Epidemiology and Burden of Progressive Familial Intrahepatic Cholestasis: Systematic Review

V. VALCHEVA[1], T. JONES-HUGHES[2], J. CAMPBELL[2], L. CRATHORNE[2]
1Albireo Pharma; 2Roboleo & Co, Leeds, UK

INTRODUCTION

Progressive familial intrahepatic cholestasis (PFIC) is a rare but severe group of liver disorders of autosomal recessive inheritance, affecting an estimated 1 in 50,000 to 100,000 children worldwide.

PFIC is subgrouped according to the genetic defect, clinical presentation, laboratory findings, and liver histology (PFIC1 to PFIC6). The major mutations are in the *ATP8B1*, *ABCB11*, *ABCB4*, *TJP2*, *NR1H4*, or *Myo5b* genes, although some patients have no identified mutation.

As a rapidly progressing disease characterized by pruritus, malabsorption and ultimately leading to liver failure, PFIC is extremely distressing for children and parents. The significant pruritus can lead to severe cutaneous mutilation and, without surgical biliary diversion (SBD) or liver transplant (LT), PFIC is typically fatal by age 20.

AIM

To conduct a systematic review of published evidence on (i) the epidemiology and (ii) the disease burden of PFIC.

METHOD

Databases including MEDLINE and Embase were searched for publications on PFIC prevalence, incidence or natural history, and the economic burden or health-related quality of life of patients with PFIC.

Preferred Reporting Items for Systematic Reviews and Meta-Analyses guidelines were followed.

RESULTS

Three systematic reviews and twenty-two studies were eligible for inclusion for the epidemiology review of PFIC including a total of 2,603 patients.

