AGING WITH HEMOPHILIA: ENHANCING DIALOGUE WITH OUR PATIENTS

PRESENTING AUTHOR: **Gerard Dolan, MD**
Nottingham University Hospitals, UK

CO-AUTHORS: **Ruud Bos**
UMC Utrecht / Van Creveld Clinic, the National Hemophilia Center
Randall Curtis
Factor VIII Computing, USA
Karin Lindvall
Department for Coagulation Disorders, University Hospital, Malmö, Sweden
Karen Wulff
Louisiana Center for Bleeding and Clotting Disorders, Tulane University School of Medicine, USA
Nichan Zourikian
Sainte Justine Pediatric/Adult Comprehensive Care Hemostasis and Inhibitor Center, Montreal, Canada

INTRODUCTION AND OBJECTIVES:

With the advances in treatment of the last 30 years, people with hemophilia are living longer and experiencing health and lifestyle challenges associated with normal aging. However, as highlighted by many health care providers (HCPs) through publications and presentations, there are very few resources available on these topics for HCPs who treat and manage the care of older adults with hemophilia. To address this need, we created an online resource to support this growing section of the hemophilia community, to help HCPs enhance their dialogue with this aging population.

MATERIALS AND METHODS:

In 2011, an international, multidisciplinary editorial board was established to develop the Above And Beyond Hemophilia program with the goal of providing information and resources to better understand and address the physical and social challenges of aging with hemophilia.

Controlled exclusively by an editorial board and sponsored by Bayer HealthCare, **AboveAndBeyondHemophilia.net** is specifically designed to facilitate HCPs' dialogue with their older patients on topics related to aging.

RESULTS:

Since launching the website in 2012, the board has guided the creation of nearly 40 articles on topics ranging from common medical issues associated with aging such as cardiovascular disease, joint issues and sexual health, to the psychosocial impact of aging. Also available for download is the "Talking About Aging" booklet, which HCPs can offer to their older adult patients to help them be more proactive in preparing for and managing doctor visits.

To date, AboveAndBeyondHemophilia.net has seen more than 2,000 visitors. Additional booklets and content are in development.

Plans for the content to be localized in different countries are underway. Both the brochure and website have already been introduced to older adults with hemophilia at the Van Creveld Clinic in the Netherlands. The content has been well-received, many considering it to be empowering and helpful in advocating for themselves on health matters beyond, but potentially impacted by, their bleeding disorder. There are also plans to translate the brochure into Dutch.

The content has been well-received, many considering it to be empowering and helpful in advocating for themselves on health matters beyond, but potentially impacted by, their bleeding disorder. There are also plans to translate the brochure into Dutch.

CONCLUSION:

AboveAndBeyondHemophilia.net is a first-of-its-kind resource to inform and support HCPs in their work with this growing and challenging segment of the hemophilia community.

Hemophilia Solutions
Never Losing Sight of the Human Factor

WFH2014
18--P-W
Ageing (Medical)
Gerard Dolan, MD
DOI: 10.3252/pso.eu.WFH2014.2014
Poster presented at: WFH 2014 World Congress, Melbourne, Australia · May 11-15

PosterSessionOnline.com