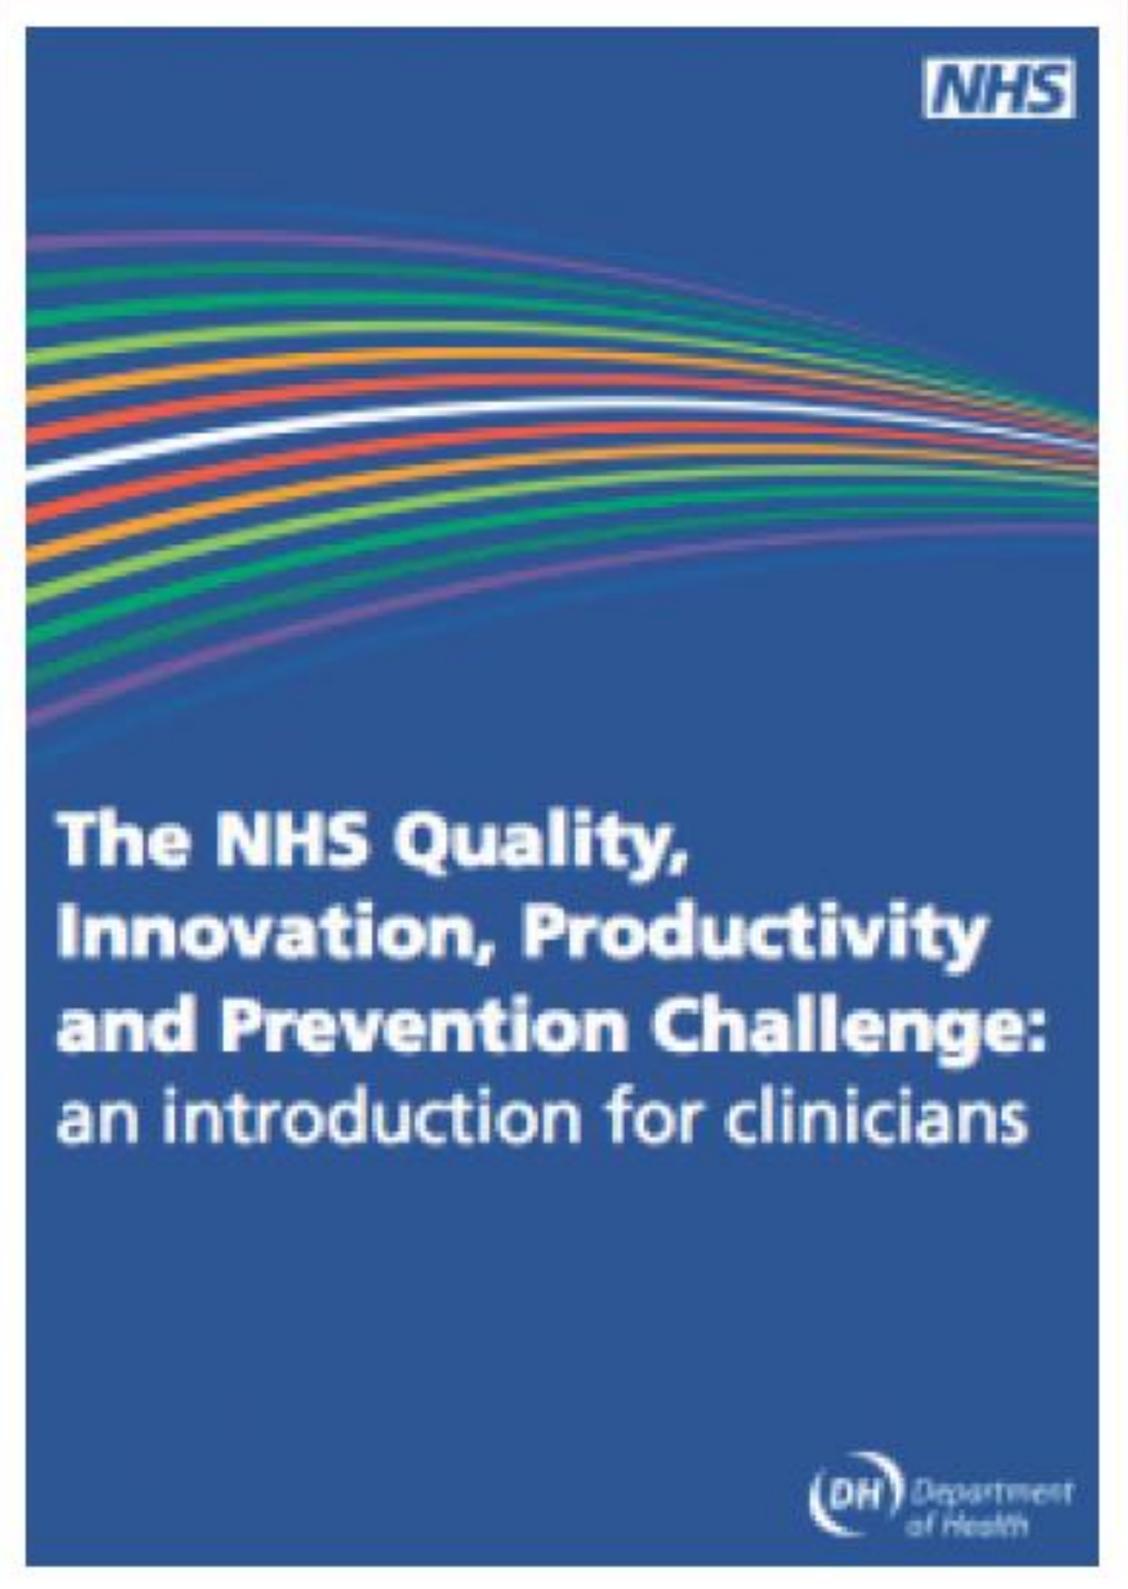
NHS
The NHS Quality, Innovation, Productivity and Prevention Challenge: an introduction for clinicians
DH Department of Health

At a political level, the Newcastle Initiative has enlisted the support of parliamentarians to ensure that the highest standards of care are reflected in updated guidance to the NHS, with a view to learning from the past to inform the future.

Further information:

Chris James - chris@haemophilia.org.uk

The Newcastle Initiative has been supported by unrestricted grants from Baxter Healthcare Limited, Bayer PLC, Novo Nordisk Limited and Pfizer Limited.

WFH2012 73--Mo

Hemophilia Programs and Organizations

Chris James

DOI: 10.3252/pso.eu.WFH2012.2012

Poster presented at: WFH 2012 World Congress, Paris, France, Sunday, July 8 - Thursday, July 12

PosterSessionOnline.com