Figure 1. PRISMA flowchart of included studies for epidemiology review

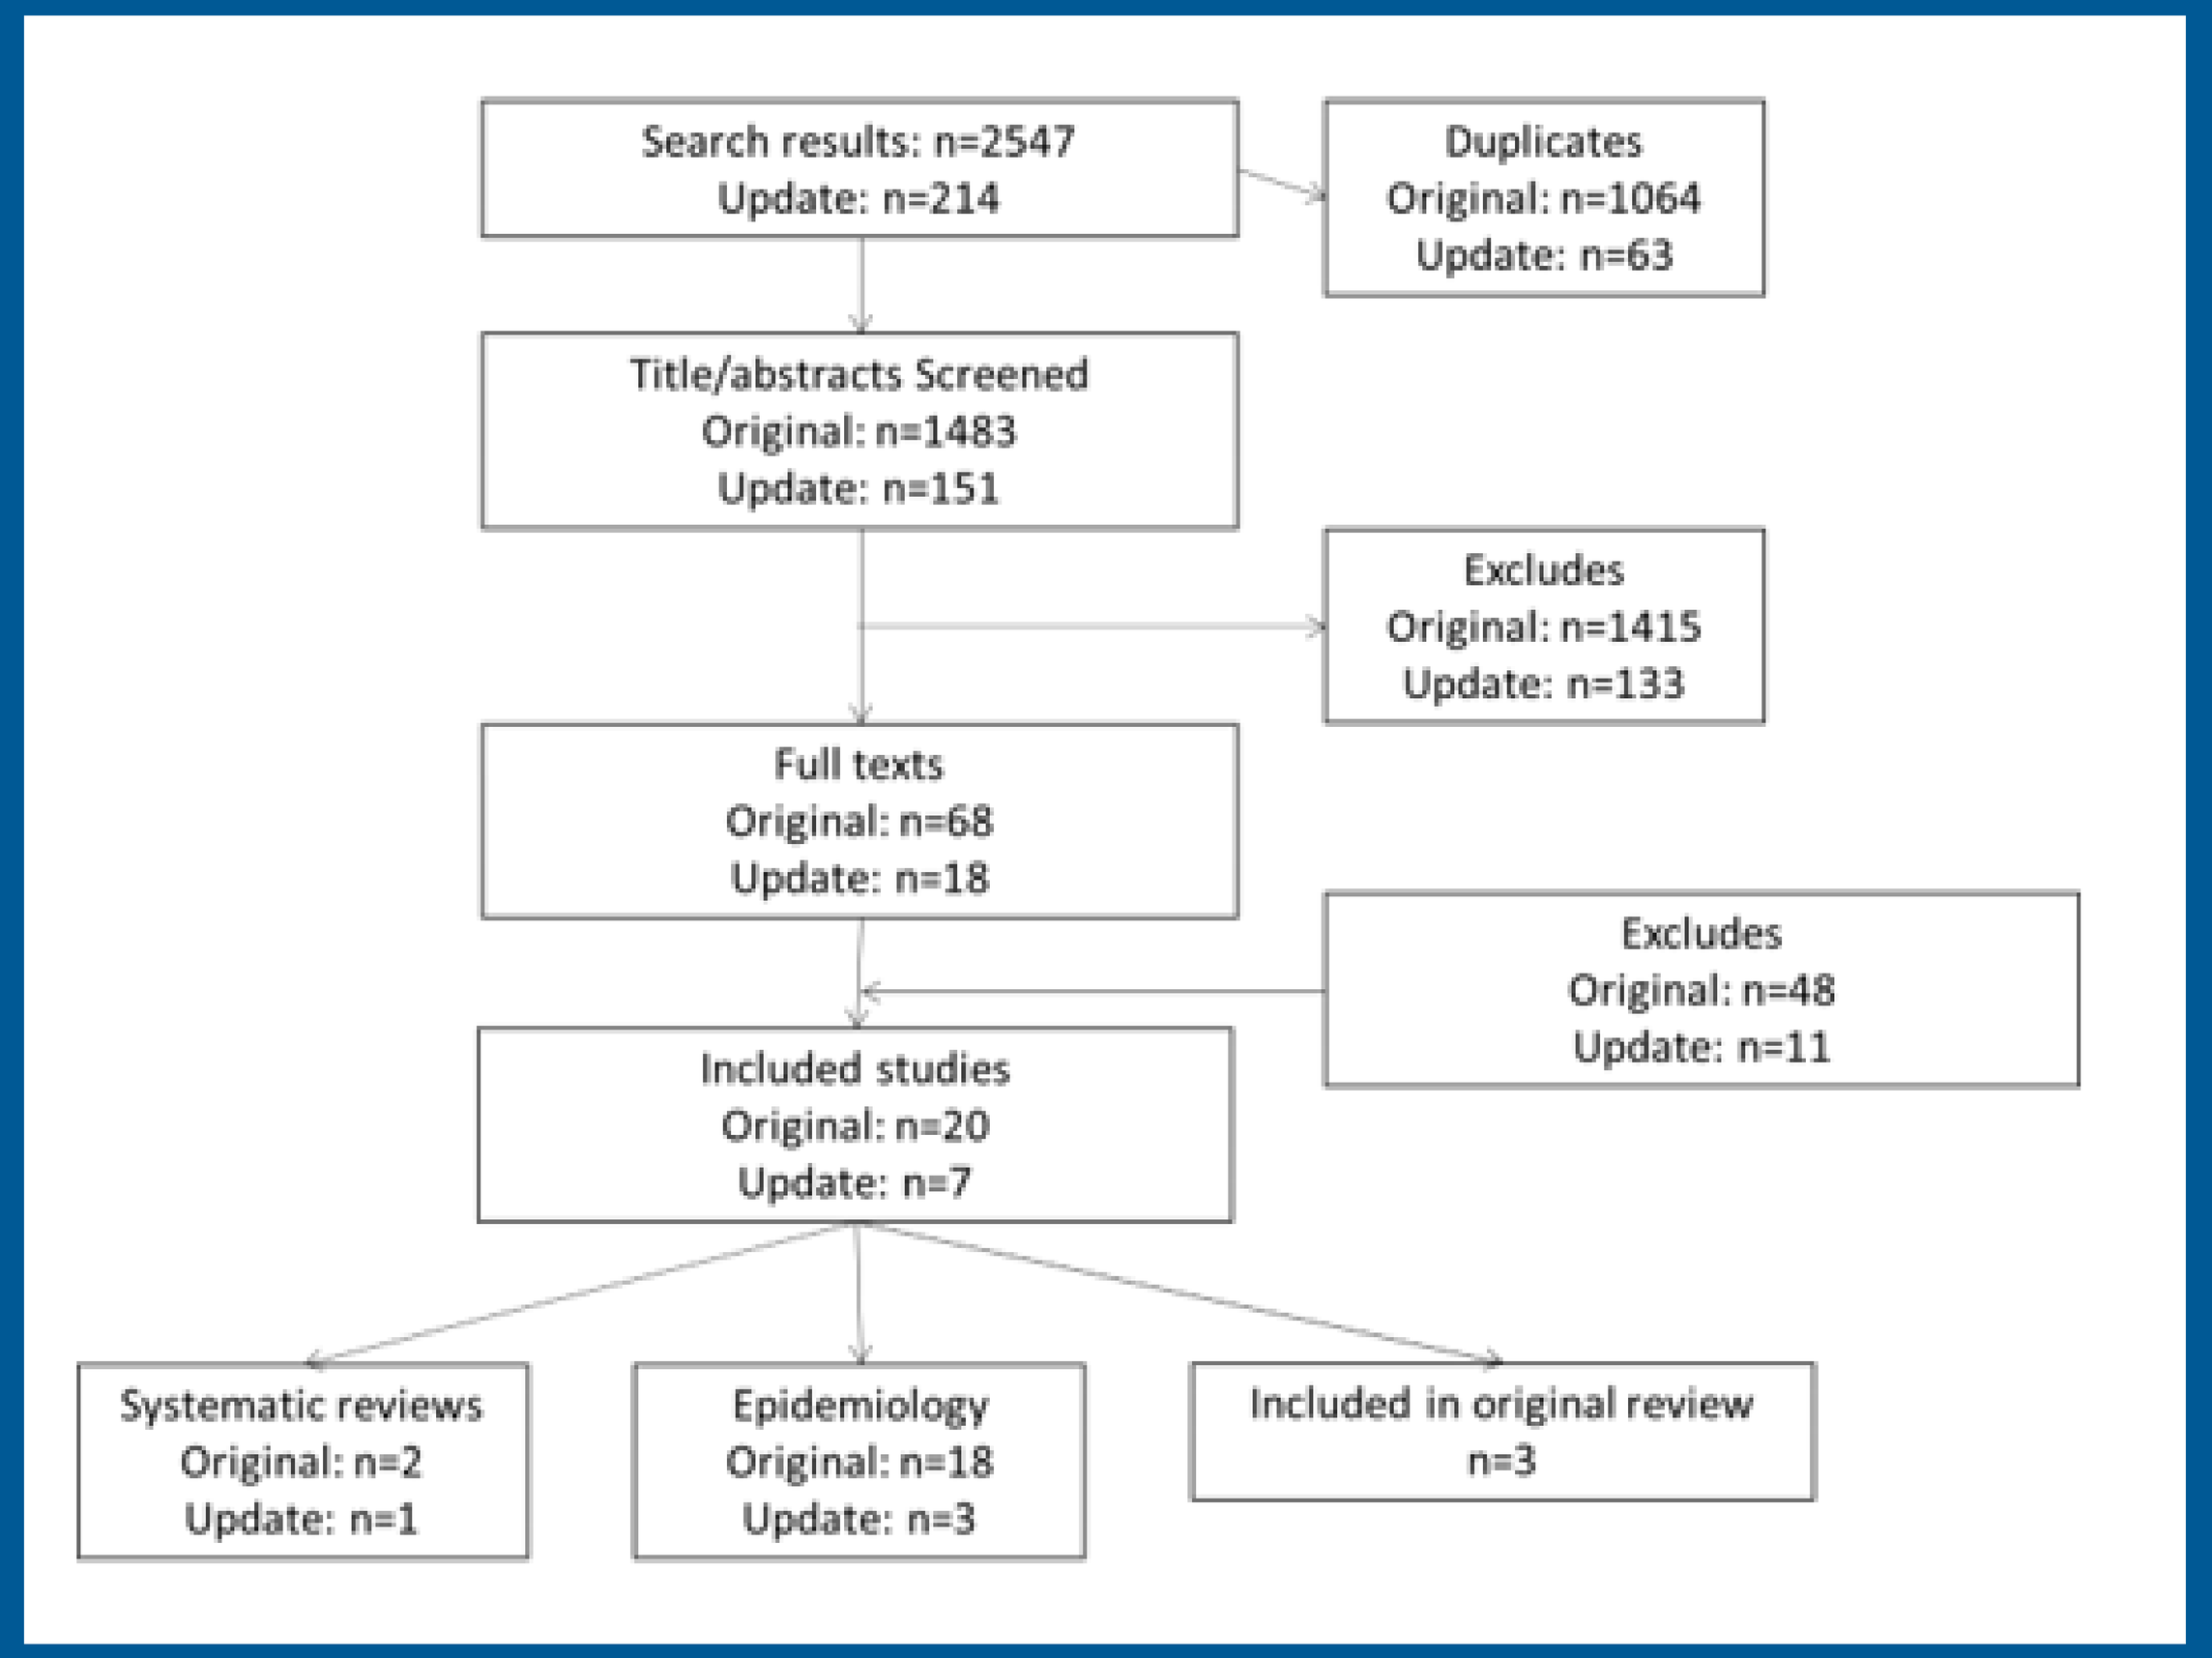
Search results: n=2547
Update: n=214
Duplicates
Original: n=1064
Update: n=63
Title/abstracts Screened
Original: n=1483
Update: n=151
Excludes
Original: n=1415
Update: n=133
Full texts
Original: n=68
Update: n=18
Excludes
Original: n=48
Update: n=11
Included studies
Original: n=20
Update: n=7
Systematic reviews
Original: n=2
Update: n=1
Epidemiology
Original: n=18
Update: n=3
Included in original review
n=3

Notes: The 3 systematic reviews included Baker et al. 2019, which provided the basis for this update review; Davis et al. 2009 and Verkade et al. 2020, where included studies were scrutinised but no additional studies were identified for inclusion.[1,2,3]

Study periods ranged from 3 to 33 years with data on PFIC 1 and 2 most widely reported.

Local population prevalence of PFIC was reported in three studies, ranging from 9.0% to 12.0% of children admitted with cholestasis, acute liver failure, or splenomegaly.

The most detailed data come from the Natural Course and Prognosis of PFIC and Effect of Biliary Diversion (NAPPED) study where findings for BSEP-deficient patients show only a third of patients reached adulthood with their native liver. However, a serum bile acid concentration <102 µmol/L or decrease of at least 75%, following surgical biliary diversion, predicted native liver survival of ≥15 years.

The review on disease burden identified 31 eligible studies and two systematic reviews, including a total of 1,583 patients. Burden of disease was mainly reported through rates of surgery and survival, with limited data on Health Related Quality Of Life (HRQOL). The leading indications for LT were intractable pruritus and progressive liver disease. Rates of SBD and LT varied widely depending on study period, sample size and PFIC type, with many patients having multiple surgeries and progressing to LT. In NAPPED, 68% of the FIC1 deficiency patients had undergone SBD by 18 years of age.

Only 2 studies reported HRQOL outcomes. Patients post LT showed a statistically significant impairment in school functioning and LT was associated with more frequent post-surgery complications than surgical biliary diversion.

Figure 2. PRISMA flowchart of included studies for disease burden review

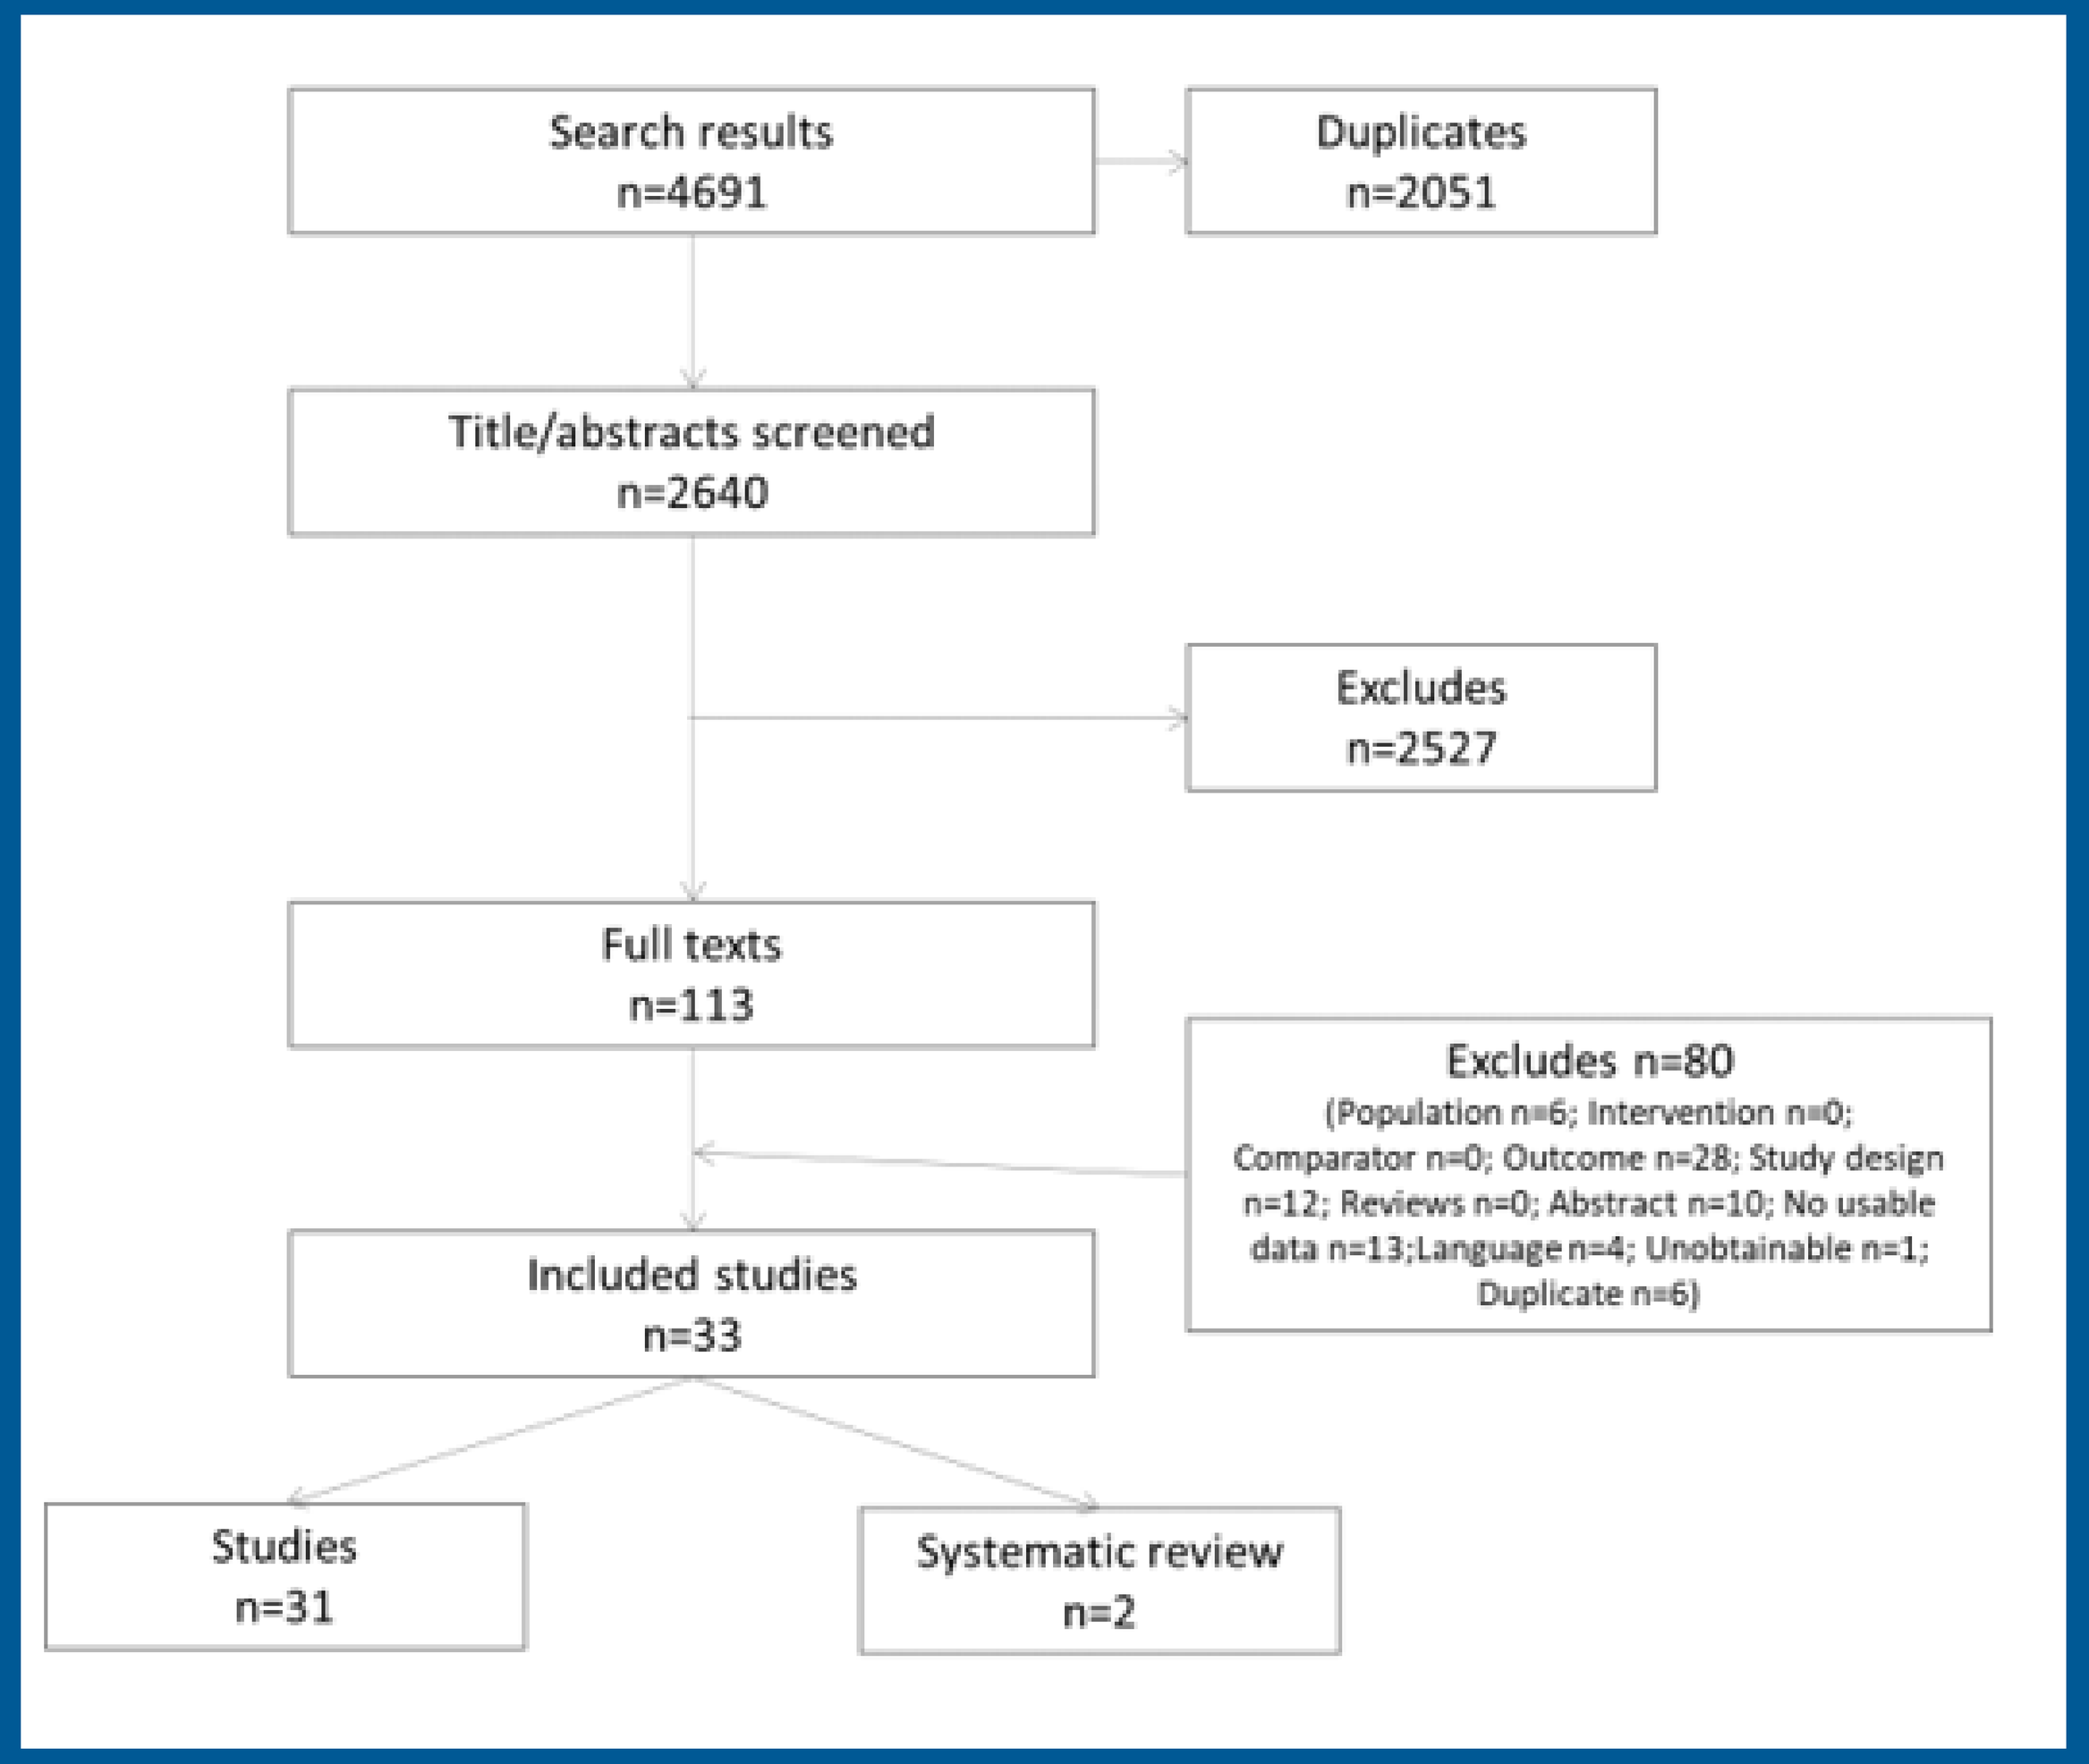
Search results
n=4691
Duplicates
n=2051
Title/abstracts screened
n=2640
Excludes
n=2527
Full texts
n=113
Excludes n=80
(Population n=6; Intervention n=0; Comparator n=0; Outcome n=28; Study design n=12; Reviews n=0; Abstract n=10; No usable data n=13;Language n=4; Unobtainable n=1; Duplicate n=6)
Included studies
n=33
Studies
n=31
Systematic review
n=2

Notes: The 2 systematic reviews included Baker et al. 2019, which provided the basis for this update review and Davis et al. 2009, where included studies were scrutinised but no additional studies were identified for inclusion.[1,3]

CONCLUSIONS

Using robust and transparent methods, this systematic review summarises our current knowledge of PFIC. The epidemiology is difficult to determine and there is large variation dependent on presentation and PFIC types and subtypes. This variation is affecting choice of intervention where one type of surgery may be beneficial in one subtype yet may be detrimental in another. This poses significant clinical challenges when considering appropriate treatment and management options.

REFERENCES

1. Davis AR, Rosenthal P, Newman TB. Nontransplant surgical interventions in progressive familial intrahepatic cholestasis. *Journal of pediatric surgery*. 2009;44(4):821-7.
2. Verkade HJ, Thompson RJ, Arnell H, Fischler B, Gillberg PG, Mattsson JP, et al. Systematic Review and Meta-analysis: Partial External Biliary Diversion in Progressive Familial Intrahepatic Cholestasis. *Journal of pediatric gastroenterology and nutrition.* 2020;71(2):176-83.
3. Baker A, Kerkar N, Todorova L, Kamath BM, Houwen RHJ. Systematic review of progressive familial intrahepatic cholestasis. *Clinics and Research in Hepatology and Gastroenterology.* 2019;43(1):20-36.

CONTACT INFORMATION

Velichka Valcheva, velichka.valcheva@albireopharma.com

DISCLOSURES

V. VALCHEVA is an employee of Albireo Pharma, Inc
T. JONES-HUGHES, J. CAMPBELL, L. CRATHORNE are employees of Roboleo & Co, Leeds, UK

ACKNOWLEDGMENTS

This study was sponsored by Albireo. Medical writing and editorial assistance were provided by Roboleo & Co. and were funded by Albireo Pharma, Inc.

Presented at the International Liver Congress; June 23–26, 2021 (Virtual)

Scan to download the poster
General hepatology
Velichka Valcheva
PO-1722
ILC2021